Amyloid and tau pathology are associated with white matter properties in cognitively unimpaired older adults at risk of AD dementia

Ting Qiu[1,2], François Rheault[3], Jean-Paul Soucy[1], R. Nathan Spreng[1], Alexa Pichet Binette[4], Maxime Descoteaux[3], Sylvia Villeneuve[1,2], for the PREVENT-AD Research Group.

1. McGill University, Montreal, Canada; 2. Douglas Mental Health University Institute, Montreal, Canada;3. Université de Sherbrooke, Sherbrooke, Canada; 4. Lund University, Lund, Sweden

BACKGROUND

White matter changes are hypothesized to be among the earliest changes occurring in the course of Alzheimer's disease (AD). We investigated associations between AD pathology (Aβ & tau) and white matter integrity in cognitively unimpaired individuals at risk of AD dementia. We expected higher levels of pathology to be related to lower free-water-corrected fractional anisotropy (FA_T, T stands for tissue) and lower neurite density index (NDI).

PARTICIPANTS

PREVENT-AD Research Group

189 cognitively unimpaired older adults with a familial history of AD; each underwent Aβ- and tau-PET, T1w/diffusion-w MRI scan.

	whole sample (N=189)	A+ (N=54)
Mean age (± SD)	67.75 (±4.7)	68.56 (±5.03)
Mean education (± SD)	15.5 (±3.1)	15.06 (±2.92)
Female (%)	72.34%	75.92%
global Aβ SUVR (± SD)	1.29 (±0.29)	1.67 (±0.33)
meta ROI tau SUVR (± SD)	1.15 (±0.10)	1.42 (±0.15)

(A+ group was classified based on Gaussian Mixture Model, cutoff: 1.30)

PET IMAGING

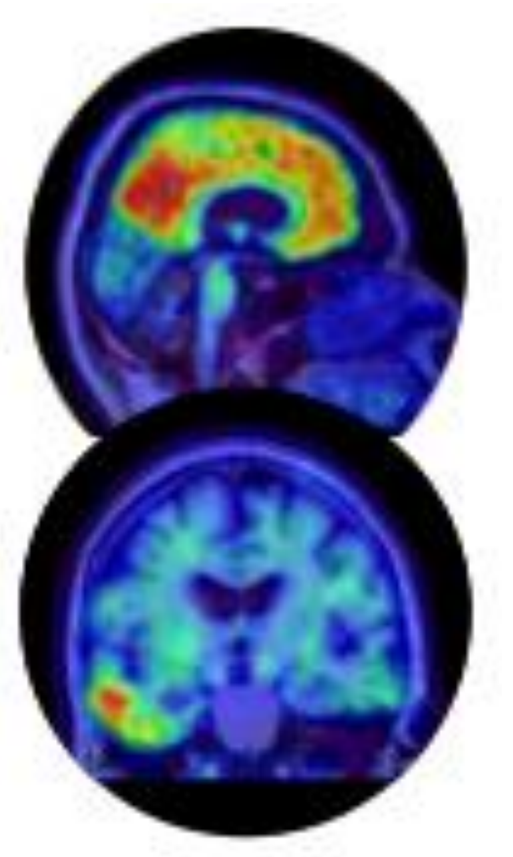

Aβ

[18F]NAV4694

(ref.region: cerebellum GM)

SUVR

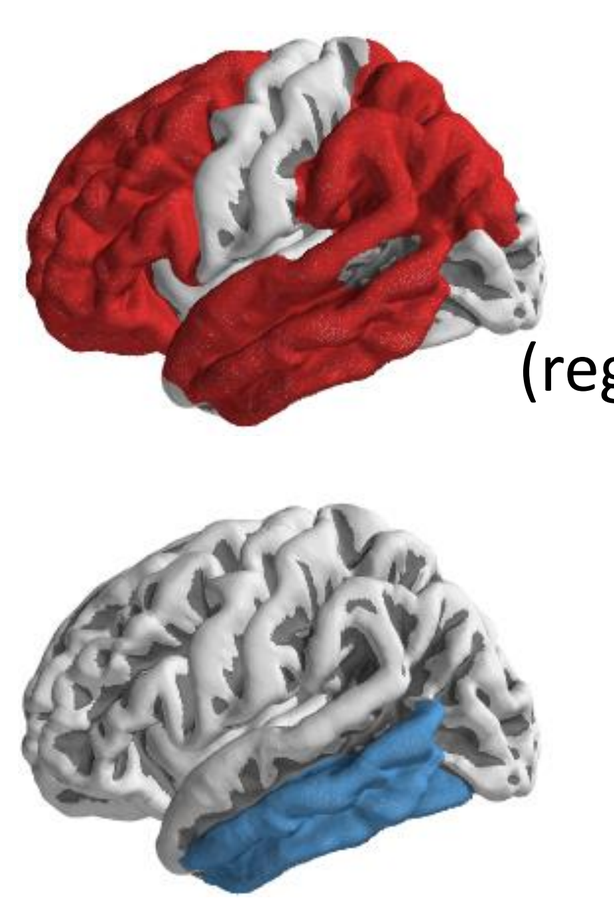

40 cortical GM regions

(regions for calculating global amyloid)

tau

[18F] AV-1451

(ref.region: inferior cerebellum GM)

SUVR

12 cortical GM regions

(meta-ROI)

Standardized uptake value ratio (SUVR) were extracted in regions from Freesurfer Desikan atlas.

DIFFUSION-WEIGHTED MRI

Diffusion-weighted MRI (multi-shell): 64 directions, b = [0, 300, 1000, 2000] s/mm^2

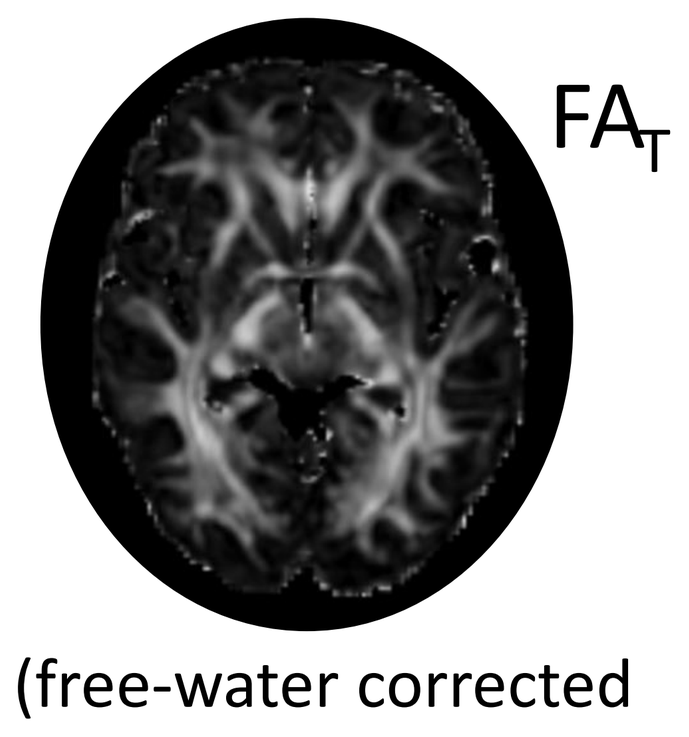
FA_T

(free-water corrected DTI derived measure)

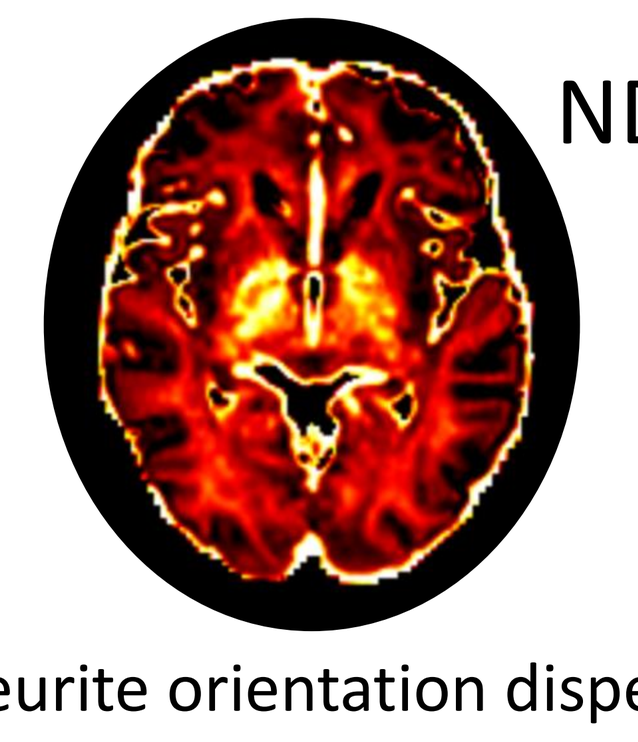
NDI

(neurite orientation dispersion and density imaging derived measure)

FA_T and NDI were used to represent white matter integrity and mapped on each bundle

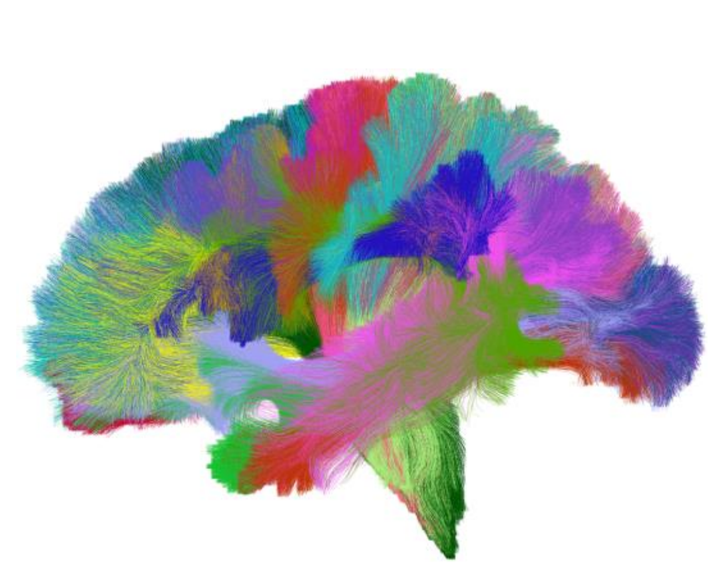

(49 bundles were extracted using TractSeg)

ANALYTICAL METHODS

- MULTIVARIATE ANALYSES

Partial least square (PLS) analyses were performed for both the whole sample and Aβ positive (A+) group

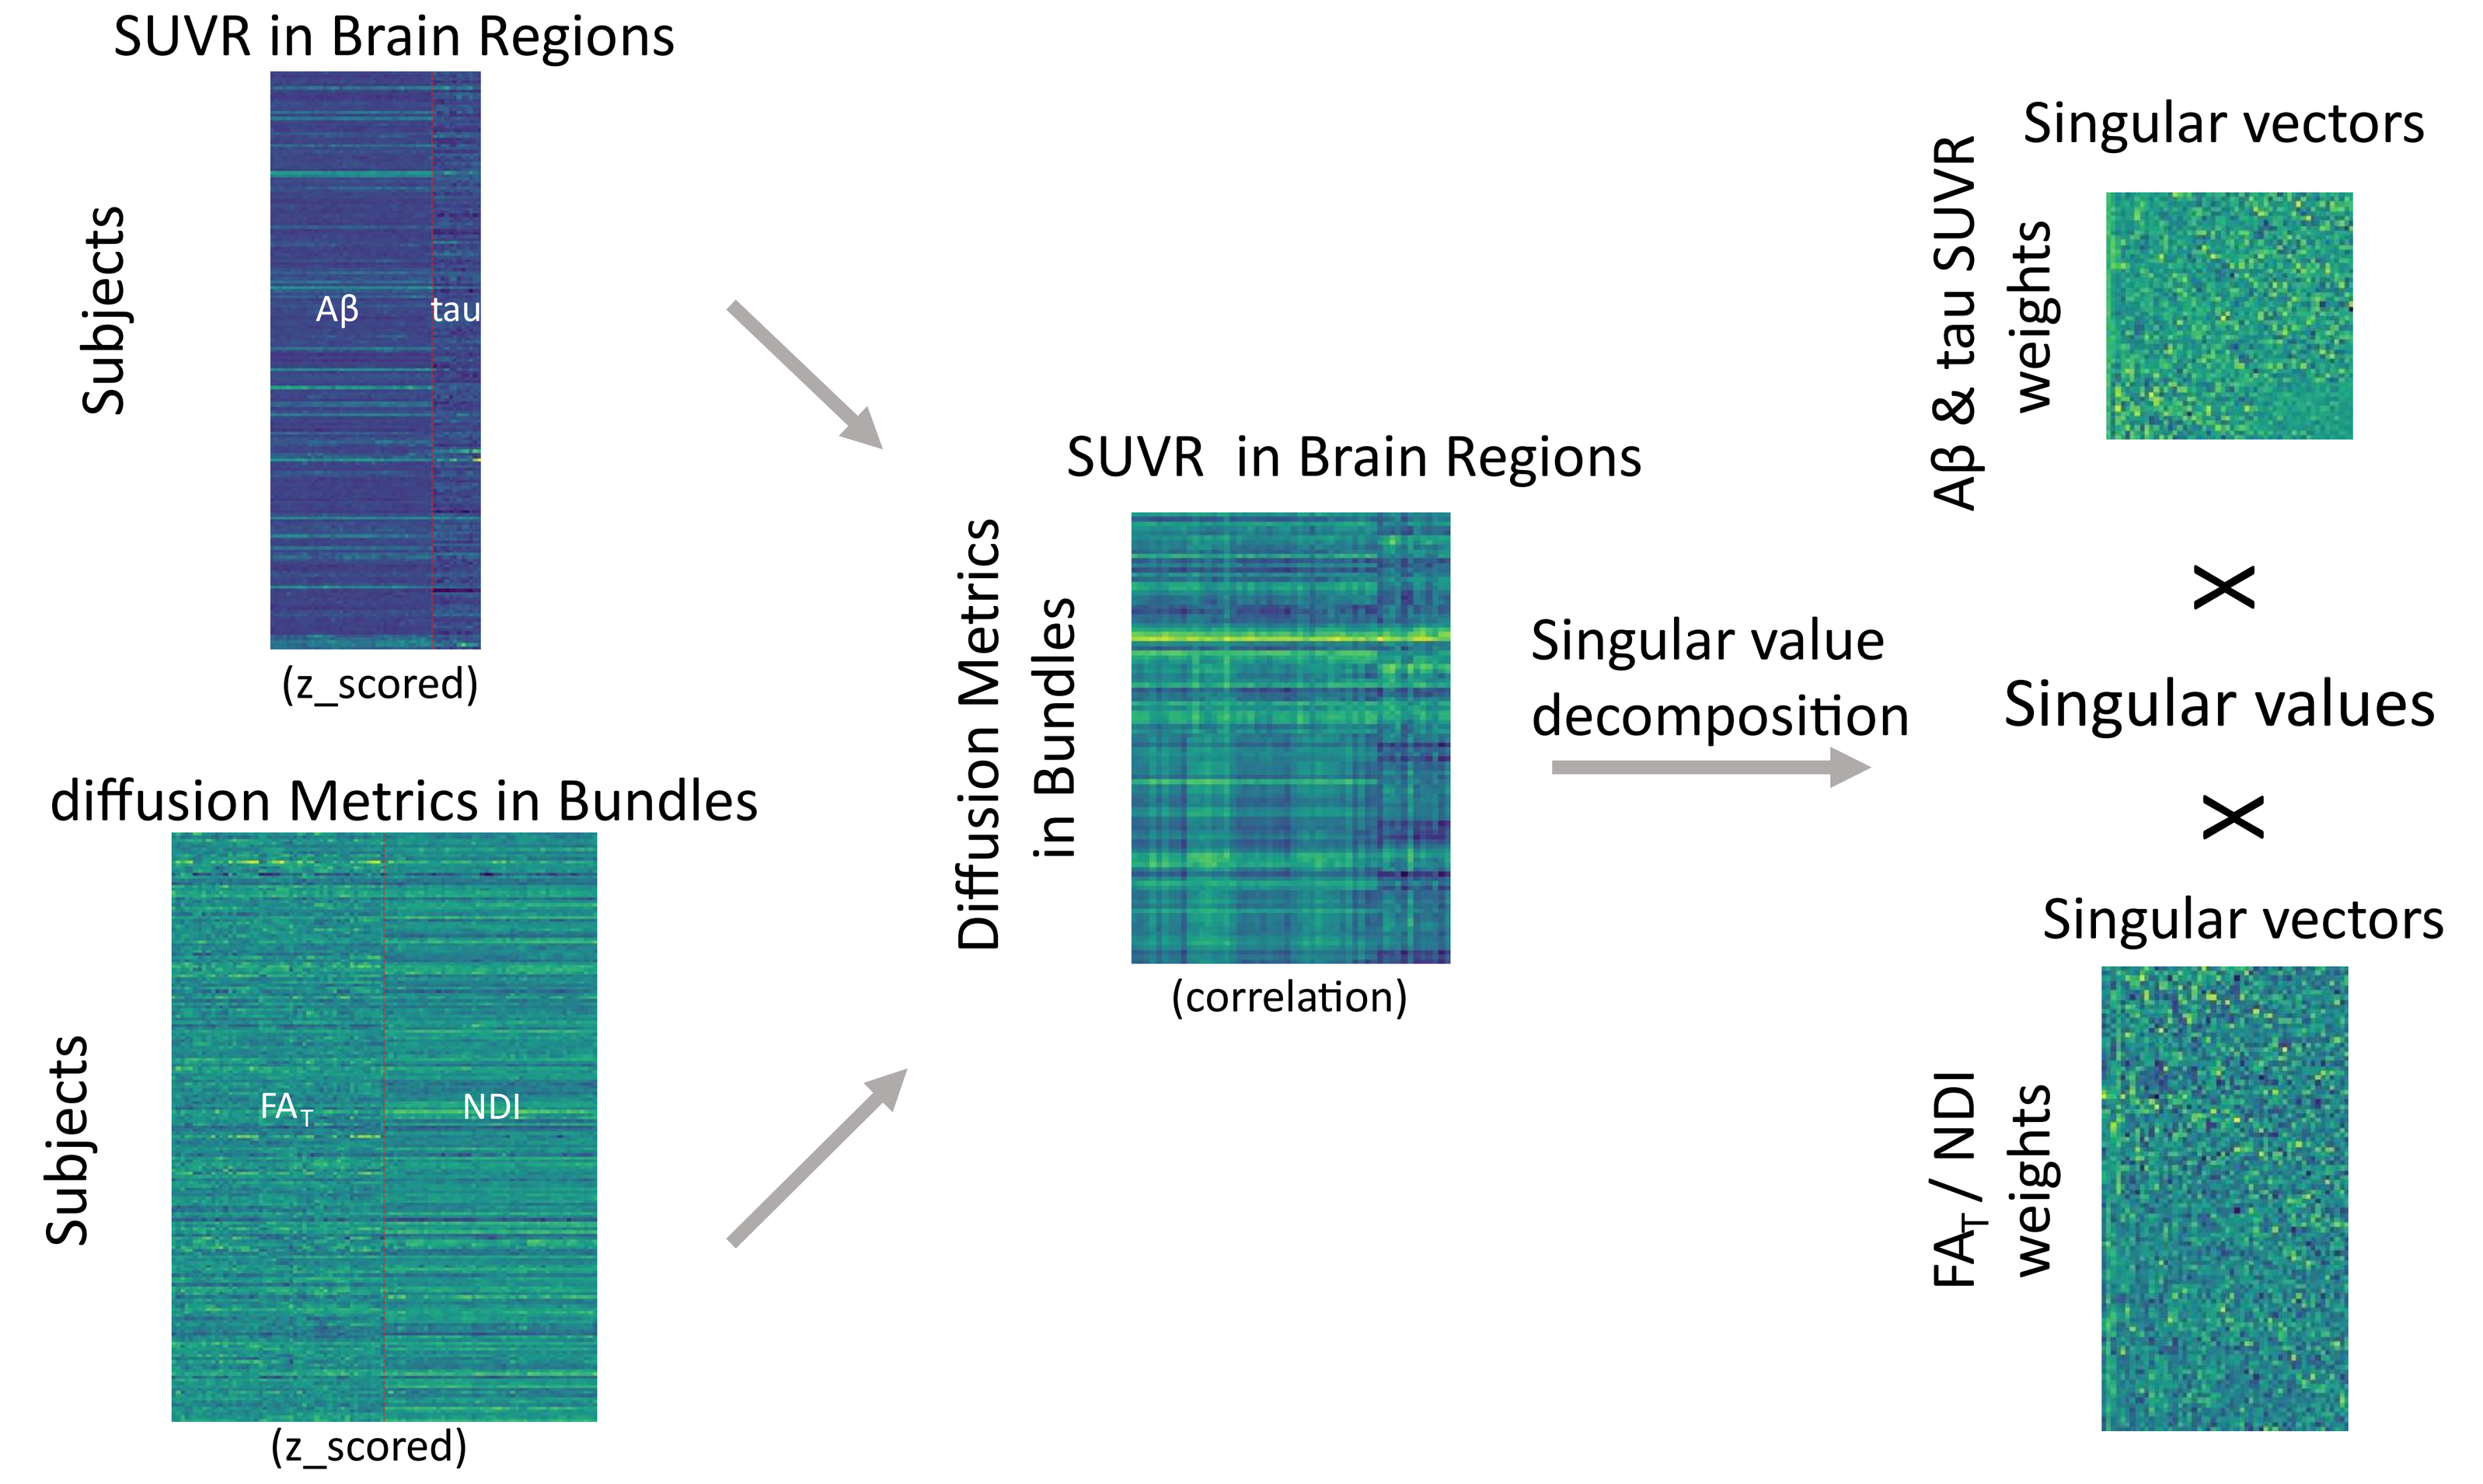
SUVR in Brain Regions
Subjects
Aβ
tau
(z_scored)
diffusion Metrics in Bundles
Subjects
FA_T
NDI
(z_scored)
SUVR in Brain Regions
Diffusion Metrics in Bundles
(correlation)
Singular value decomposition
Singular vectors
Aβ & tau SUVR weights
×
Singular values
×
Singular vectors
FA_T / NDI weights

- UNIVARIATE ANALYSES

Linear regression models were performed to examine the association between global Aβ SUVR (or meta-ROI tau SUVR) and diffusion measures (i.e., FA_T, NDI) in each bundle for both the whole sample and Aβ positive (A+) group.

RESULTS- PLS ANALYSIS (whole sample)

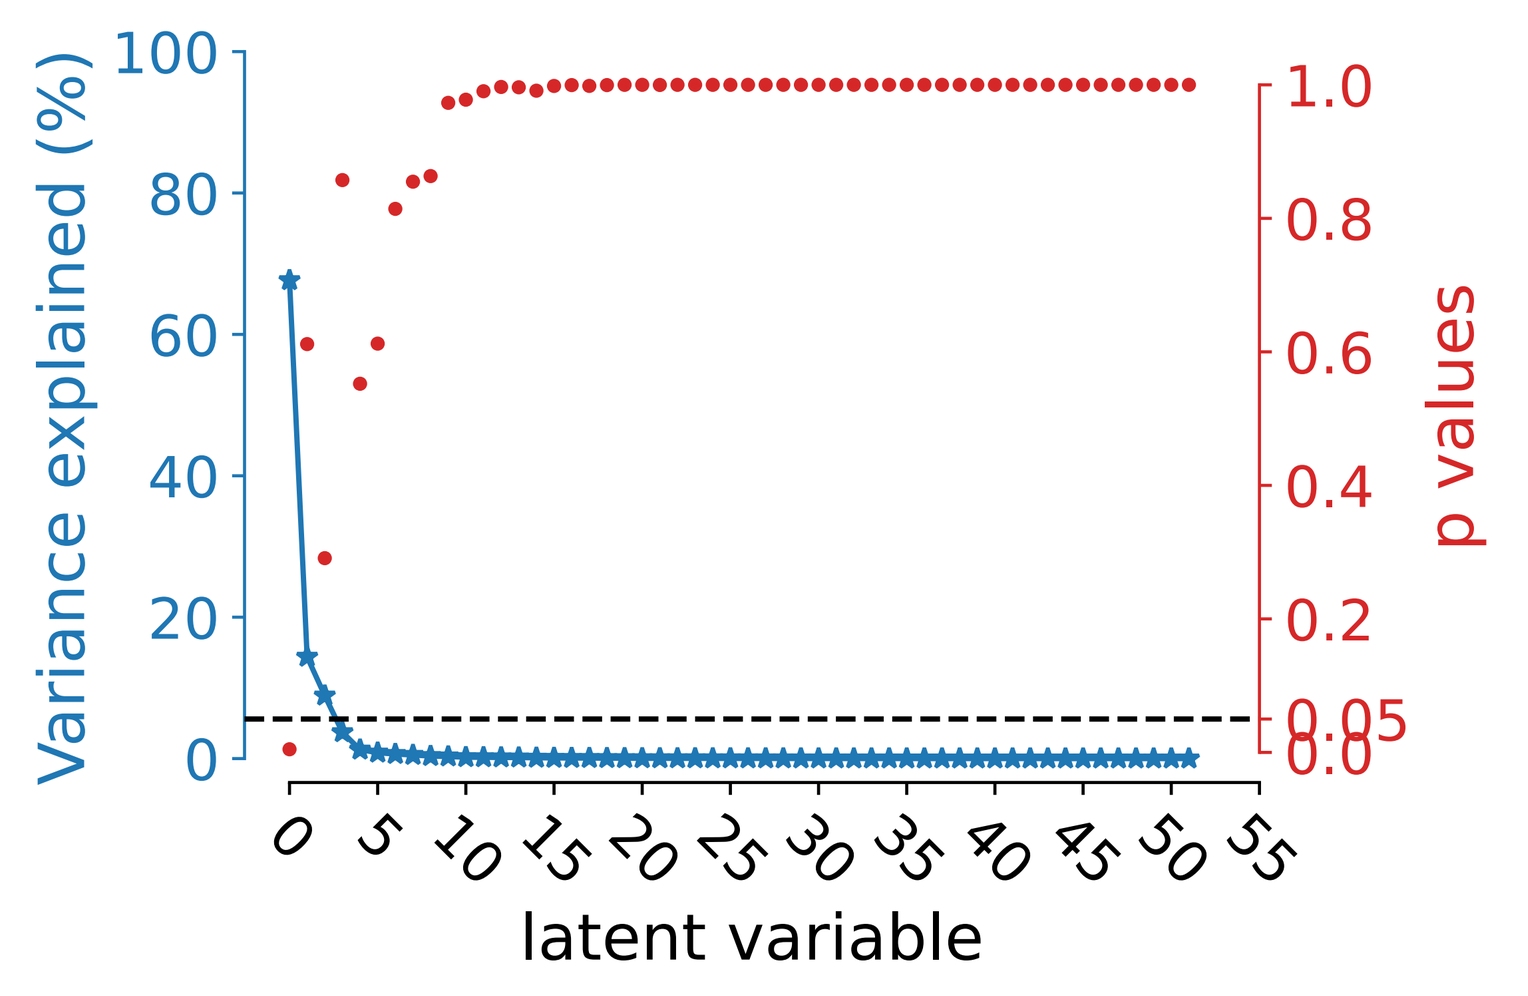
Variance explained (%)
p values
latent variable
0.05

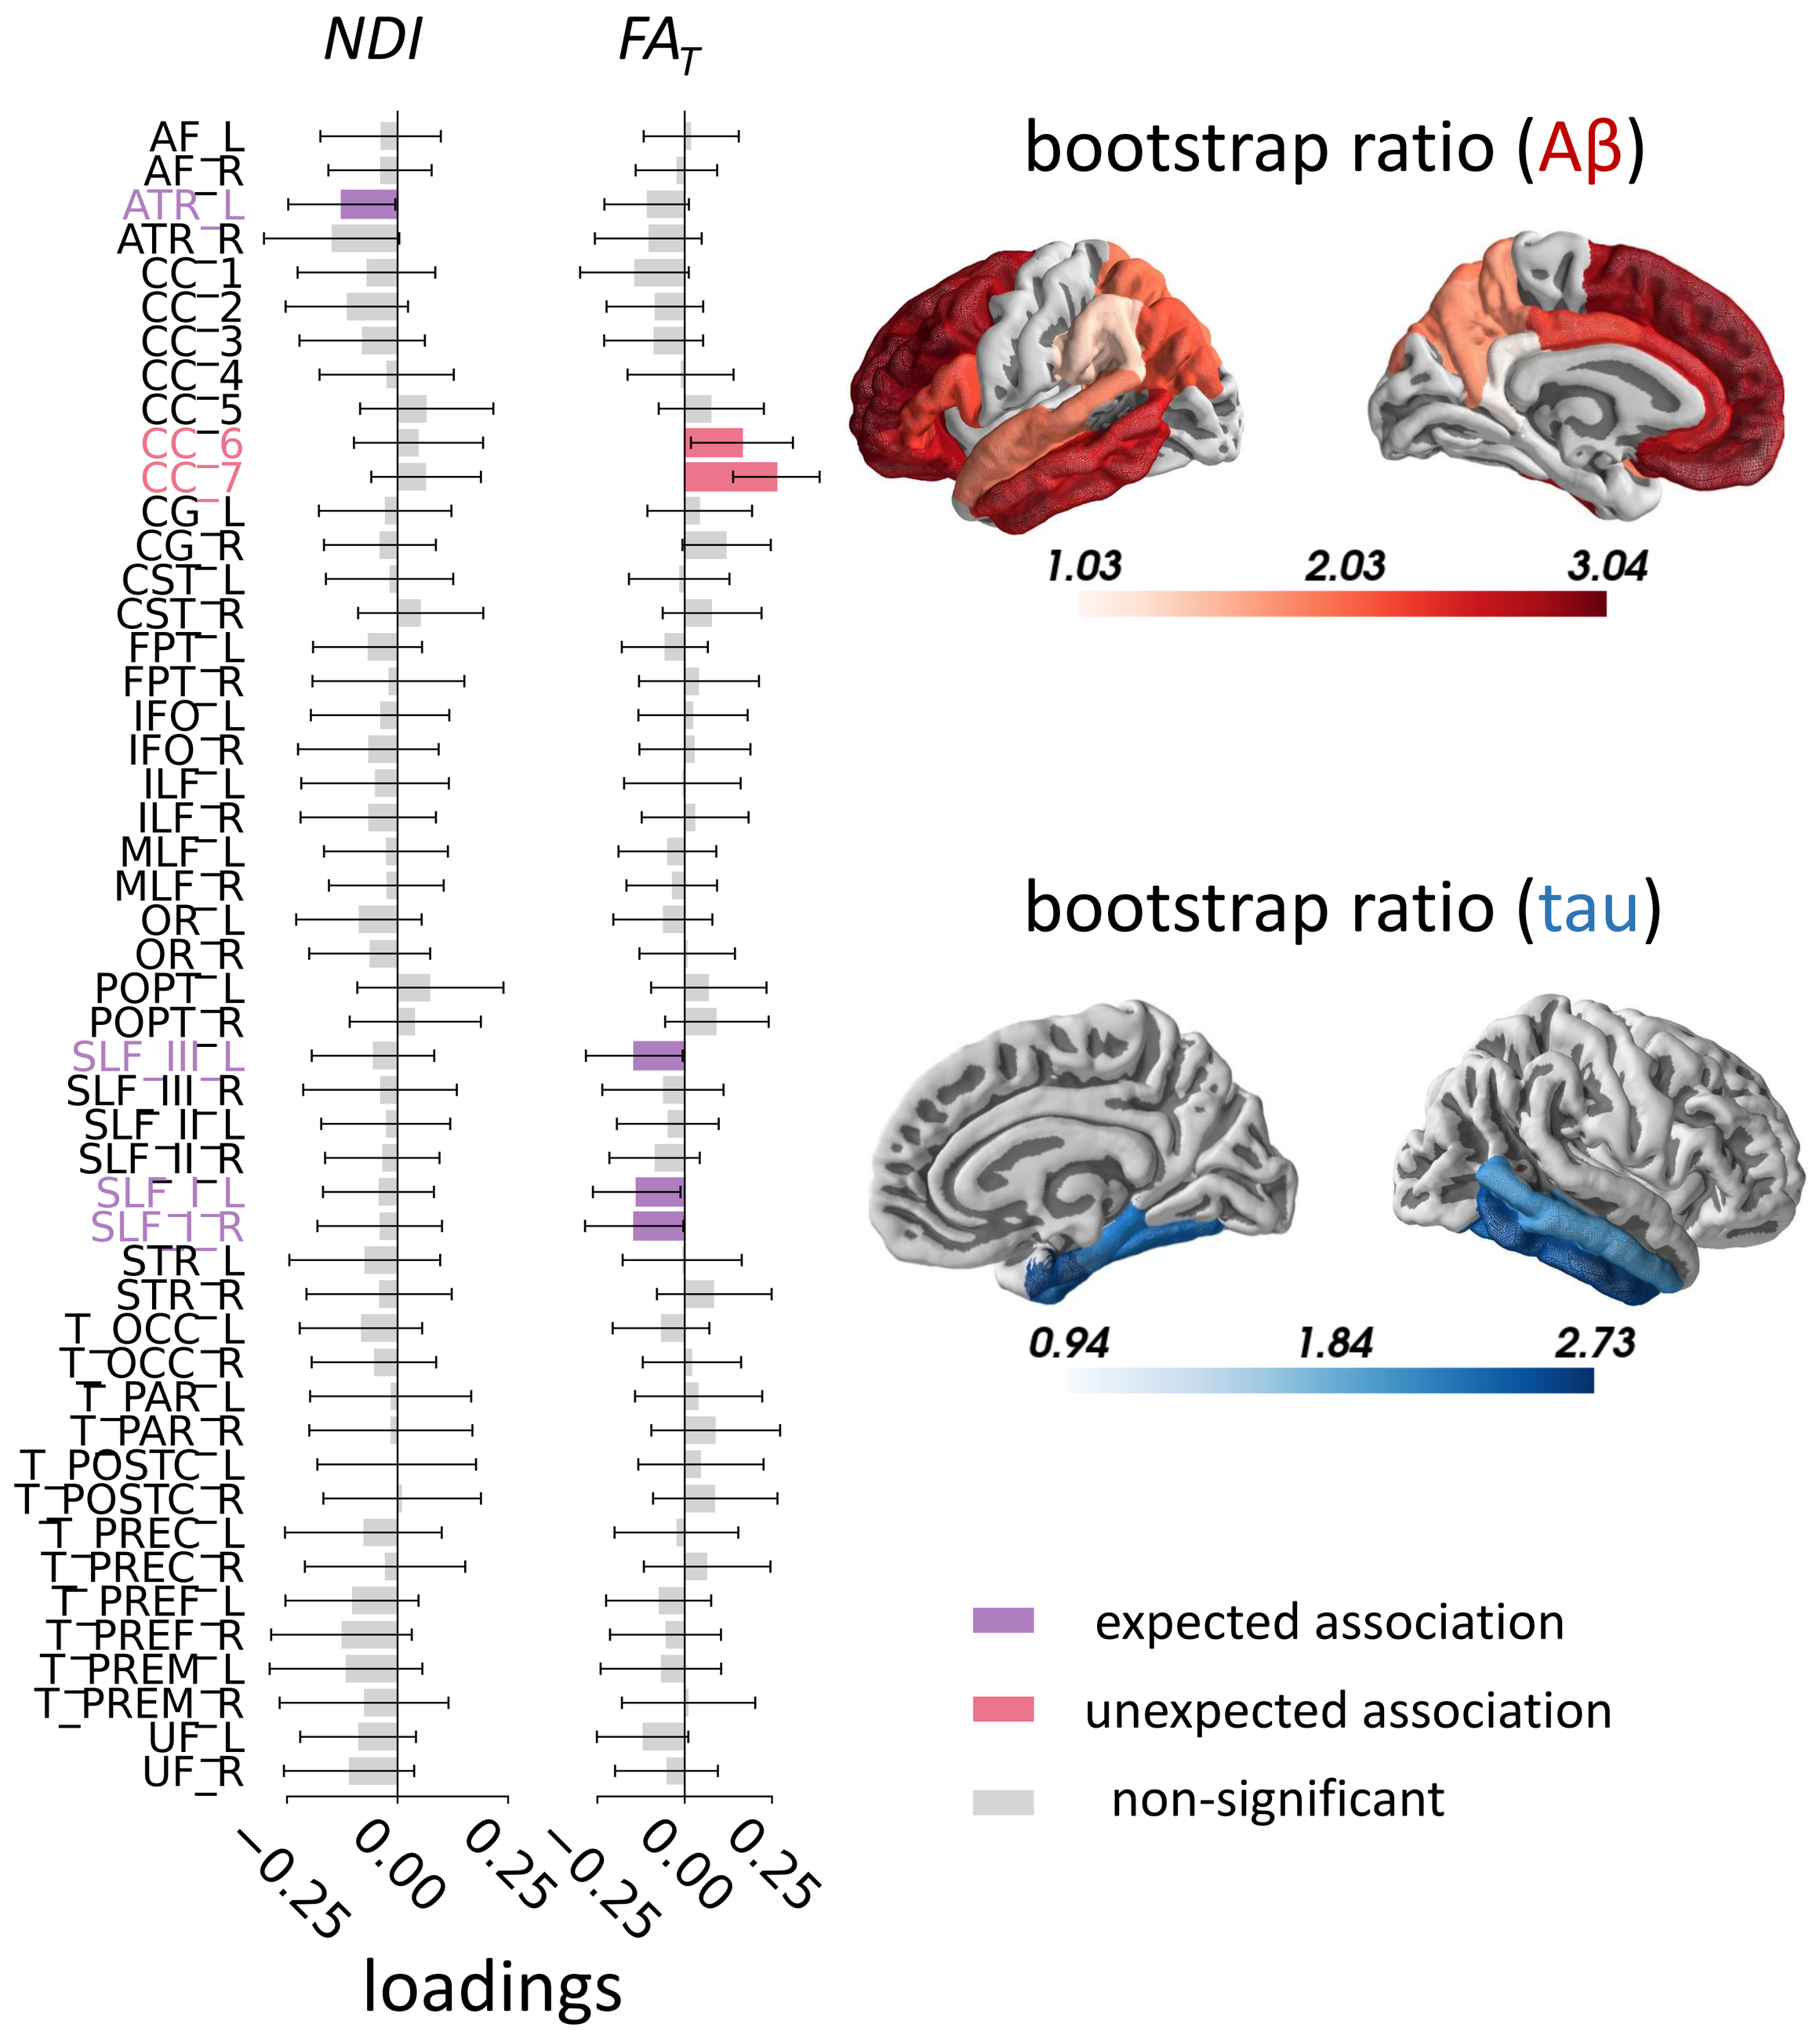
NDI
FA_T
loadings
bootstrap ratio (Aβ)
1.03
2.03
3.04
bootstrap ratio (tau)
0.94
1.84
2.73
expected association
unexpected association
non-significant

In the whole sample, a significant latent variable linked AD pathology to diffusion measures in brain bundles. Expectedly, higher pathology levels were associated with lower NDI/FA_T in some vulnerable bundles (ATR/SLF), while an unexpected association showed a relationship between greater pathology burden and higher FA_T values in part of CC.

RESULTS- PLS ANALYSIS (A+ group)

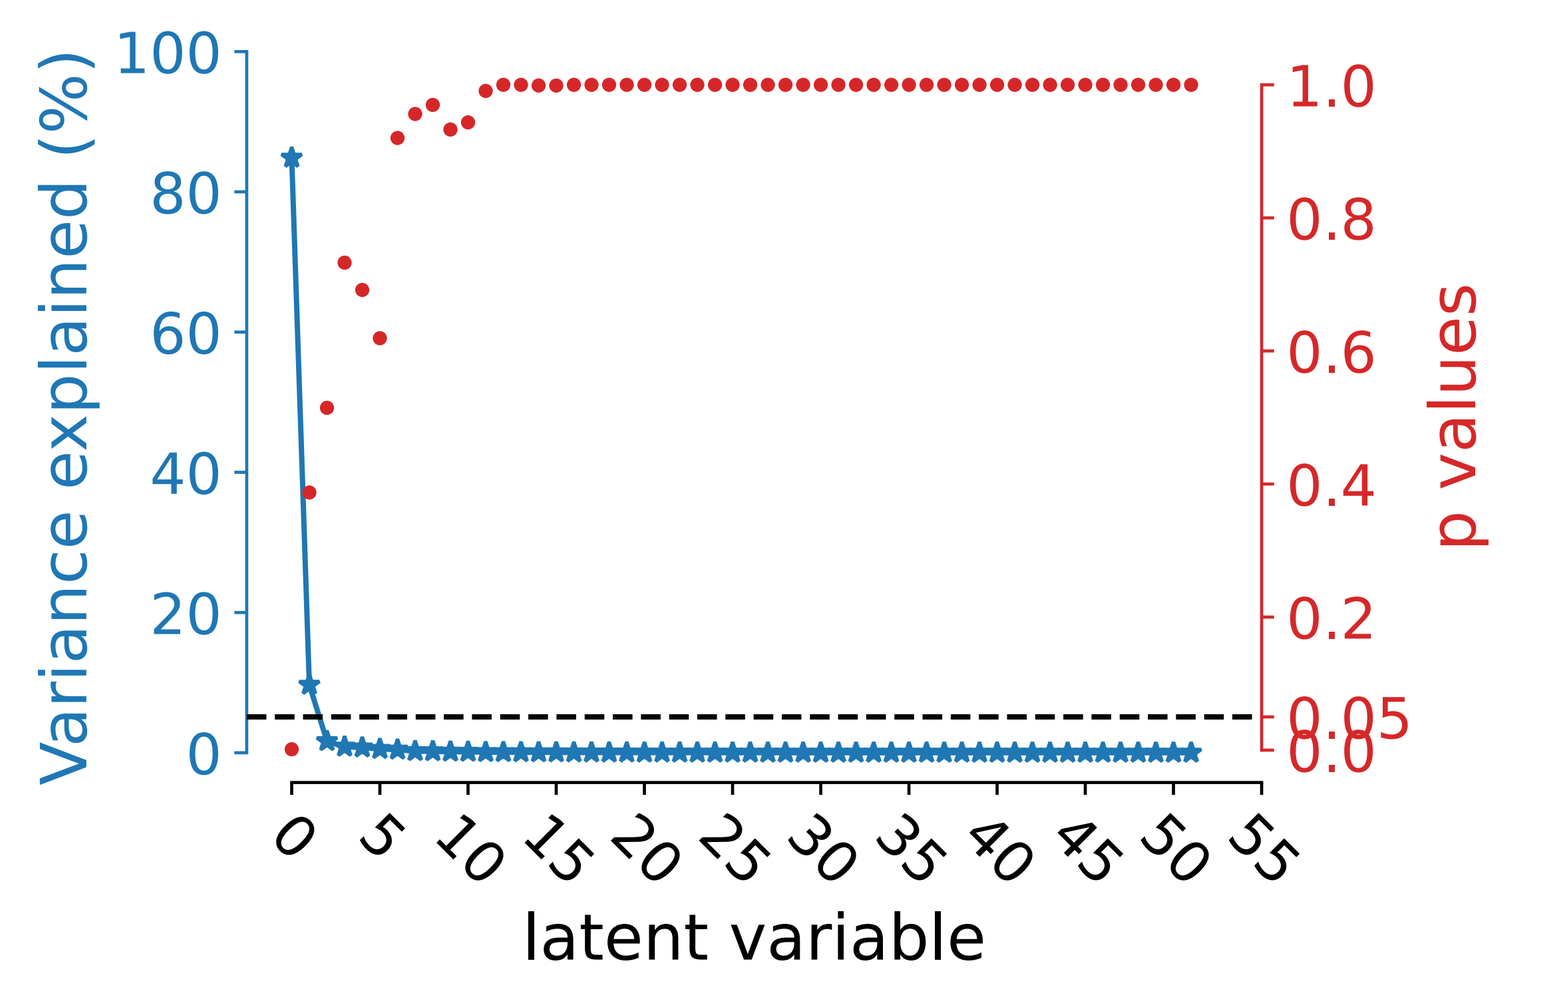
Variance explained (%)
p values
latent variable
0.05

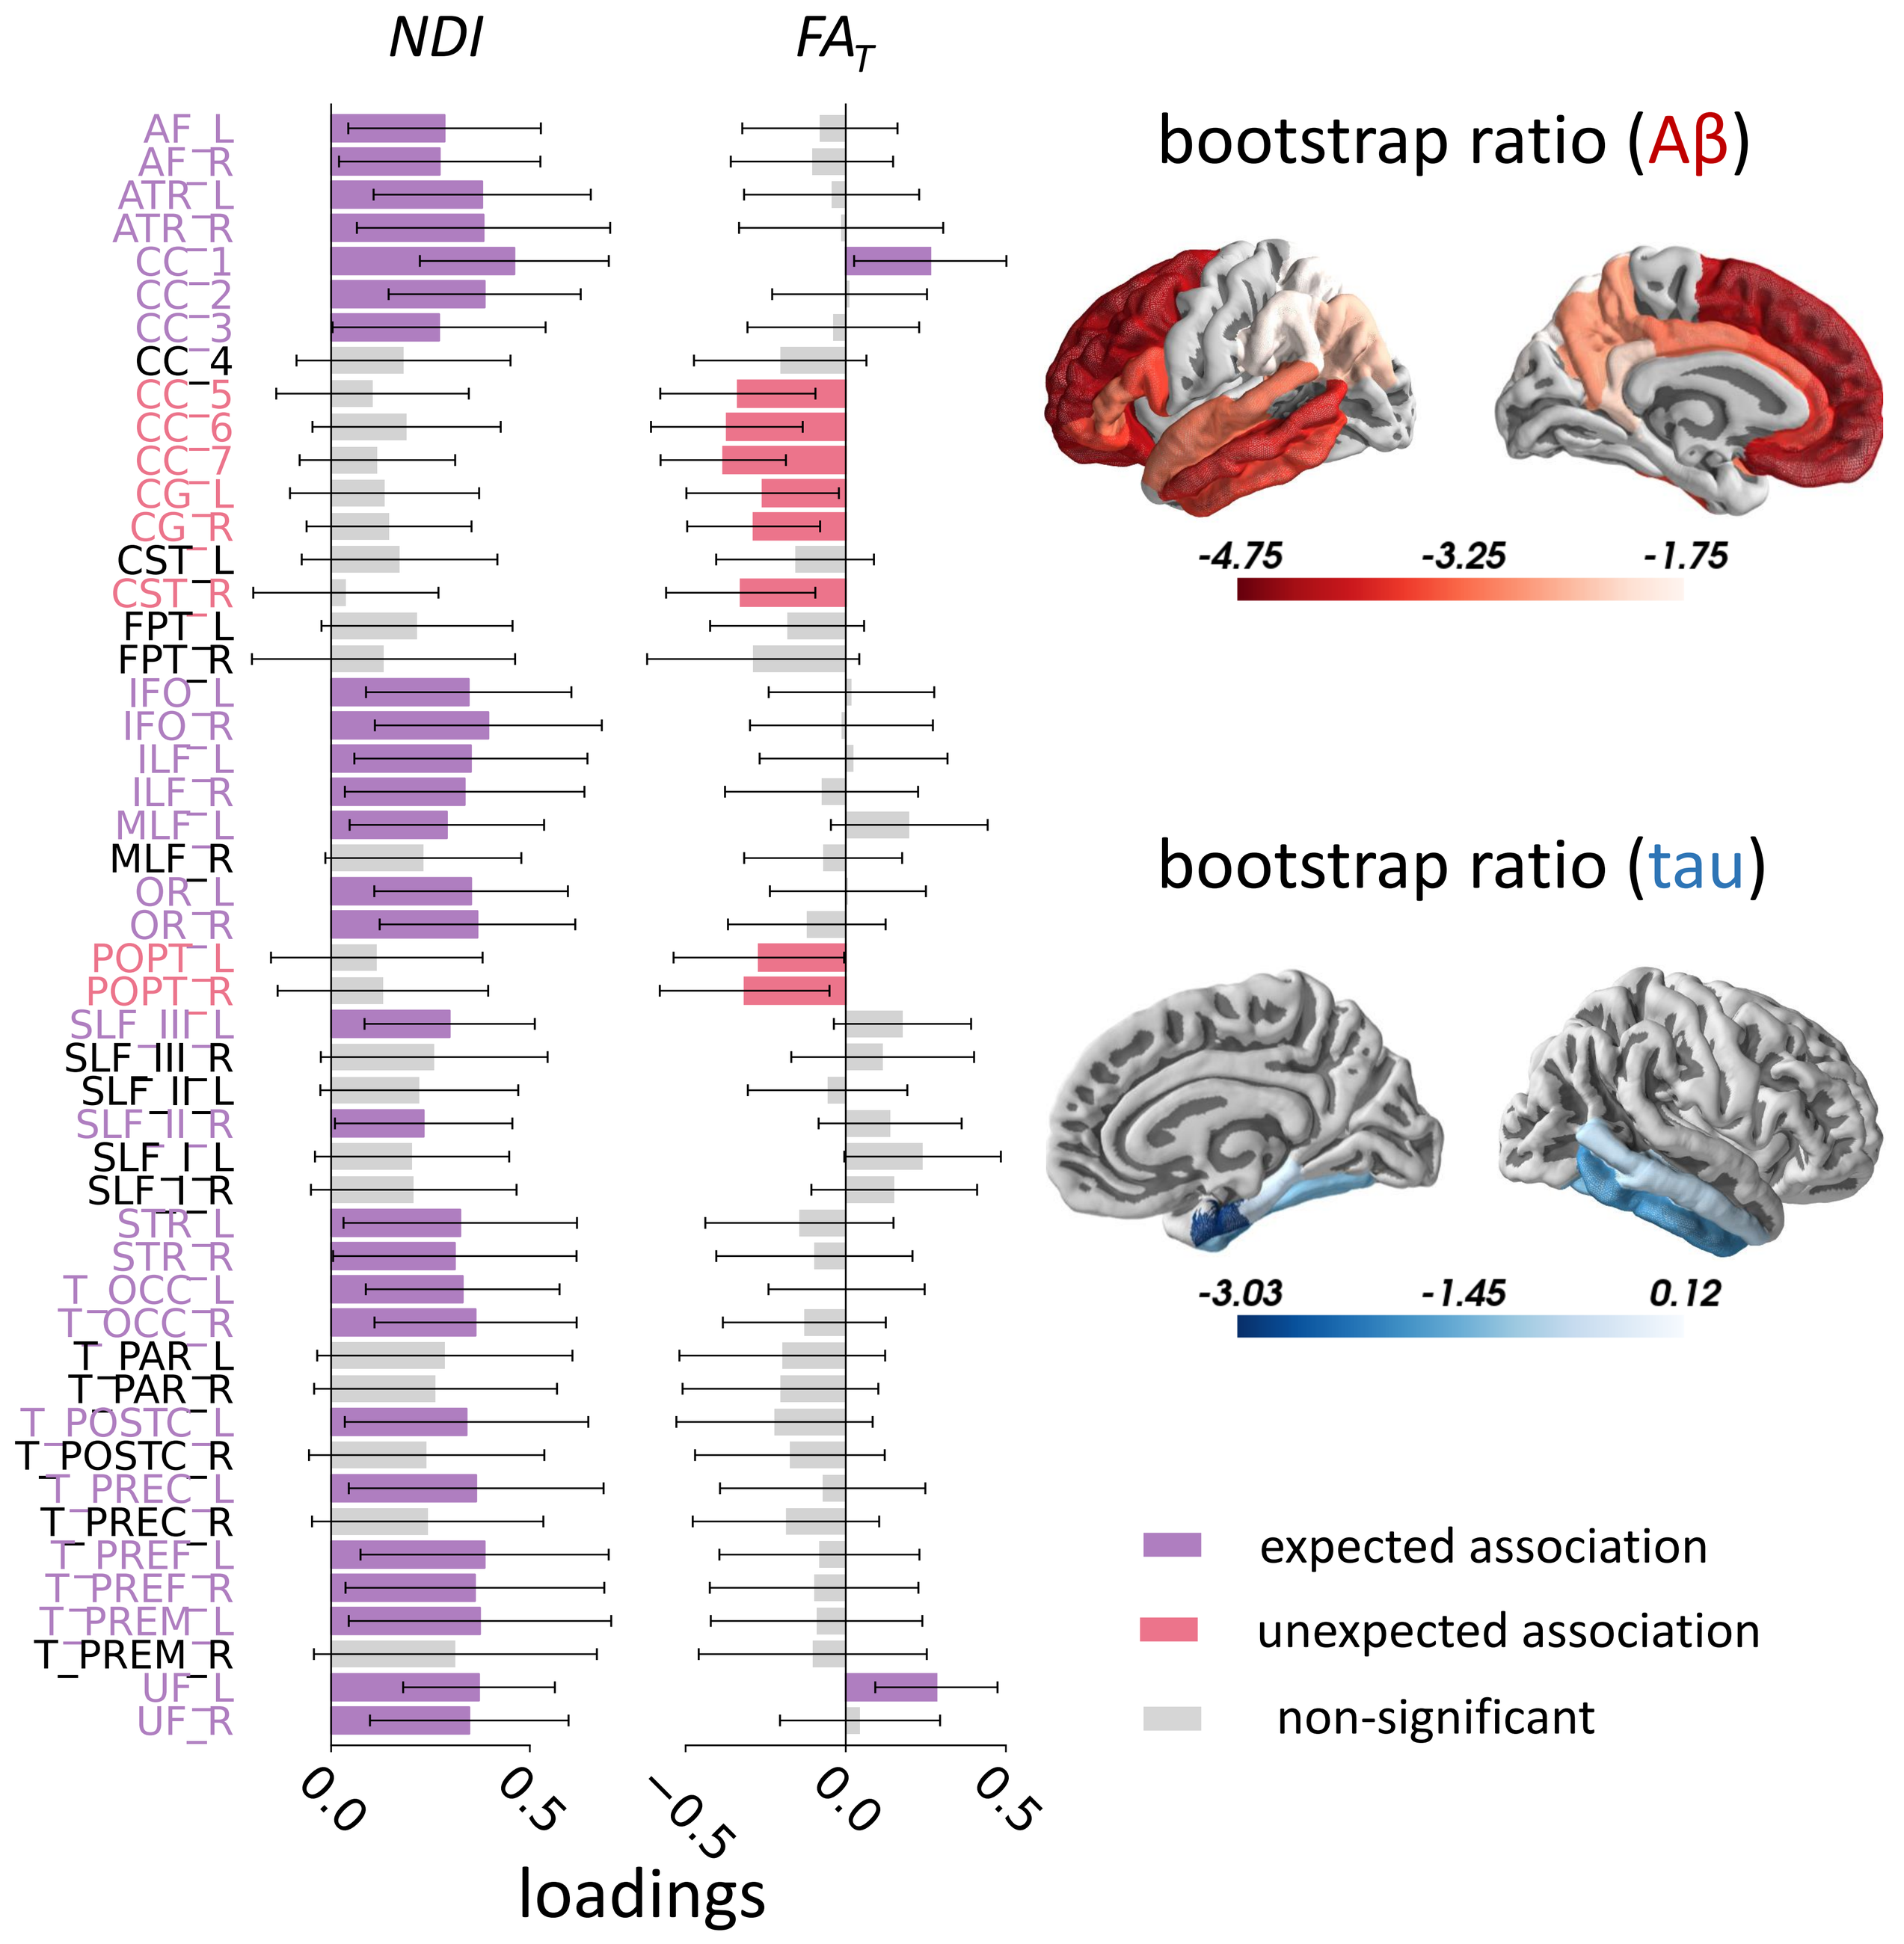
NDI
FA_T
loadings
bootstrap ratio (Aβ)
-4.75
-3.25
-1.75
bootstrap ratio (tau)
-3.03
-1.45
0.12
expected association
unexpected association
non-significant

In the A+ group, we observed one significant latent variable relating AD pathology to diffusion measures in brain bundles. **More vulnerable bundles** were detected contributing to this pattern. Most vulnerable bundles showed a decrease in NDI with higher AD pathology, as expected. However, we observed unexpected associations for FA_T in eight bundles, mainly located in CC or CST bundles.

RESULTS- UNIVARIATE ANALYSES

Diffusion measures (FA_T/NDI) ~ global Aβ

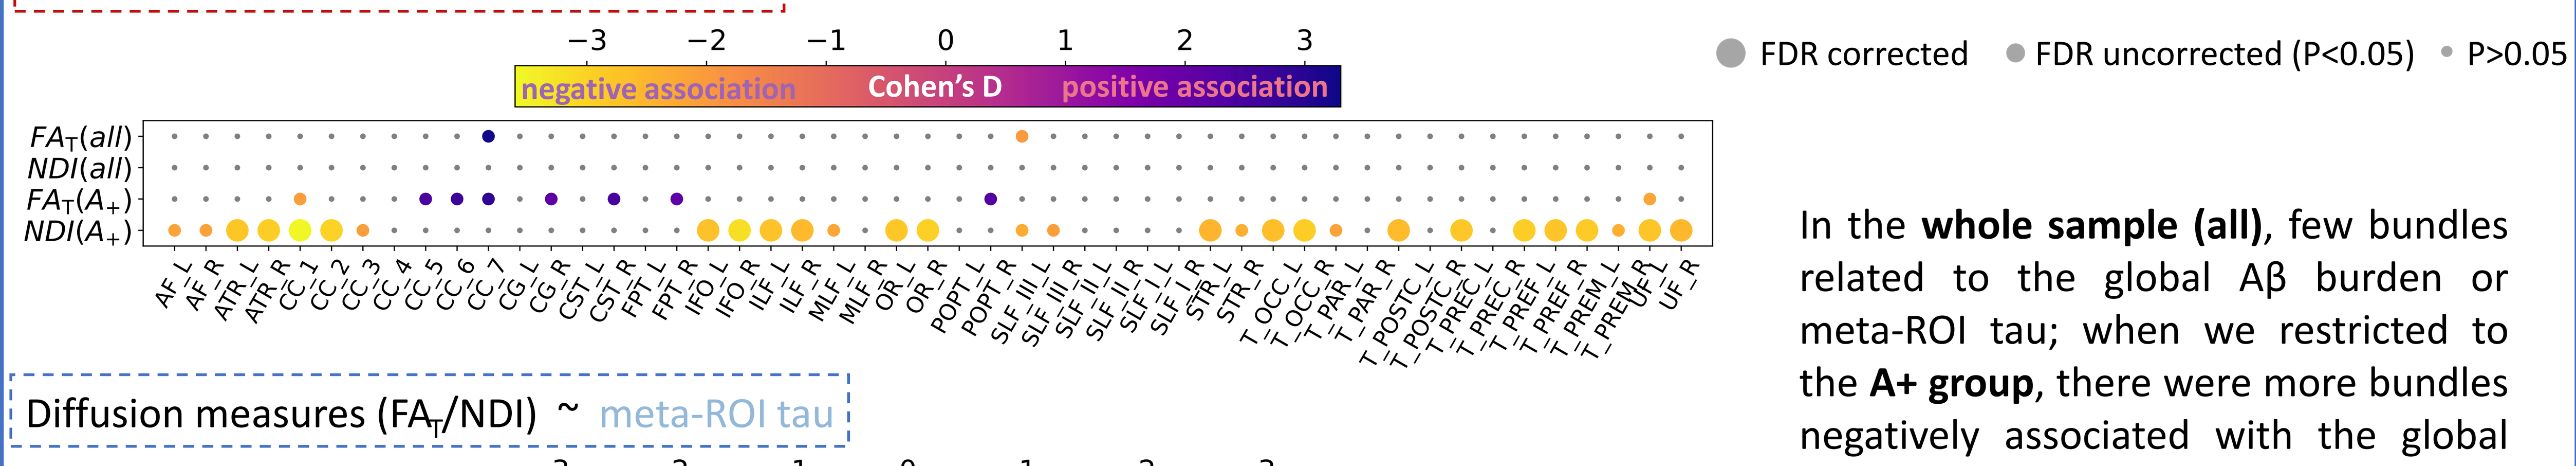
negative association
Cohen's D
positive association
FA_T(all)
NDI(all)
FA_T(A+)
NDI(A+)
FDR corrected
FDR uncorrected (P<0.05)
P>0.05

Diffusion measures (FA_T/NDI) ~ meta-ROI tau

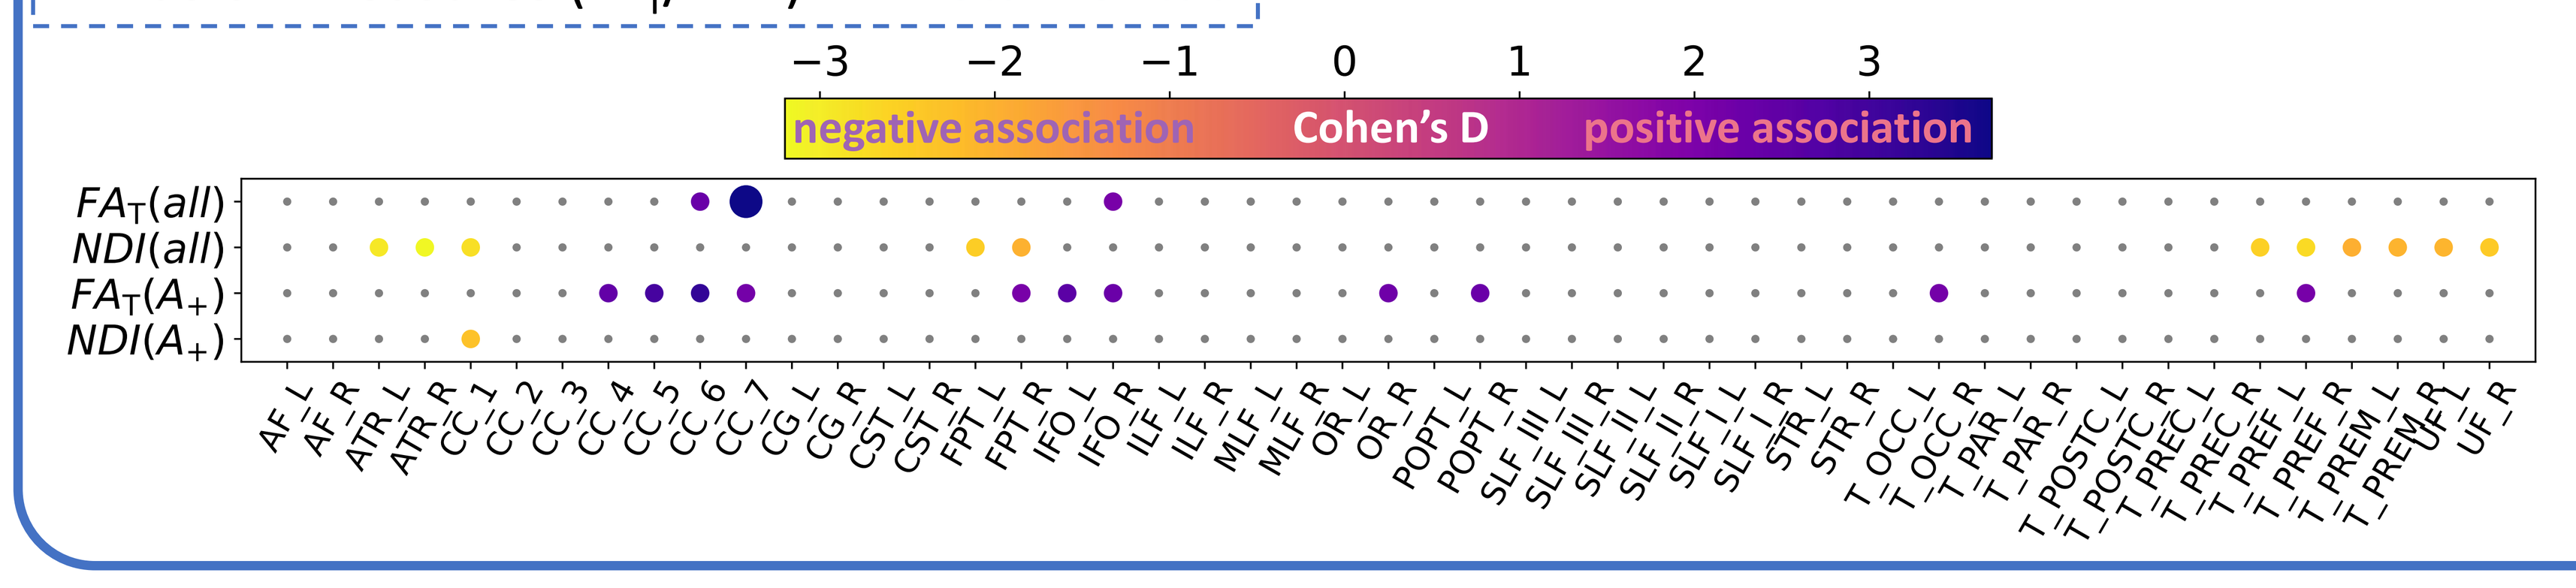
negative association
Cohen's D
positive association
FA_T(all)
NDI(all)
FA_T(A+)
NDI(A+)

In the **whole sample (all)**, few bundles related to the global Aβ burden or meta-ROI tau; when we restricted to the **A+ group**, there were more bundles negatively associated with the global Aβ burden, especially for NDI in those bundles. Similarly, we found higher FA_T in the CC/CST was related to greater AD pathology.

CONCLUSION

We reveal topographical associations between pathology and microstructural alterations in white matter during the preclinical stage of AD. NDI and FA_T offer distinct information about the bundle-specific relationship with Aβ and tau pathology.

These findings emphasize the susceptibility of several WM bundles (e.g. uncinate fasciculus, thalamic bundles, corpus callosum) to the early presence of Aβ and tau, indicating that white matter integrity measures in those bundles have potential as early biomarkers for AD.

Poster downloadable @ villeneuvelab.com ting.qiu@mail.mcgill.ca @Ting12460123