SEX AND FAMILY HISTORY DIFFERENCES IN Aβ AND TAU VULNERABILITY IN PRECLINICAL ALZHEIMER'S DISEASE

Villeneuve Lab
Multimodal Imaging of the Aging Brain

Valentin Ourry, PhD[1, 2] ✉; Frédéric St-Onge, MSc[2, 3]; Bery Mohammediyan, BA[1, 2]; Yara Yakoub, MSc[2, 3]; Jean-Paul Soucy, MD, MSc[4]; Judes Poirier, PhD[1, 2]; John C.S. Breitner, MD, MPH[1, 2]; Sylvia Villeneuve, PhD[1, 2, 4]; for the PREVENT-AD Research Group

[1] Department of Psychiatry, Faculty of Medicine, McGill University, Montreal, Quebec, Canada. [2] Douglas Mental Health University Institute, Montreal, Quebec, Canada. [3] Integrated Program in Neuroscience, Faculty of Medicine, McGill University, Montreal, Quebec, Canada. [4] McConnell Brain Imaging Center, Montreal Neurological Institute, McGill University, Montreal, Quebec, Canada.

Introduction

Background

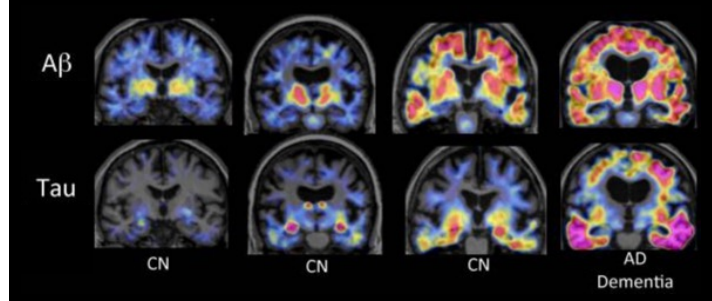
Aβ
Tau
CN
CN
CN
AD
Dementia

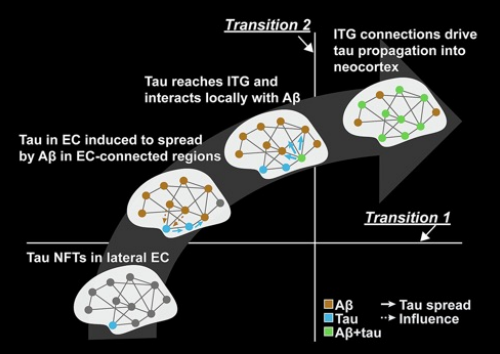
Transition 2
ITG connections drive tau propagation into neocortex
Tau reaches ITG and interacts locally with Aβ
Tau in EC induced to spread by Aβ in EC-connected regions
Transition 1
Tau NFTs in lateral EC
Aβ
Tau
Aβ+tau
Tau spread
Influence

Alzheimer's disease (AD) biomarkers [1, 2, 3]

Questions

Biological sex ♀ ♂

ApoE4 status

AD family history

Objectives

In cognitively unimpaired older adults with longitudinal data, we assessed:

1) between-sex (a) and AD family history (b) differences in Aβ and tau
2) the interaction between sex (a) or AD family history (b) and Aβ on tau
3) the interaction between ApoE and Aβ on tau separated by sex (a) or AD family history (b)

Methods

Participants

We included **234 cognitively unimpaired (at baseline) older adults** from the longitudinal PREVENT-AD cohort [4].

Variable	Level	Female (n=166)	Male (n=71)	Total (n=237)	p-value
Age (y)	mean (sd)	67.8 (4.8)	69.3 (5.4)	68.3 (5)	0.05
Education (y)	mean (sd)	15.4 (3.2)	15.6 (3.3) [10 – 24]	15.5 (3.2)	0.6
MMSE	mean (sd)	28.7 (1.2)	29 (1.3)	28.8 (1.3)	0.36
ApoE	ApoE 4 non-carrier (%)	103 (62)	40 (56.3)	143 (60.3)	
	ApoE 4 carrier (%)	63 (38)	31 (43.7)	94 (39.7)	0.5
PET NAV Follow Up (y)	mean (sd)	4.3 (0.5)	4.4 (0.4)	4.3 (0.4)	0.51
Missing	n	89	42	131	
PET TAU Follow Up (y)	mean (sd)	4.3 (0.5)	4.3 (0.4)	4.3 (0.5)	0.59
Missing	n	91	42	133	
AD family history	Maternal	102 (61.4)	36 (50.7)	138 (58.2)	
	Paternal	38 (22.9)	18 (25.4)	56 (23.6)	
	Both	20 (12)	13 (18.3)	33 (13.9)	
	Other	6 (3.6)	4 (5.6)	10 (4.2)	0.39
MCI progression	no (%)	152 (91.6)	62 (87.3)	214 (90.3)	
	yes (%)	14 (8.4)	9 (12.7)	23 (9.7)	0.44

Acquisition and preprocessing

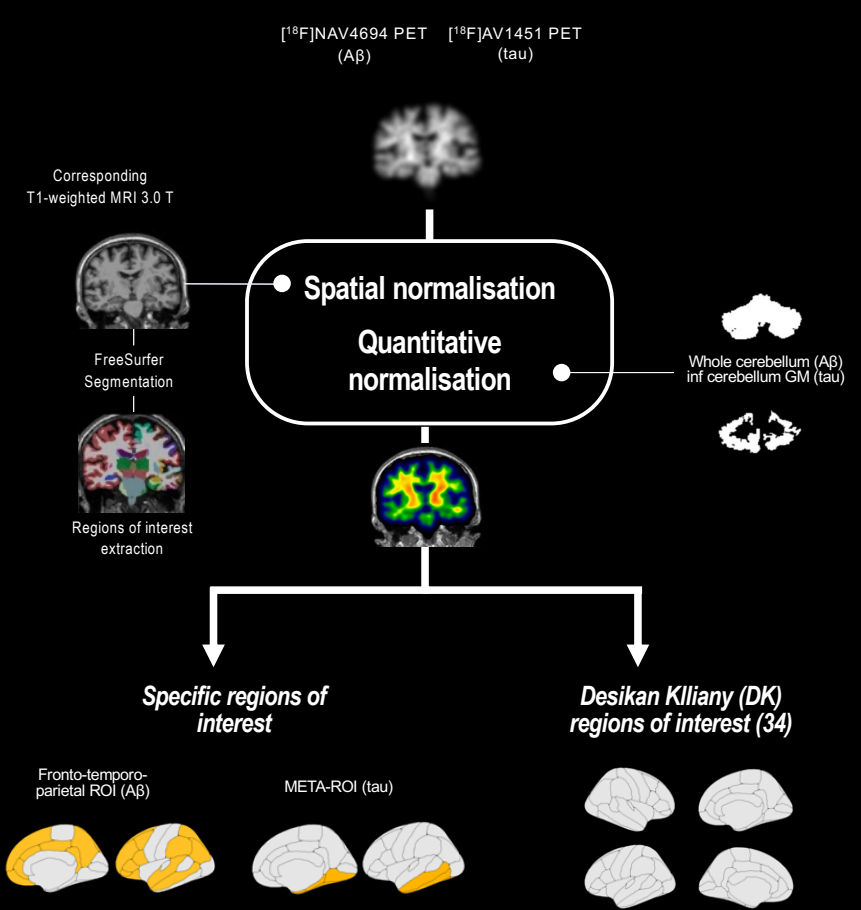
[18F]NAV4694 PET (Aβ)
[18F]AV1451 PET (tau)
Corresponding T1-weighted MRI 3.0 T
FreeSurfer Segmentation
Regions of interest extraction
Spatial normalisation
Quantitative normalisation
Whole cerebellum (Aβ)
inf cerebellum GM (tau)
Specific regions of interest
Fronto-temporo-parietal ROI (Aβ)
META-ROI (tau)
Desikan Klliany (DK) regions of interest (34)

Statistical analysis

1) T-test between male and female (a) or paternal and maternal family history (b) on SUVr at baseline and annual SUVr changes (follow up scan – baseline scan / follow up duration).

2) Interaction between Sex (a) or AD family history (b) and Aβ SUVr at baseline on tau SUVr at baseline and annual tau SUVr changes

3) Interaction between ApoE status and Aβ SUVr at baseline on tau SUVr at baseline and annual tau SUVr changes separated by sex (a) or AD family history (b)

Adjusting our models by age and education did not change the results.

Results

1. Females have higher tau at baseline in frontal, parietal and temporal lobes

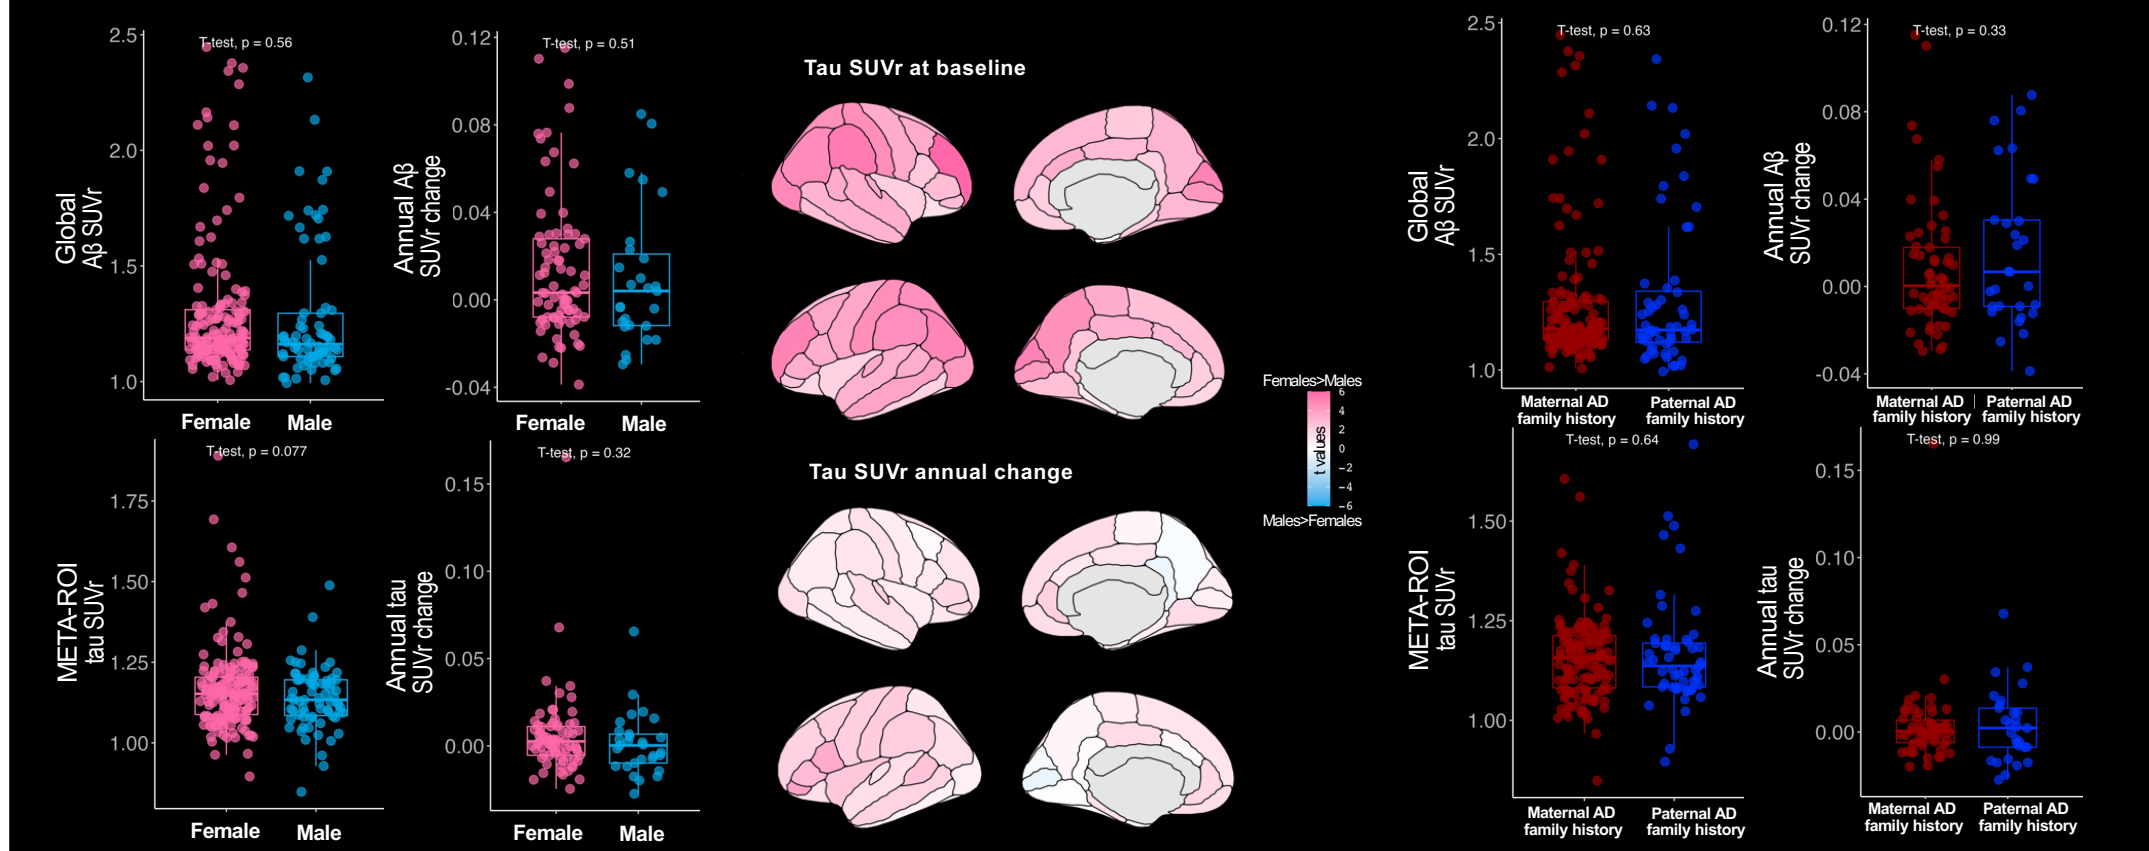
a) Sex differences
T-test, p = 0.56
T-test, p = 0.51
T-test, p = 0.077
T-test, p = 0.32
Global Aβ SUVr
Annual Aβ SUVr change
META-ROI tau SUVr
Annual tau SUVr change
Female
Male
Tau SUVr at baseline
Tau SUVr annual change
Females>Males
t values
Males>Females
b) AD family history differences
T-test, p = 0.63
T-test, p = 0.33
T-test, p = 0.64
T-test, p = 0.99
Maternal AD family history
Paternal AD family history

2. Females and paternal AD family history have stronger tau-Aβ association

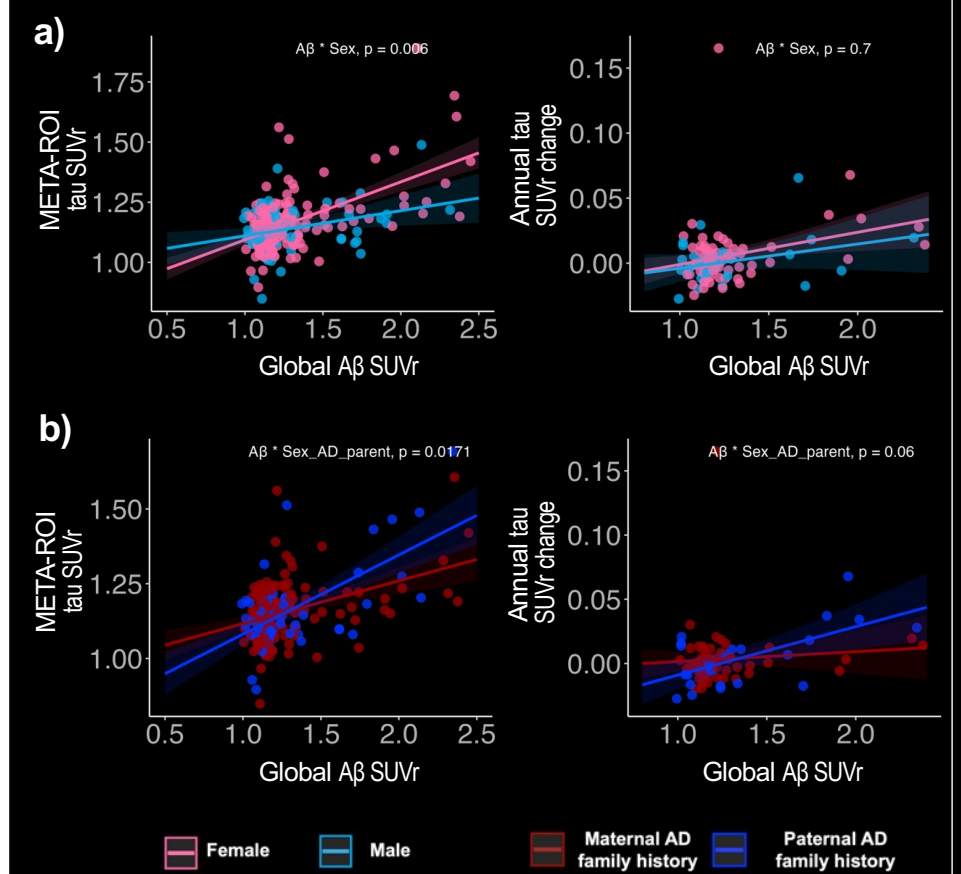
a)
Aβ * Sex, p = 0.006
Aβ * Sex, p = 0.7
b)
Aβ * Sex_AD_parent, p = 0.0171
Aβ * Sex_AD_parent, p = 0.06
META-ROI tau SUVr
Annual tau SUVr change
Global Aβ SUVr
Female
Male
Maternal AD family history
Paternal AD family history

3. ApoE4 status does not moderate the association when separated by sex or AD family history

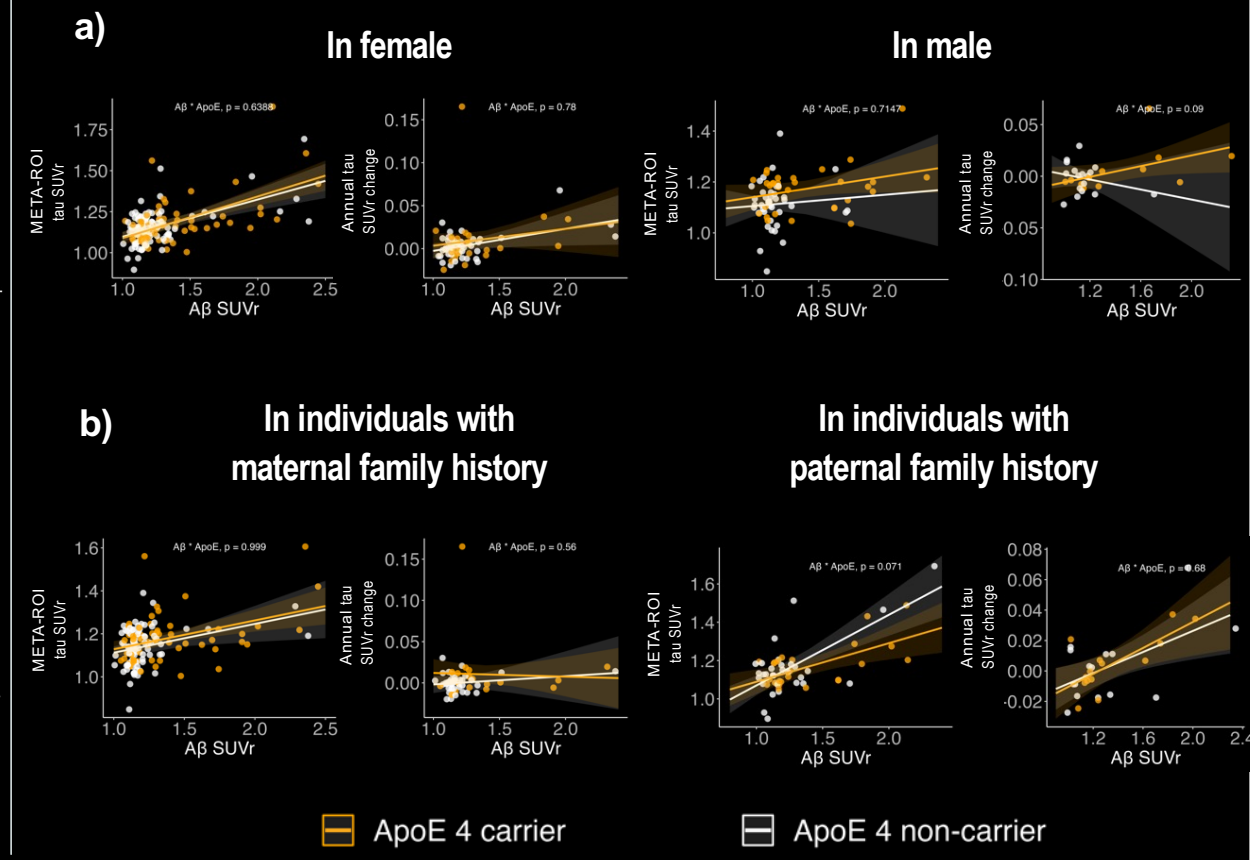
a)
In female
In male
Aβ * ApoE, p = 0.6388
Aβ * ApoE, p = 0.78
Aβ * ApoE, p = 0.7147
Aβ * ApoE, p = 0.09
b)
In individuals with maternal family history
In individuals with paternal family history
Aβ * ApoE, p = 0.999
Aβ * ApoE, p = 0.56
Aβ * ApoE, p = 0.071
META-ROI tau SUVr
Annual tau SUVr change
Aβ SUVr
ApoE 4 carrier
ApoE 4 non-carrier

Conclusion

Females and individuals with a paternal family history of AD might need less Aβ before developing AD-related tau but did not accumulate tau faster. Identifying individuals more vulnerable to AD pathology, by disentangling sex-specific effects, would help to design personalized interventions for AD.

References : [1] Sperling et al., 2014. Neuron. doi: 10.1016/j.neuron.2014.10.038. **[2]** Buckley et al., 2019. JAMA Neurol. doi:10.1001/jamaneurol.2018.4693. **[3]** Lee et al., 2022. Brain. doi: 10.1016/j.neuron.2022.03.034 **[4]** Tremblay-Mercier et al., 2021. Neuroimage Clin. doi: 10.1016/j.nicl.2021.102733.

Authors have no disclosure.

Aknowlegment : Thanks to all participants, neuro-imaging staff for data acquisitions and all members of the Douglas and Villeneuve lab who contributed to this study.

Funding: PREVENT-AD Research Group; neuro Montreal Neurological Institute-Hospital; CIHR IRSC Canadian Institutes of Health Research / Instituts de recherche en santé du Canada; Québec Fonds de recherche – Nature et technologies, Fonds de recherche – Santé, Fonds de recherche – Société et culture; Fondation famille Lemaire; Alzheimer's Association

valentin.ourry@gmail.com
#OurryValentin
www.villeneuvelab.com

ALZHEIMER'S ASSOCIATION AAIC 23