Genetic and environmental factors are differentially related to Aβ burden in the presymptomatic phase of autosomal dominant and sporadic Alzheimer's disease

Julie Gonneaud, PhD,[1,2,3] Christophe Bedetti, MSc,[1,2] Alexa Pichet Binette, M.Sc,[1,2,3] Tammie L.S. Benzinger, MD, PhD,[4,5] John C. Morris, MD,[4,5] Randall J. Bateman, MD,[4,5]
Judes Poirier, PhD,[1,2,3] John C.S. Breitner, MD, MPH,[1,3] and Sylvia Villeneuve, PhD,[1,2,3] for The DIAN Study Group and PREVENTAD Research Group

[1] McGill University, Montreal, QC, Canada; [2] Douglas Mental Health Research Institute, Montreal, QC, Canada; [3] Centre for Studies on Prevention of Alzheimer's disease (StoP-AD Centre), Douglas Mental Health Institute, Montreal, QC, Canada; [4] Washington University School of Medicine, St. Louis, MO, USA; [5] Knight Alzheimer's Disease Research Center, St. Louis, MO, USA

Background and objectives

Autosomal dominant Alzheimer's disease (ADAD) is considered the gold standard to evaluate the cascade of biomarker changes occurring in the preclinical phase of Alzheimer's disease (AD), since years from symptom onset can be estimated in presymptomatic mutation carriers taking parental age at symptom onset as a reference.[1] However, while the preclinical phase of sporadic AD (sAD) is known to be influenced by environmental and genetic factors, ADAD pathophysiology is directly triggered by the mutation of a gene involved in beta-amyloid (Aβ) production (*PSEN*1, *PSEN*2 or *APP*),[2] questioning the similarity of the two variants in their preclinical stage.

Our objective is to evaluate whether factors known to affect Aβ trajectories in presymptomatic ADAD mutation carriers also affect asymptomatic individuals with a parental history (PH) of sAD, and vice-versa. More specifically, we want to test if 1) parental EYO, as developed in ADAD, can help predicting cerebral Aβ burden in sAD and 2) genetic and environmental factors known to affect Aβ accumulation in asymptomatic individuals at risk of sAD (apolipoproteinE4 [*APOE*4][3,4] and education)[5,6] also affect Aβ trajectories in presymptomatic ADAD mutation carriers.

Participants and Methods

Cognitively normal participants were recruited from the DIAN observational study, which includes children of individuals with a known mutation for ADAD, and the PREVENT-AD cohort, which includes individuals with a PH of sAD.

Demographics

	DIAN: **117** presymptomatic ADAD mutation carriers		PREVENT-AD: **106** asymptomatic elderly with a parental history of sAD		
Age (mean ± SD [range)]	35.04 ± 9.43	[18 – 61]	67.28 ± 4.72	[58 – 83]	***
F/M ratio (%)	64/53	(55%-45%)	78/28	(74%-26%)	**
EYO (mean ± SD [range)]	-13.73 ± 8.92	[-30 – +7]	-6.23 ± 7.26	[-21 – +13]	***
Education (y) (mean ± SD [range)]	15.21 ± 2.99	[10 – 24]	38	(31%)	*ns*
APOE4 carriers (%)	36	(31%)	15.37 ± 3.29	[7 – 24]	*ns*
*PSEN*1/*PSEN*2/*APP*	85/17/15	(73%-15%-12%)	-		-
Aβ SUVR (mean ± SD [range)]	1.02 ± .18	[.78 – 1.59]	1.08 ± .24	[.86 – 2.28]	-
Aβ positive (%)	46	(39%)	16	(15%)	***
MMSE (mean ± SD [range)]	29.11 ± 1.25	[24 – 30]	28.86 ± 1.18	[25 – 30]	*ns*

** $p < .01$, *** $p < .001$

"Estimated Years from expected symptom Onset" (EYO) calculation

For all individuals we calculated an EYO as follows: participant's age – parent's age at symptom onset

Imaging

All participants underwent structural MRI and Aβ-PET scans.

T1-weighted images were acquired following ADNI protocol, on different 3T scanners for DIAN (due to the multicenter nature of the study), and on a Siemens 3T scanner for the Prevent-AD.

30-minute Aβ-PET scans were acquired ~40 minutes after the intravenous injection of 8 to 18 mCi of C^{11}-PIB tracer for DIAN participants and of ~5.4mCi of F^{18}-NAV4694 in PREVENT-AD cohort.

PET processing

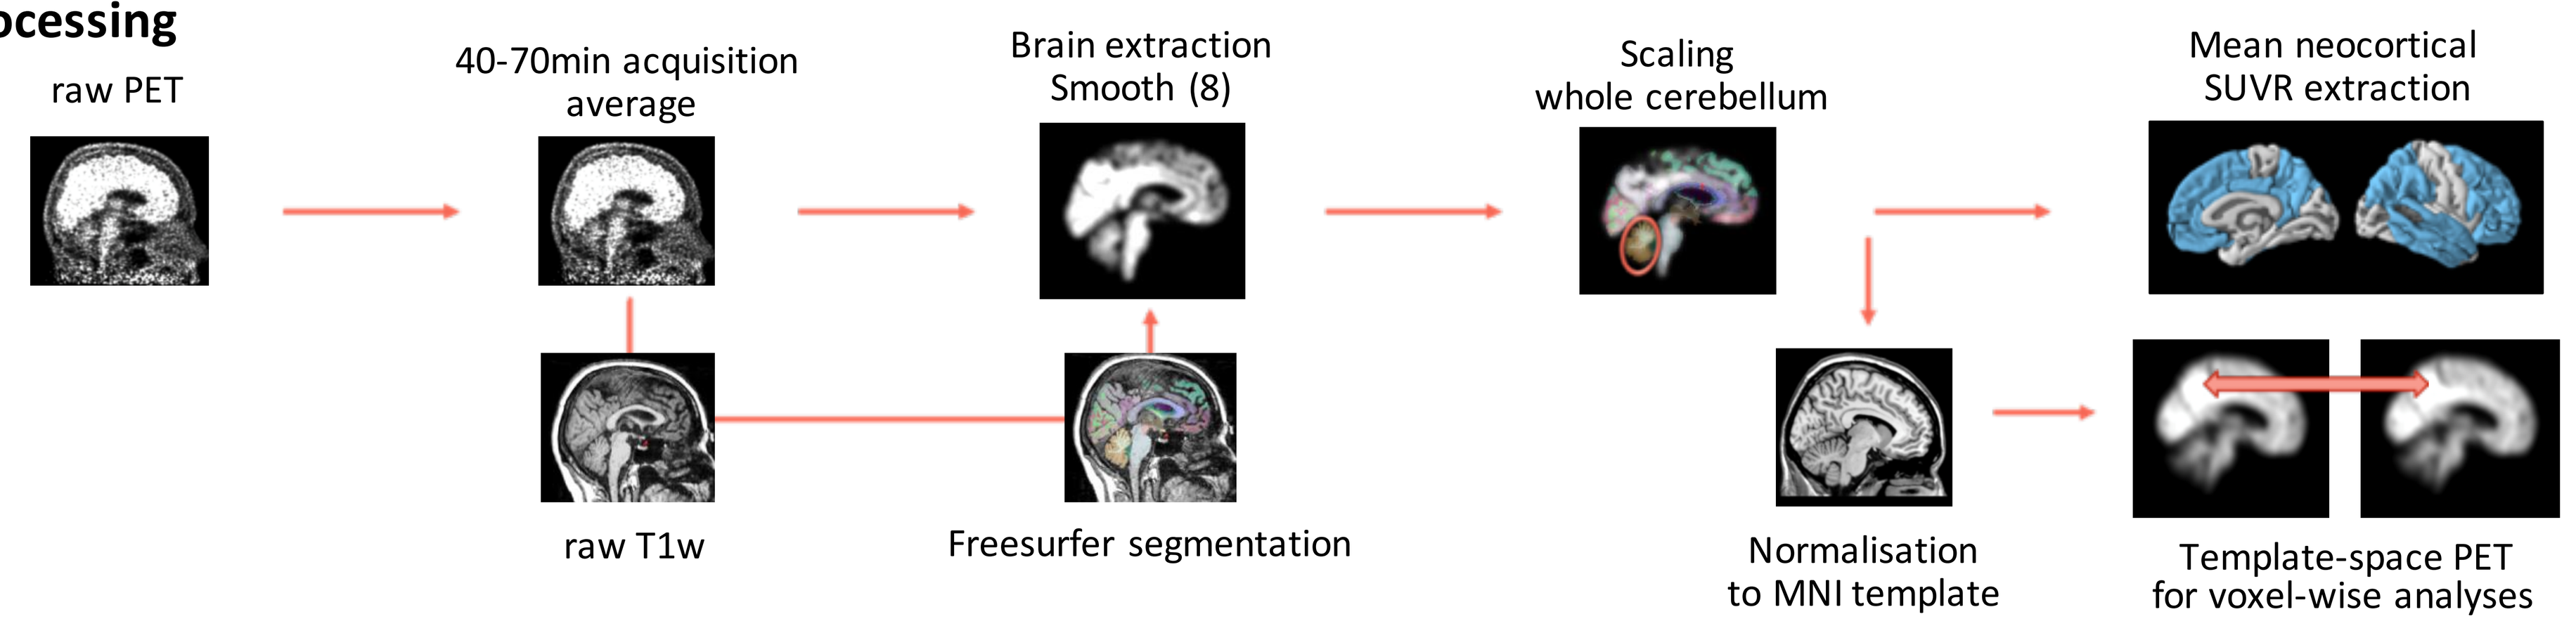
raw PET
40-70min acquisition average
Brain extraction Smooth (8)
Scaling whole cerebellum
Mean neocortical SUVR extraction
raw T1w
Freesurfer segmentation
Normalisation to MNI template
Template-space PET for voxel-wise analyses

Analyses

1- We assessed the effect of EYO on Aβ burden in both cohorts using partial correlations, controlling for age and sex (+ site and mutation type in ADAD)

2- We assessed the effect of *APOE*4 status, education, and their interaction on Aβ burden in both cohorts using general linear models, controlling for age, sex and MMSE (+ site and mutation type in ADAD)

Analyses were replicated voxel-wise using SPM12 .

Results

1) Can a parental EYO, as developed in ADAD, predict cerebral Aβ burden in preclinical sAD?

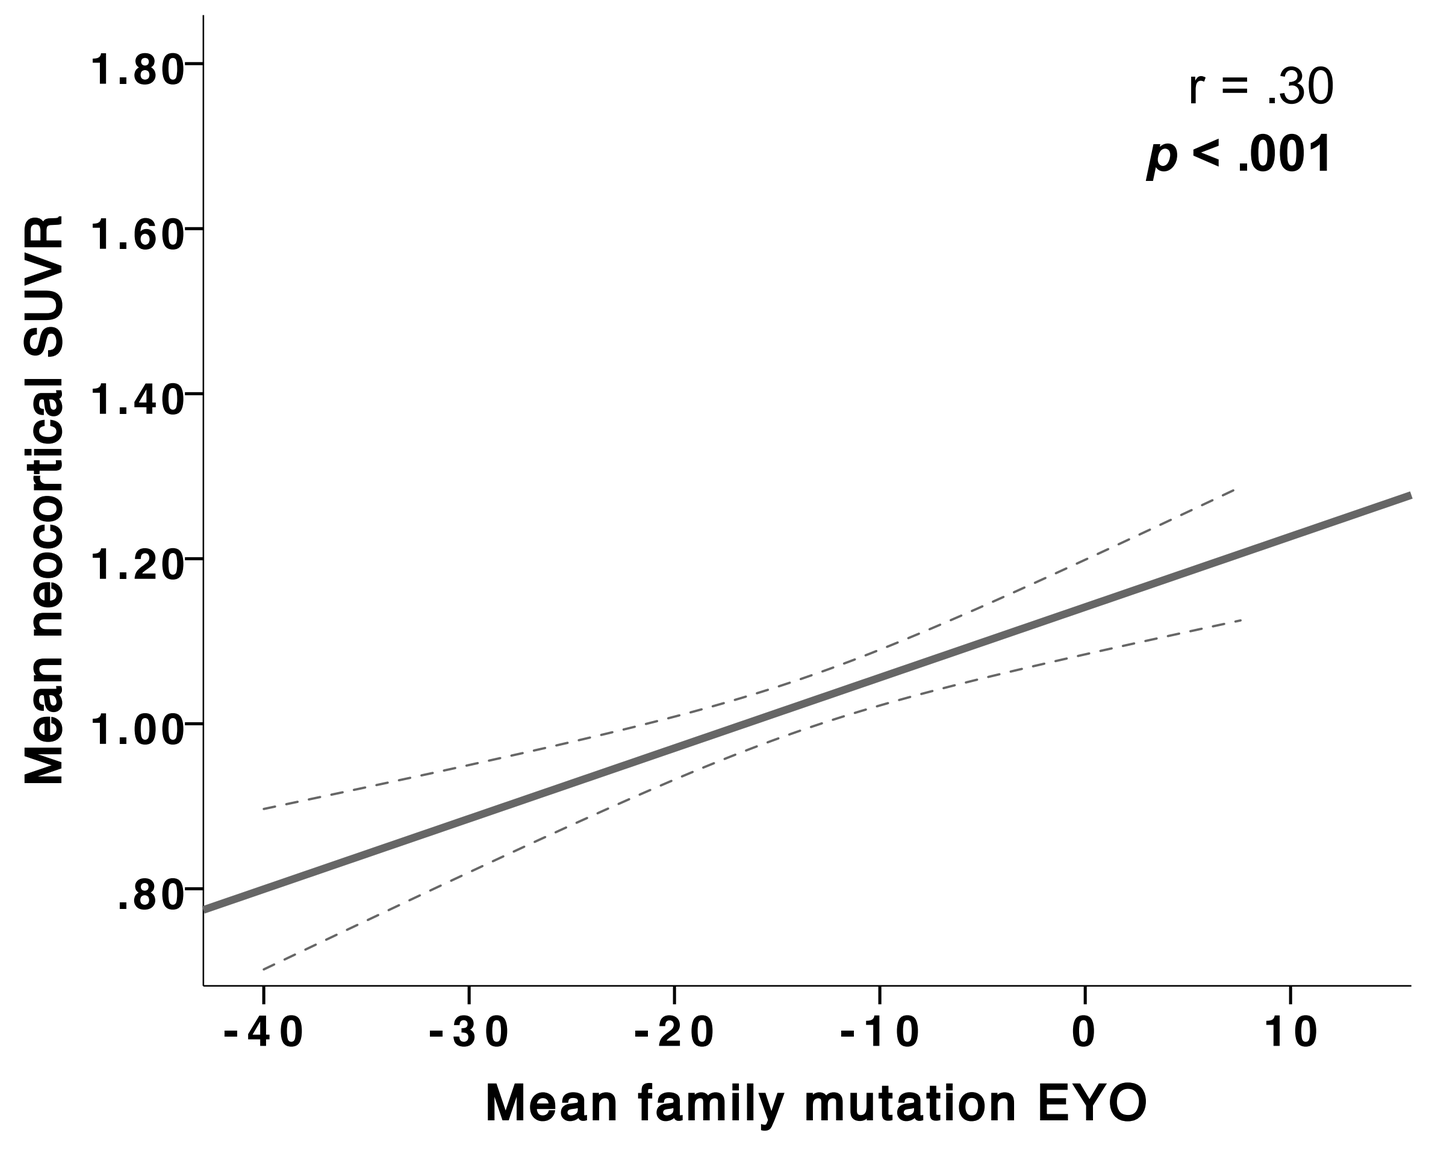
r = .30
p < .001
Mean neocortical SUVR
1.80 1.60 1.40 1.20 1.00 .80
-40 -30 -20 -10 0 10
Mean family mutation EYO

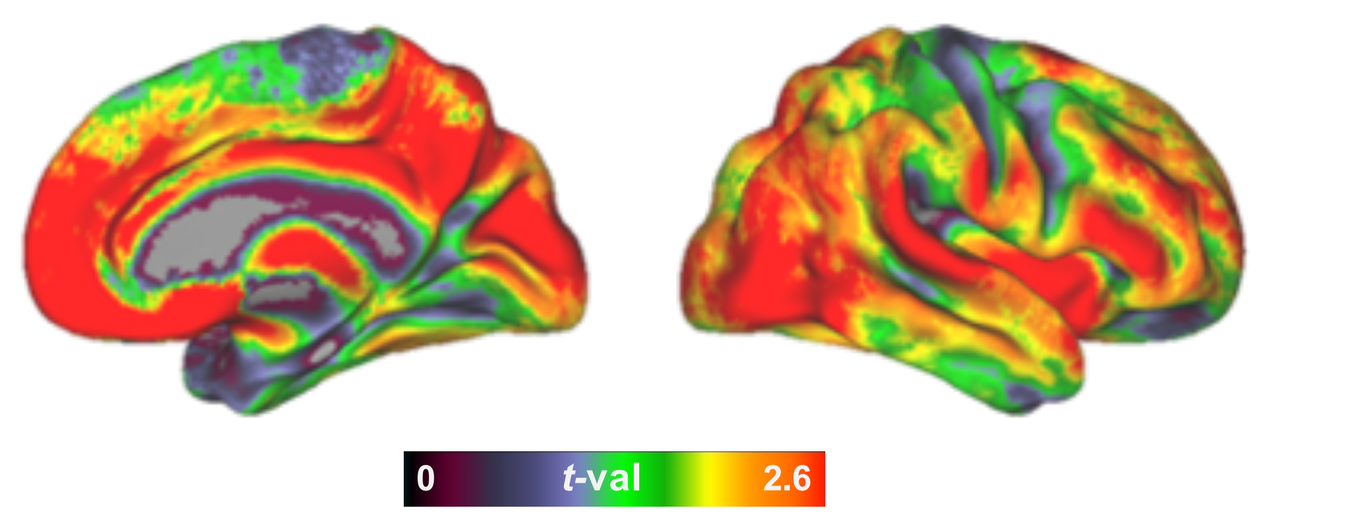
0 t-val 2.6

Aβ burden increases as presymptomatic ADAD mutation carriers approach their expected age of symptom onset.

This effect is largely distributed across the AD-sensitive regions.

controlled for age, sex, site and mutation type

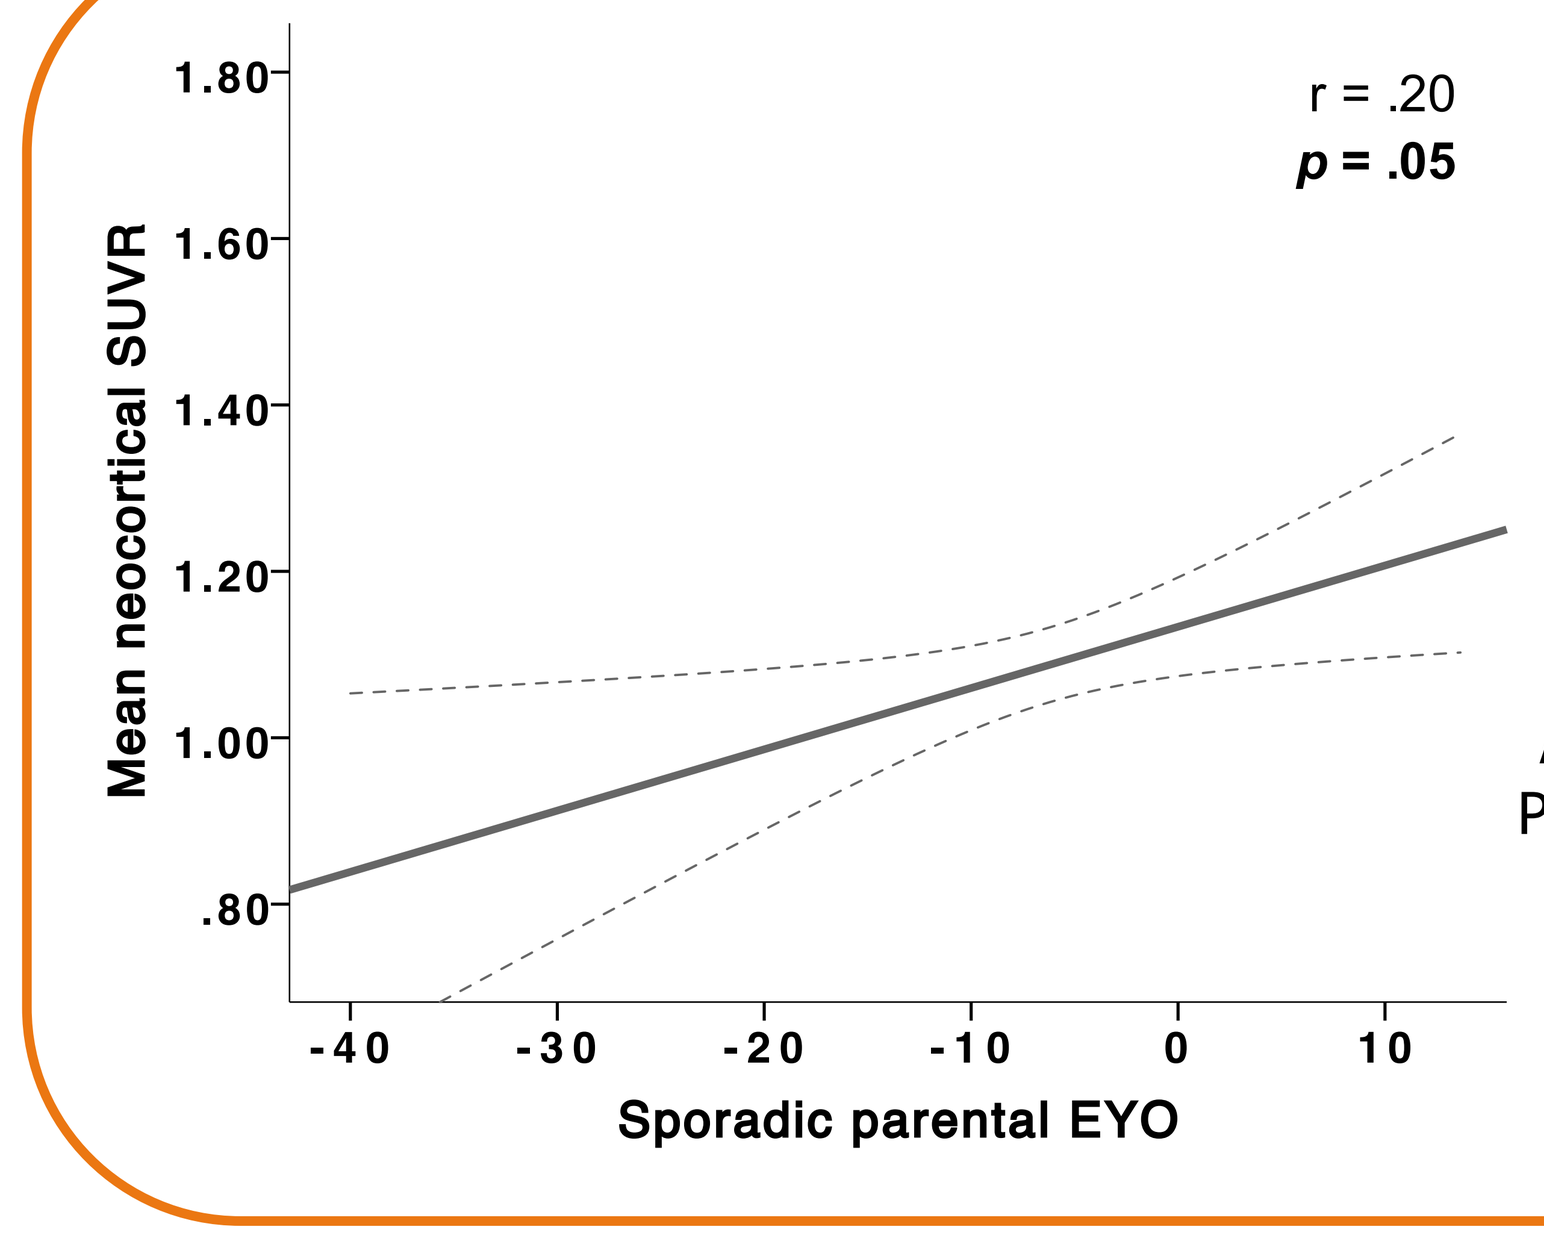
r = .20
p = .05
Mean neocortical SUVR
1.80 1.60 1.40 1.20 1.00 .80
-40 -30 -20 -10 0 10
Sporadic parental EYO

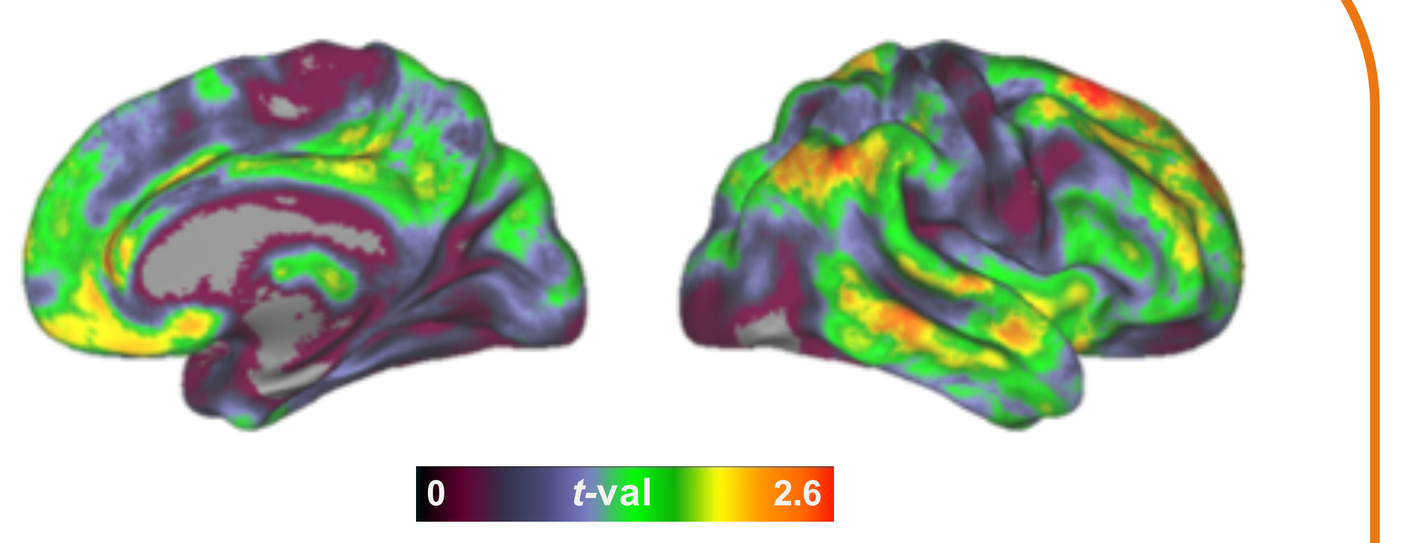
0 t-val 2.6

Aβ burden increases as asymptomatic individuals with a PH of sAD approach their parent's age at symptom onset.

This effect is distributed across the AD-sensitive regions.

controlled for age and sex

2) Do factors known to affect Aβ accumulation in asymptomatic individuals at risk of sAD (i.e. *APOE*4 and education) also affect Aβ trajectories in presymptomatic ADAD mutation carriers?

In presymptomatic ADAD mutation carriers, Aβ burden is not affected by *APOE*4 status, but it decreases as years of education increase.

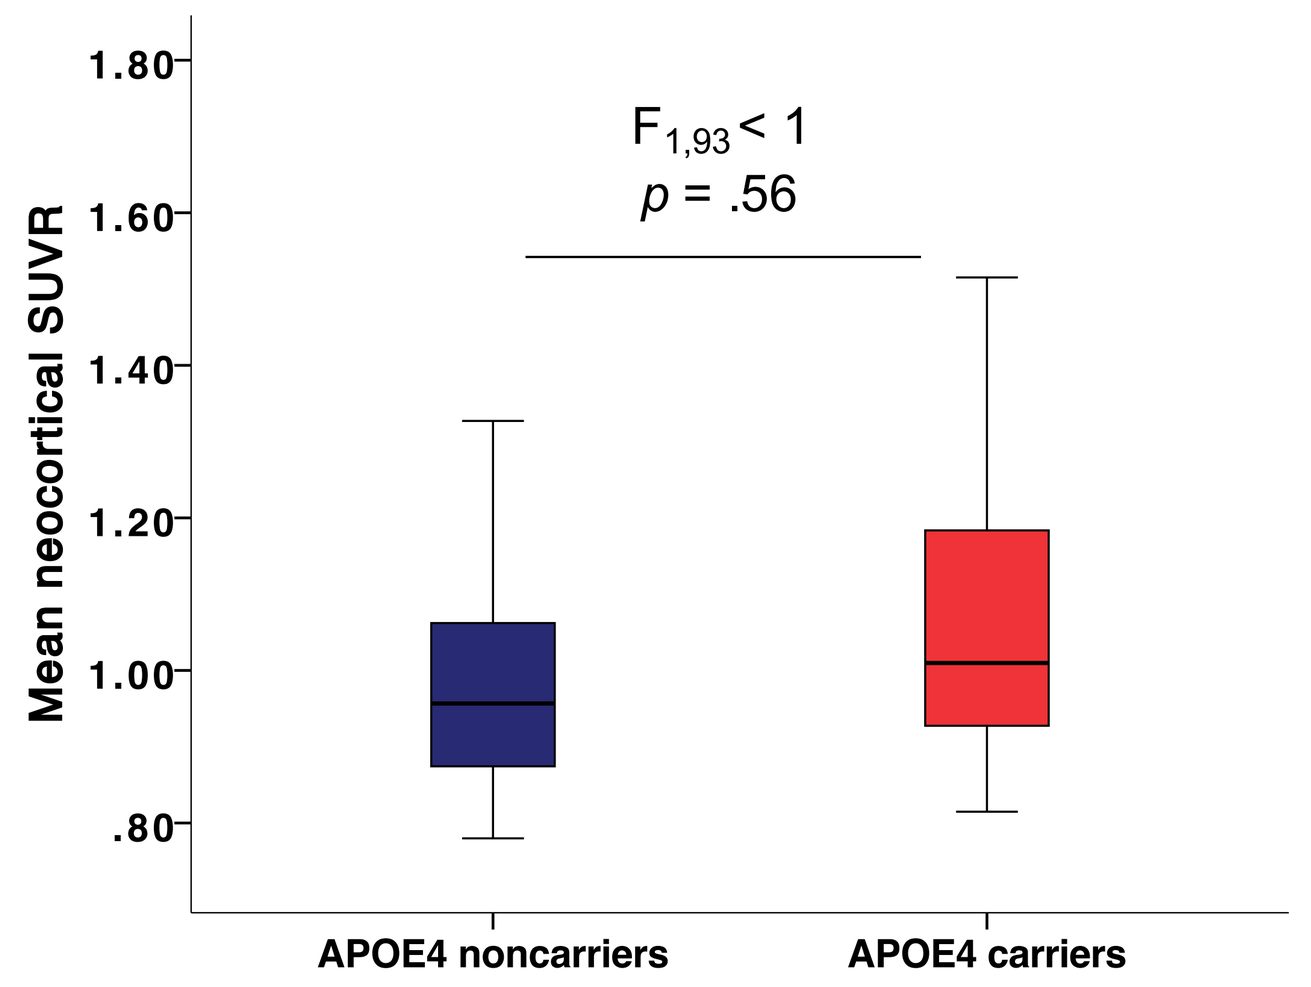
F1,93 < 1
p = .56
Mean neocortical SUVR
APOE4 noncarriers
APOE4 carriers

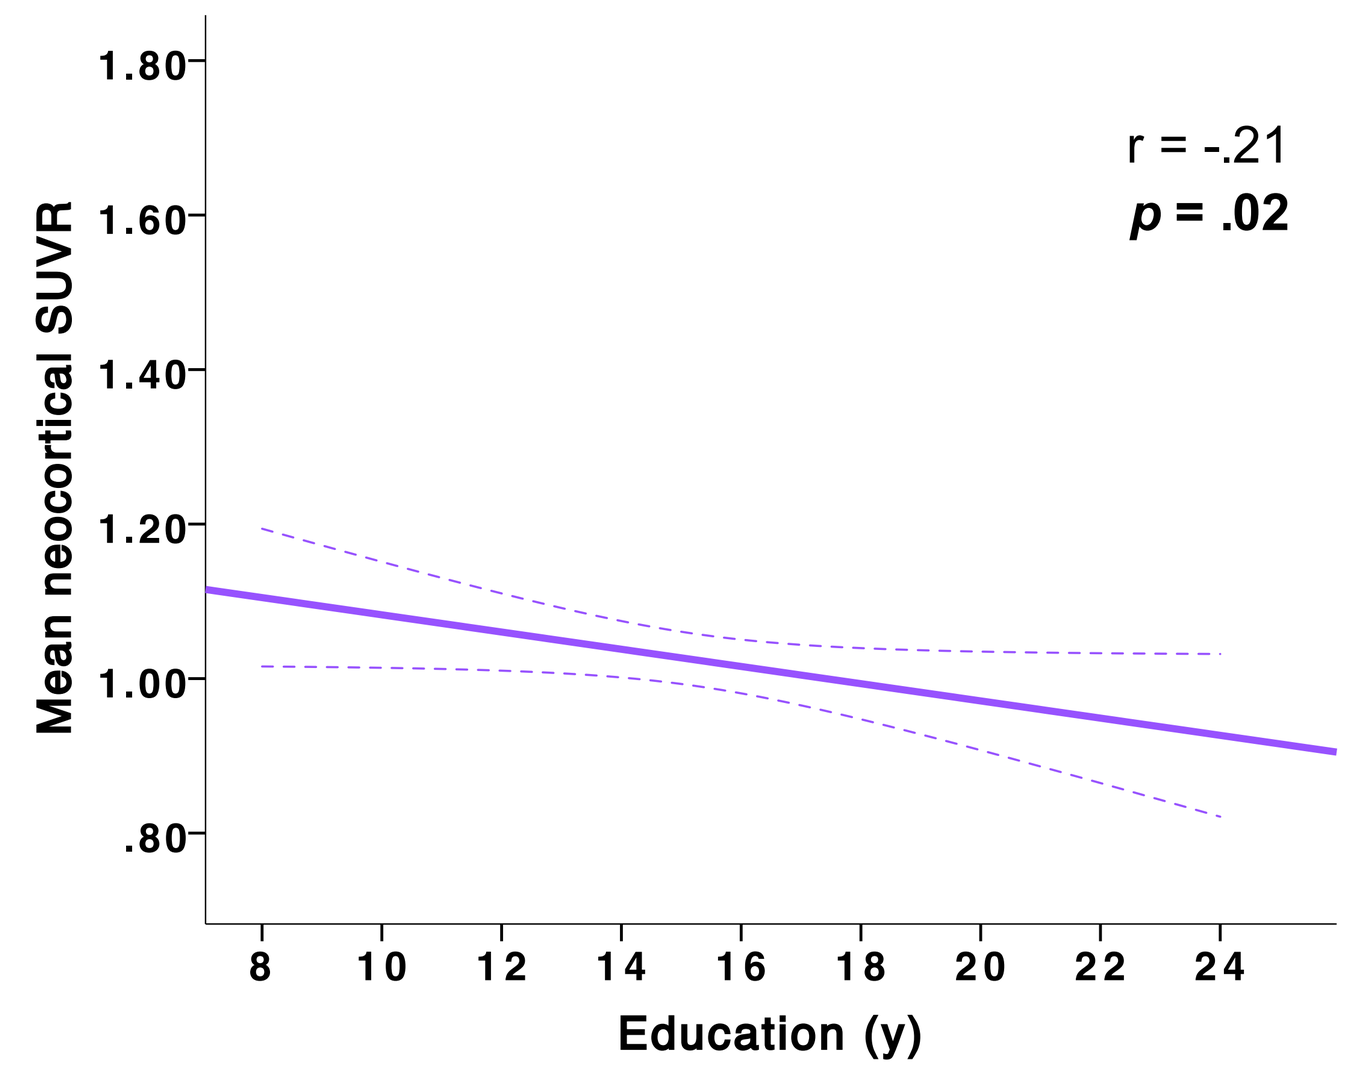
r = -.21
p = .02
Mean neocortical SUVR
Education (y)

There is no *APOE*4 status * education interaction

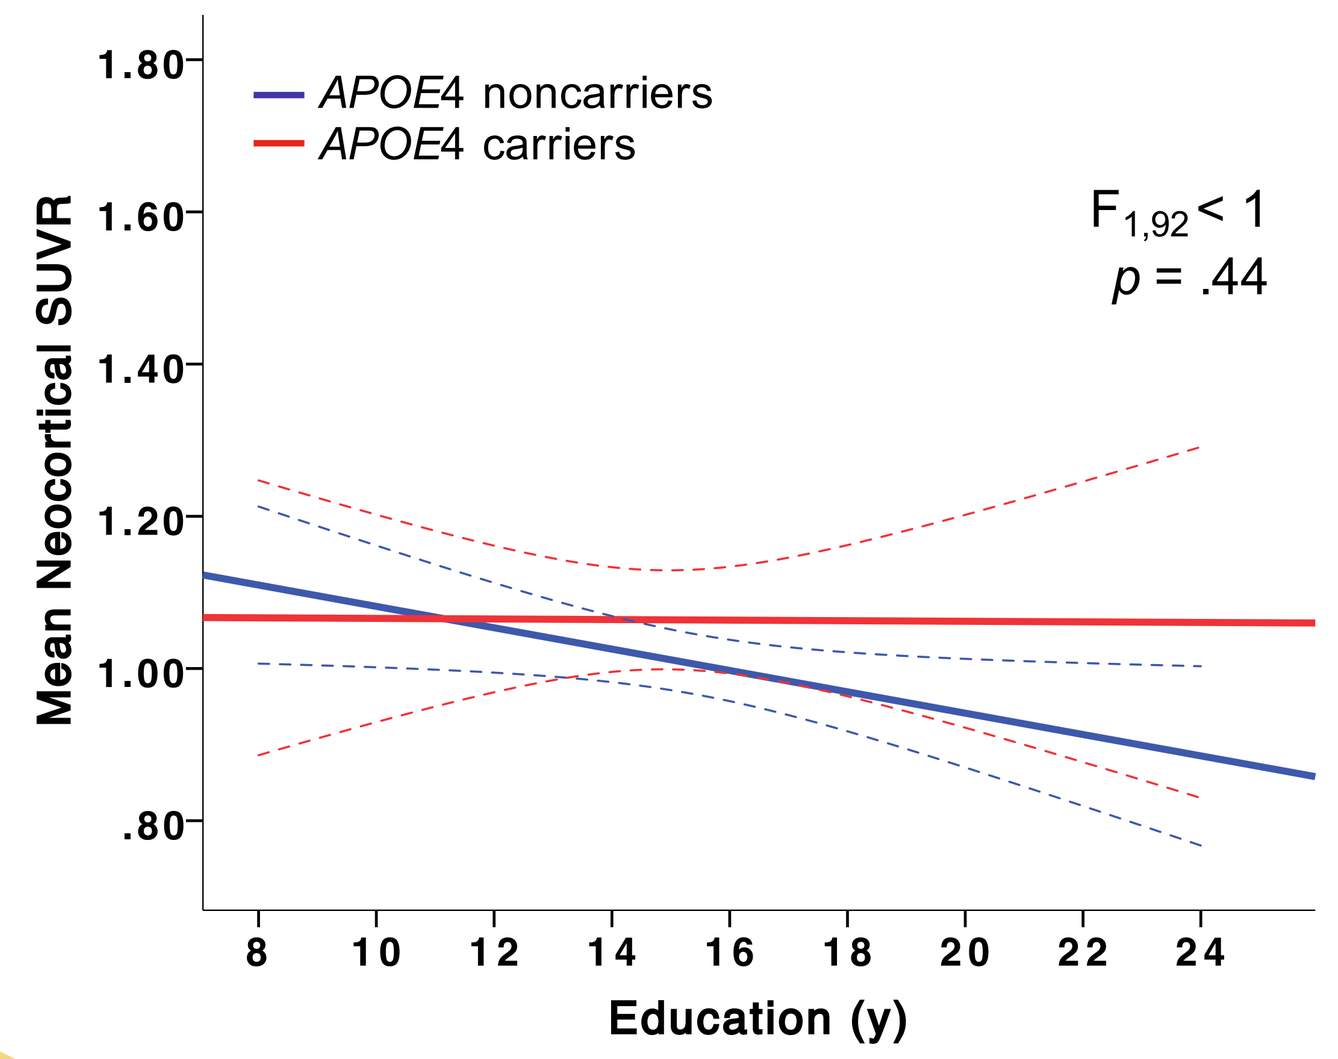
APOE4 noncarriers
APOE4 carriers
F1,92 < 1
p = .44
Mean Neocortical SUVR
Education (y)

Effect of education on Aβ in ADAD presymptomatic mutation carriers

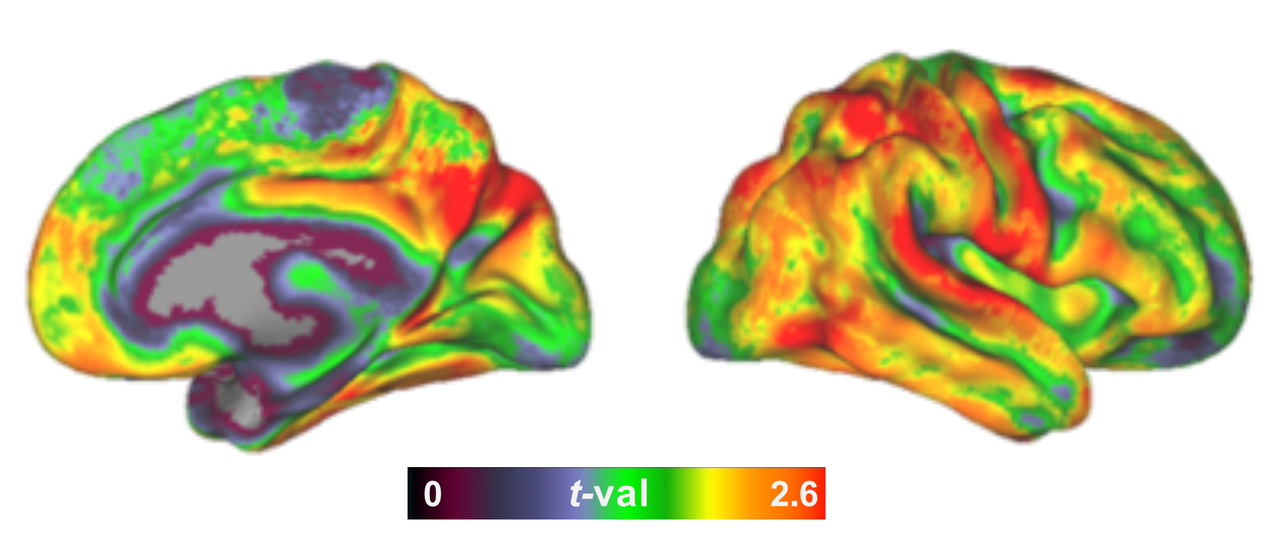
0 t-val 2.6

controlled for age, sex, MMSE, site and mutation type

In asymptomatic individuals with a PH of sAD, Aβ burden is higher in *APOE*4 carriers than in non-carriers, and it decreases as a function of years of education.

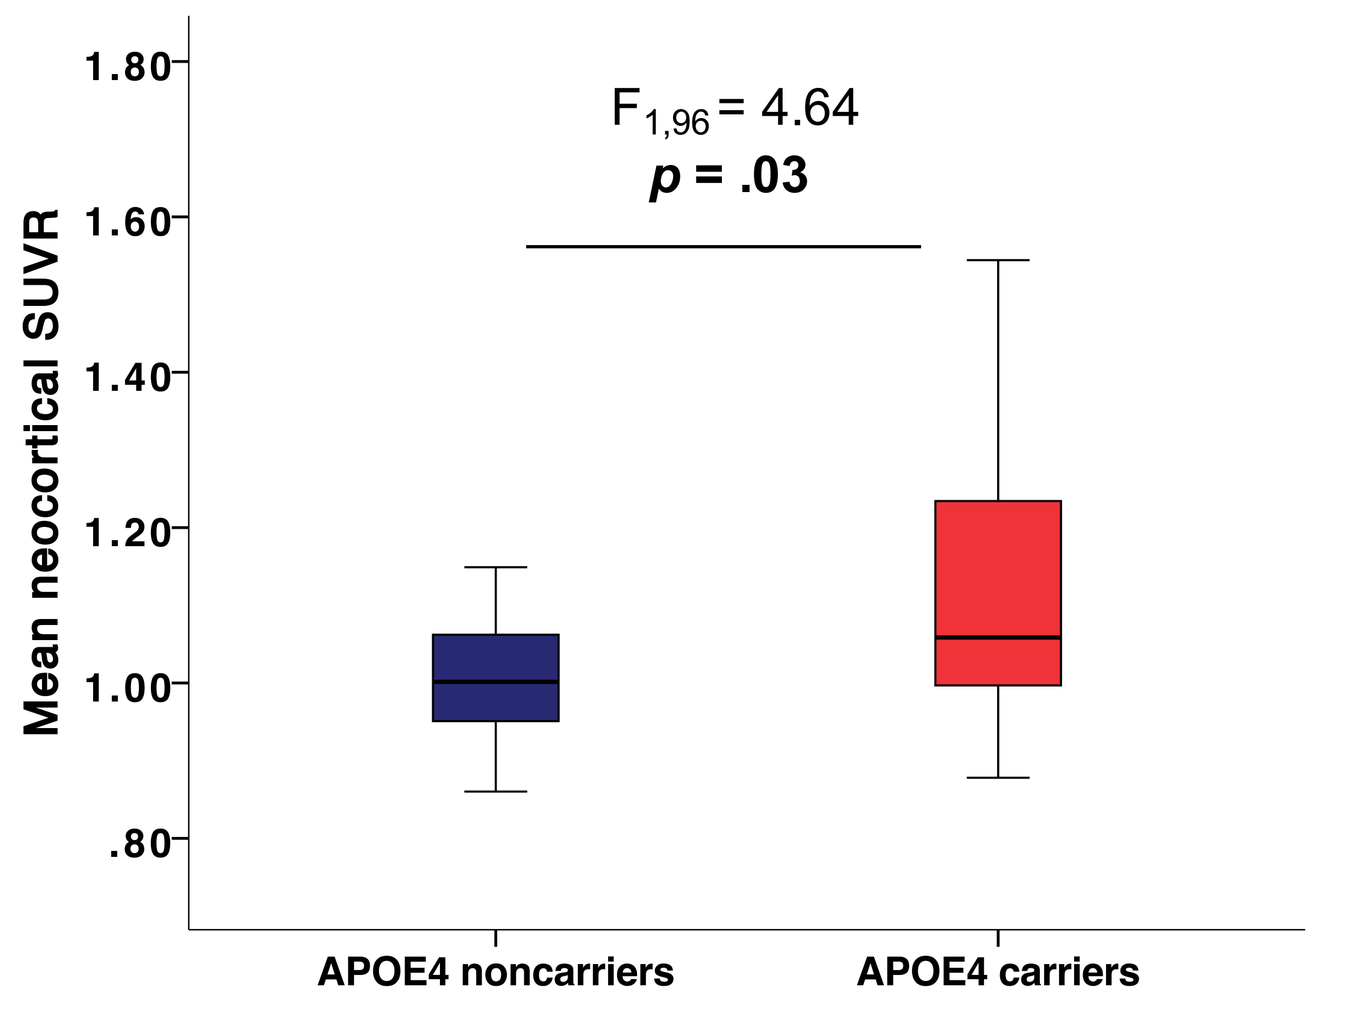
F1,96 = 4.64
p = .03
Mean neocortical SUVR
APOE4 noncarriers
APOE4 carriers

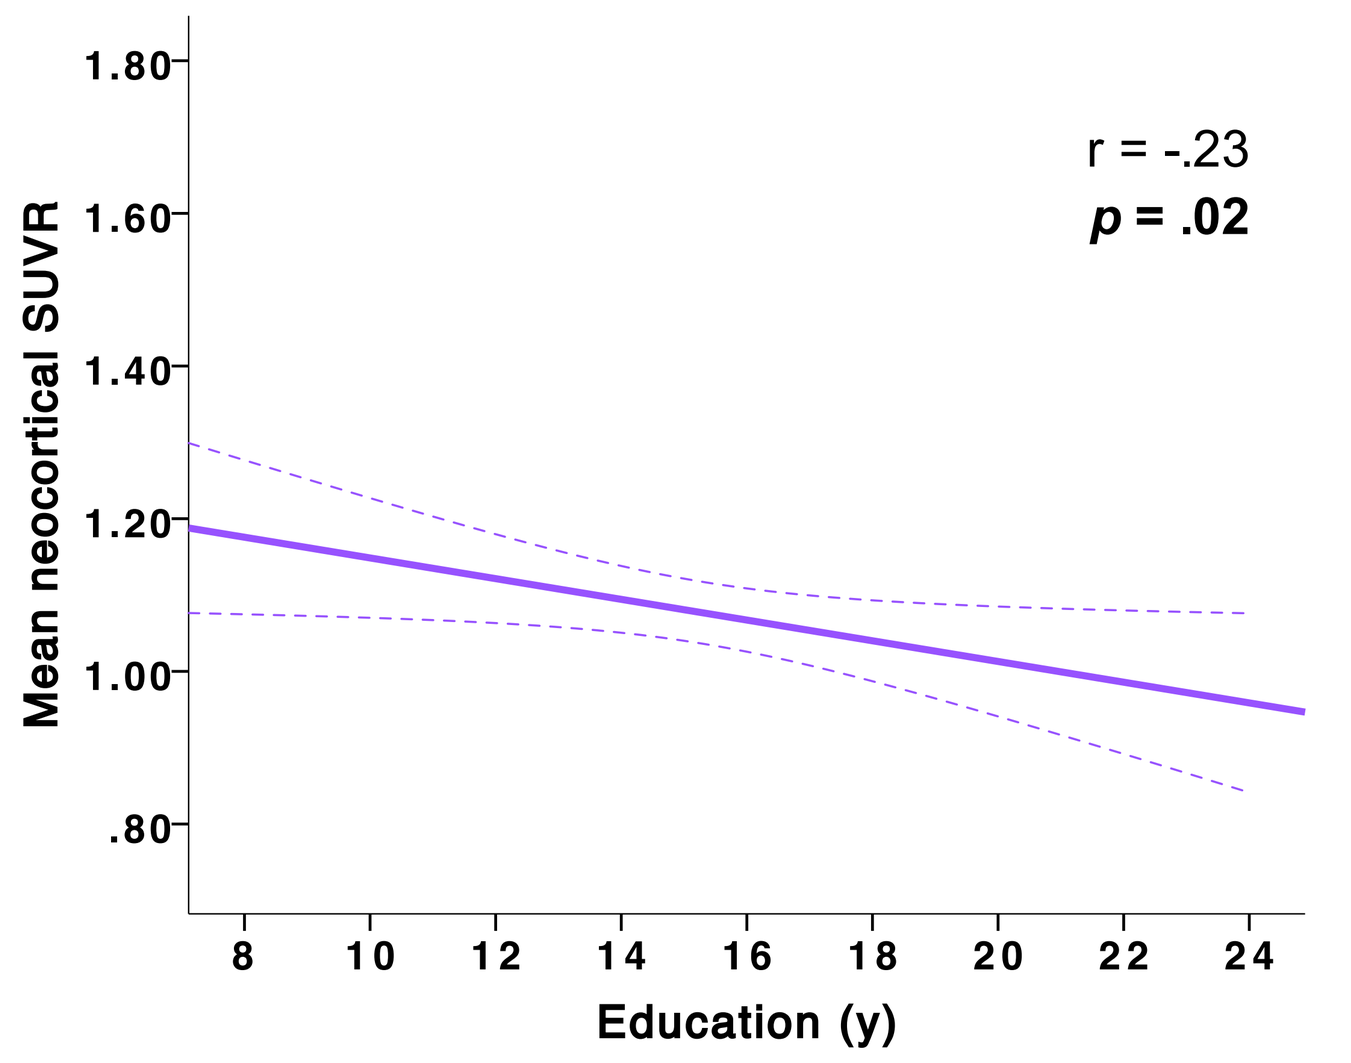
r = -.23
p = .02
Mean neocortical SUVR
Education (y)

There is an *APOE*4 status * education interaction, such that the effect of education is found only in *APOE*4 carriers

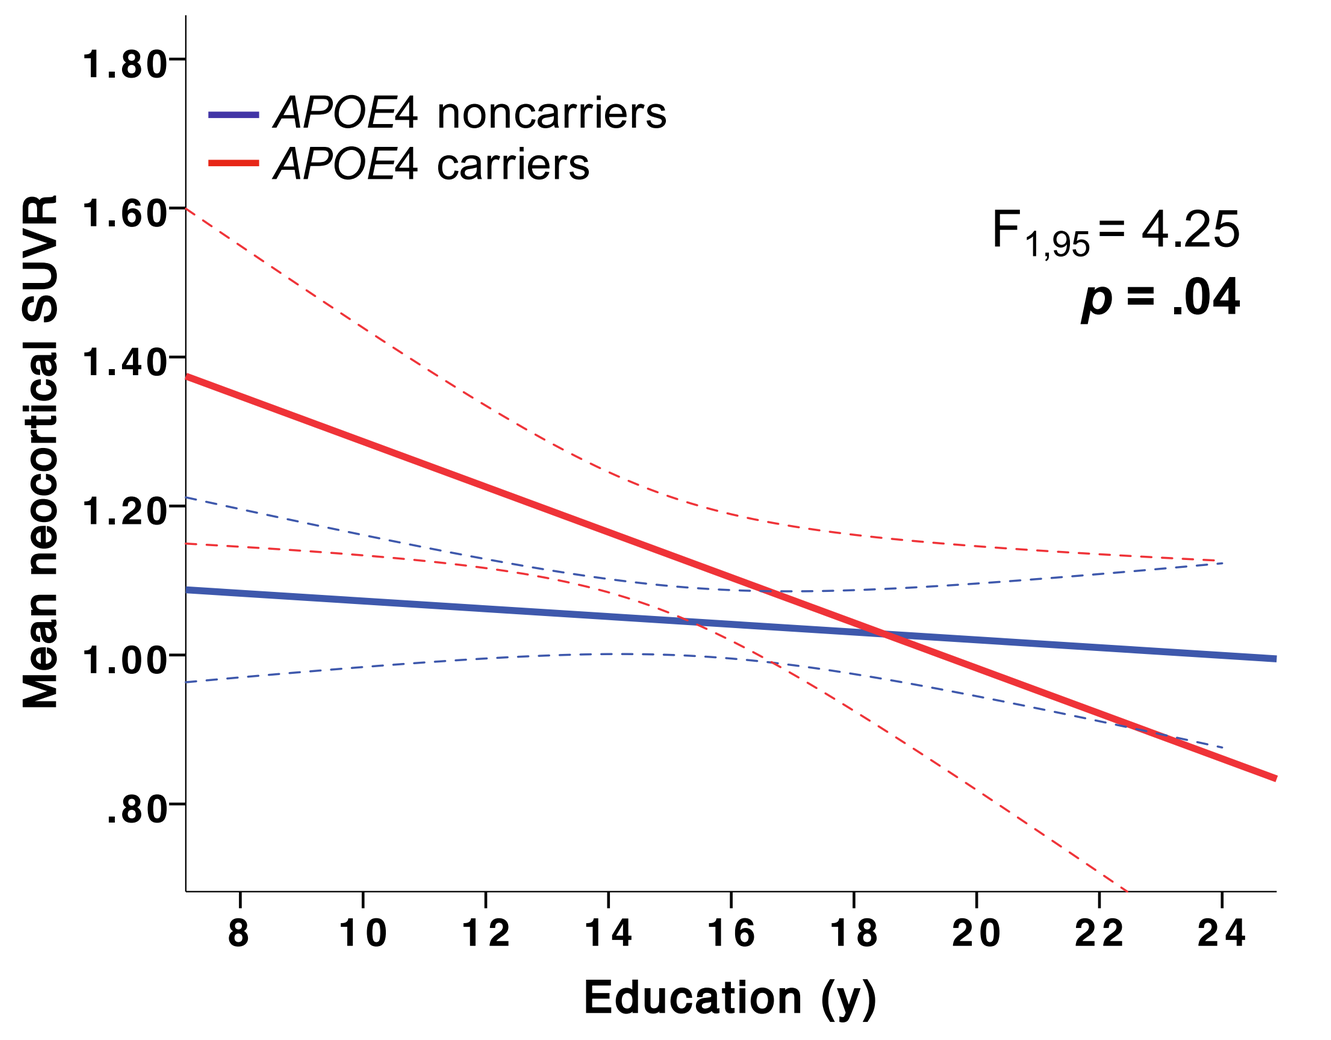
APOE4 noncarriers
APOE4 carriers
F1,95 = 4.25
p = .04
Mean neocortical SUVR
Education (y)

Effect of education on Aβ in *APOE*4 carriers

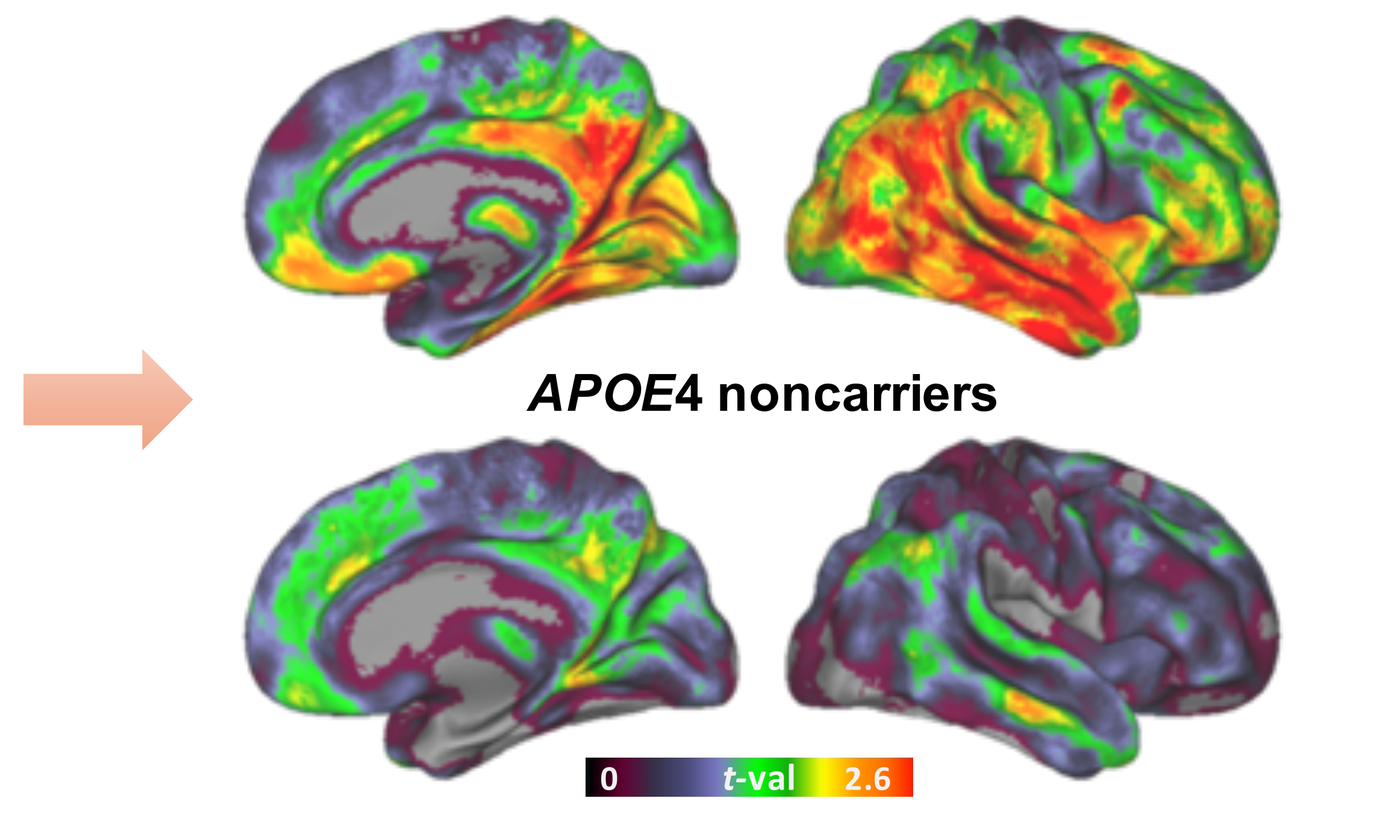
APOE4 noncarriers
0 t-val 2.6

controlled for age, sex and MMSE

Summary and conclusions

Our results suggest that, as it is the case in ADAD, a parental EYO index might help to predict Aβ accumulation in asymptomatic individuals at risk of sAD.[6,7] This might have important implications for the characterization of the preclinical phase of sAD and the recruitment of prevention/clinical trials.

While *APOE*4 is highly associated with Aβ burden in people at risk of sAD, it has no impact in ADAD.

By contrast, environmental factors, approximated here using years of education, could affect biomarker progression in both variants of the disease. Furthermore, education seems able to counteract the deleterious effect of *APOE*4 on Aβ burden in asymptomatic individuals with a PH of sAD.[8] These results suggest the existence of reserve mechanisms, not only in individuals at risk of sAD , but also in individuals carrying aggressive ADAD variants.

References: 1. Bekris et al. *J Geriatr Psychiatry Neurol* (2010); 2. Bateman et al. *N Engl J Med* (2012); 3. Gonneaud et al. *Neurology* (2016); 4. Morris et al. *Ann Neurol* (2010); 5. Arenaza-Urquijo et al. *Neurobiol Aging* (2017); 6. Lo et al. *Alzheimer Dis Assoc Disord* (2013); 6. Villeneuve et al. *JAMA Neurol* (2018); 7. Vogel et al. *Brain* (2018); 8. Wirth et al. *J Neurosci* (2012)

Authors have no disclosure.

Acknowledgments: We would like to thank all the participants and collaborators from the DIAN and PREVENT-AD studies and all members of the Villeneuve lab who contributed to this study.

contact : julie.gonneaud@gmail.com | **poster downloadable @ villeneuvelab.com**

Funding

Fonds de recherche Santé Québec; alzheimer's association; Société Alzheimer; Canada Research Chairs / Chaires de recherche du Canada; CIHR IRSC; INNOVATION.CA Canada Foundation for Innovation / Fondation canadienne pour l'innovation; fondation famille lemaire; RBIQ Réseau de bio-imagerie du Québec