Sleep Quality and Day-to-Day Variability: Relation to Alzheimer's pathology in Cognitively Unimpaired Older Adults At-Risk of AD Dementia

Bery Mohammediyan[1,2], BA; Andrée-Ann Baril[1,2], PhD; Frédéric St-Onge[1,2], MSc; Valentin Ourry, PhD[1,2]; Julie Carrier[3], PhD; Jean-Paul Soucy[2], MD; John Breitner[1,2], PhD; Judes Poirier[1,2], PhD; Sylvia Villeneuve [1,2], PhD; for the PREVENT-AD Research Group

[1]Douglas Mental Health University Institute, Montreal, Quebec, Canada, [2]Department of Psychiatry, Faculty of Medicine, McGill University, Montreal, Quebec, Canada, [3]Department of Psychology, University of Montreal, Montreal, Quebec, Canada

1. Introduction

Background: Given that the association between sleep and AD has been hypothesized to be bidirectional, longitudinal studies are needed to deepen the understanding of sleep and AD in the preclinical phases of the disease.

Objectives: We investigated whether 1) poorer sleep is accompanied by faster AD pathology accumulation and 2) early AD pathologic changes are accompanied by sleep changes before the onset of cognitive symptoms.

2. Methods

Participants are from the PREVENT-AD cohort, a longitudinal cohort of initially cognitively unimpaired older adults with a self-reported parental history of AD, that were tested with:

- 2 PET scans (FU: 4.31y+-0.45y, n=98)
- Pittsburgh Sleep Quality Index (PSQI) 2-3 times (FU:1.02y+-0.17y, n=183)
- Actiwatch for one week 2-3 times (FU:2.46y+-1.25y, n=109)

Demographics

	PSQI longitudinal (183)	Actigraphy longitudinal (109)	AD PET longitudinal (98)
Age (years)	68.23	68.92	67.35
Sex (%female)	72.47	73.62	75
Education (years)	15.46	15.28	15.31
APOE status (%positive)	38.02	36.22	35.35
AD pathology			
Amyloid baseline	1.29	1.28	1.26
Tau baseline	1.16	1.16	1.15
Amyloid follow-up	NA	NA	1.32
Tau follow-up	NA	NA	1.17

Actogram

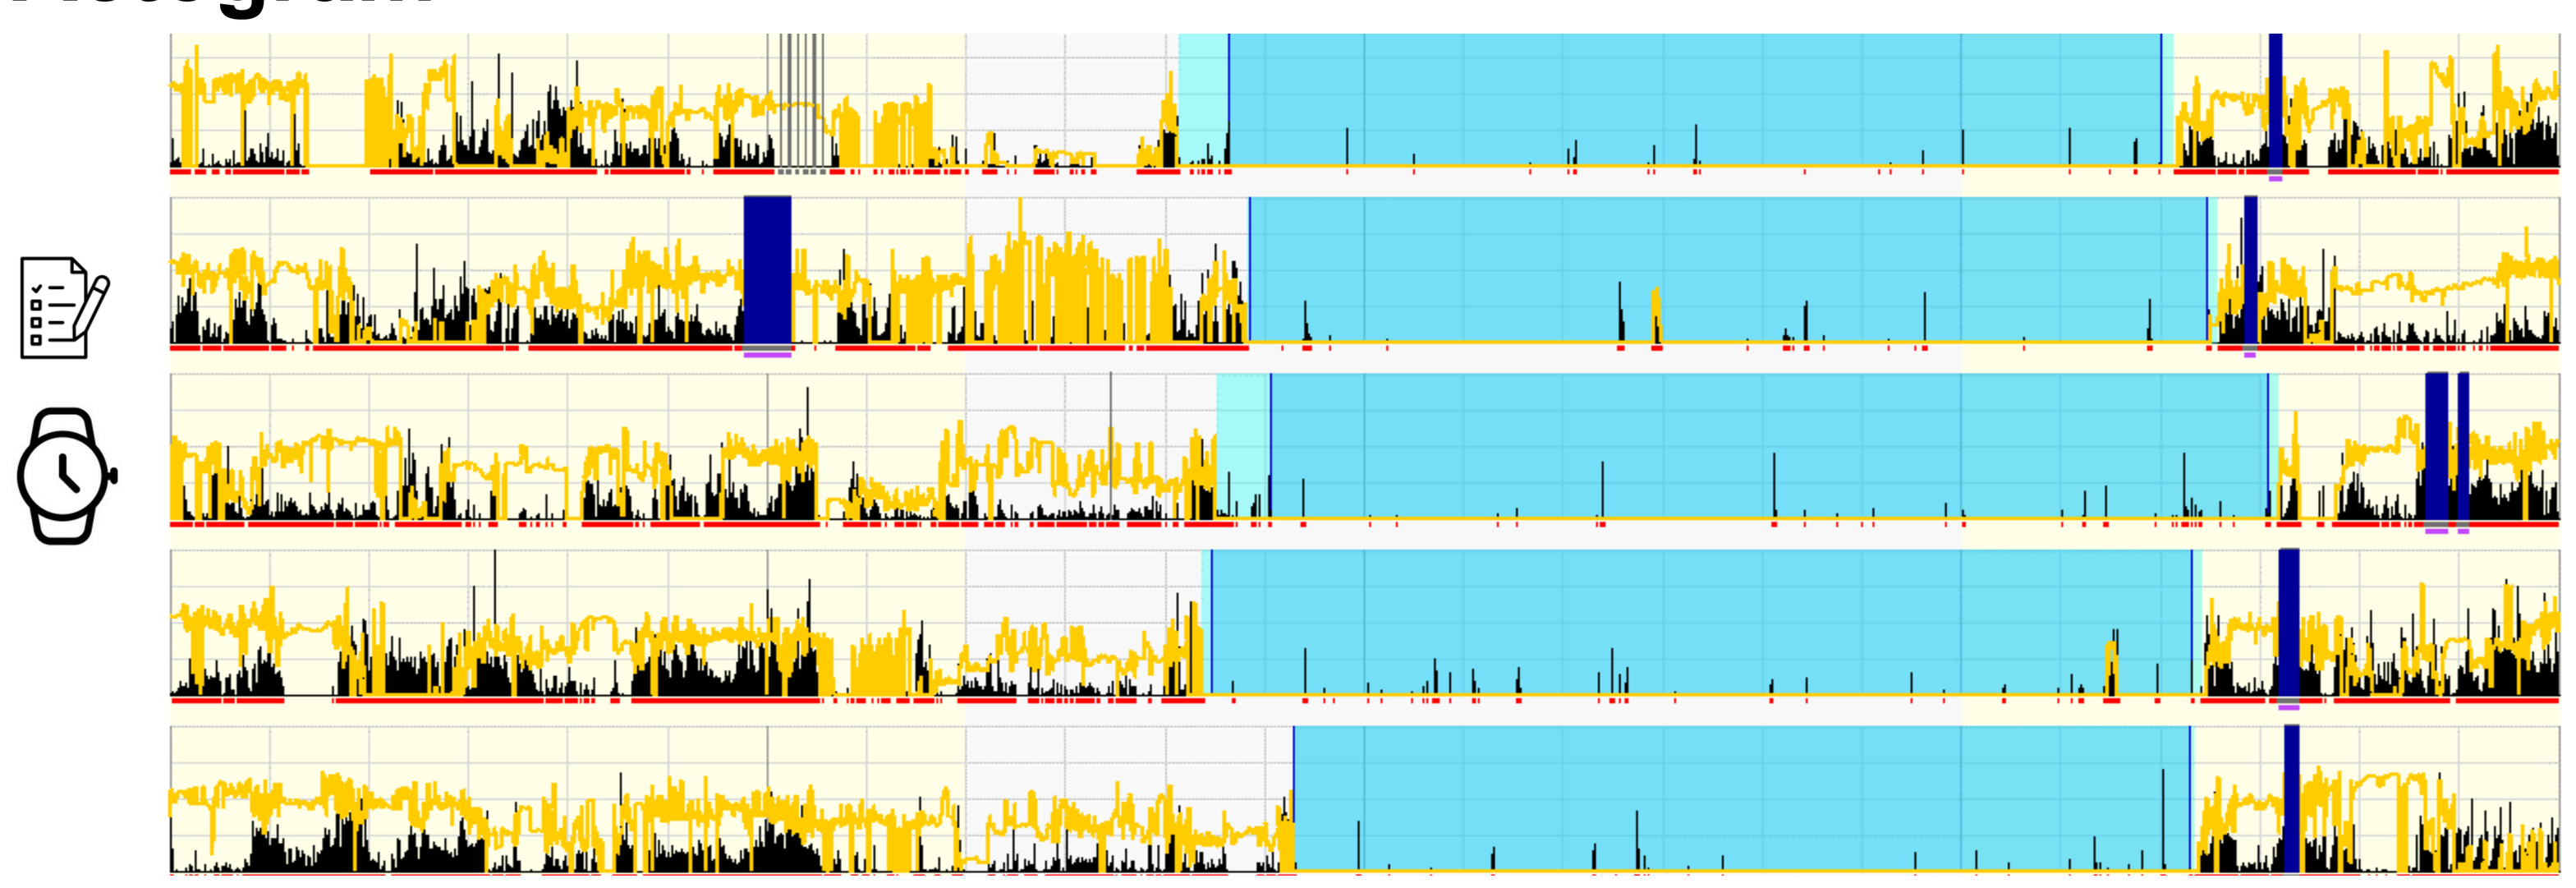

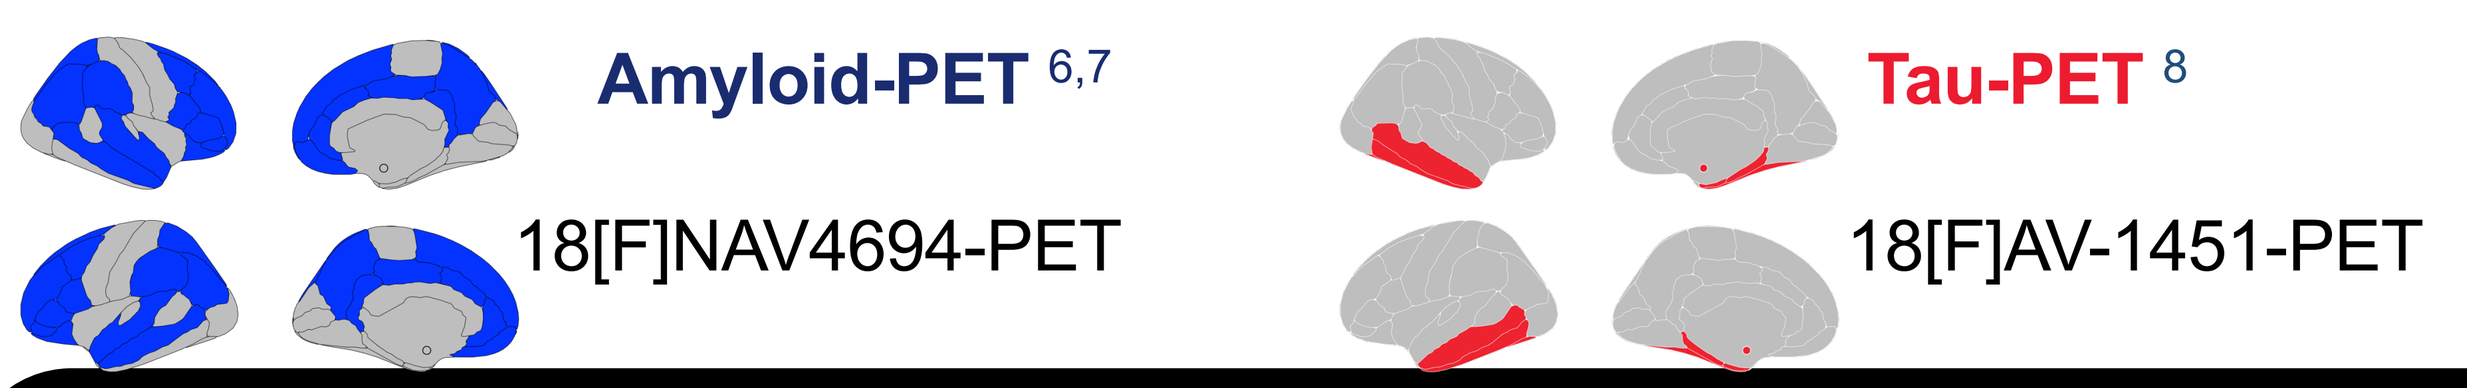
Amyloid-PET 6,7
18[F]NAV4694-PET
Tau-PET 8
18[F]AV-1451-PET

3. Results

1) Sleep -> AD pathology

Greater day-to-day sleep duration variability -> higher global amyloid across time

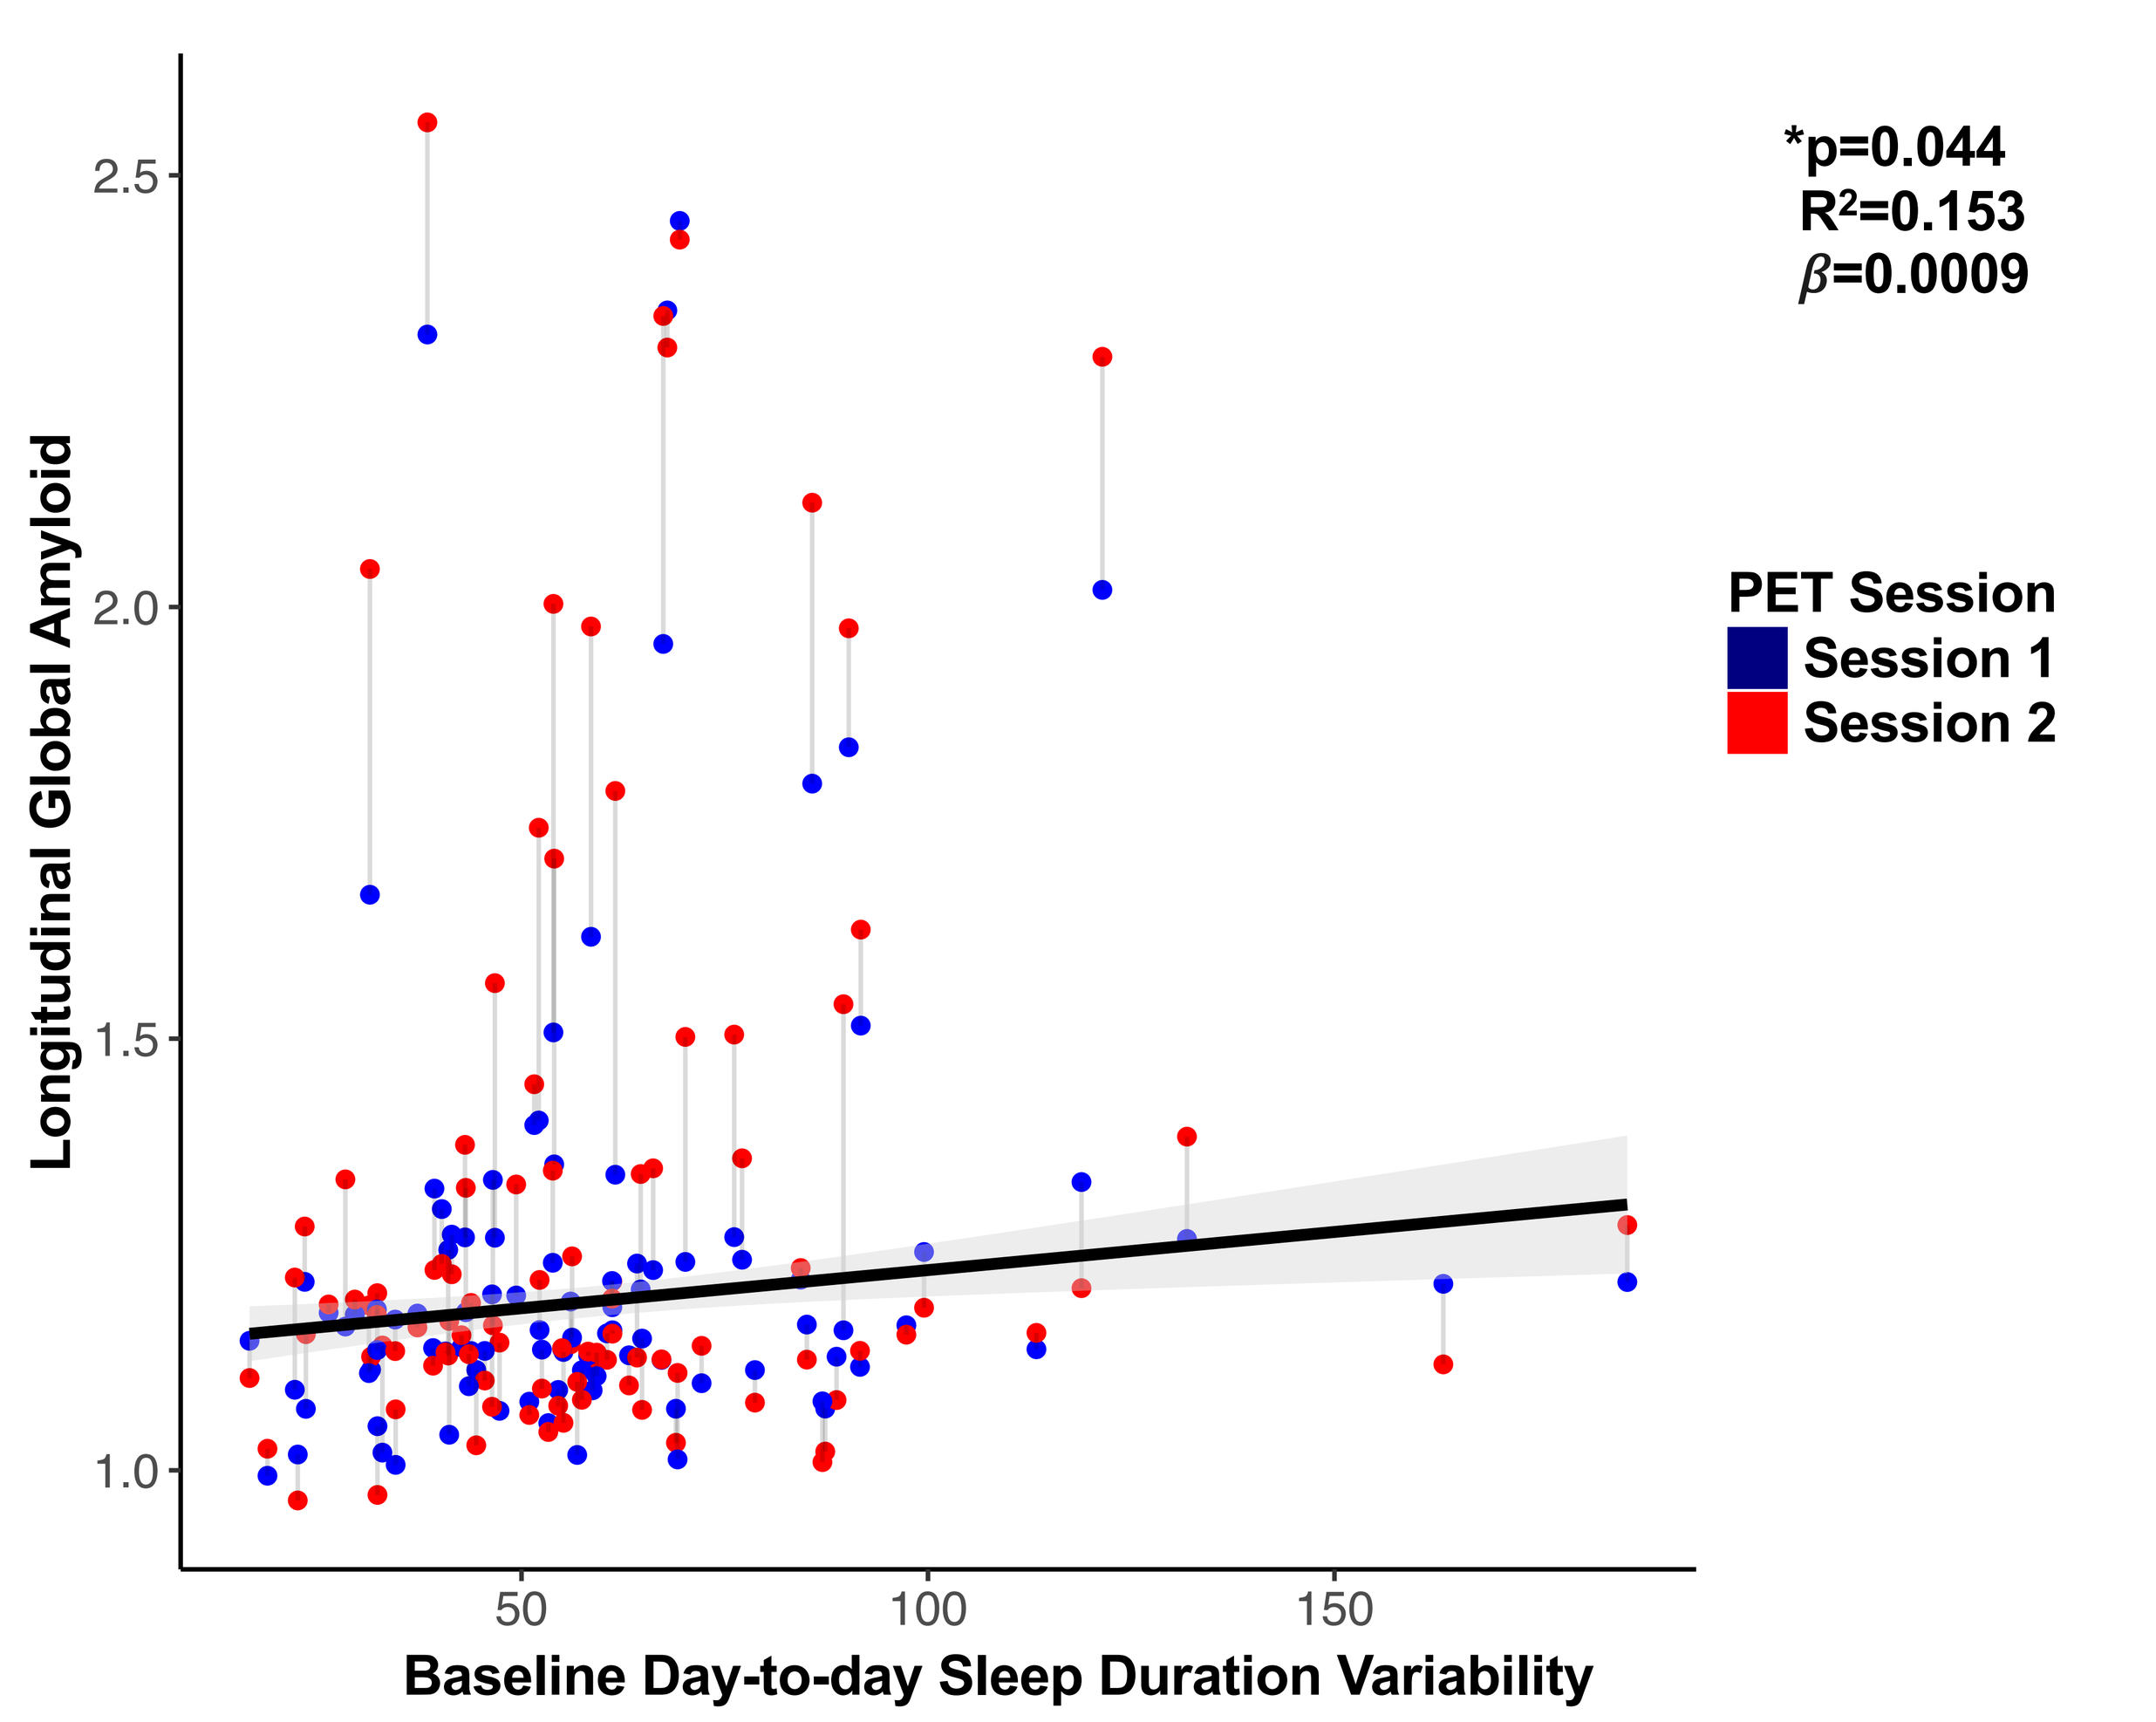
*p=0.044
R^2=0.153
β=0.0009
Longitudinal Global Amyloid
Baseline Day-to-day Sleep Duration Variability
PET Session
Session 1
Session 2

Greater day-to-day sleep efficiency variability -> higher global amyloid across time

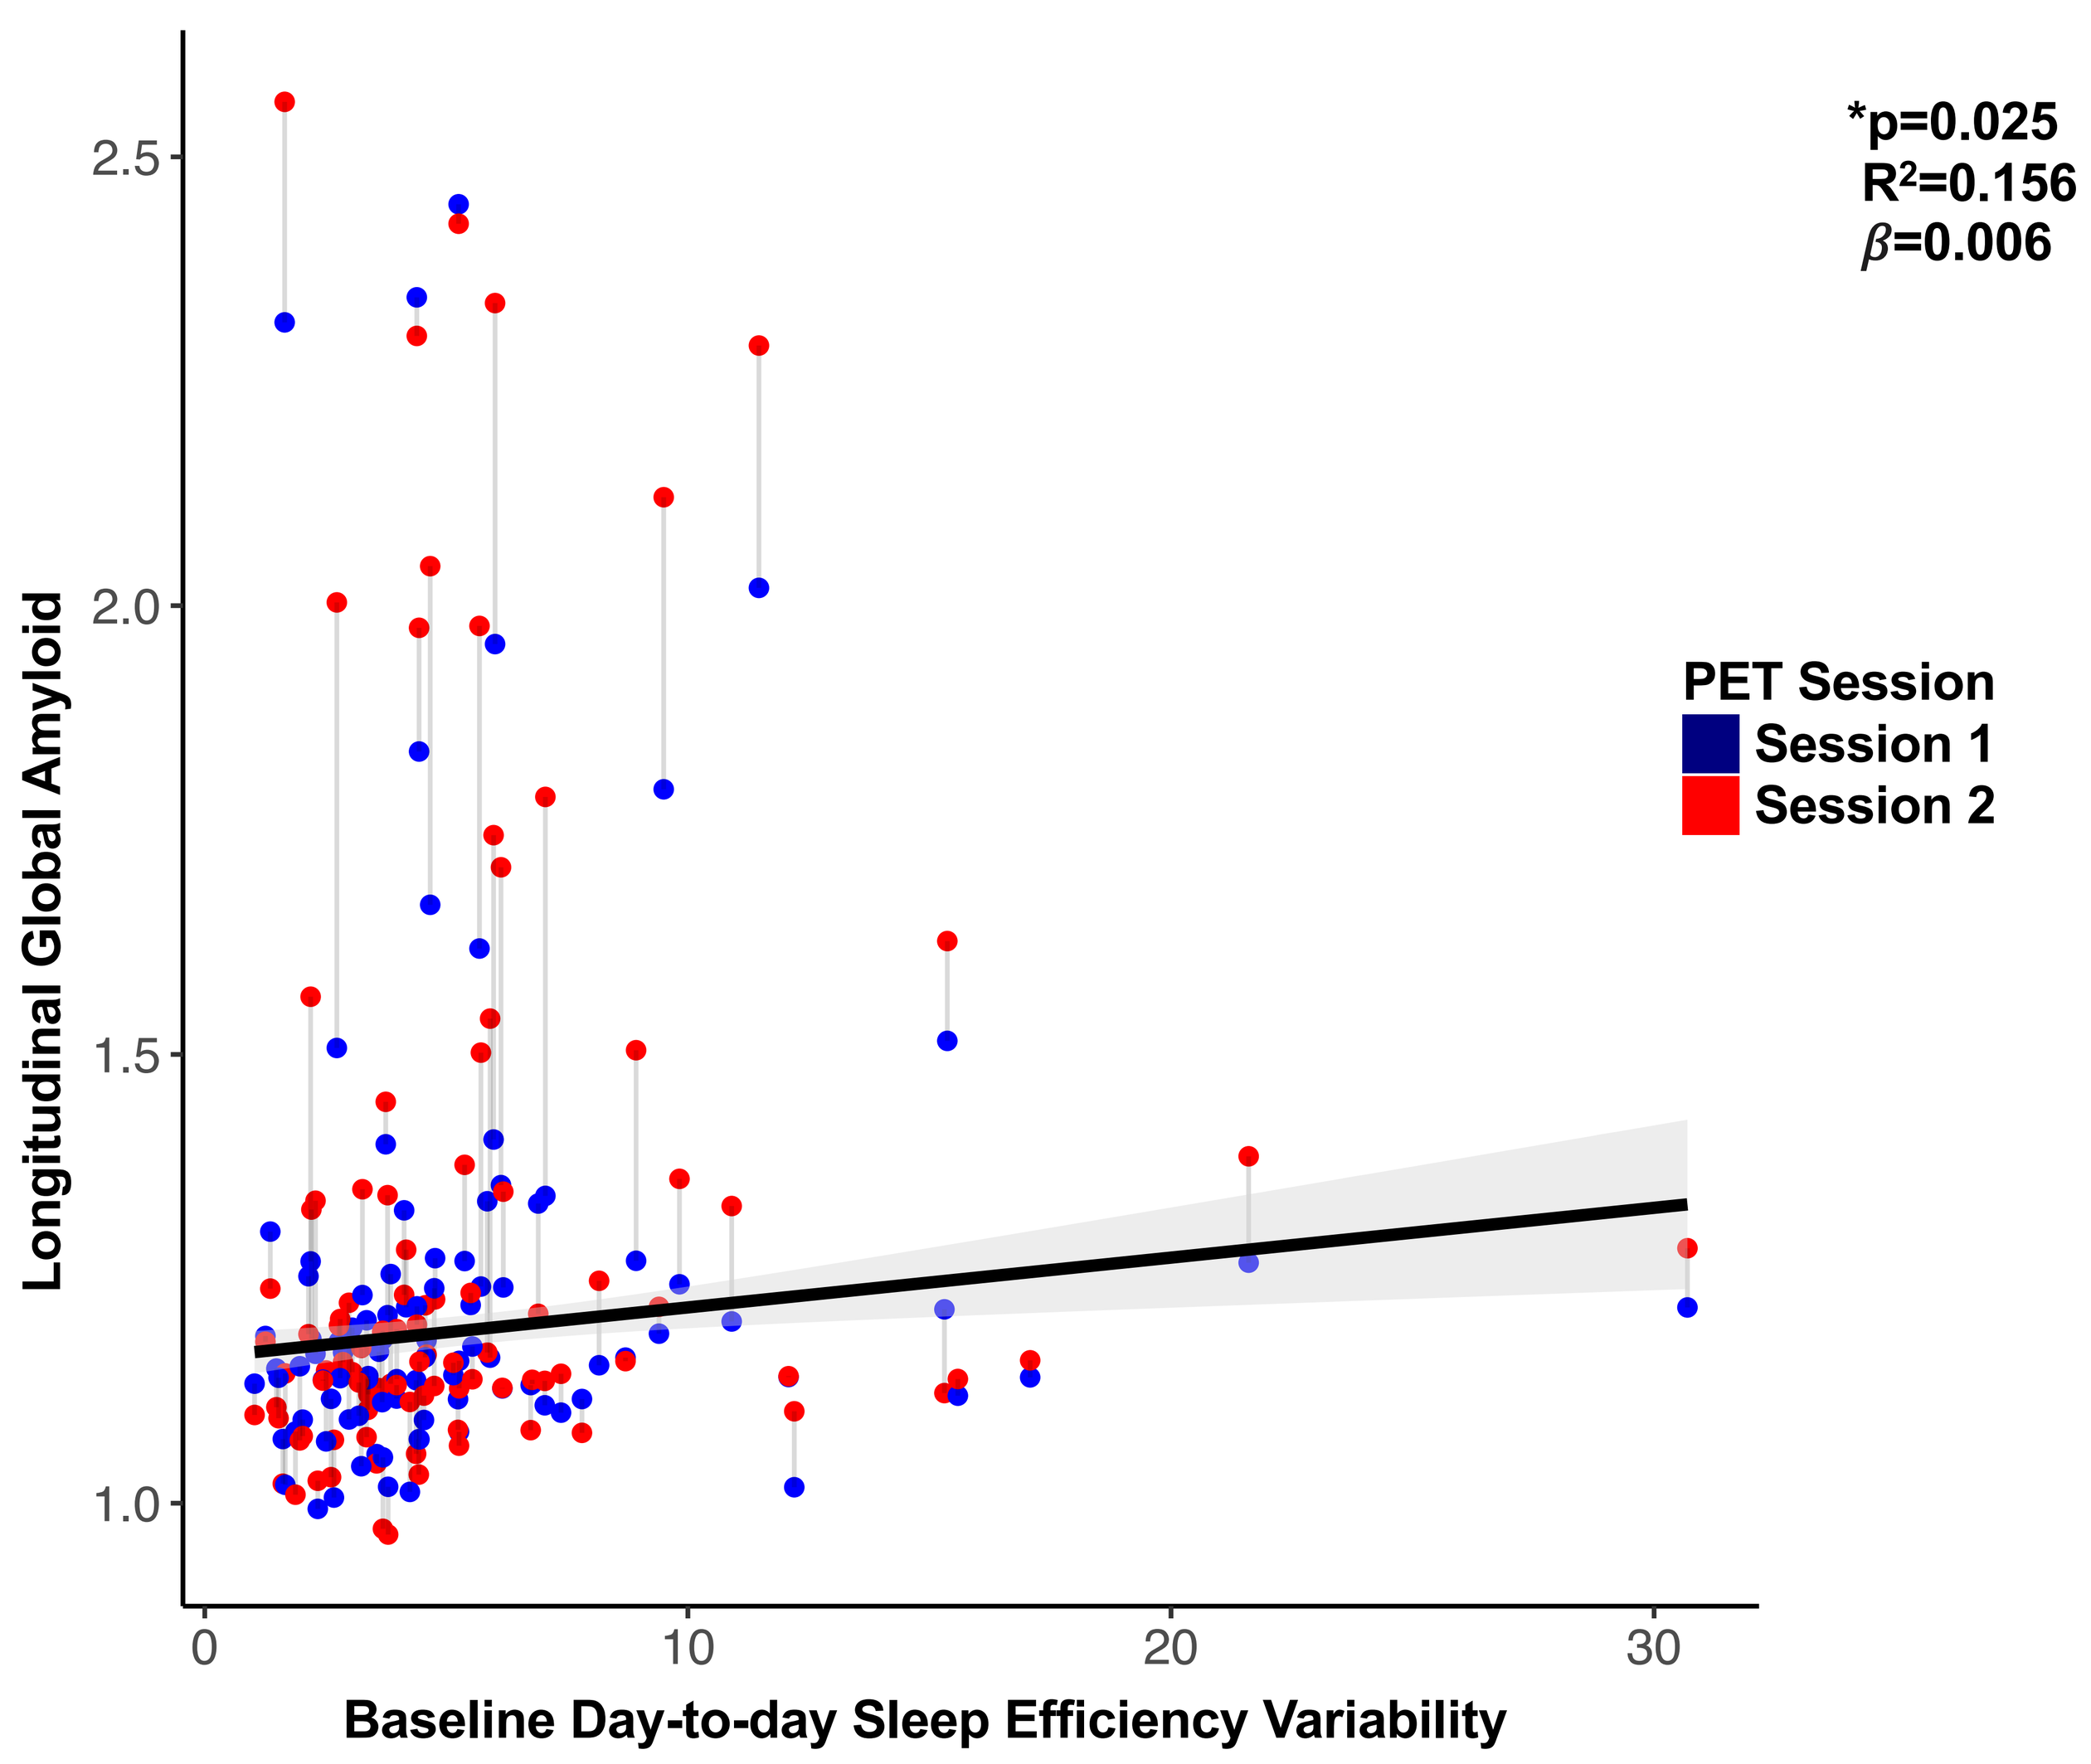
*p=0.025
R^2=0.156
β=0.006
Longitudinal Global Amyloid
Baseline Day-to-day Sleep Efficiency Variability
PET Session
Session 1
Session 2

Greater day-to-day sleep efficiency variability -> higher global medial temporal tau across time

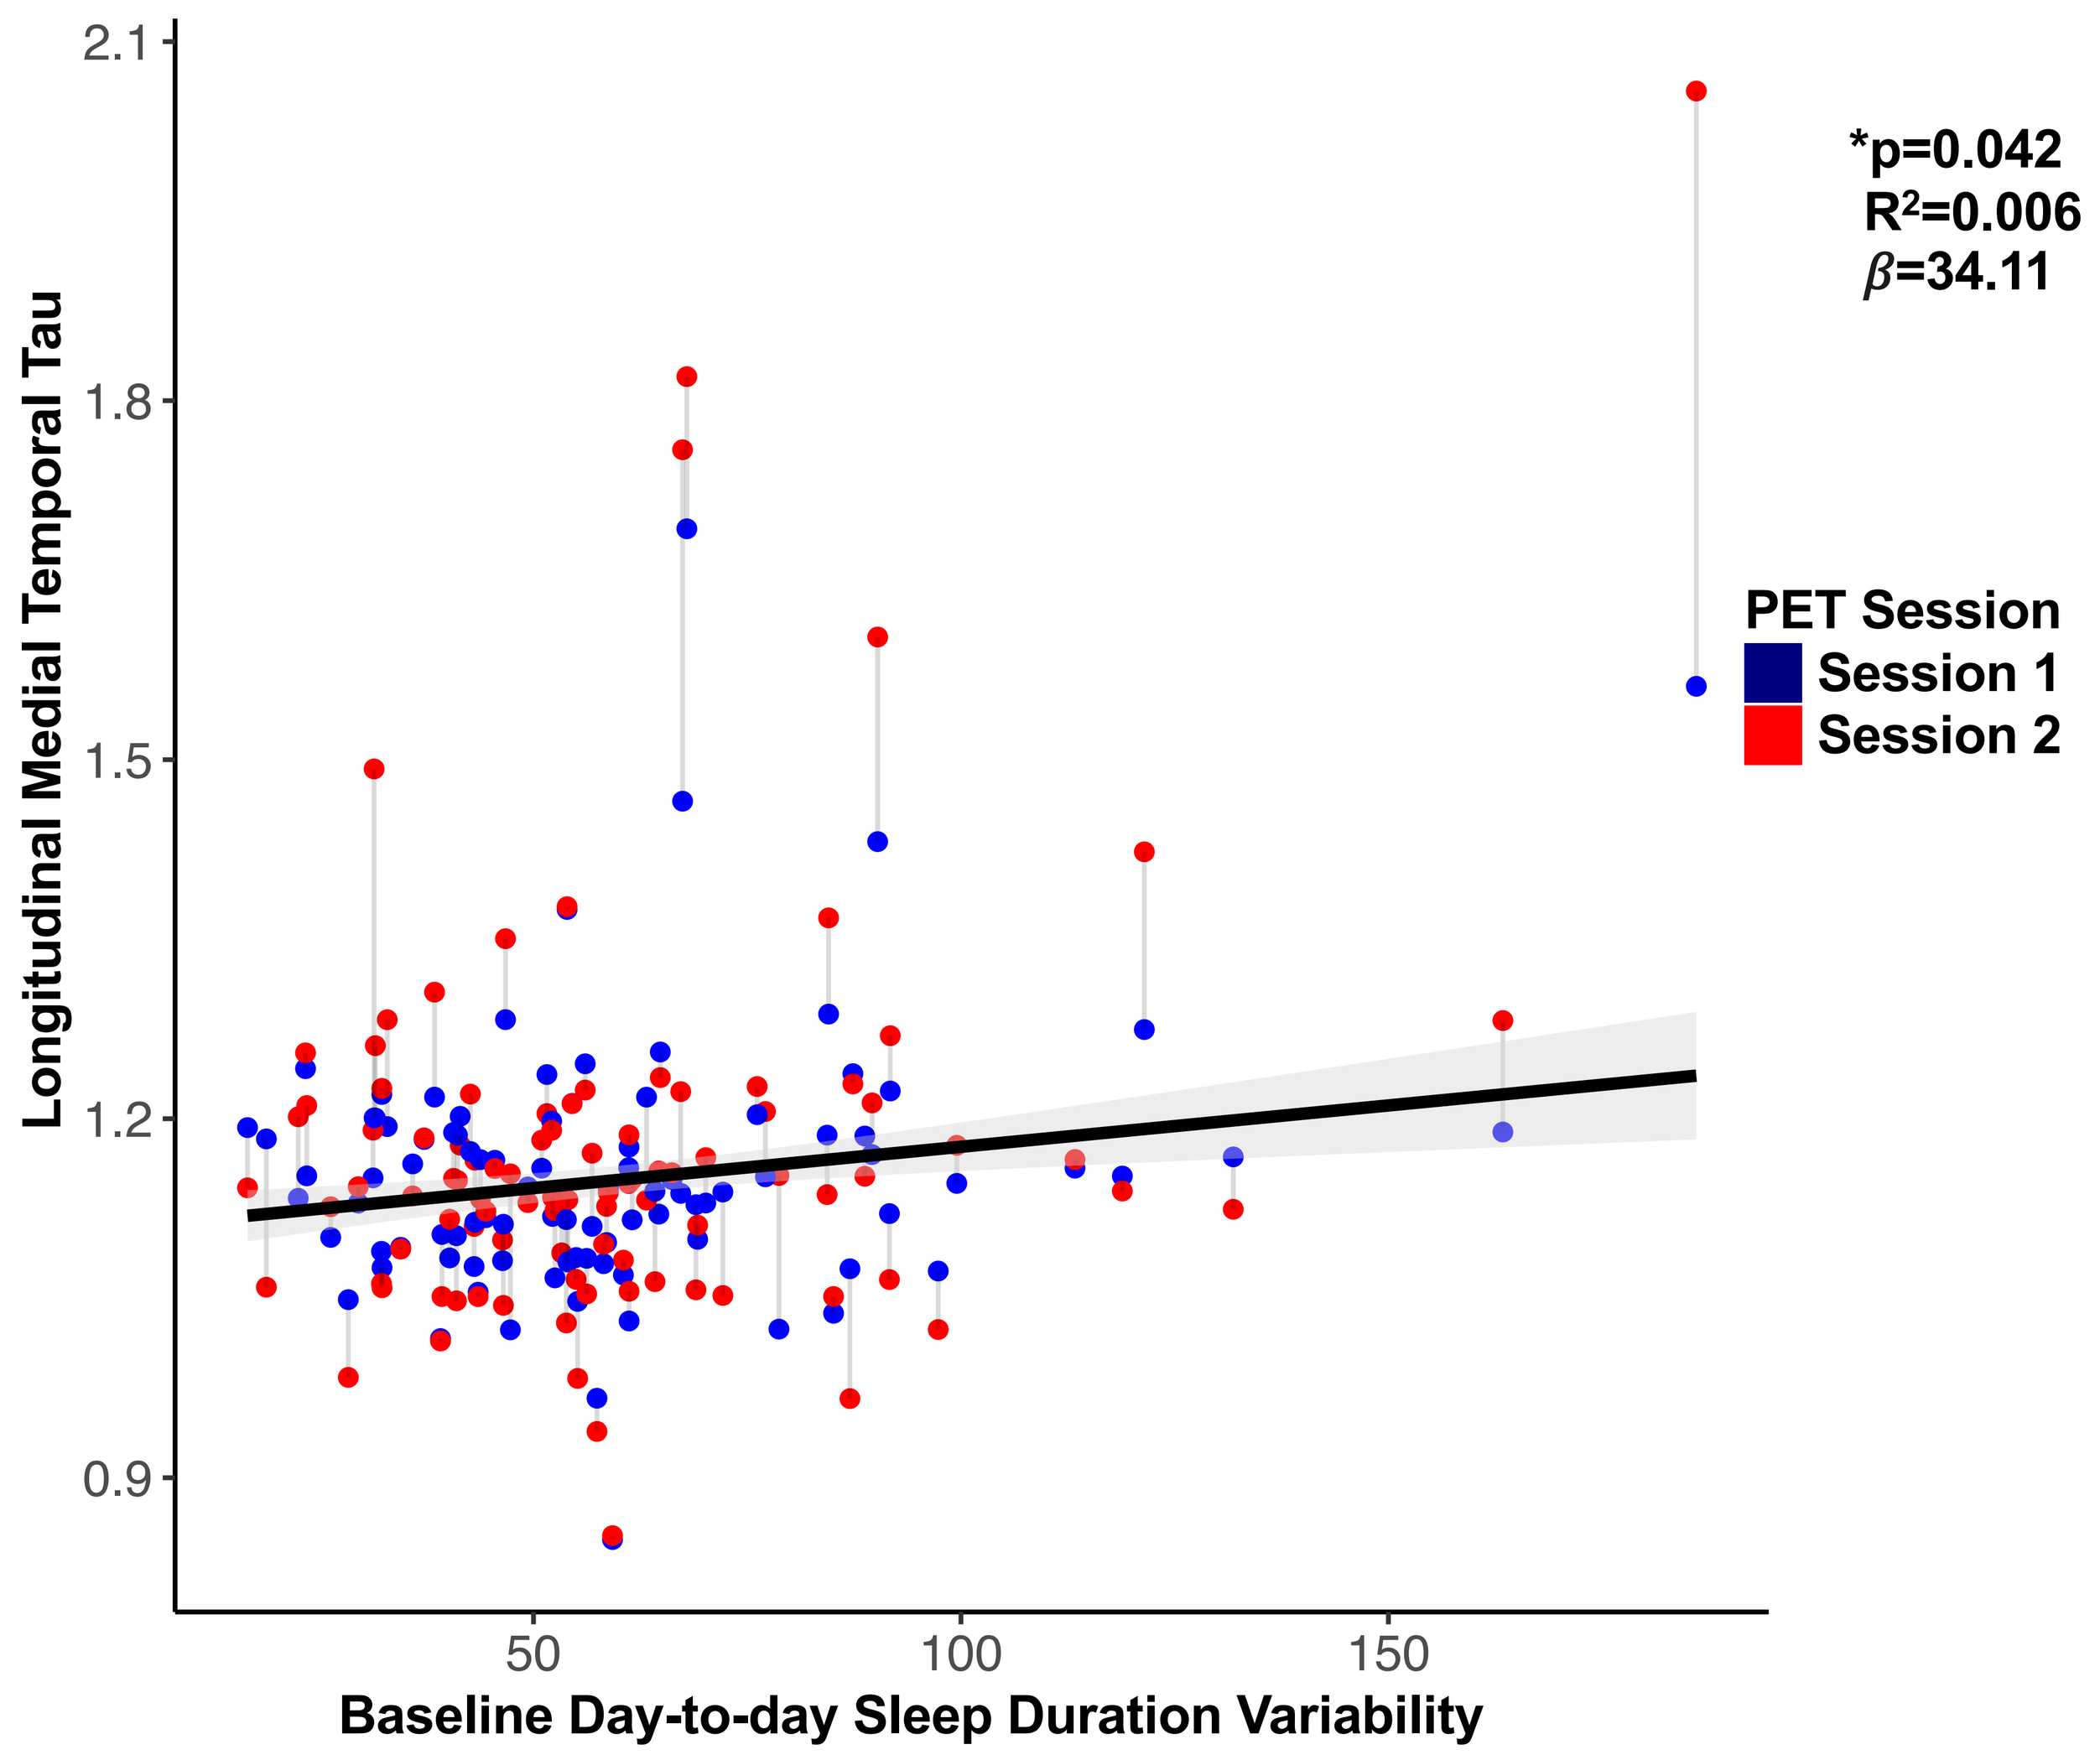
*p=0.042
R^2=0.006
β=34.11
Longitudinal Medial Temporal Tau
Baseline Day-to-day Sleep Duration Variability
PET Session
Session 1
Session 2

2) AD pathology -> Sleep

Higher global amyloid -> greater day-to-day sleep duration variability across time

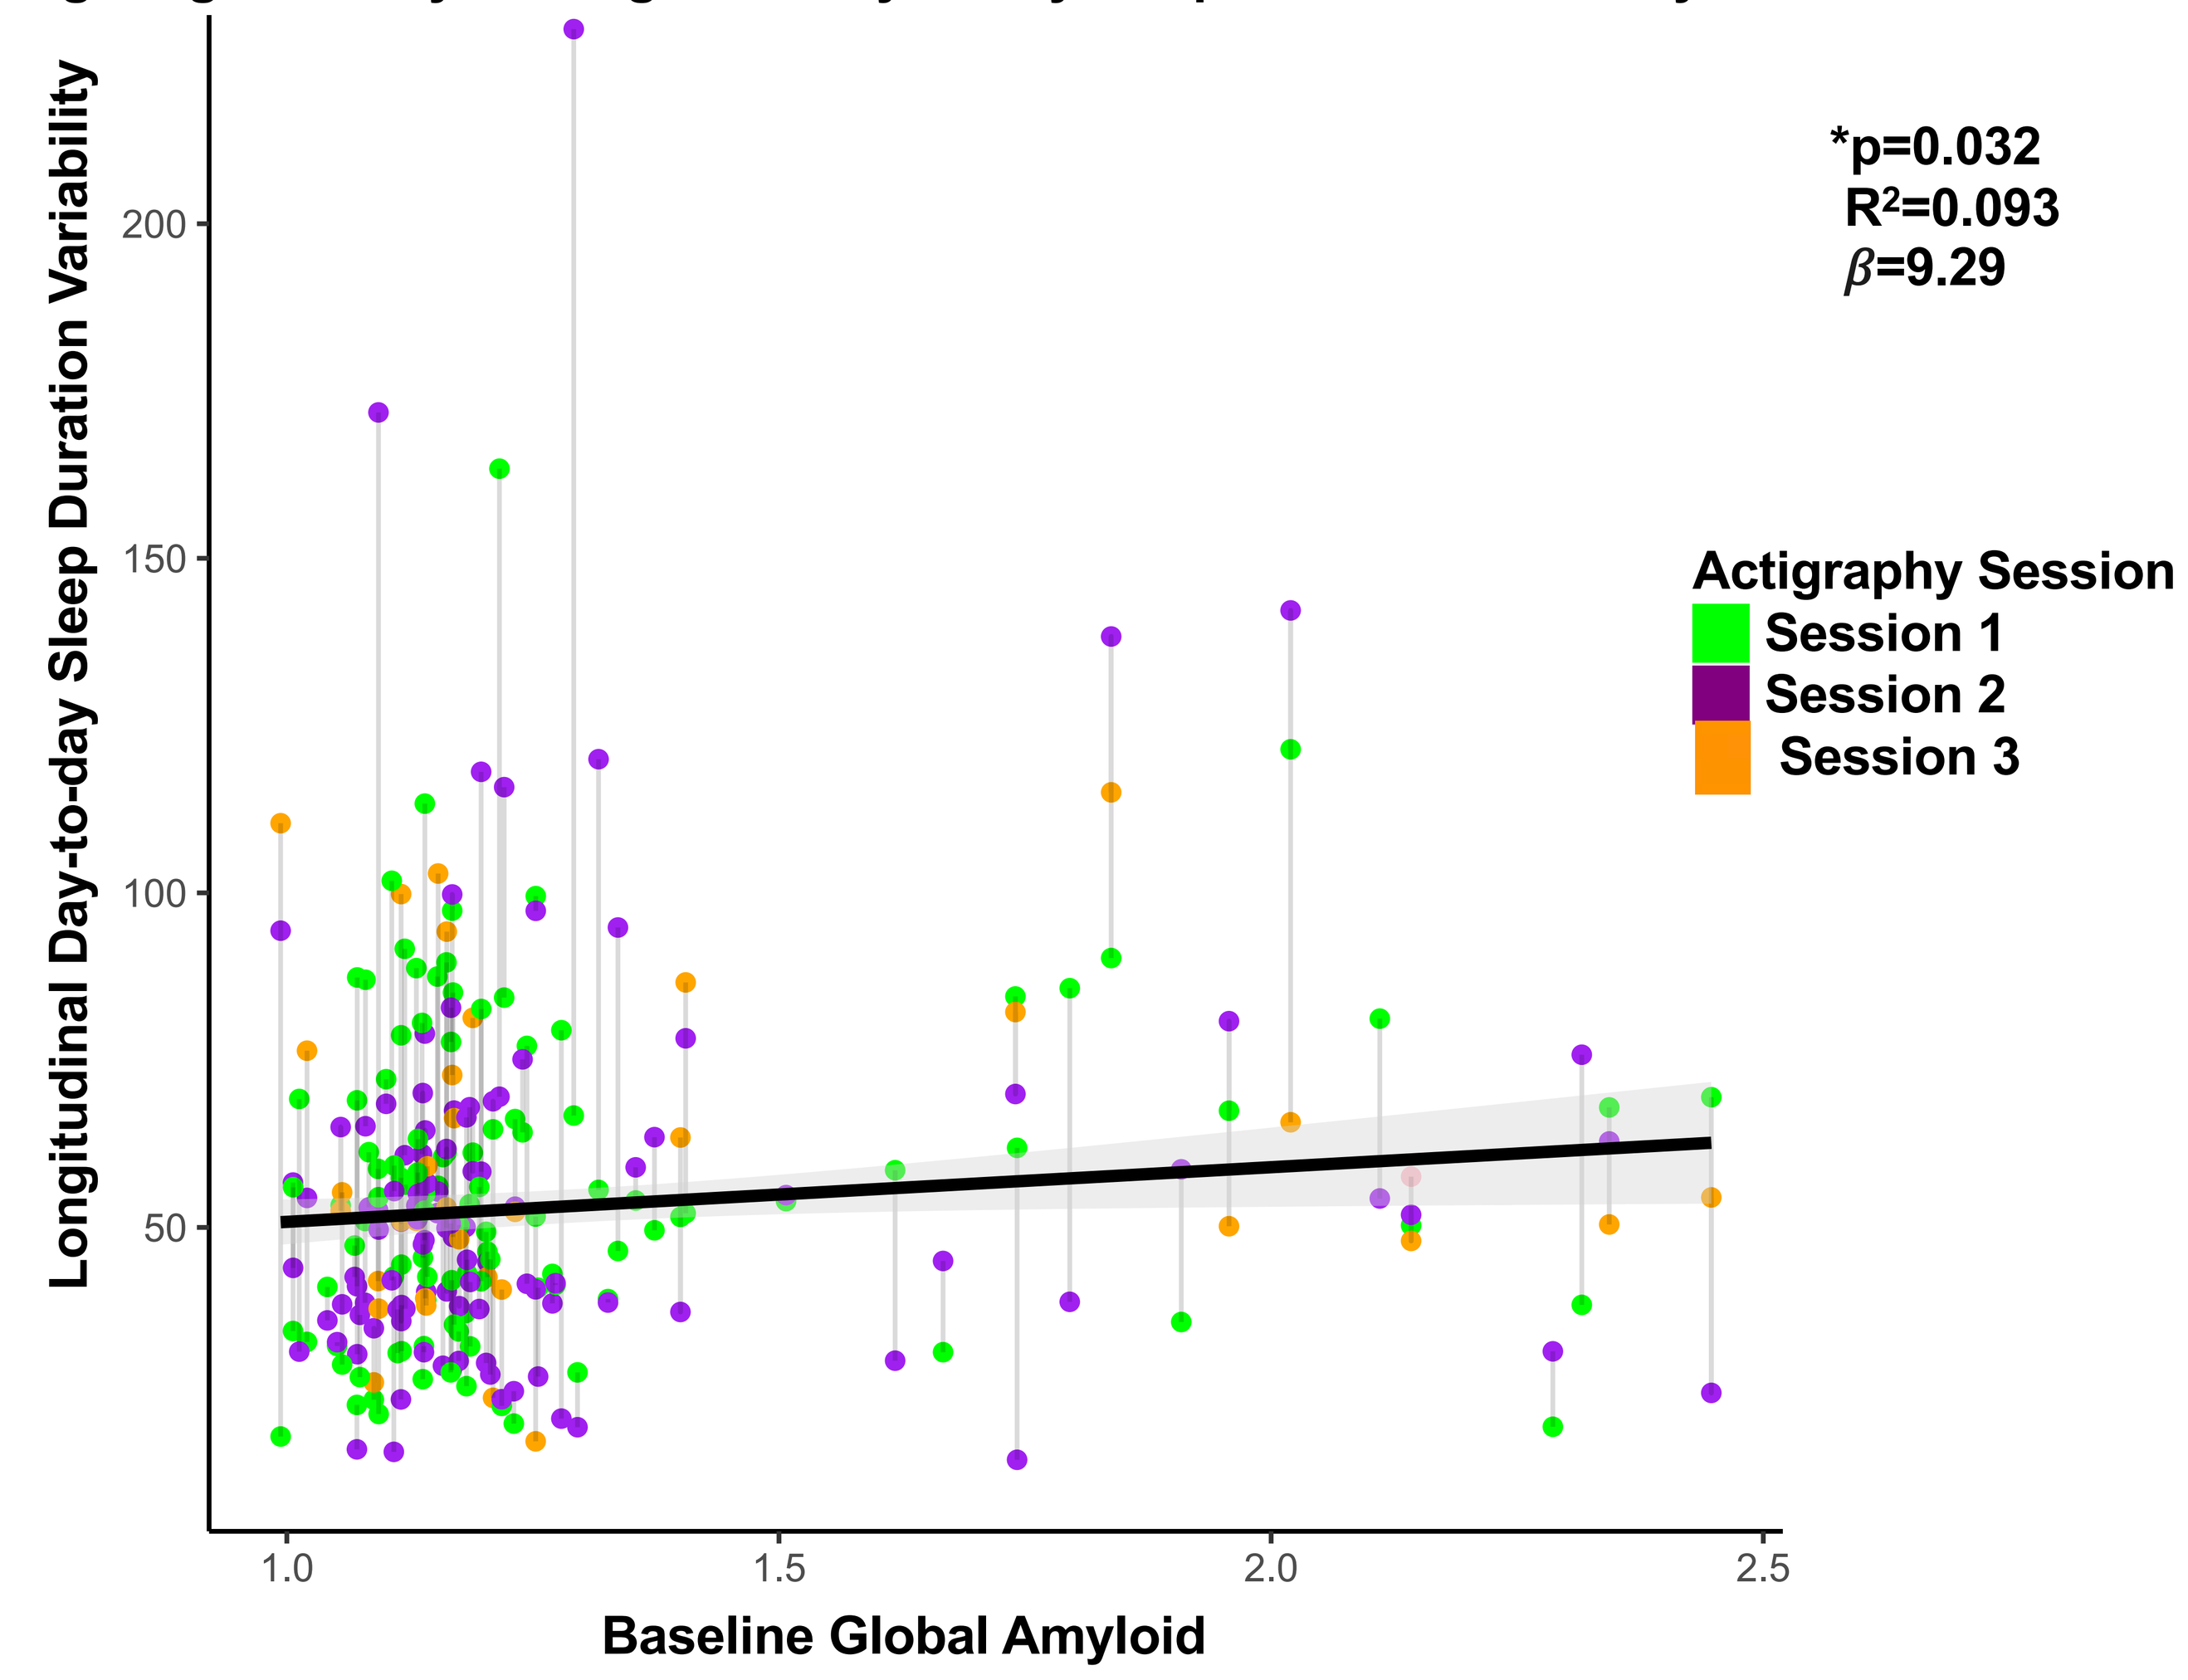
*p=0.032
R^2=0.093
β=9.29
Longitudinal Day-to-day Sleep Duration Variability
Baseline Global Amyloid
Actigraphy Session
Session 1
Session 2
Session 3

Higher medial temporal tau -> greater day-to-day sleep duration variability across time

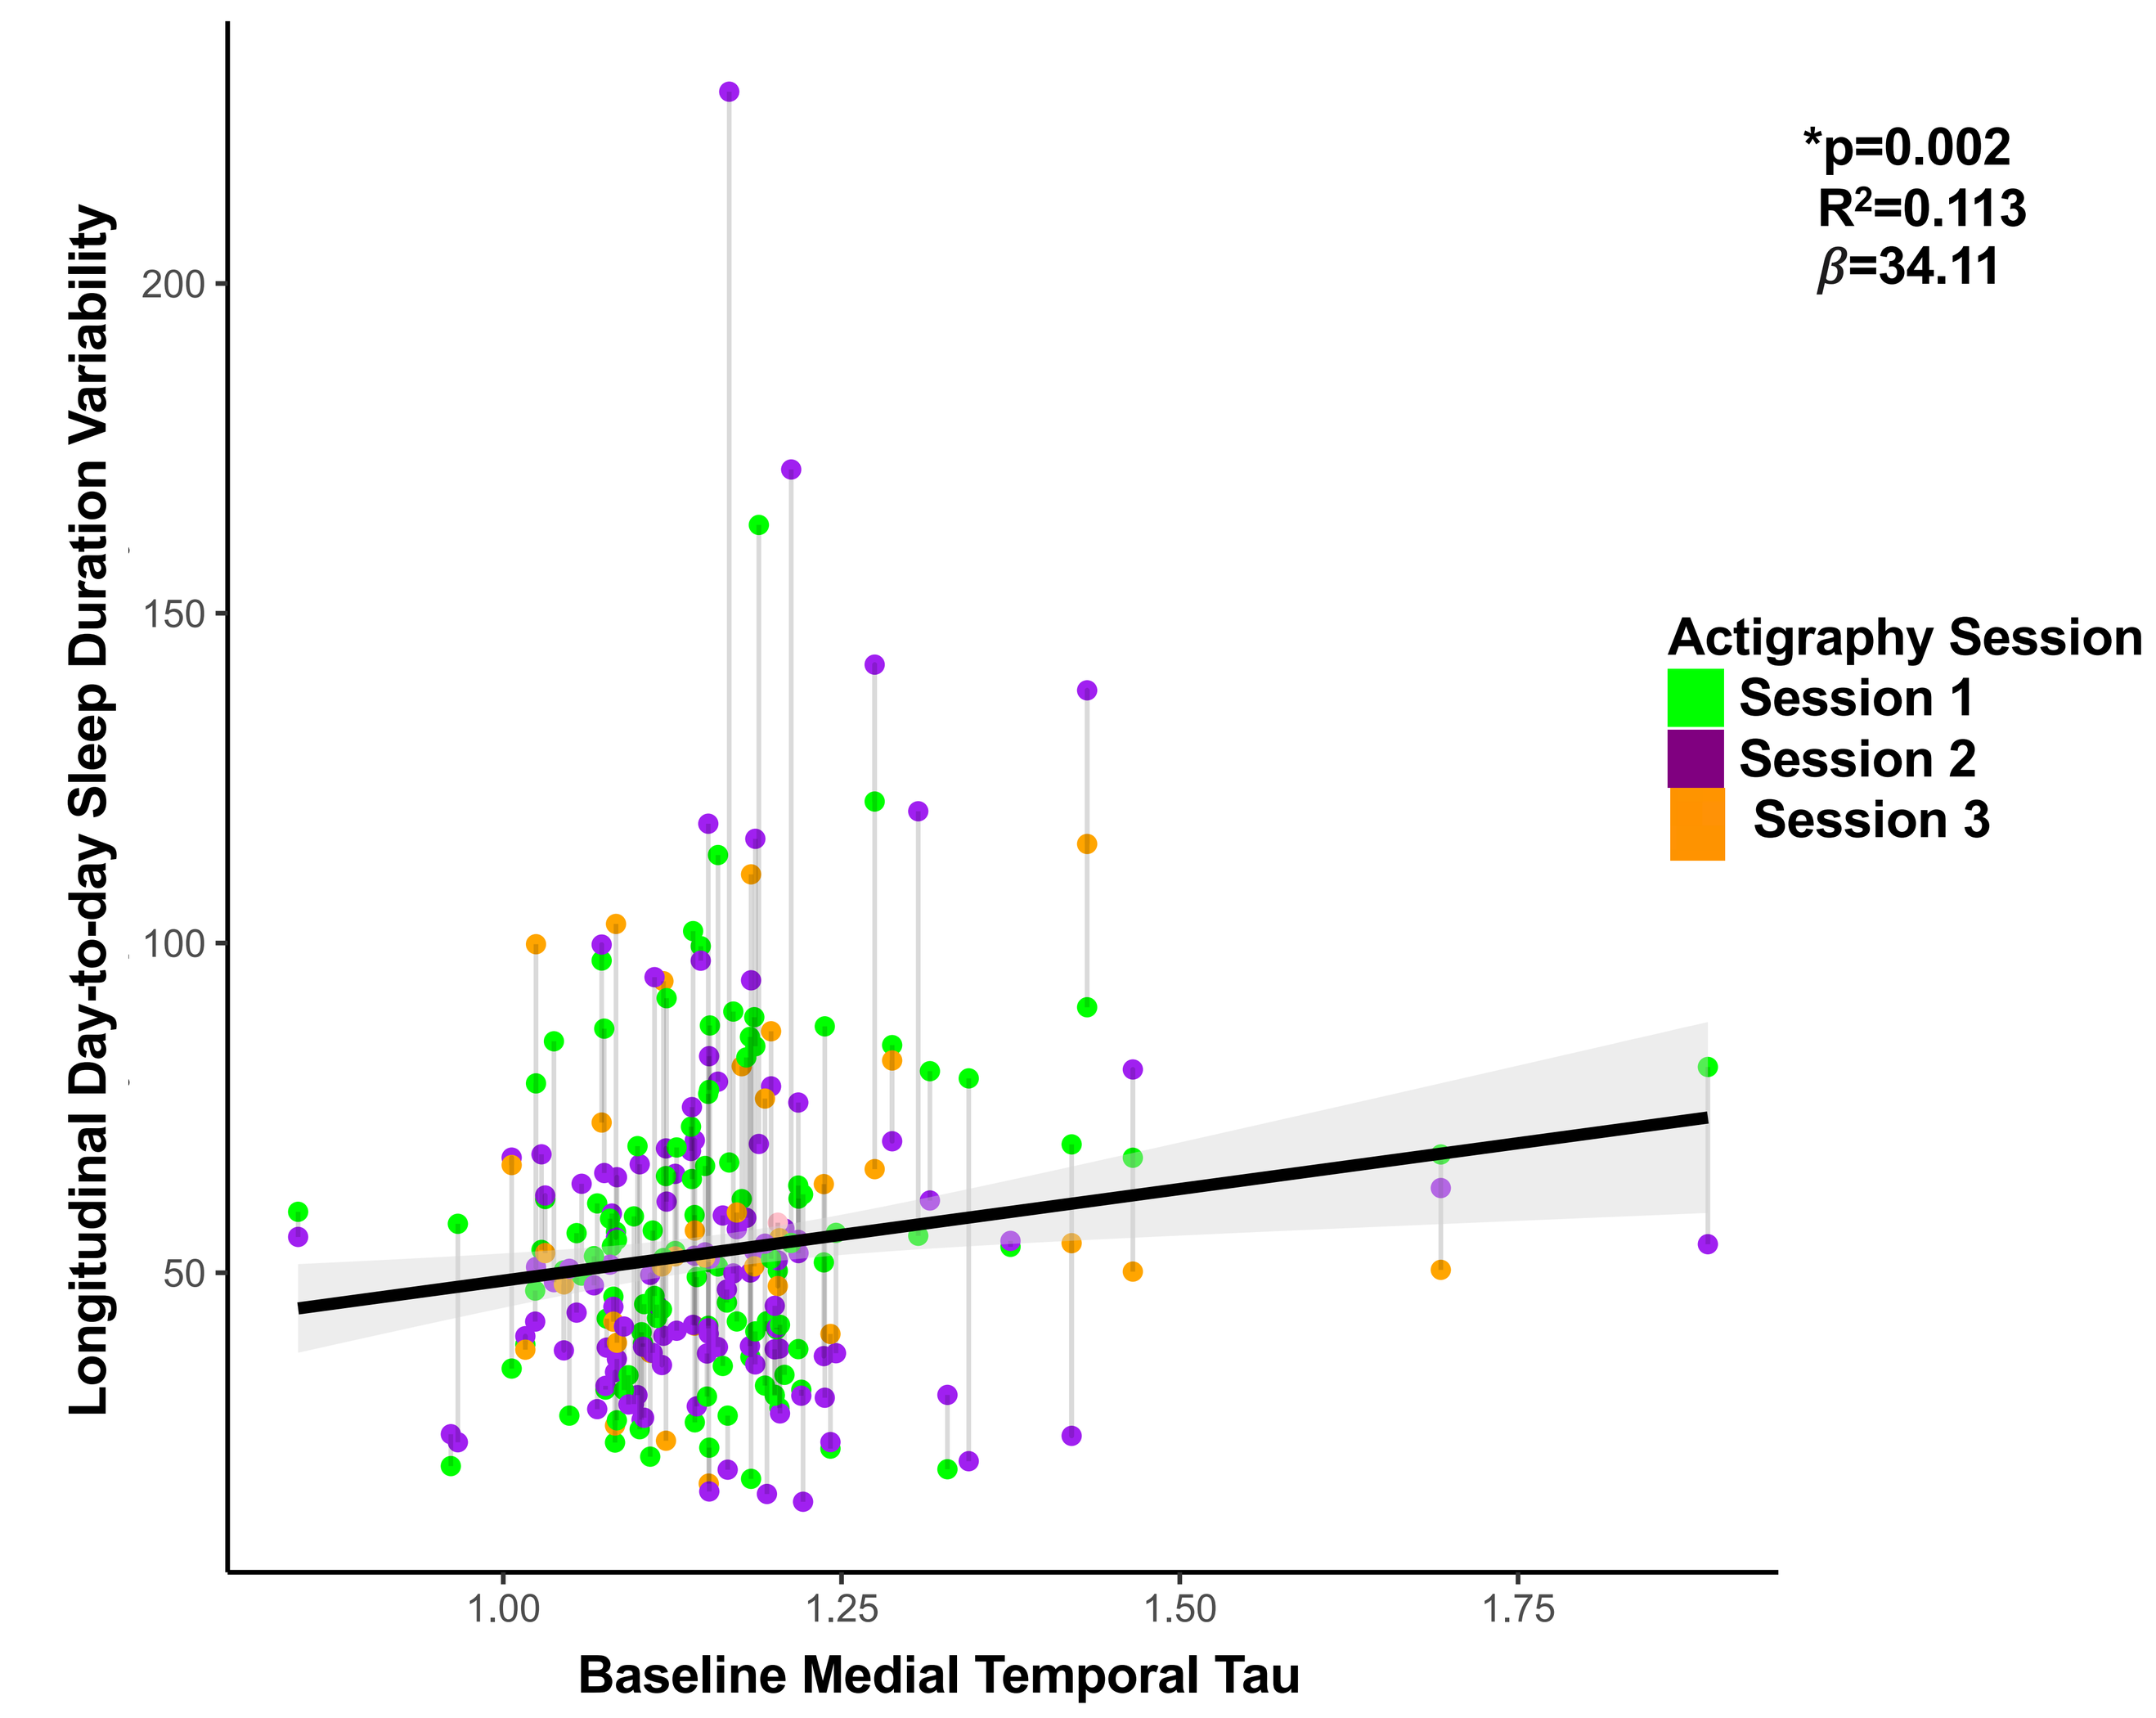
*p=0.002
R^2=0.113
β=34.11
Longitudinal Day-to-day Sleep Duration Variability
Baseline Medial Temporal Tau
Actigraphy Session
Session 1
Session 2
Session 3

Higher medial temporal tau -> worst self-reported sleep quality

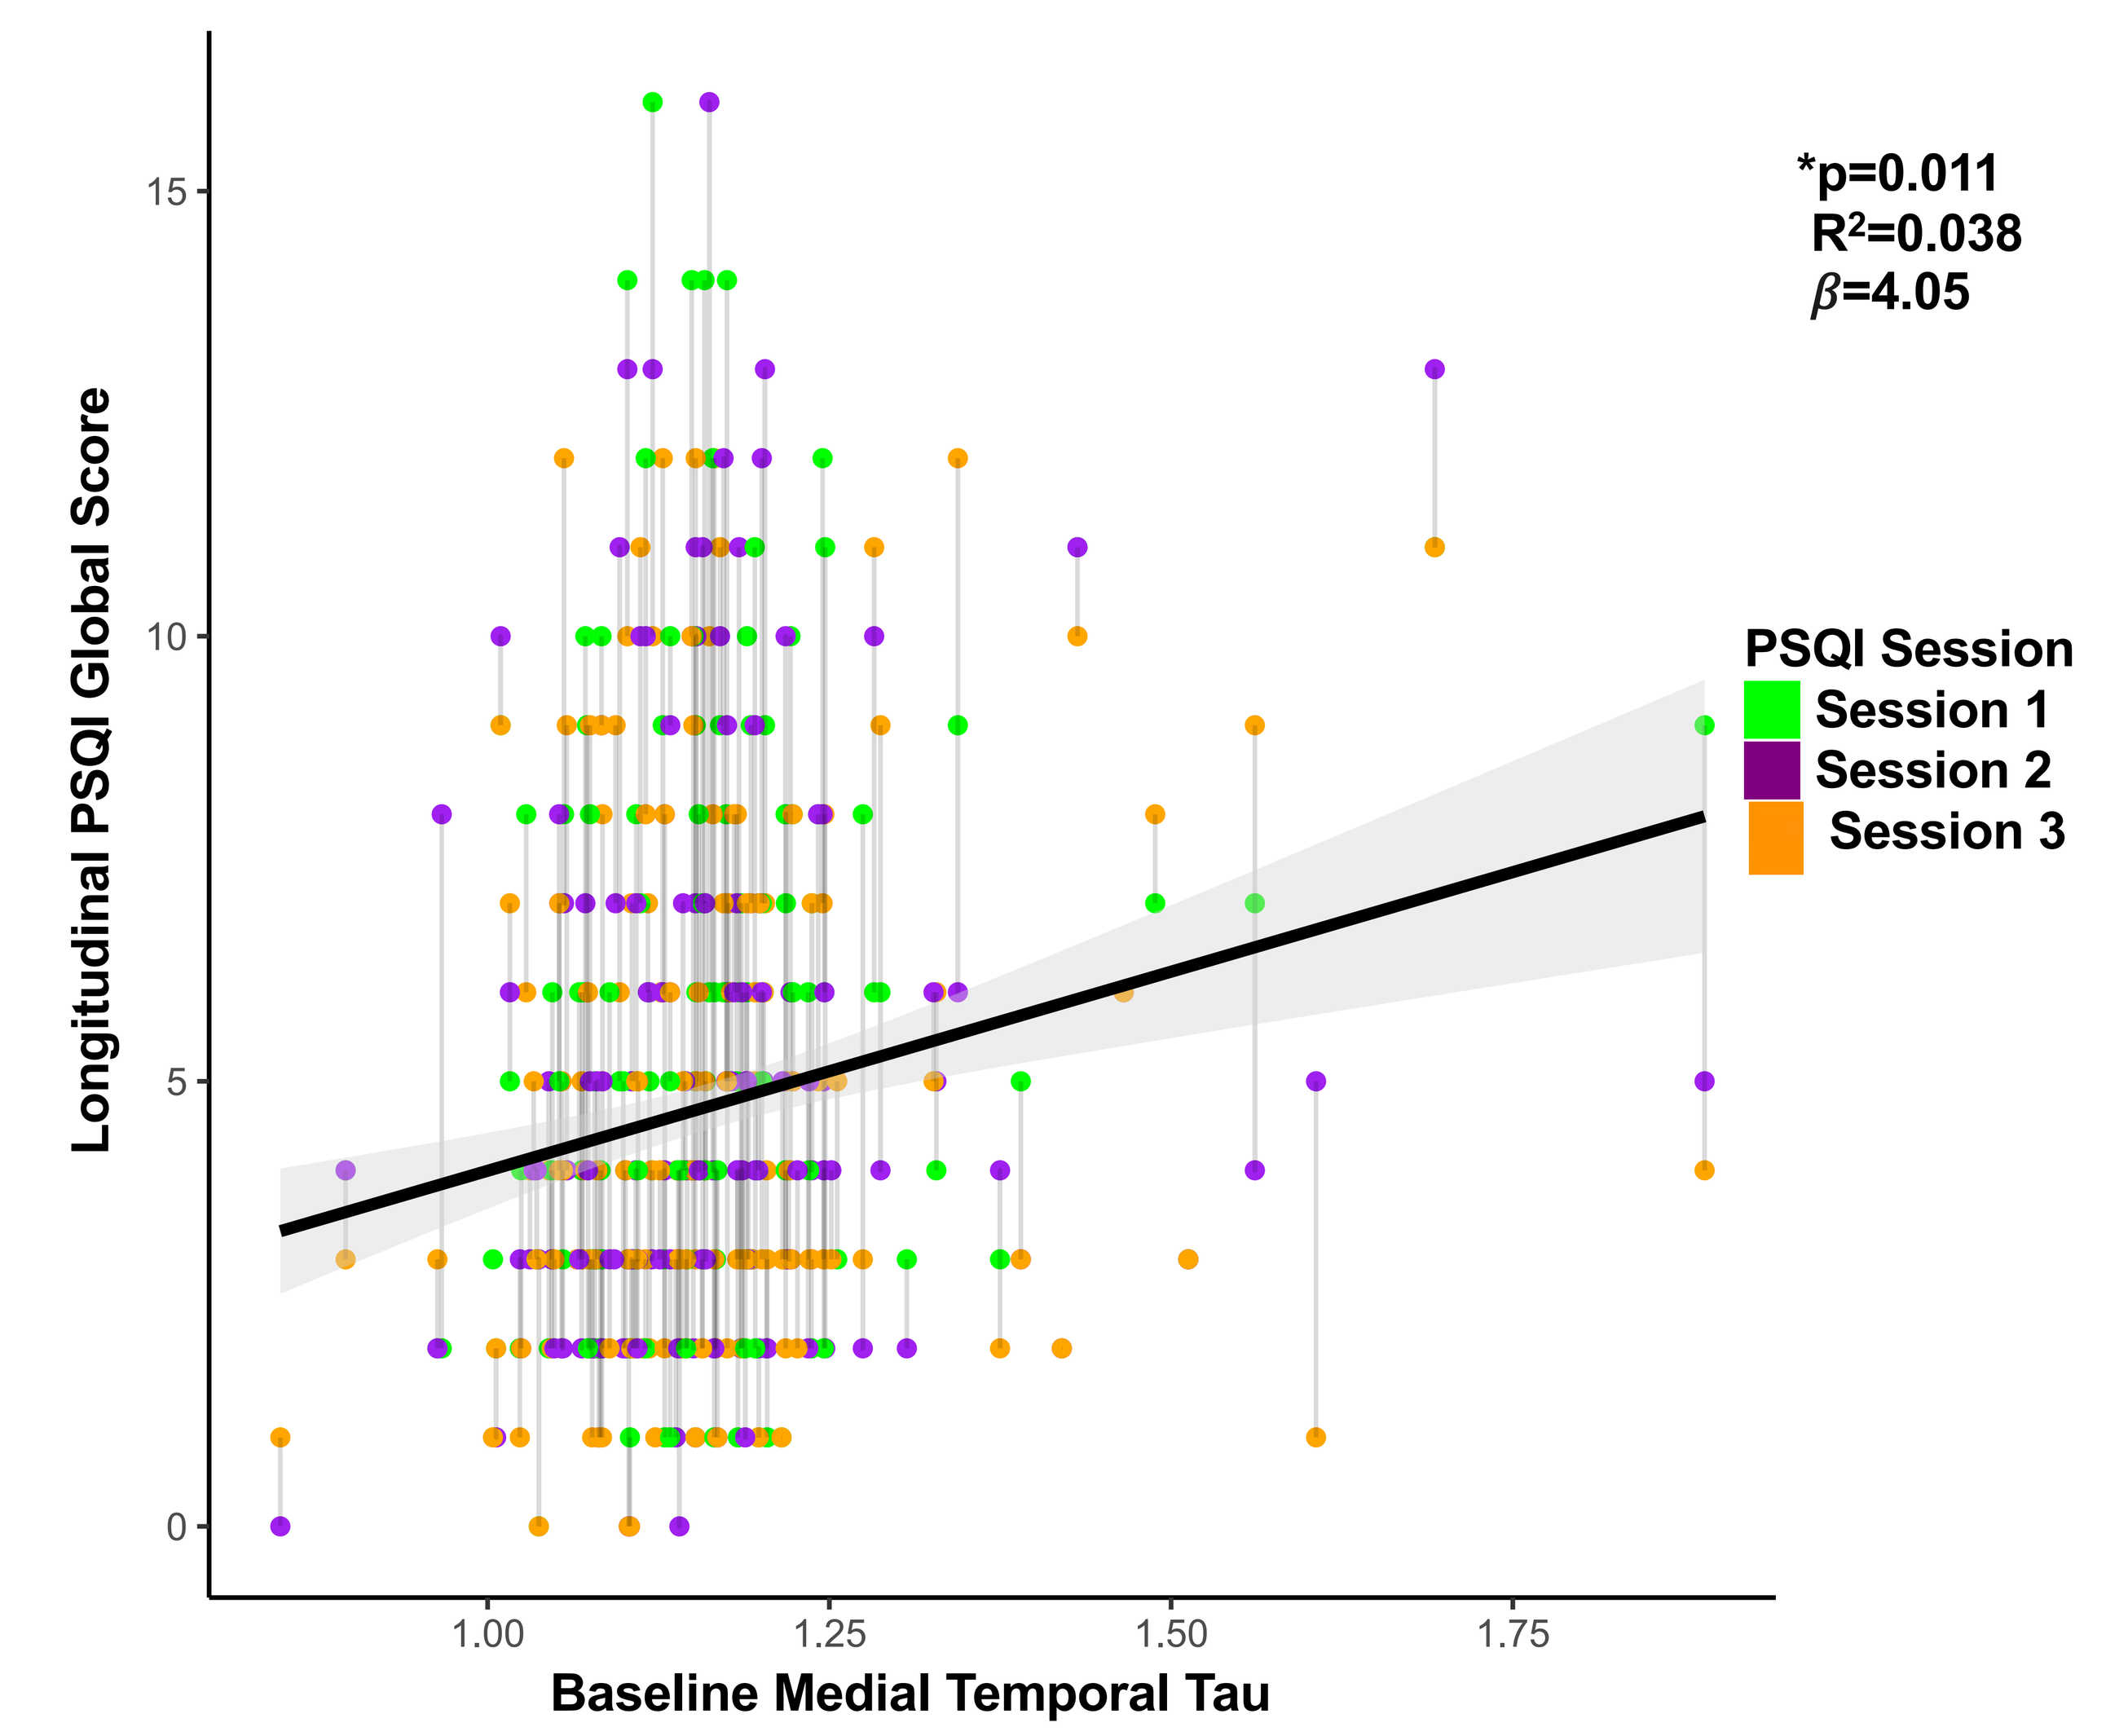
*p=0.011
R^2=0.038
β=4.05
Longitudinal PSQI Global Score
Baseline Medial Temporal Tau
PSQI Session
Session 1
Session 2
Session 3

4. Conclusions

We found an association between higher day-to-day sleep duration and efficiency variability and higher levels of amyloid across time. We found that higher day-to-day sleep duration variability is associated with higher levels of tau across time. Inversely, we found that higher levels of baseline amyloid is associated with greater subsequent day-to-day sleep duration variability. We also found that higher levels of tau is associated with greater subsequent day-to-day sleep duration variability and worst self-reported sleep quality. **This supports the idea of a bi-directional association between AD pathology and sleep quality in the preclinical stages of AD. Sleep impairments in older age could contribute to AD pathology accumulation, which could in turn disrupt sleep further.**

bery.mohammediyan@mail.mcgill.ca
@m_bery_