Years to Parental Symptom Onset Predicts Amyloid Burden in Healthy Elderly with a Parental History of Sporadic Alzheimer's Disease.

Villeneuve S,[1] Vogel JW,[1] Gonneaud J,[1] Pichet Binette A,[1] Rosa-Neto P,[1] Gauthier S,[1] Bateman RJ,[2] Fagan AM,[2] Morris JC,[2] Benzinger TL,[2] Johnson SC,[3] Breitner JC,[1] Poirier J,[1] Prevent-AD group.

[1] McGill University; [2] Washington University; [3] University of Wisconsin

Background and objectives

- In the autosomal dominant genetic forms of Alzheimer's disease (AD), symptom onset is predictable across generations. It is possible to estimate the number of years until symptom onset (EYO) in individuals that have a genetic mutation causing AD by subtracting their parent's symptom onset age from their current age.[1]
- Whether the age of the parent's symptom onset can help predict biomarker abnormalities in sporadic AD is not known.
- We sought to assess whether amyloid-beta (Aβ) burden, a pathological hallmark of AD, appears in a similarly predictable manner among cognitively normal persons with a parental history of sporadic AD.
- Because apolipoprotein E (*APOE*) ε4 and female gender increase the risk of sporadic AD, we further assessed whether the relationship between spEYO and AD biomarkers are influenced by these risk factors.

Method: Participants

- Analyses were performed on cognitively normal older adults from the Pre-symptomatic Evaluation of Novel or Experimental Treatments for Alzheimer's Disease (PREVENT-AD) cohort, McGill University, Montreal, Canada.[2]
- The main analyses were repeated in two independent cohorts [the Knight Alzheimer's Disease Research Center Adult Children Study (ACS)[3] and the Wisconsin Registry for Alzheimer's Prevention (WRAP)[4]] using cerebrospinal fluid (CSF) and Pittsburgh compound B positron emission tomography (PIB-PET) Aβ biomarkers.

	Prevent-AD (n = 101)	**ACS** (n = 128)	**WRAP** (n = 135)	Group differences
Age	61.75 ± 5.11	63.39 ± 5.08	59.88 ± 6.00	a,b,c
Male	29.41%	27.68%	31.76%	
Education	14.99 ± 2.97	15.87 ± 2.33	16.49 ± 2.51	a,b
***APOE*4**	38.24%	43.75%	42.35%	
CDR = 0	100%	100%	70.97%	
MMSE	-	29.02 ± 1.29	29.21 ± 1.16	
MOCA	27.87 ± 1.50	-	-	
spEYO	-12.52 ± 6.68	-10.21 ± 8.70	-13.87 ± 7.39	a,c

a Difference between Prevent-AD and ACS; **b** Difference between Prevent-AD and WRAP; **c** Difference between ACS and WRAP, $p < 0.05$

Method: Aβ assessment

- CSF $A\beta_{1\text{-}42}$ levels from the PREVENT-AD and the ACS cohort were determined by INNOTEST ELISA. CSF $A\beta_{1\text{-}42}$ levels from the WRAP were quantified by electrochemiluminescence using an Aβ triplex assay.
- PIB-PET was not available for the PREVENT-AD cohort; PIB-PET from the ACS cohort [60-minute, cerebellar ref. region]; PIB-PET from the WRAP cohort [70-minute, cerebellar ref. region].
- PIB-PET annual rate of change was calculated as follows:
 [Aβ scores at follow-up – Aβ scores at baseline] ÷ interval (in years) between the two scans

Method: spEYO calculation

The "Sporadic Parental Estimated Years from Symptom Onset" (spEYO) score was calculated as follows:

age of the participant at assessment – age of the parent at symptom onset

Proximity to parental symptom onset and amyloid burden in the PREVENT-AD cohort.

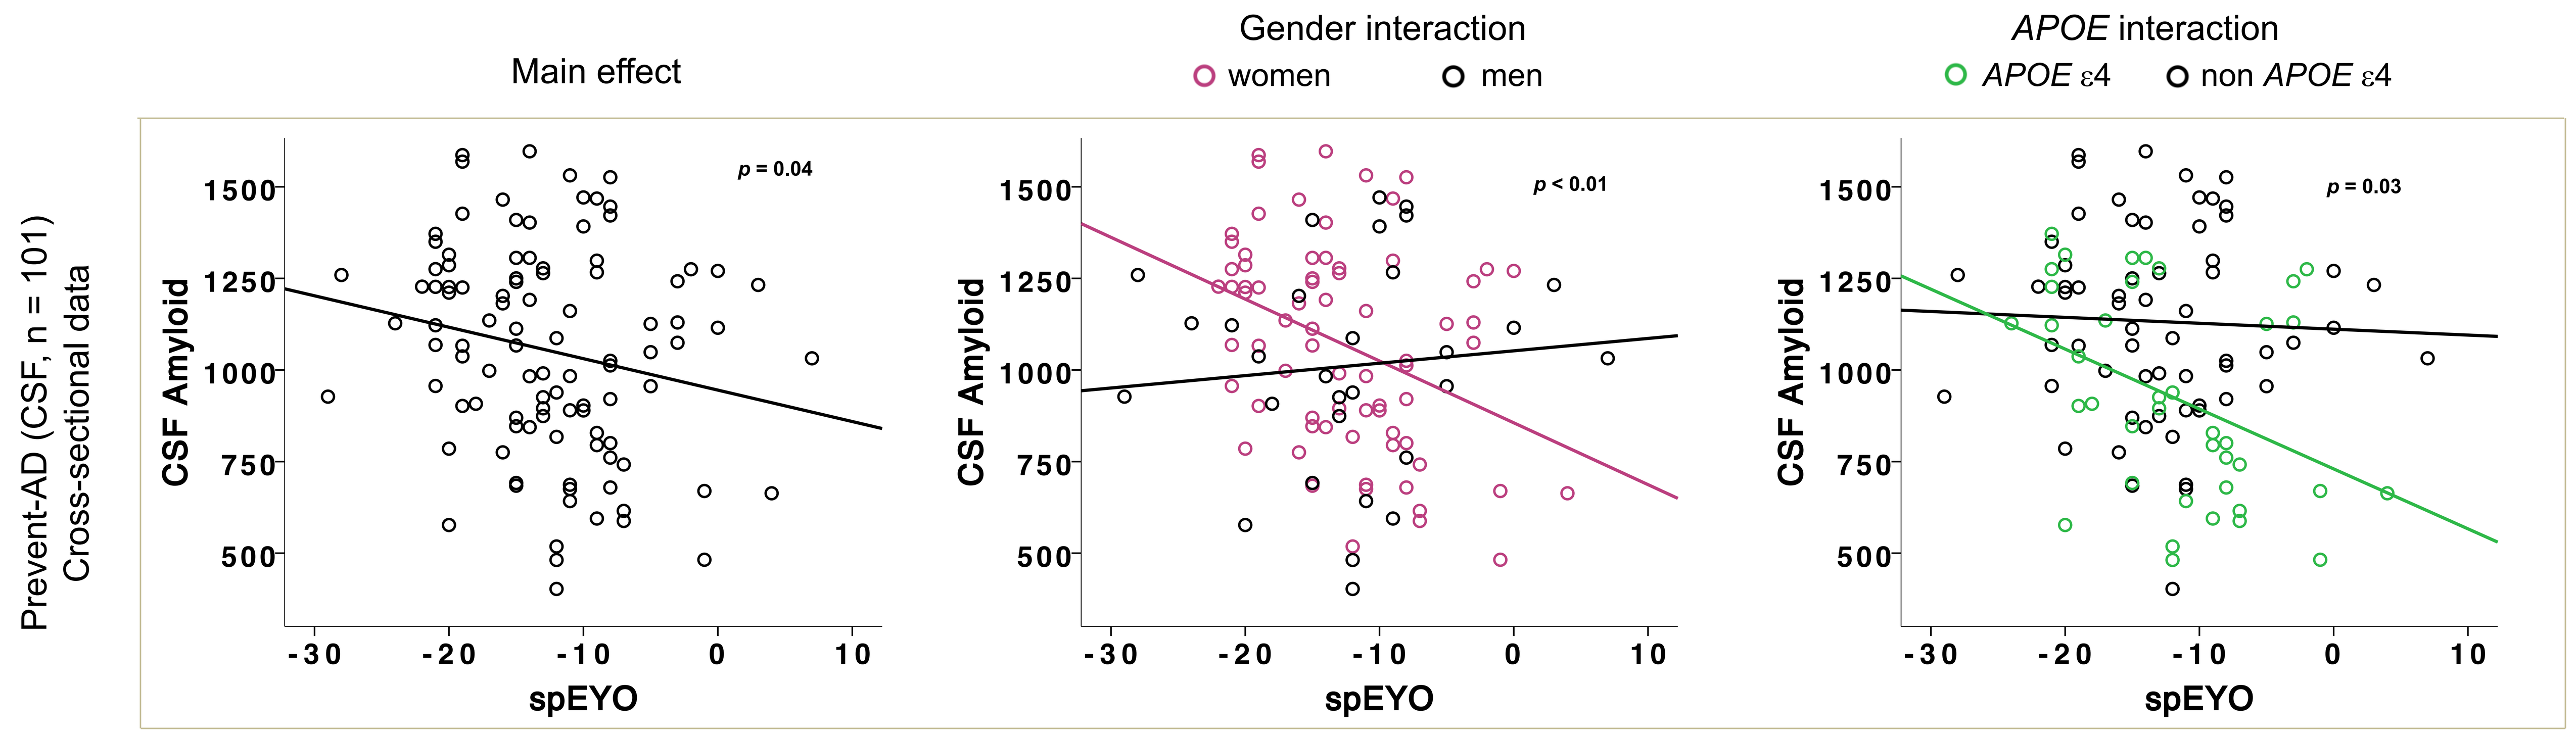

Main effect
Gender interaction
women
men
APOE interaction
APOE ε4
non APOE ε4
Prevent-AD (CSF, n = 101) Cross-sectional data
CSF Amyloid
spEYO
p = 0.04
p < 0.01
p = 0.03

- As individuals get closer to their parent's symptom age of onset they show decreased concentrations of CSF $A\beta_{1\text{-}42}$ (indicating increased brain Aβ deposition).
- These relationships are stronger in women (pink, second row) and in *APOE*4 carriers (green, third row).
- Analyses were controlled for age, gender and education.

ACS cohort

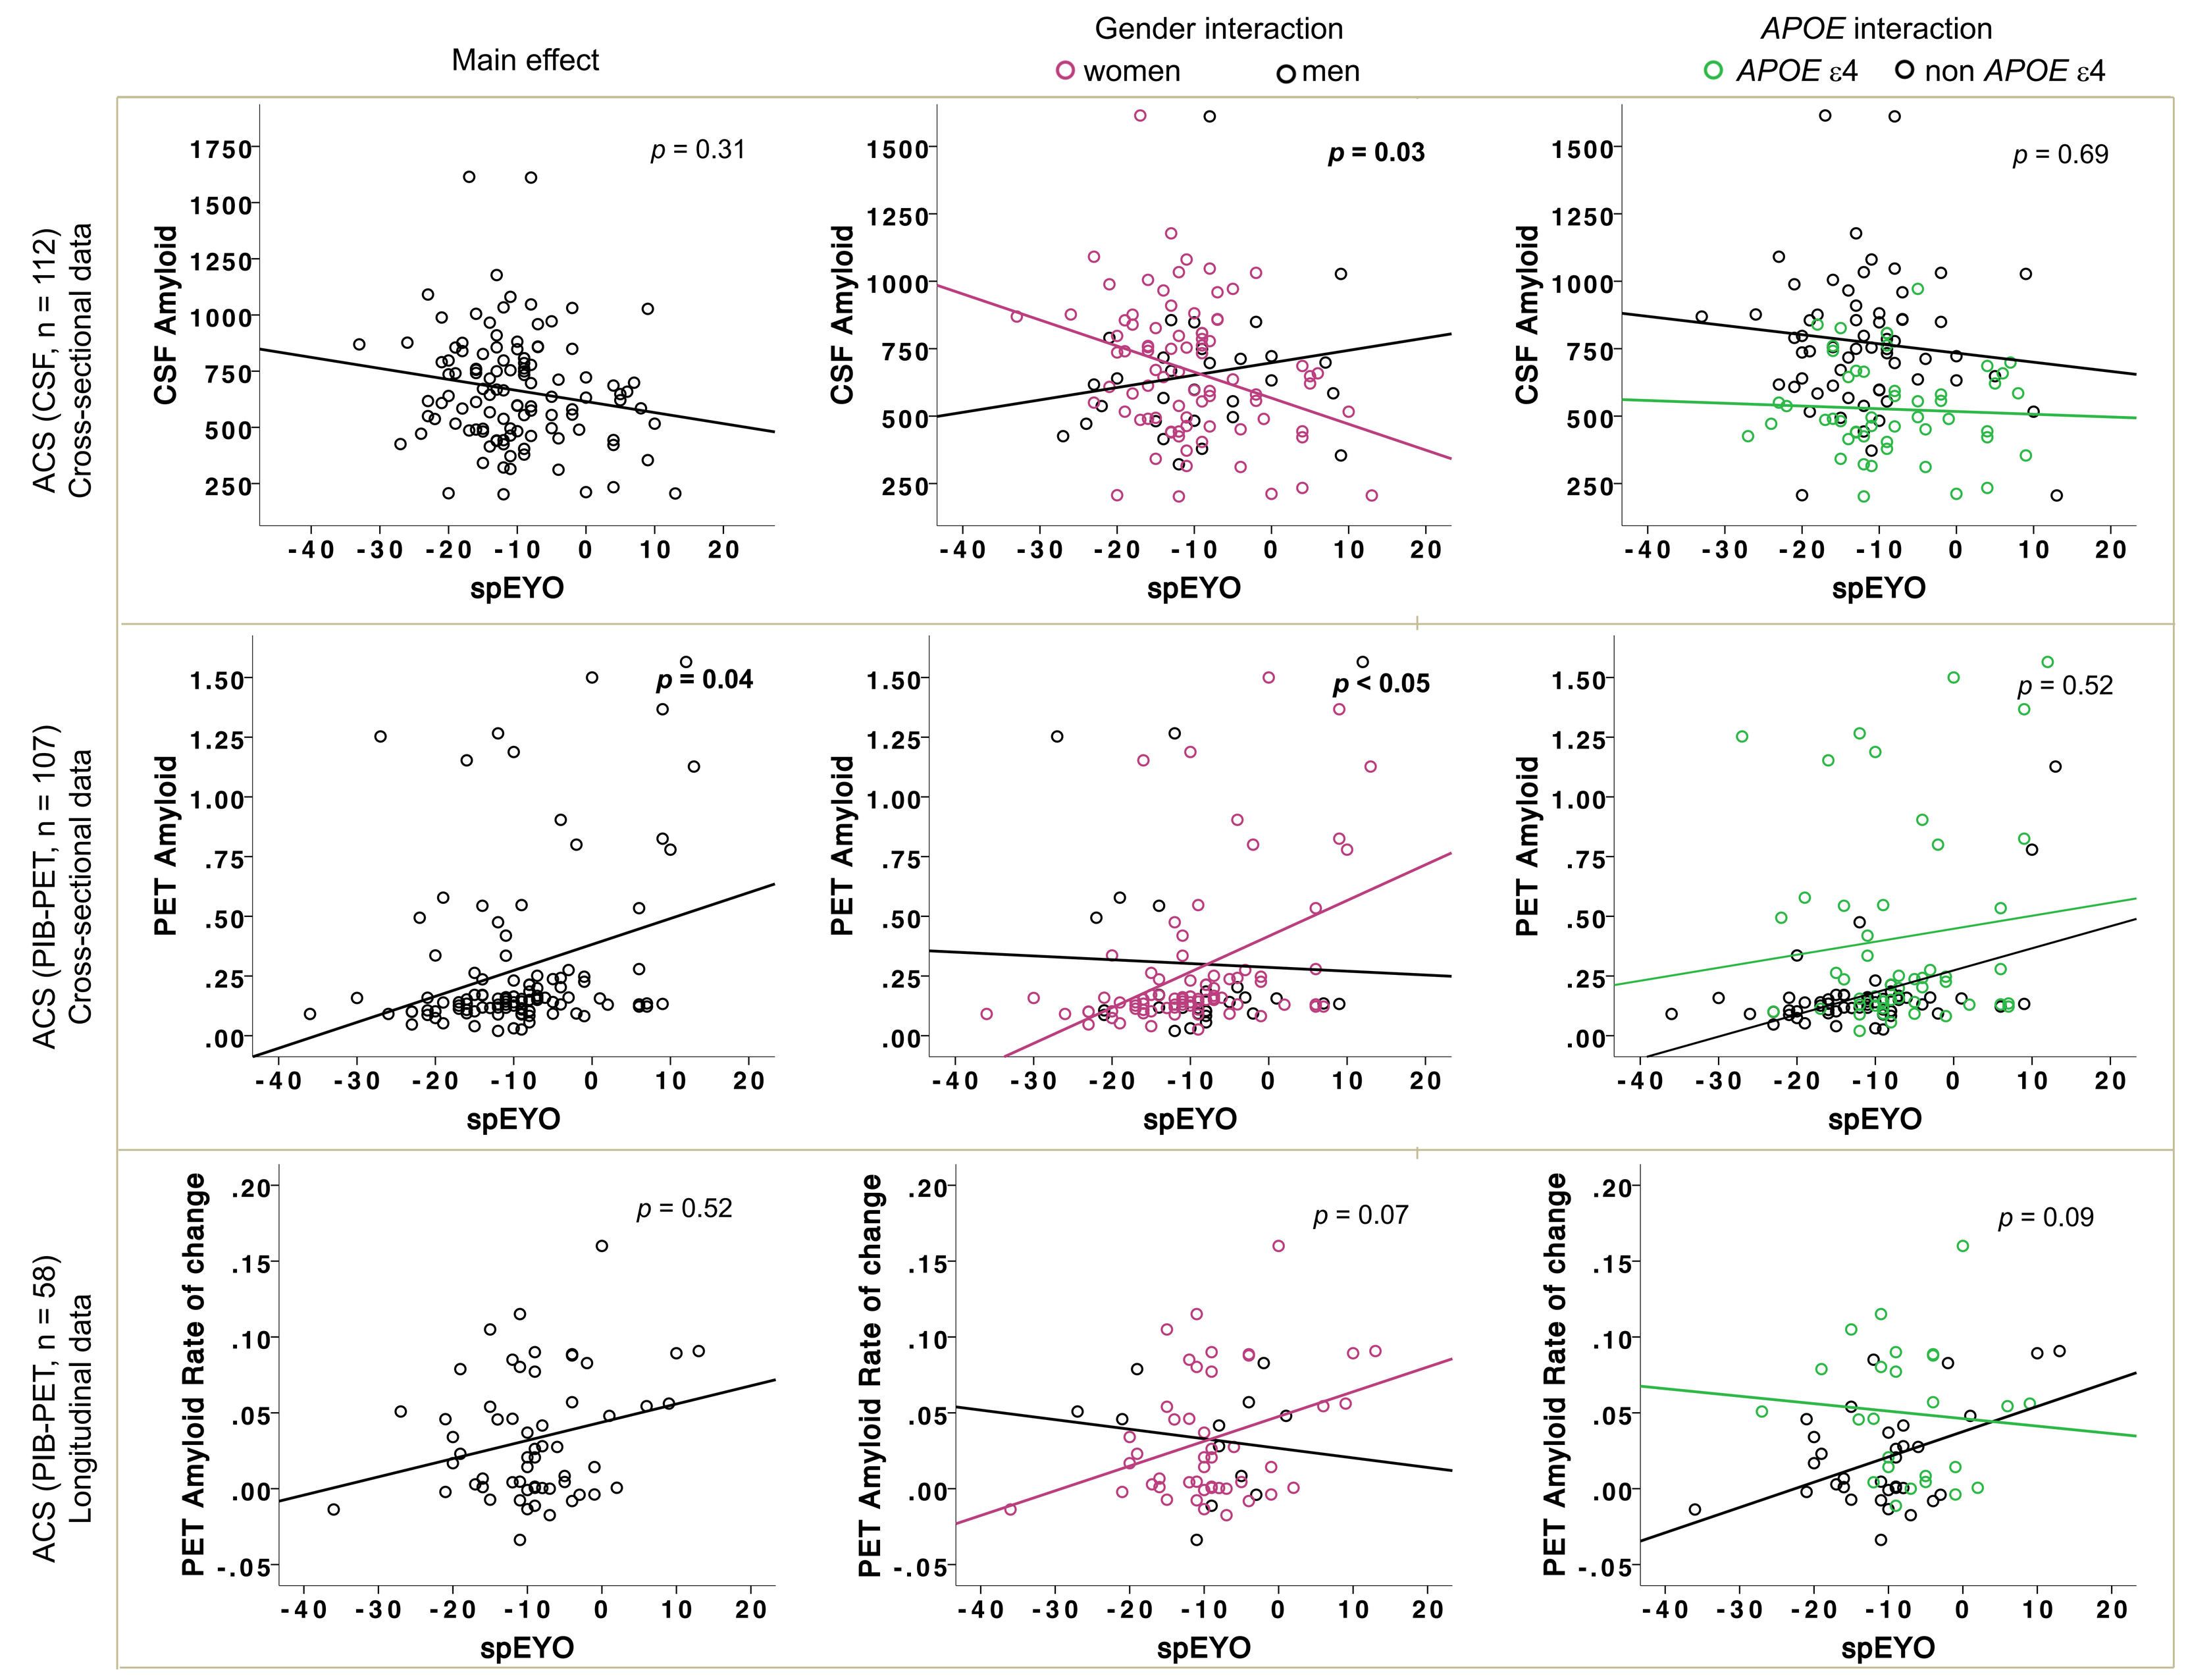

Main effect
Gender interaction
women
men
APOE interaction
APOE ε4
non APOE ε4
ACS (CSF, n = 112) Cross-sectional data
ACS (PIB-PET, n = 107) Cross-sectional data
ACS (PIB-PET, n = 58) Longitudinal data
CSF Amyloid
PET Amyloid
PET Amyloid Rate of change
spEYO
p = 0.31
p = 0.03
p = 0.69
p = 0.04
p < 0.05
p = 0.52
p = 0.52
p = 0.07
p = 0.09

- The relationship between spEYO and Aβ burden was replicated in the ACS PIB-PET cohort, with an increase of brain Aβ deposition as individuals approach their parent's symptom onset.
- The interaction between spEYO and sex was also replicated using both CSF $A\beta_{1\text{-}42}$ level and PIB-PET data.
- The spEYO**APOE* interaction was not replicated in the ACS cohort.
- Longitudinal ACS PIB-PET data further suggests that women tend to accumulate brain Aβ at a faster rate than men as they approach the age of their parent's onset.

WRAP cohort

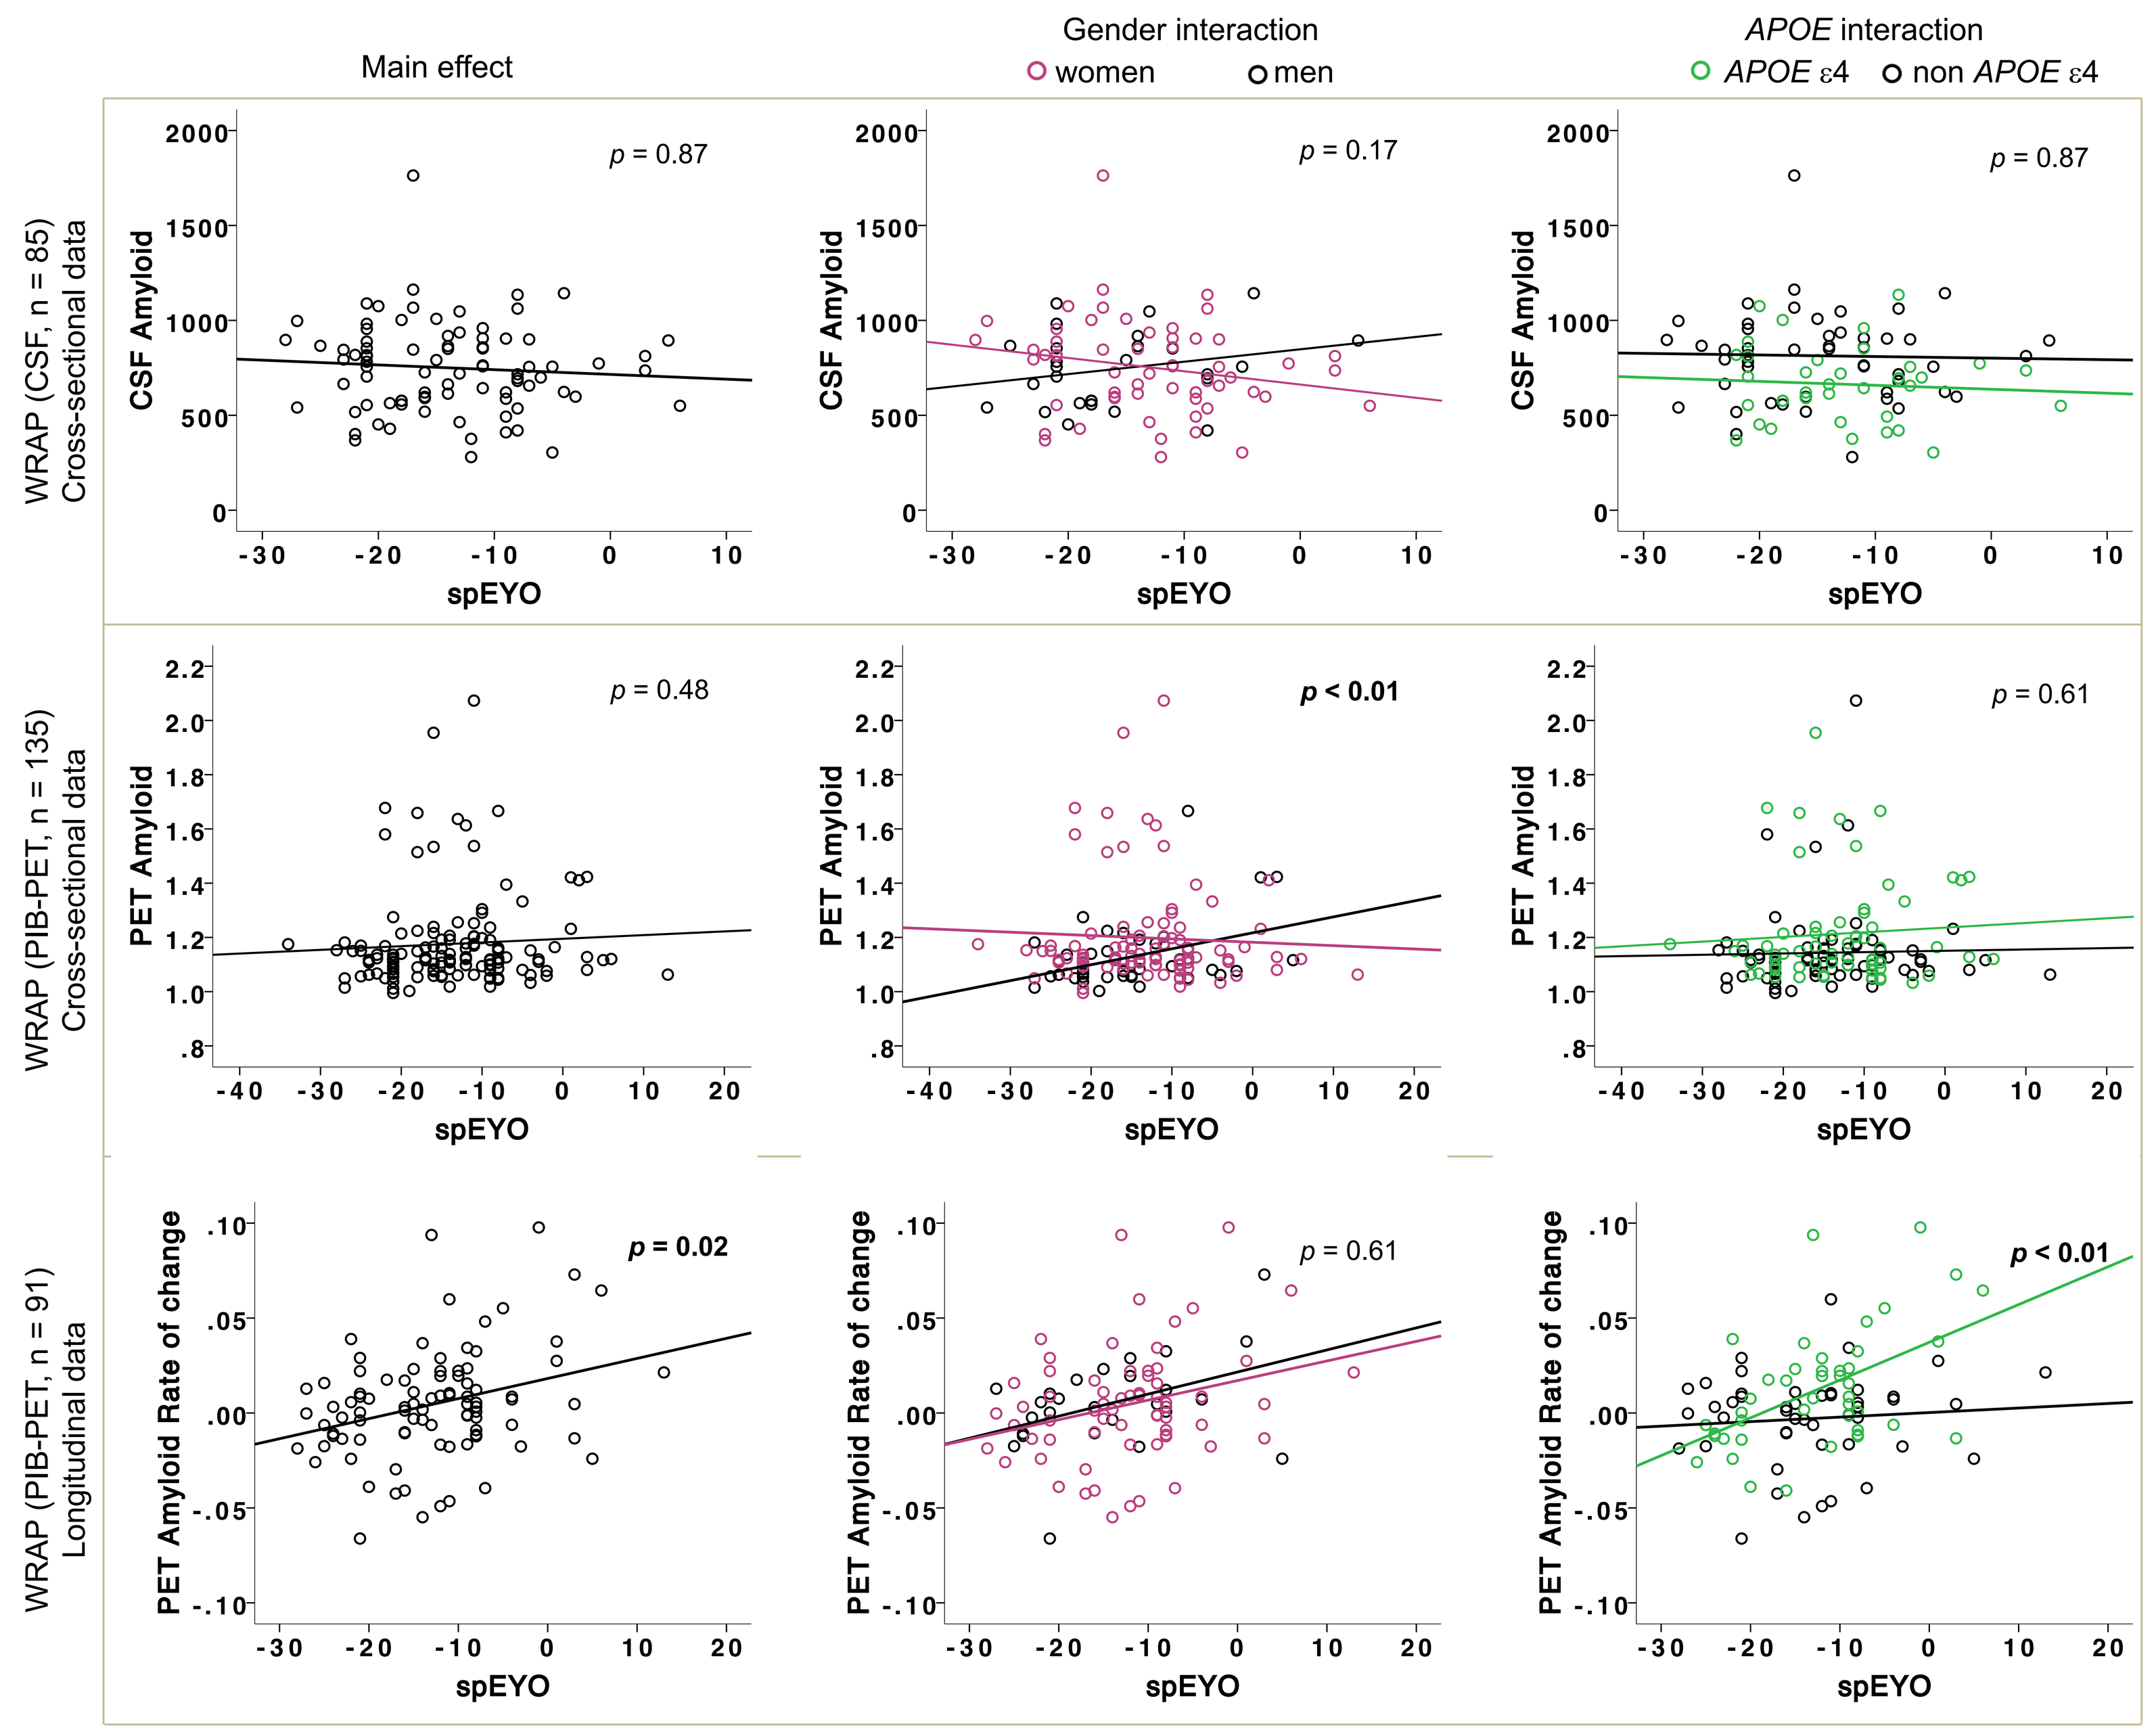

Main effect
Gender interaction
women
men
APOE interaction
APOE ε4
non APOE ε4
WRAP (CSF, n = 85) Cross-sectional data
WRAP (PIB-PET, n = 135) Cross-sectional data
WRAP (PIB-PET, n = 91) Longitudinal data
CSF Amyloid
PET Amyloid
PET Amyloid Rate of change
spEYO
p = 0.87
p = 0.17
p = 0.87
p = 0.48
p < 0.01
p = 0.61
p = 0.02
p = 0.61
p < 0.01

- We did not replicate the PREVENT-AD cross-sectional findings.
- As individuals approach the age of their parent's symptom onset they however demonstrate faster rate of brain Aβ accumulation.
- The spEYO**APOE* interaction further suggests that this last association is stronger in *APOE*4 carriers than in non-*APOE*4 carriers.

Summary and Conclusions

- In the PREVENT-AD cohort, CSF $A\beta_{1\text{-}42}$ values changed with proximity to parental symptom onset, such that individuals approaching their parent's symptom onset age showed lower Aβ levels. Interaction analyses showed that this effect is stronger in both *APOE*4 carriers and women.
- In the ACS cohort, the main effect was replicated using PIB-PET data and the gender interaction was replicated using both CSF and PIB-PET data.
- In the WRAP cohort, the results were not replicated using cross-sectional data, but the main effect and the *APOE* interaction were replicated using PIB-PET longitudinal data, suggesting that *APOE*4 carriers accumulate brain Aβ at a faster rate as they approach the age of their parent's symptom onset.
- These results suggest that proximity to parental symptom onset can help estimate Aβ biomarker changes in asymptomatic individuals at risk of AD, particularly in women and *APOE*4 carriers.

References:
[1] Bateman RJ et al., NEJM 2012; [2] Breitner JC et al., JPAD 2016; [3] Coats M, Morris J Geriatr Psychiatry Neurol 2005; [4] Sager MA, et al., J Geriatr Psychiatry Neurol 2005.

CIHR IRSC Canadian Institutes of Health Research / Instituts de recherche en santé du Canada; NIH National Institute on Aging; Société Alzheimer; alzheimer's association; INNOVATION.CA Canada Foundation for Innovation / Fondation canadienne pour l'innovation