Neurophysiological activity linked to Alzheimer's Disease pathology relates to longitudinal cognition and progression to Mild Cognitive Impairment.

Jonathan Gallego Rudolf [1,2], Alex I. Wiesman [2], Sylvain Baillet [2], Sylvia Villeneuve [1], the PREVENT-AD Research Group

[1] Douglas Mental Health University Institute, Montreal, QC, Canada, [2] Montreal Neurological Institute, Montreal, QC, Canada. Contact information: jonathan.gallegorudolf@mail.mcgill.ca

1. Background

- The pathological processes underlying Alzheimer's Disease (AD) develop during a wide preclinical phase before the onset of cognitive symptoms, providing an ideal window for capturing early disease biomarkers [1].
- Identifying aberrant signaling markers of early neurophysiological dysfunction linked to amyloid-beta (Aβ) and tau pathologies could be an alternative for predicting progression from the asymptomatic to the Mild Cognitive Impairment (MCI) phase of the disease [2].

2. Study aims

- 1. To examine whether **neurophysiological (MEG) changes** related to the deposition of **Aβ and tau pathologies (PET)** are associated with participants **demographics** and **longitudinal cognitive performance**.
- 2. To assess the added value of **neurophysiological activity features** for predicting progression from the asymptomatic to the Mild Cognitive Impairment stage of AD.

Logistic regression models for predicting MCI progression

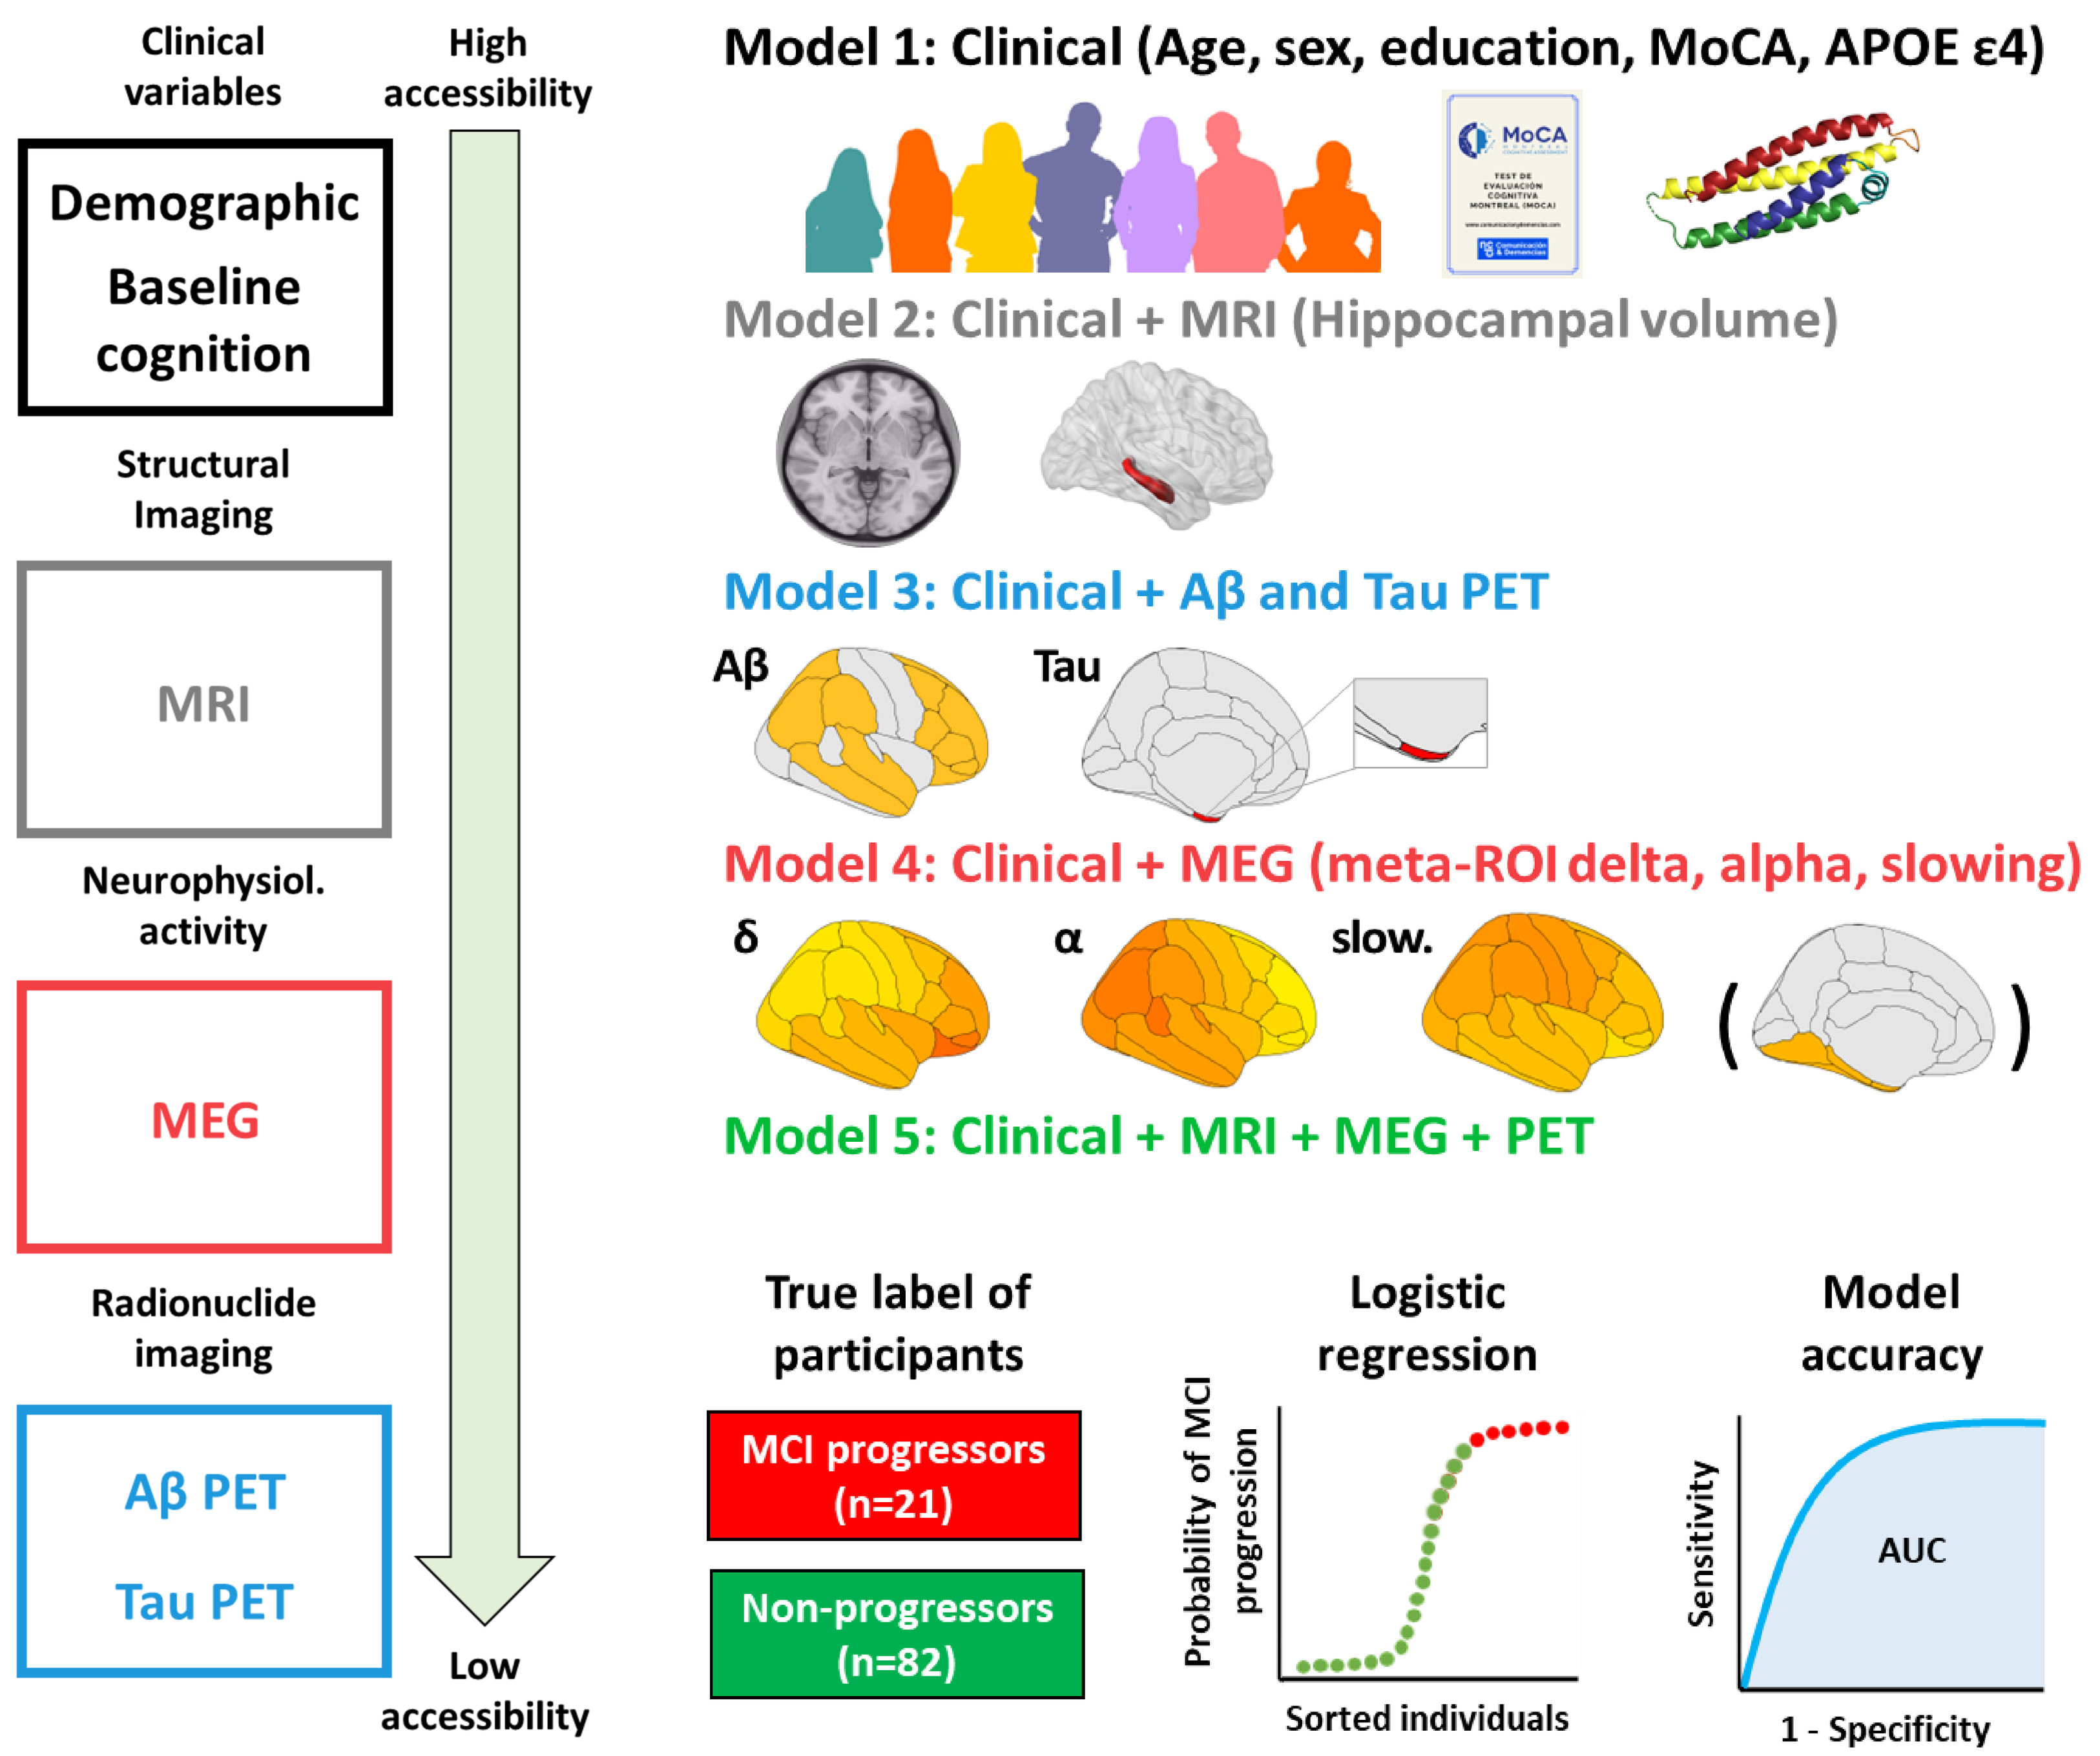

Clinical variables
High accessibility
Demographic
Baseline cognition
Structural Imaging
MRI
Neurophysiol. activity
MEG
Radionuclide imaging
Aβ PET
Tau PET
Low accessibility
Model 1: Clinical (Age, sex, education, MoCA, APOE ε4)
Model 2: Clinical + MRI (Hippocampal volume)
Model 3: Clinical + Aβ and Tau PET
Aβ
Tau
Model 4: Clinical + MEG (meta-ROI delta, alpha, slowing)
δ
α
slow.
Model 5: Clinical + MRI + MEG + PET
True label of participants
MCI progressors (n=21)
Non-progressors (n=82)
Logistic regression
Probability of MCI progression
Sorted individuals
Model accuracy
Sensitivity
AUC
1 - Specificity

3. Methods

PREVENT-AD cohort

103 participants (all cognitively unimpaired at enrolment)

20.4% MCI (mean time for progression 2.9 years)

Neurophysiological spectral features

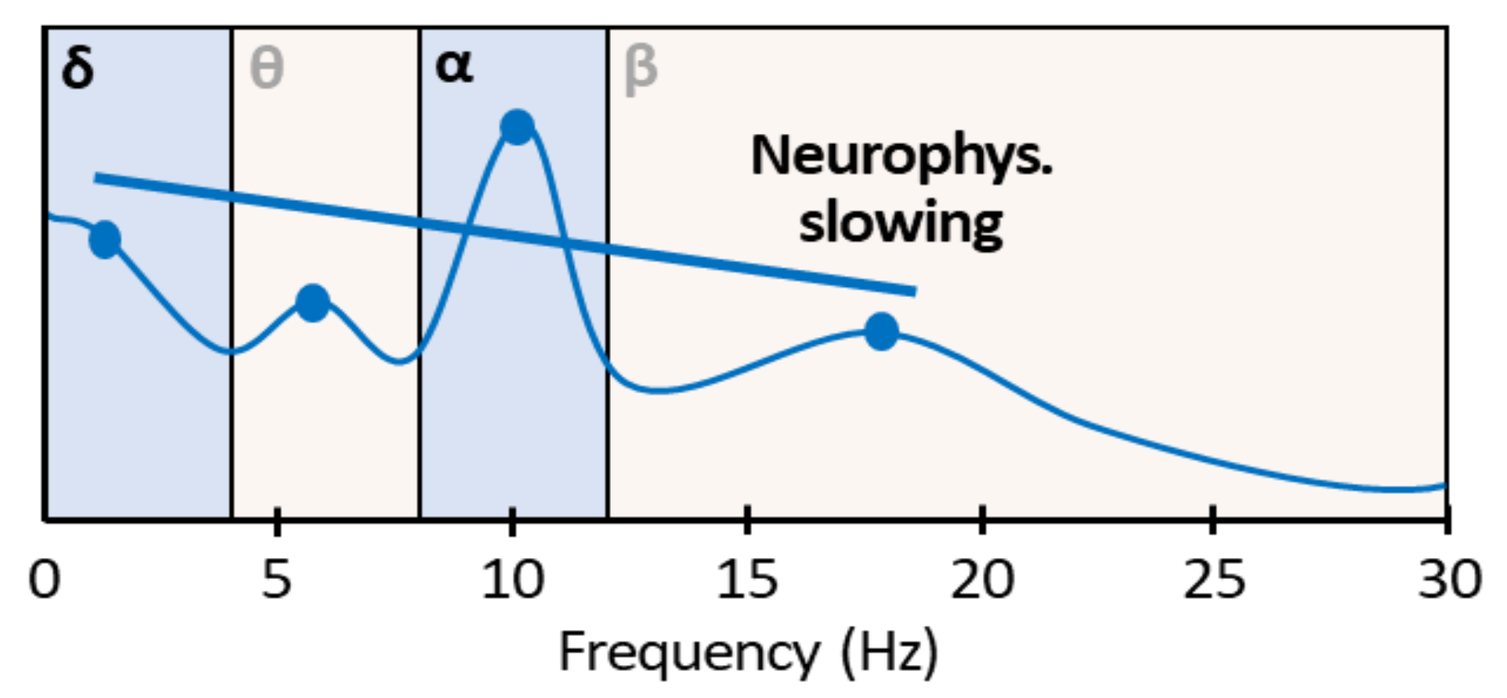

δ
θ
α
β
Neurophys. slowing
0
5
10
15
20
25
30
Frequency (Hz)

PLS analysis to assess association between neurophysiology, Aβ, tau and behavior

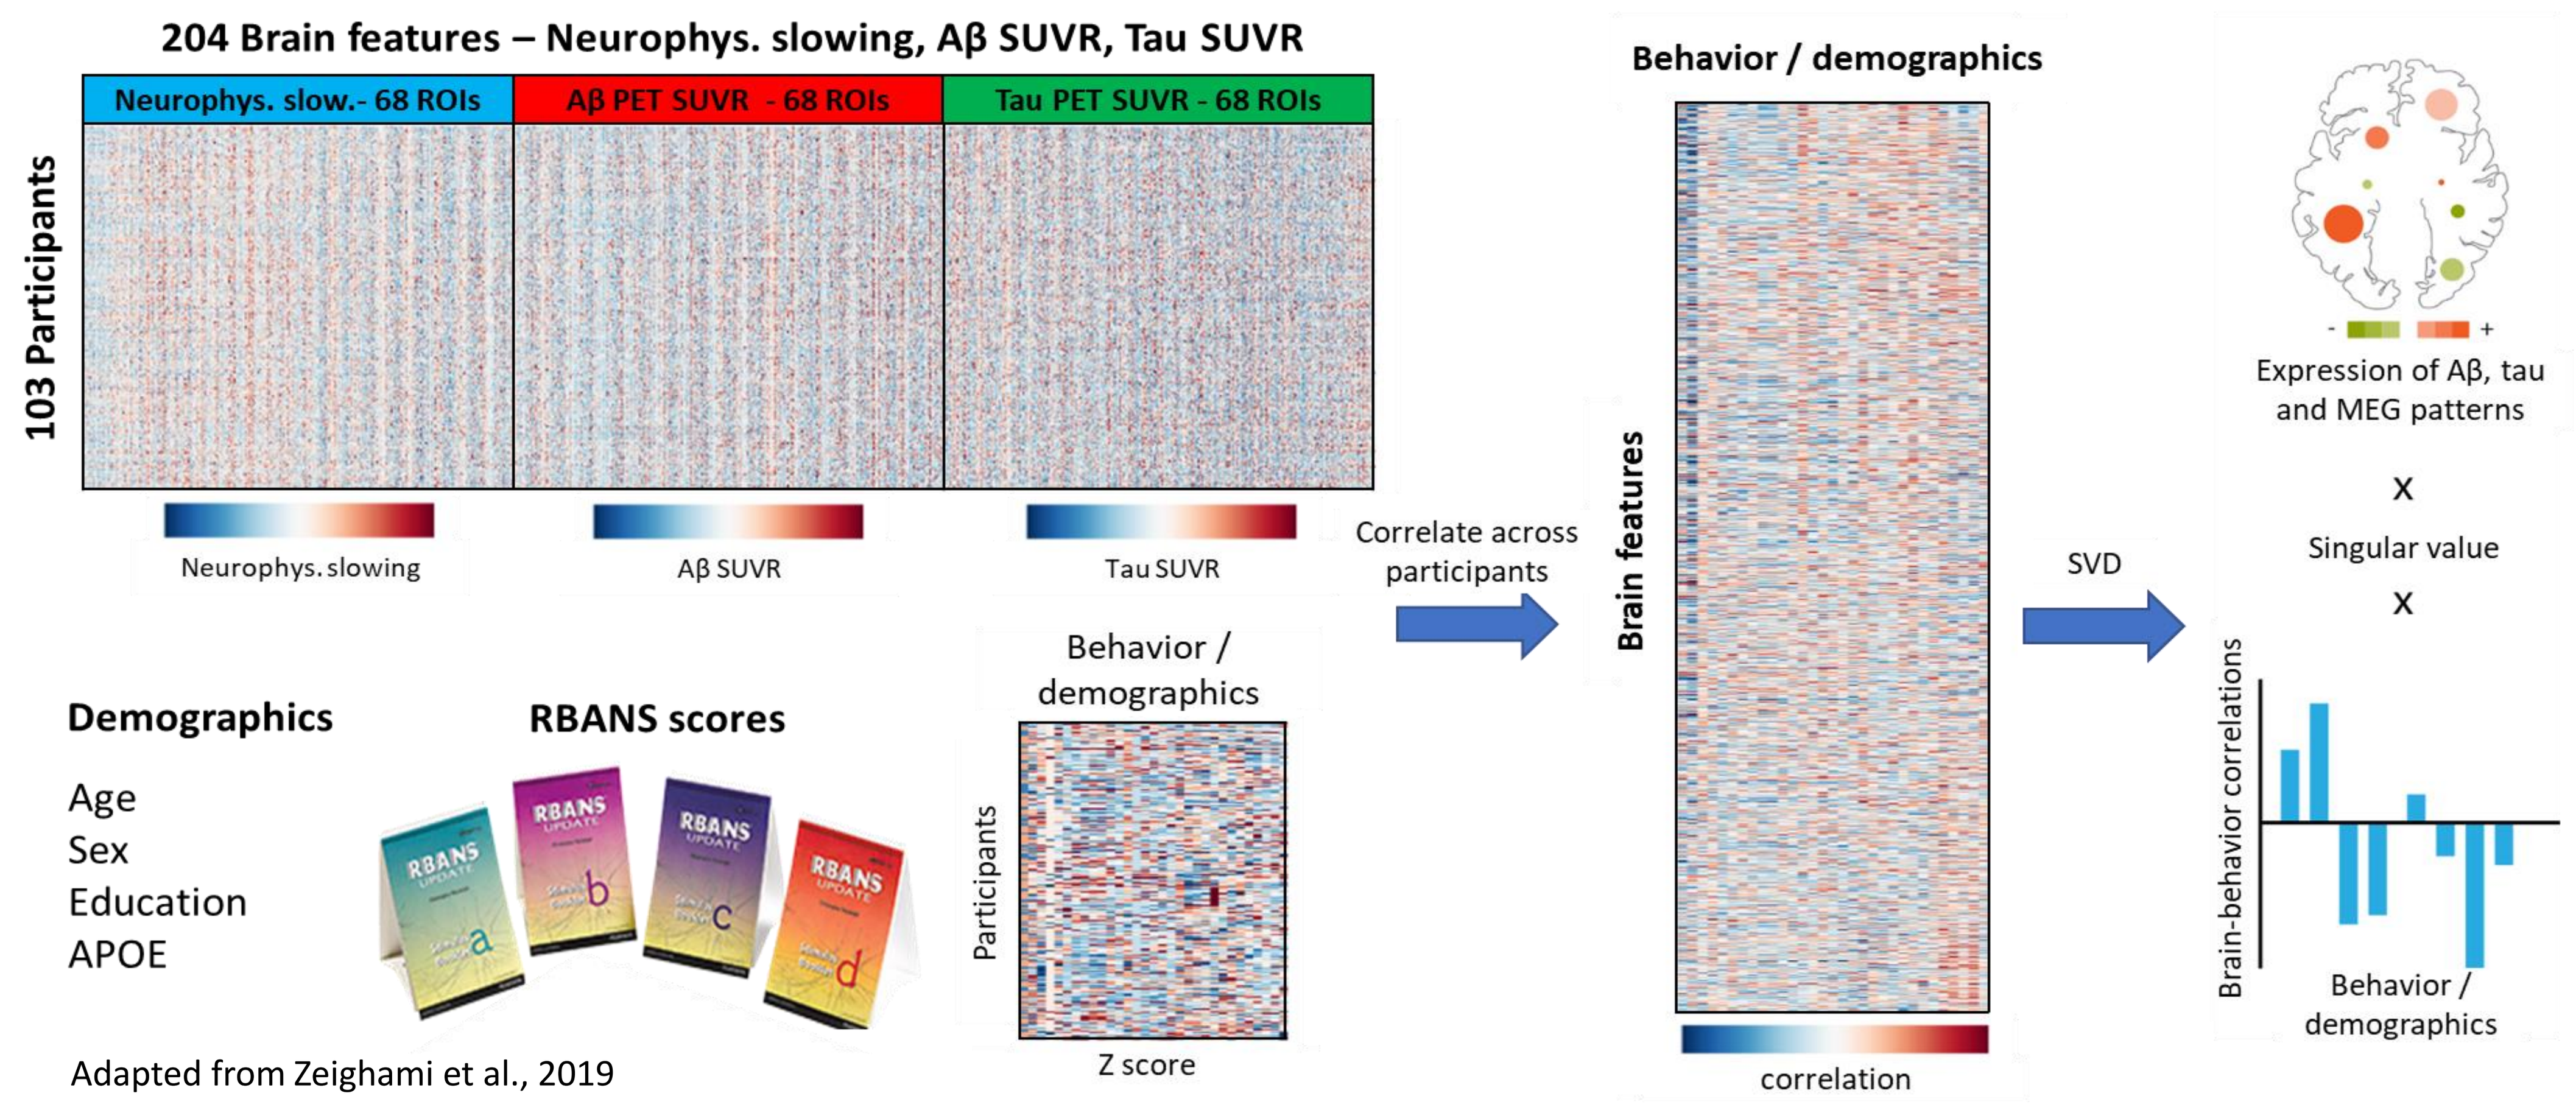

204 Brain features – Neurophys. slowing, Aβ SUVR, Tau SUVR
Neurophys. slow.- 68 ROIs
Aβ PET SUVR - 68 ROIs
Tau PET SUVR - 68 ROIs
103 Participants
Neurophys. slowing
Aβ SUVR
Tau SUVR
Demographics
Age
Sex
Education
APOE
RBANS scores
Behavior / demographics
Participants
Z score
Adapted from Zeighami et al., 2019
Correlate across participants
Behavior / demographics
Brain features
correlation
SVD
Expression of Aβ, tau and MEG patterns
x
Singular value
x
Brain-behavior correlations
Behavior / demographics

4. Results

Neurophysiological slowing in frontal regions, higher global Aβ and higher temporal tau are associated with longitudinal cognitive decline and positive APOE ε4 status

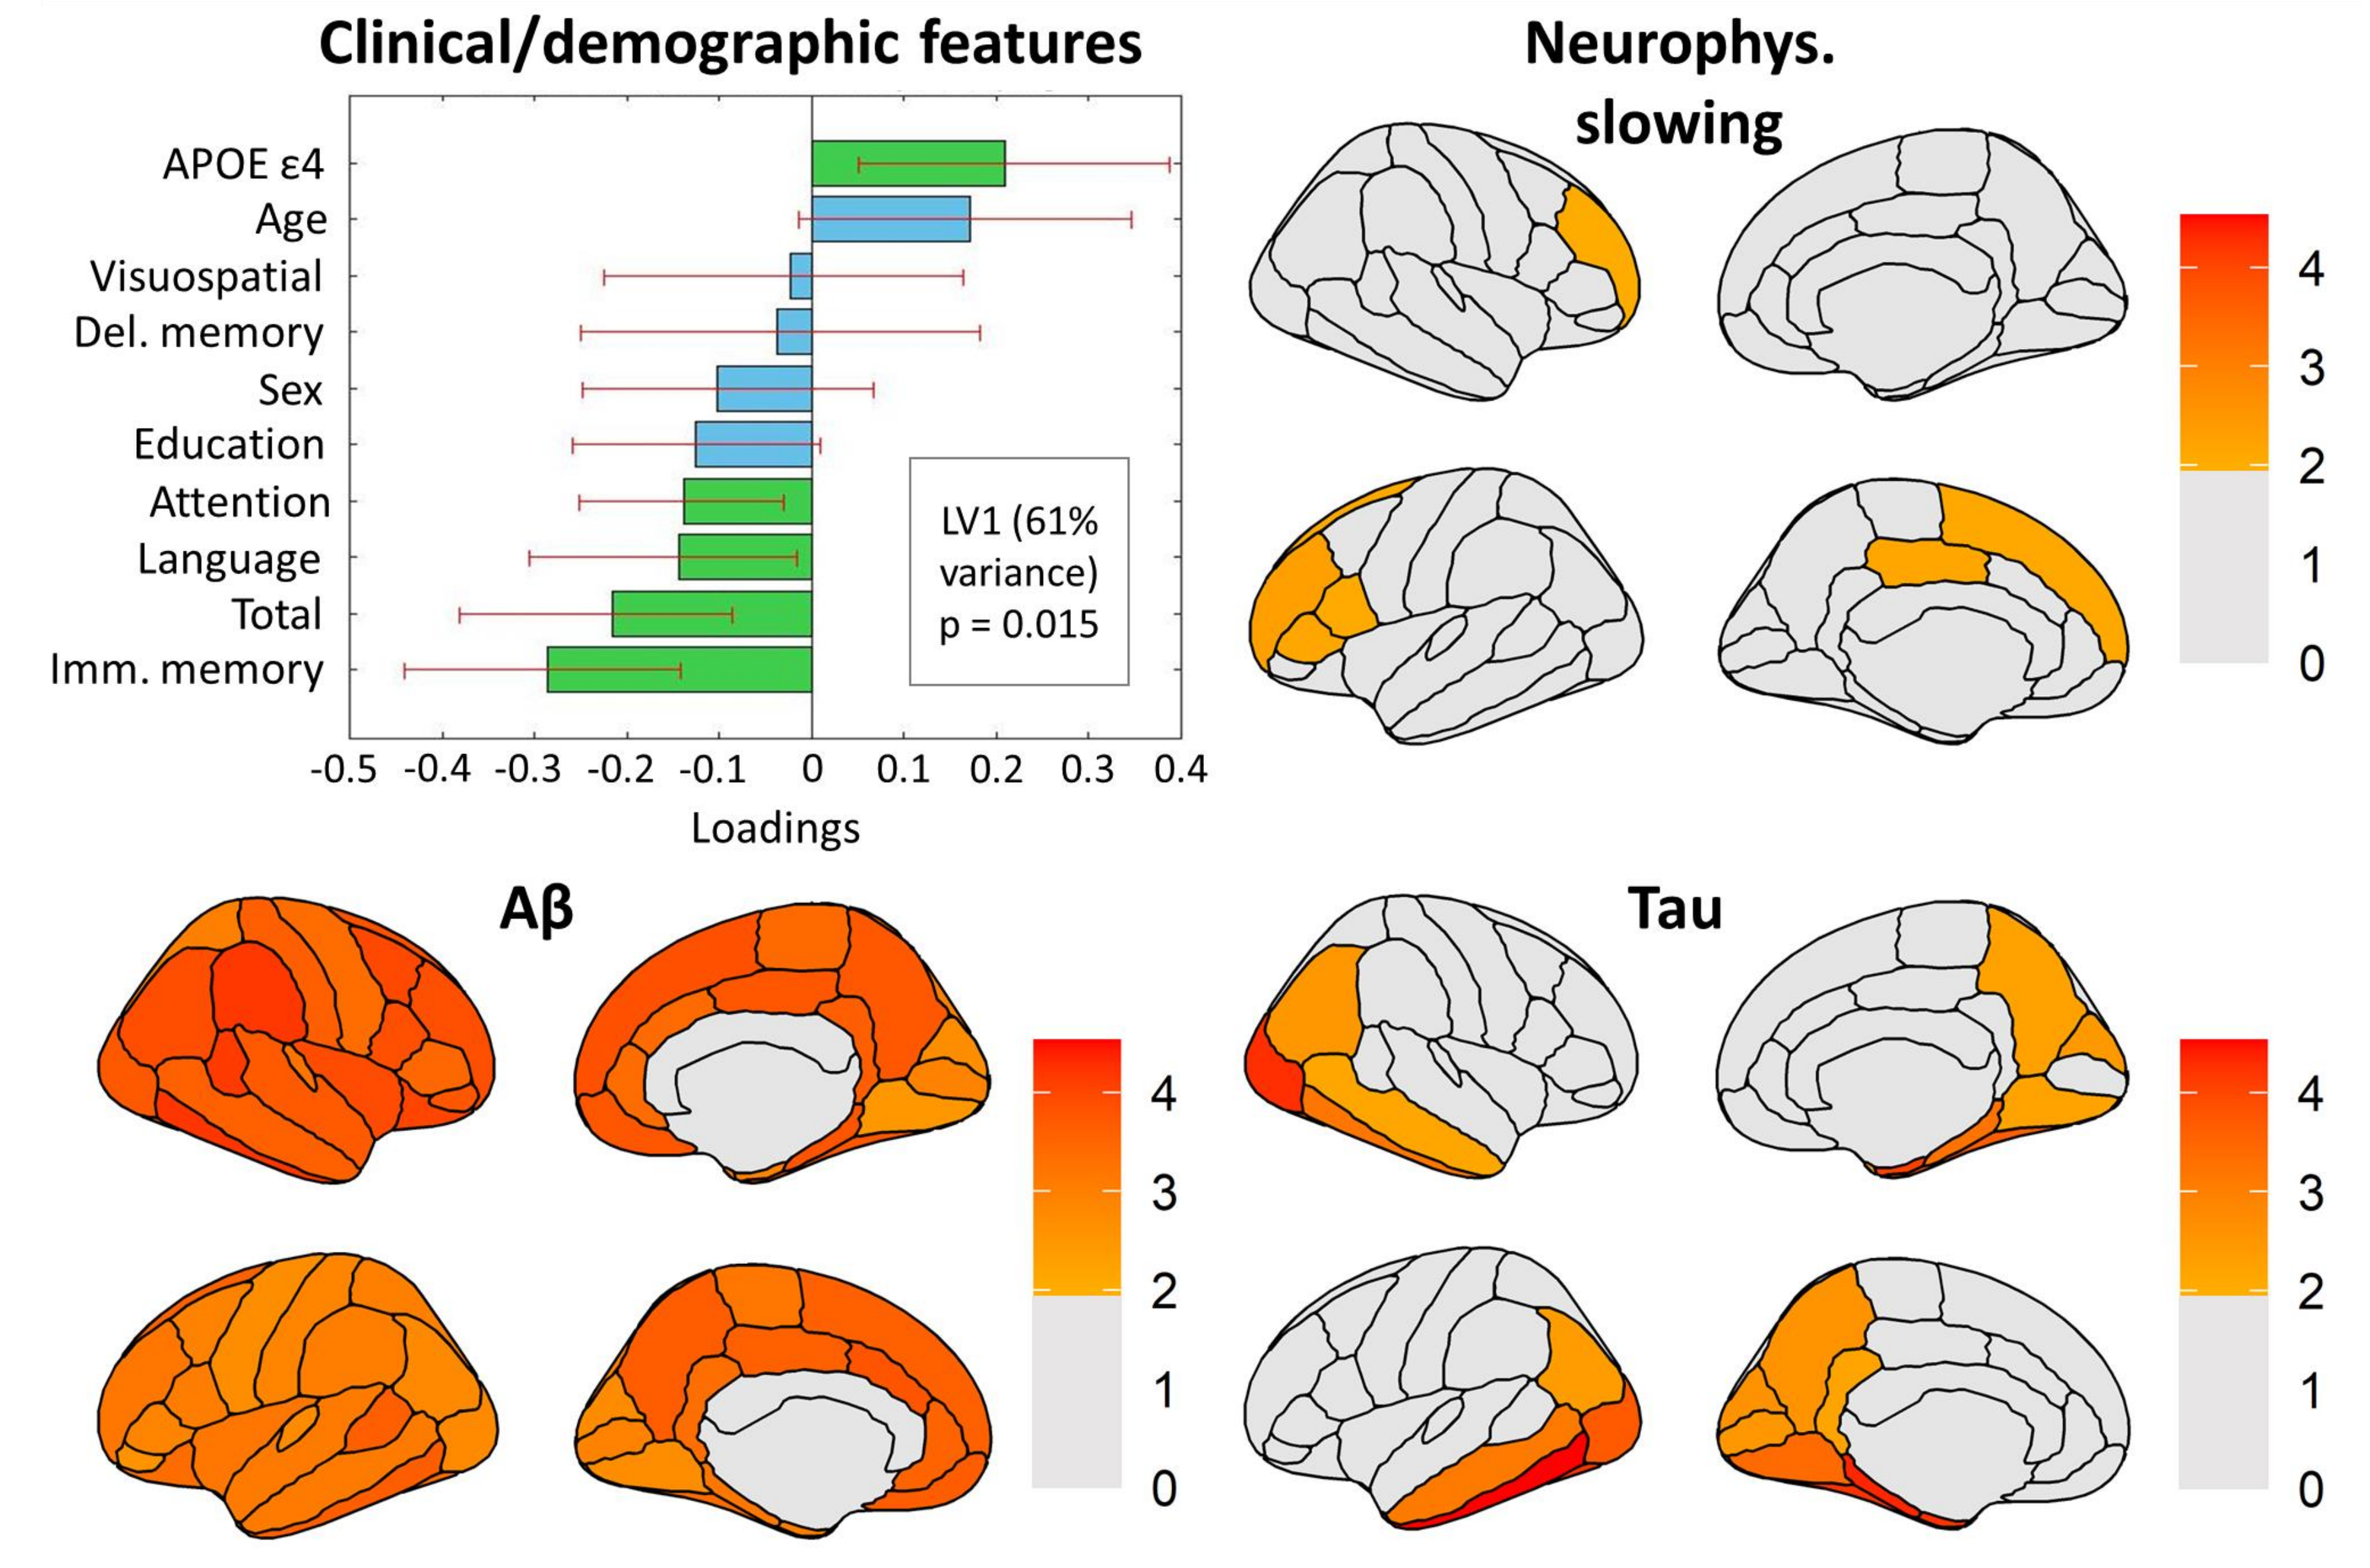

Clinical/demographic features
APOE ε4
Age
Visuospatial
Del. memory
Sex
Education
Attention
Language
Total
Imm. memory
LV1 (61% variance)
p = 0.015
-0.5 -0.4 -0.3 -0.2 -0.1 0 0.1 0.2 0.3 0.4
Loadings
Neurophys. slowing
Aβ
Tau
4
3
2
1
0

Neurophysiological activity in tau accumulating regions outperforms clinical and MRI models, matching the accuracy of Aβ and tau PET for predicting MCI progression

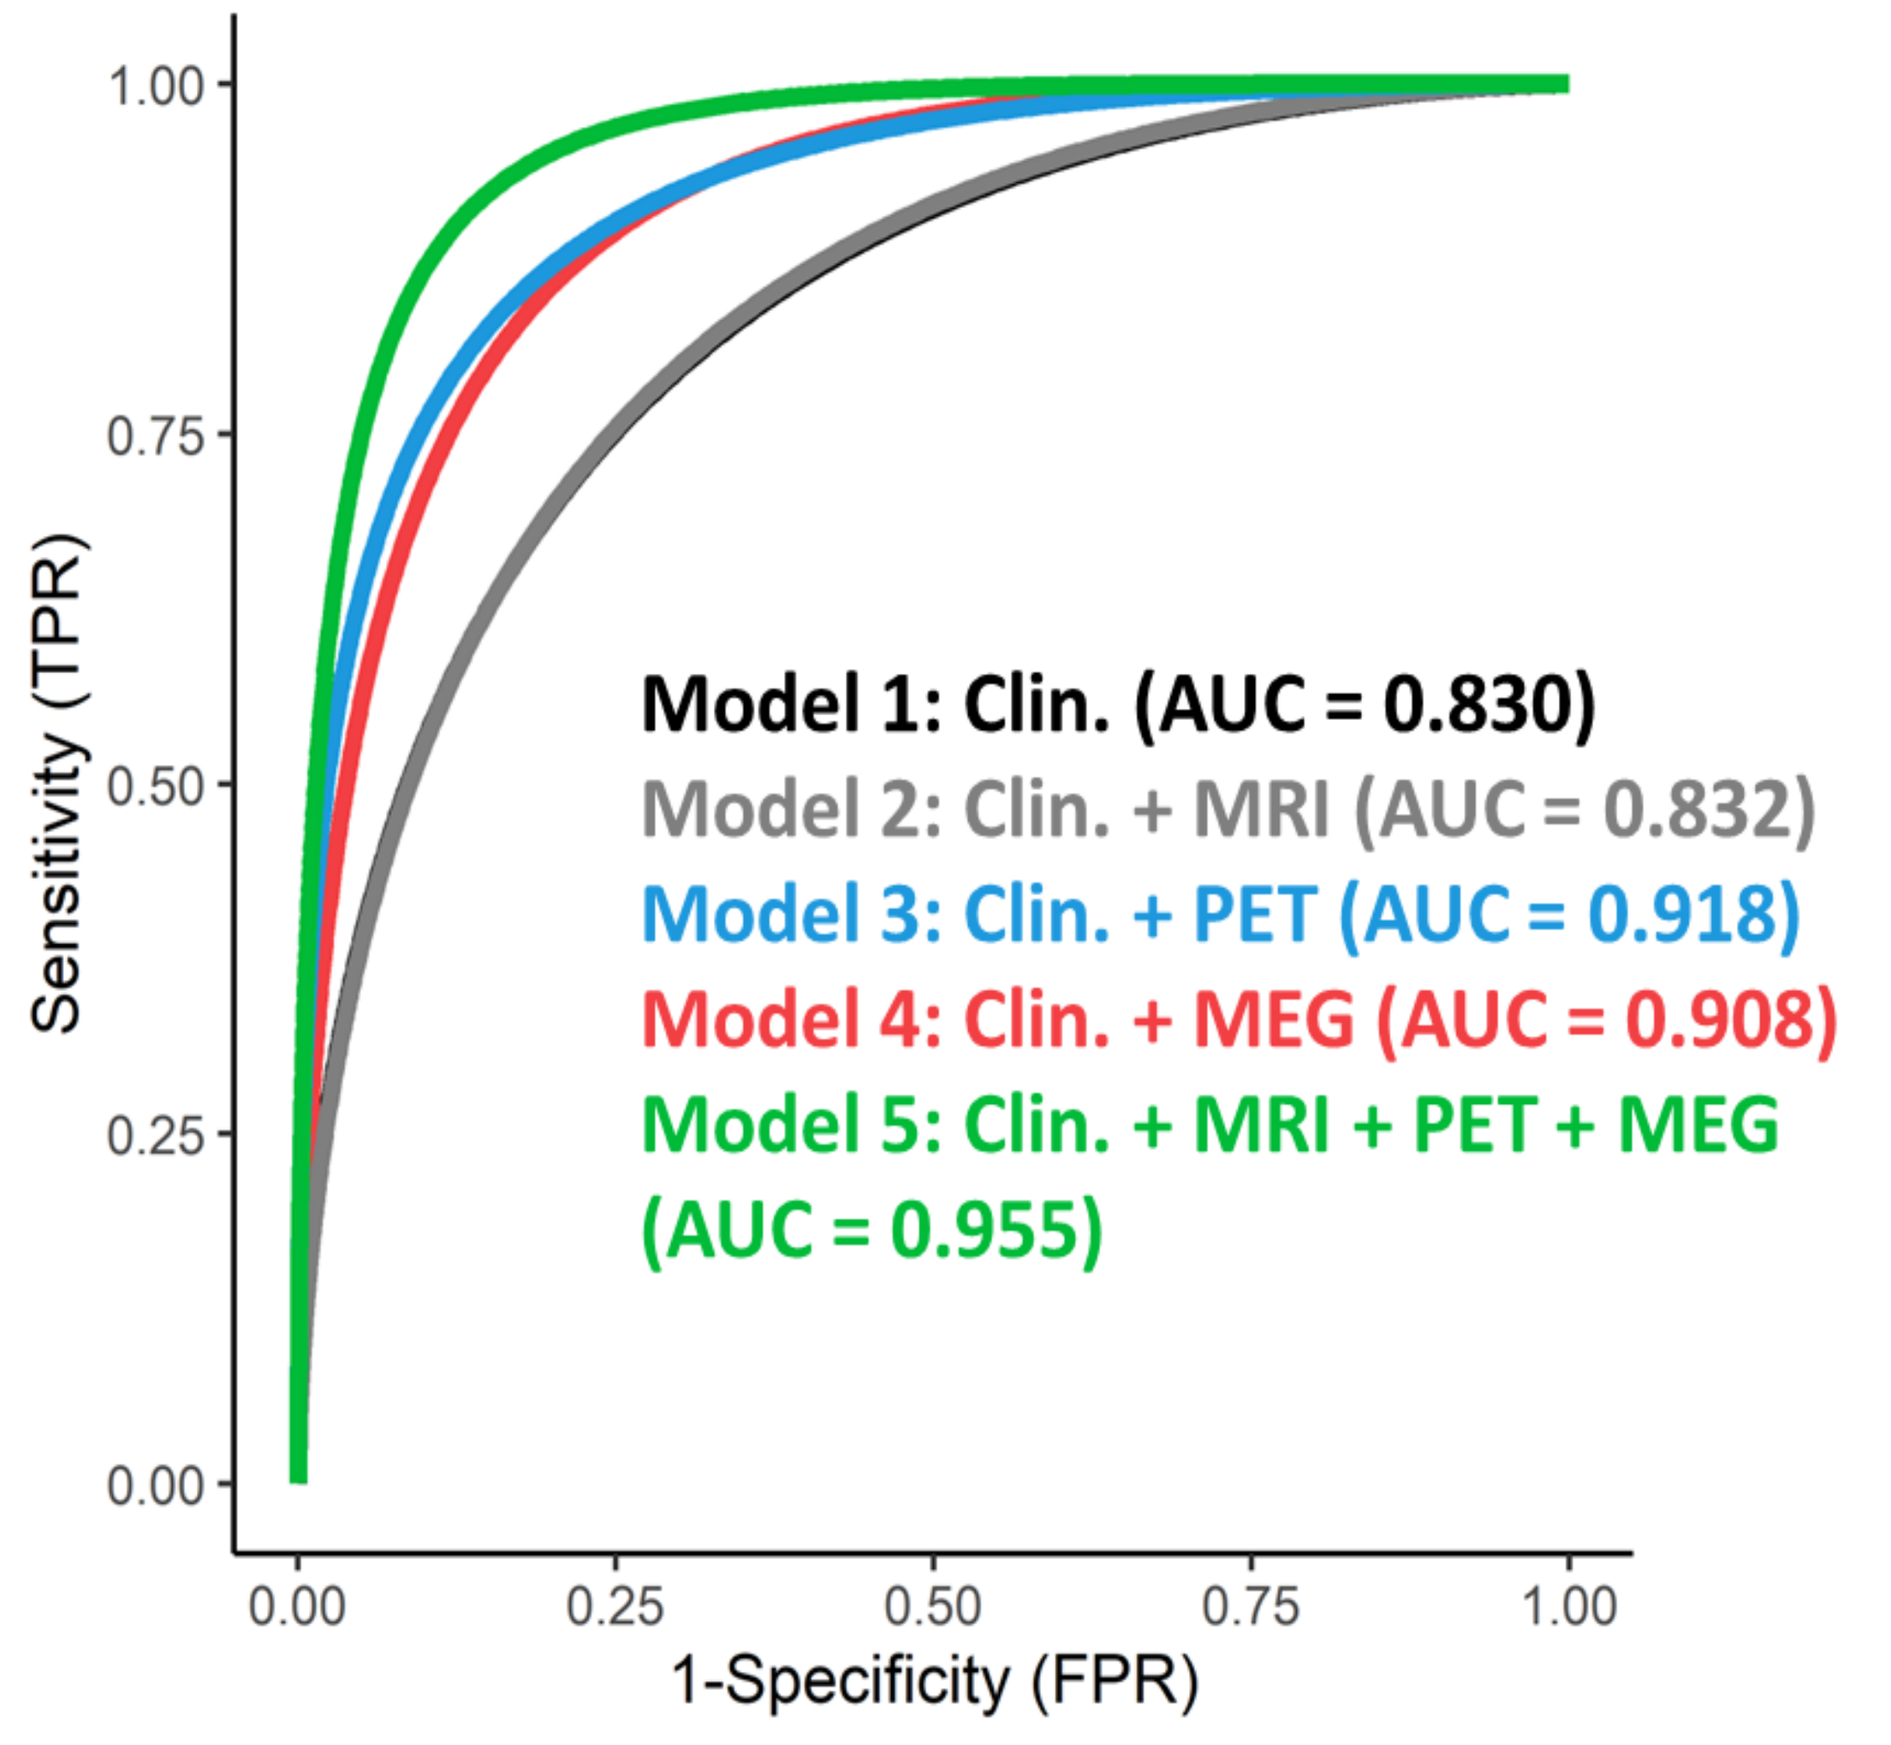

Sensitivity (TPR)
1-Specificity (FPR)
Model 1: Clin. (AUC = 0.830)
Model 2: Clin. + MRI (AUC = 0.832)
Model 3: Clin. + PET (AUC = 0.918)
Model 4: Clin. + MEG (AUC = 0.908)
Model 5: Clin. + MRI + PET + MEG (AUC = 0.955)

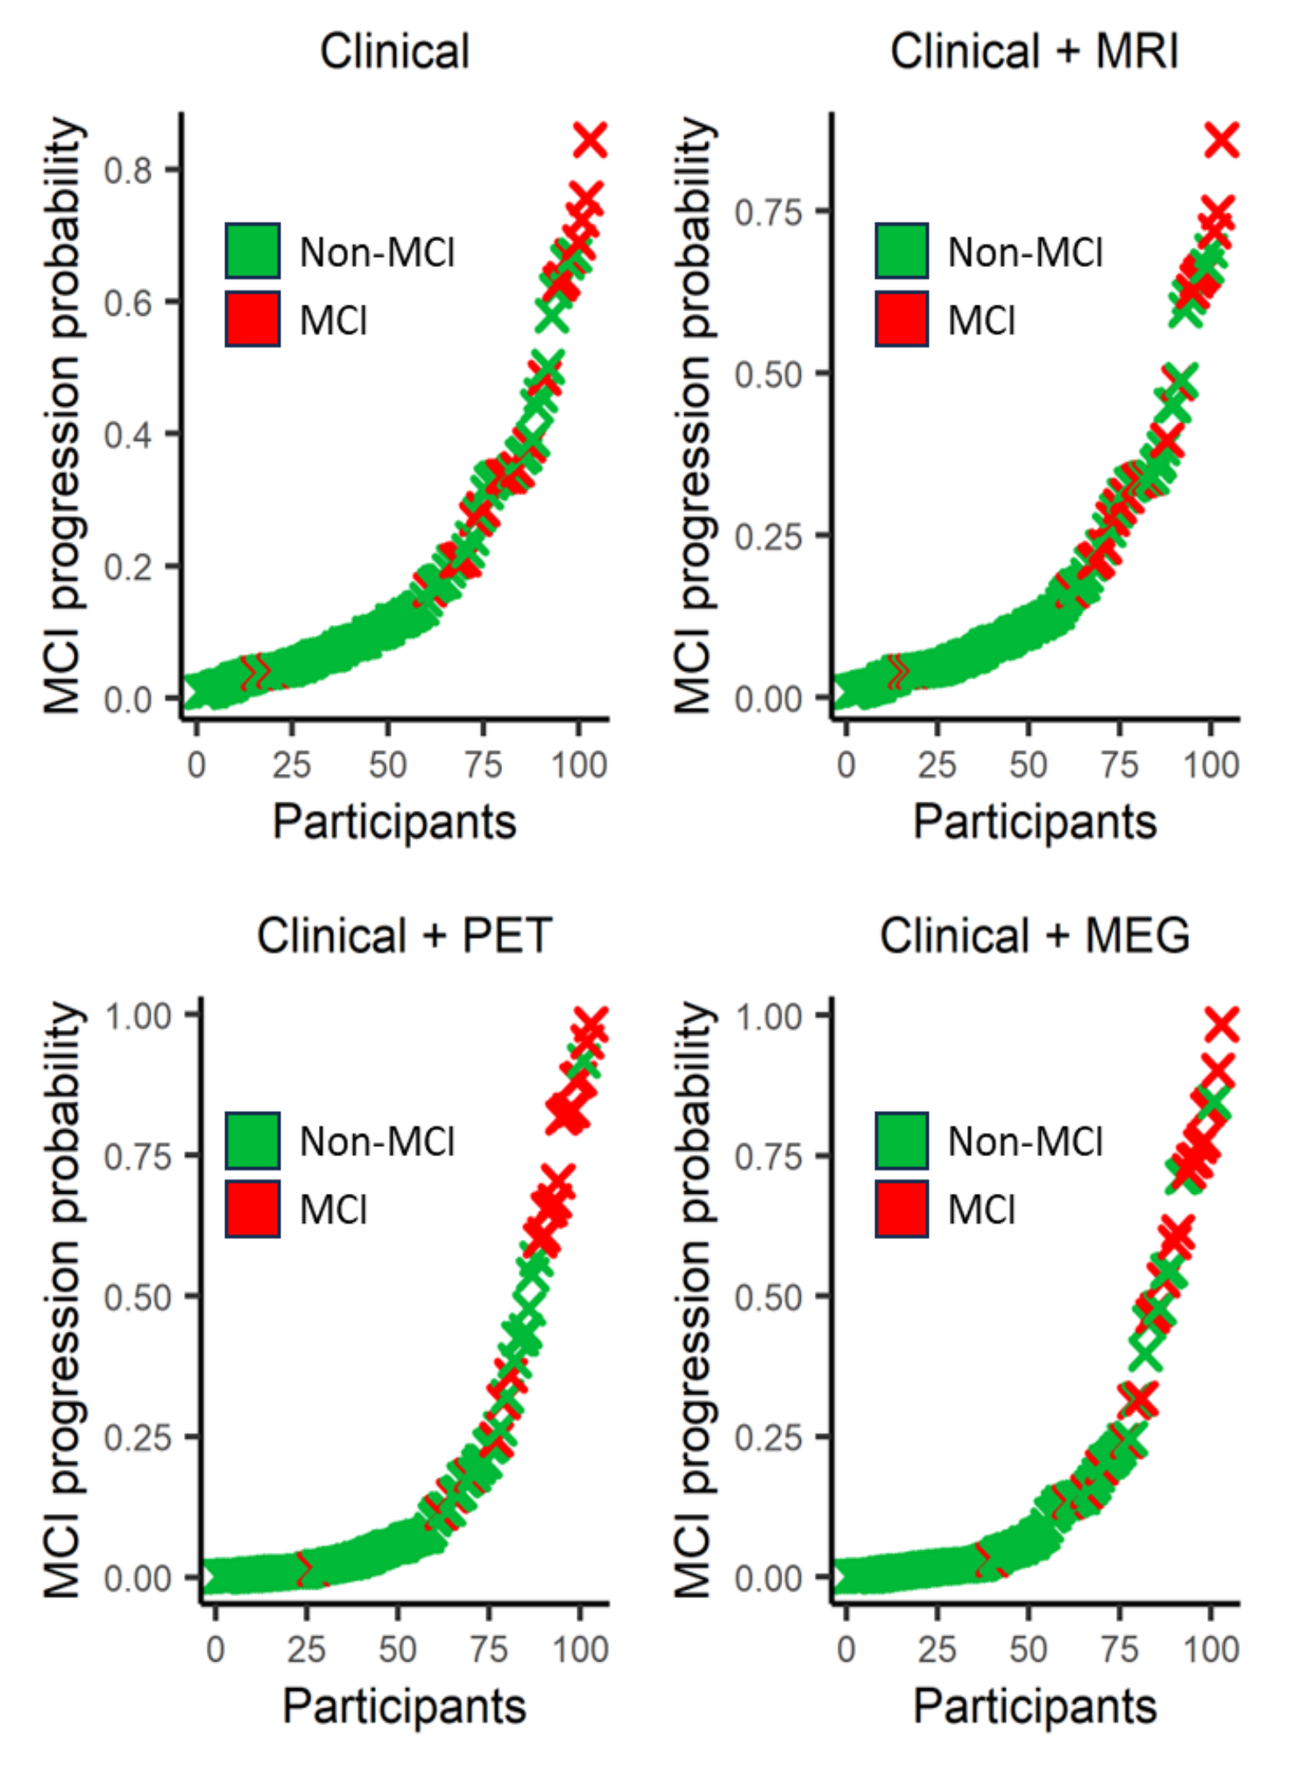

Clinical
Clinical + MRI
Clinical + PET
Clinical + MEG
Non-MCI
MCI
MCI progression probability
Participants

Stepwise logistic regression feature selection corroborates the added value of neurophysiological activity for predicting participant's cognitive status

	Full model	Reduced model
Variables included	All (Clinical + MRI + PET + MEG)	Tau, delta, slowing, MoCA, APOE, education
Area Under Curve	0.955	0.948
Akaike Information Criterion	69.54	61.14
Likelihood ratio test	χ^2 = 1.68 ; p = 0.89 ; Full model = Reduced model	

5. Conclusions

- Neurophysiological activity slowing, higher Aβ and tau burden are associated with positive APOE e4 status and cognitive decline across multiple domains.
- Early detectable neurophysiological changes may represent an alternative screening tool to select participants for specialized invasive testing for AD proteinopathy.

6. Acknowledgements

PREVENT-AD Research Group · Alzheimer's Association · CIHR IRSC · Villeneuve Lab (Multimodal Imaging of the Aging Brain) · Fonds de recherche Santé Québec · CONACYT (Consejo Nacional de Ciencia y Tecnología) · neuro speed · QBIN Quebec Bio-Imaging Network · Canada First Research Excellence Fund · Healthy Brains Healthy Lives

References

1. Dubois, B. et al., (2016). Preclinical Alzheimer's disease: Definition, natural history, and diagnostic criteria. *Alzheimer's and Dementia*, *12*(3), 292–323. https://doi.org/10.1016/j.jalz.2016.02.002.

2. López-Sanz, D., Serrano, N., & Maestú, F. (2018). The role of magnetoencephalography in the early stages of Alzheimer's disease. *Frontiers in Neuroscience*, *12*, 1–8. https://doi.org/10.3389/fnins.2018.00572.