Capturing the spatial extent of tau pathology in the Alzheimer's disease spectrum

Frédéric St-Onge[1,2], Alexa Pichet Binette[3], Marianne Chapleau[4], John CS Breitner[2,5], Sylvia Villeneuve[2,5,6]

1. Integrated program in Neuroscience, Faculty of Medicine, McGill University, Montreal, Canada; 2. Research Center of the Douglas Mental Health University Institute, Montreal Canada; 3. Clinical Memory Research Unit, Faculty of Medicine, Lund University, Lund, Sweden; 4. Faculty of medicine, University of California in San Francisco, United States; 5. Department of Psychiatry, Faculty of Medicine, McGill University, Montreal, Canada; 6. McConnell Brain Imaging Centre, Montreal Neurological Institute, Montreal, Canada

McGill

Douglas CENTRE DE RECHERCHE RESEARCH CENTRE

frederic.st-onge@mail.mcgill.ca

Villeneuve Lab

Multimodal Imaging of the Aging Brain

1. Introduction

Tau progression in the brain follows…

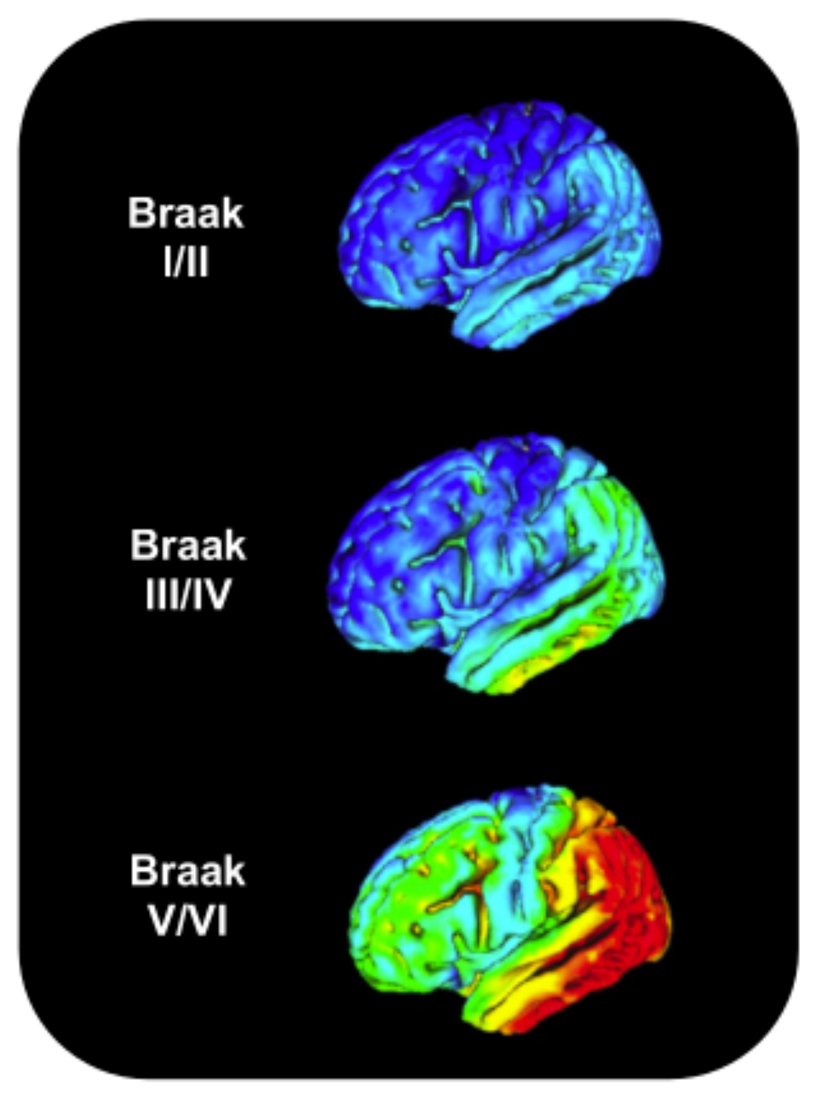

Braak I/II
Braak III/IV
Braak V/VI

?

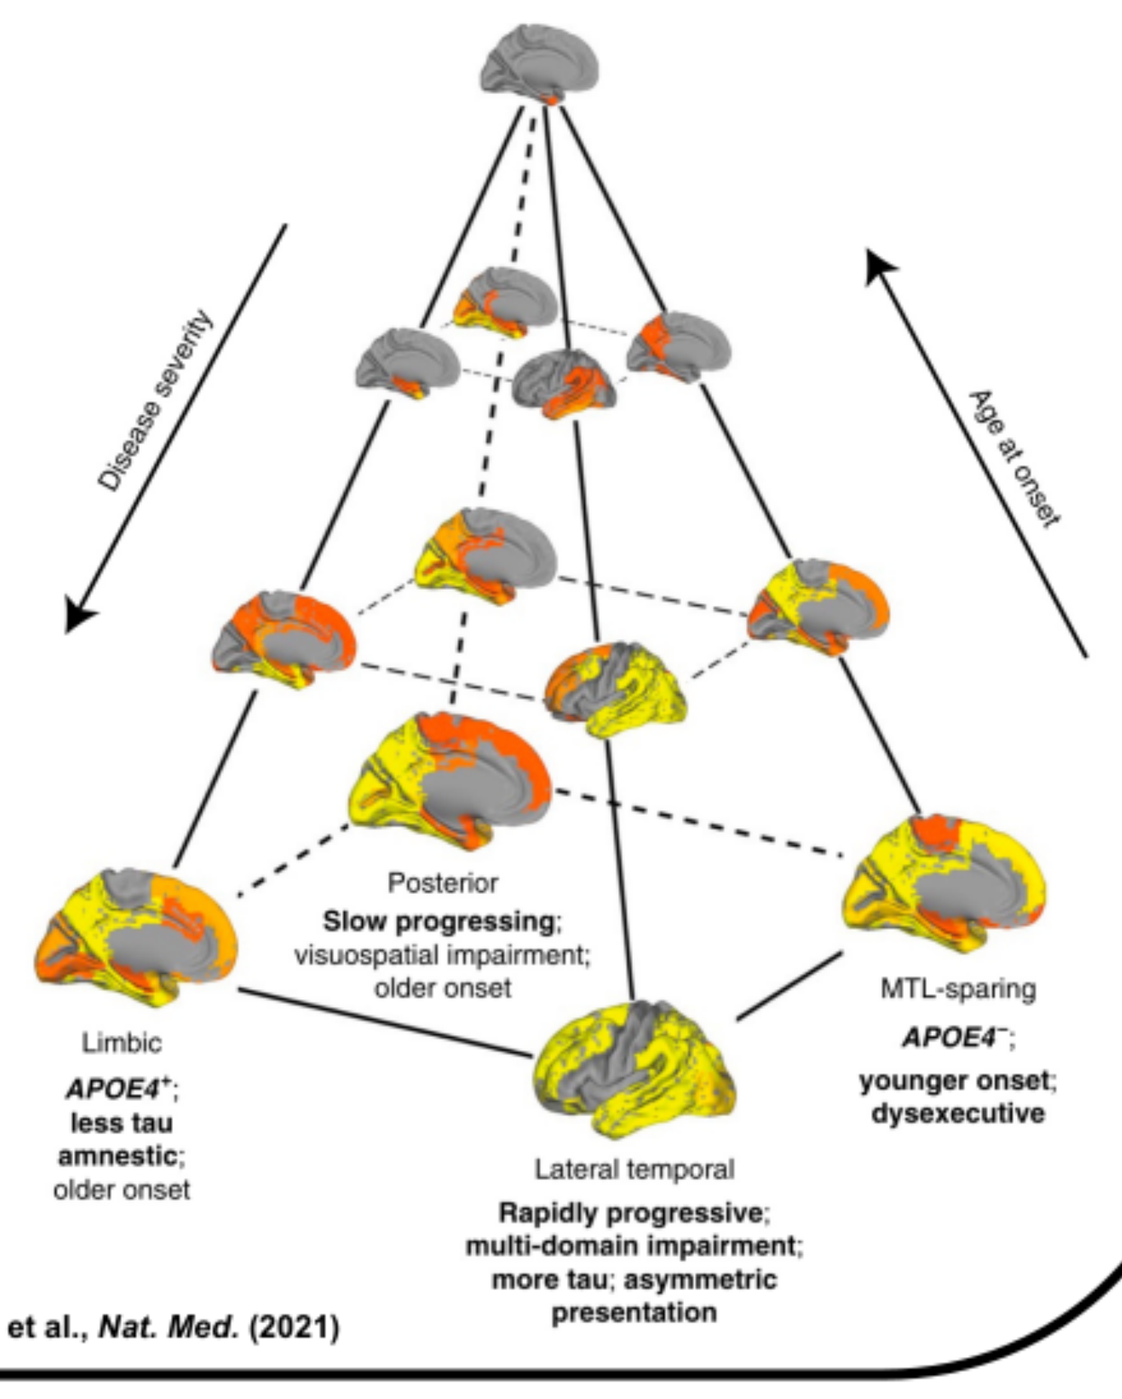

Disease severity
Age at onset
Posterior
Slow progressing; visuospatial impairment; older onset
Limbic
APOE4+; less tau amnestic; older onset
MTL-sparing
APOE4−; younger onset; dysexecutive
Lateral temporal
Rapidly progressive; multi-domain impairment; more tau; asymmetric presentation

Schöll, Lockhart et al., *Neuron* (2016) Vogel et al., *Nat. Med.* (2021)

2. Methods

Spatial Extent Methodology

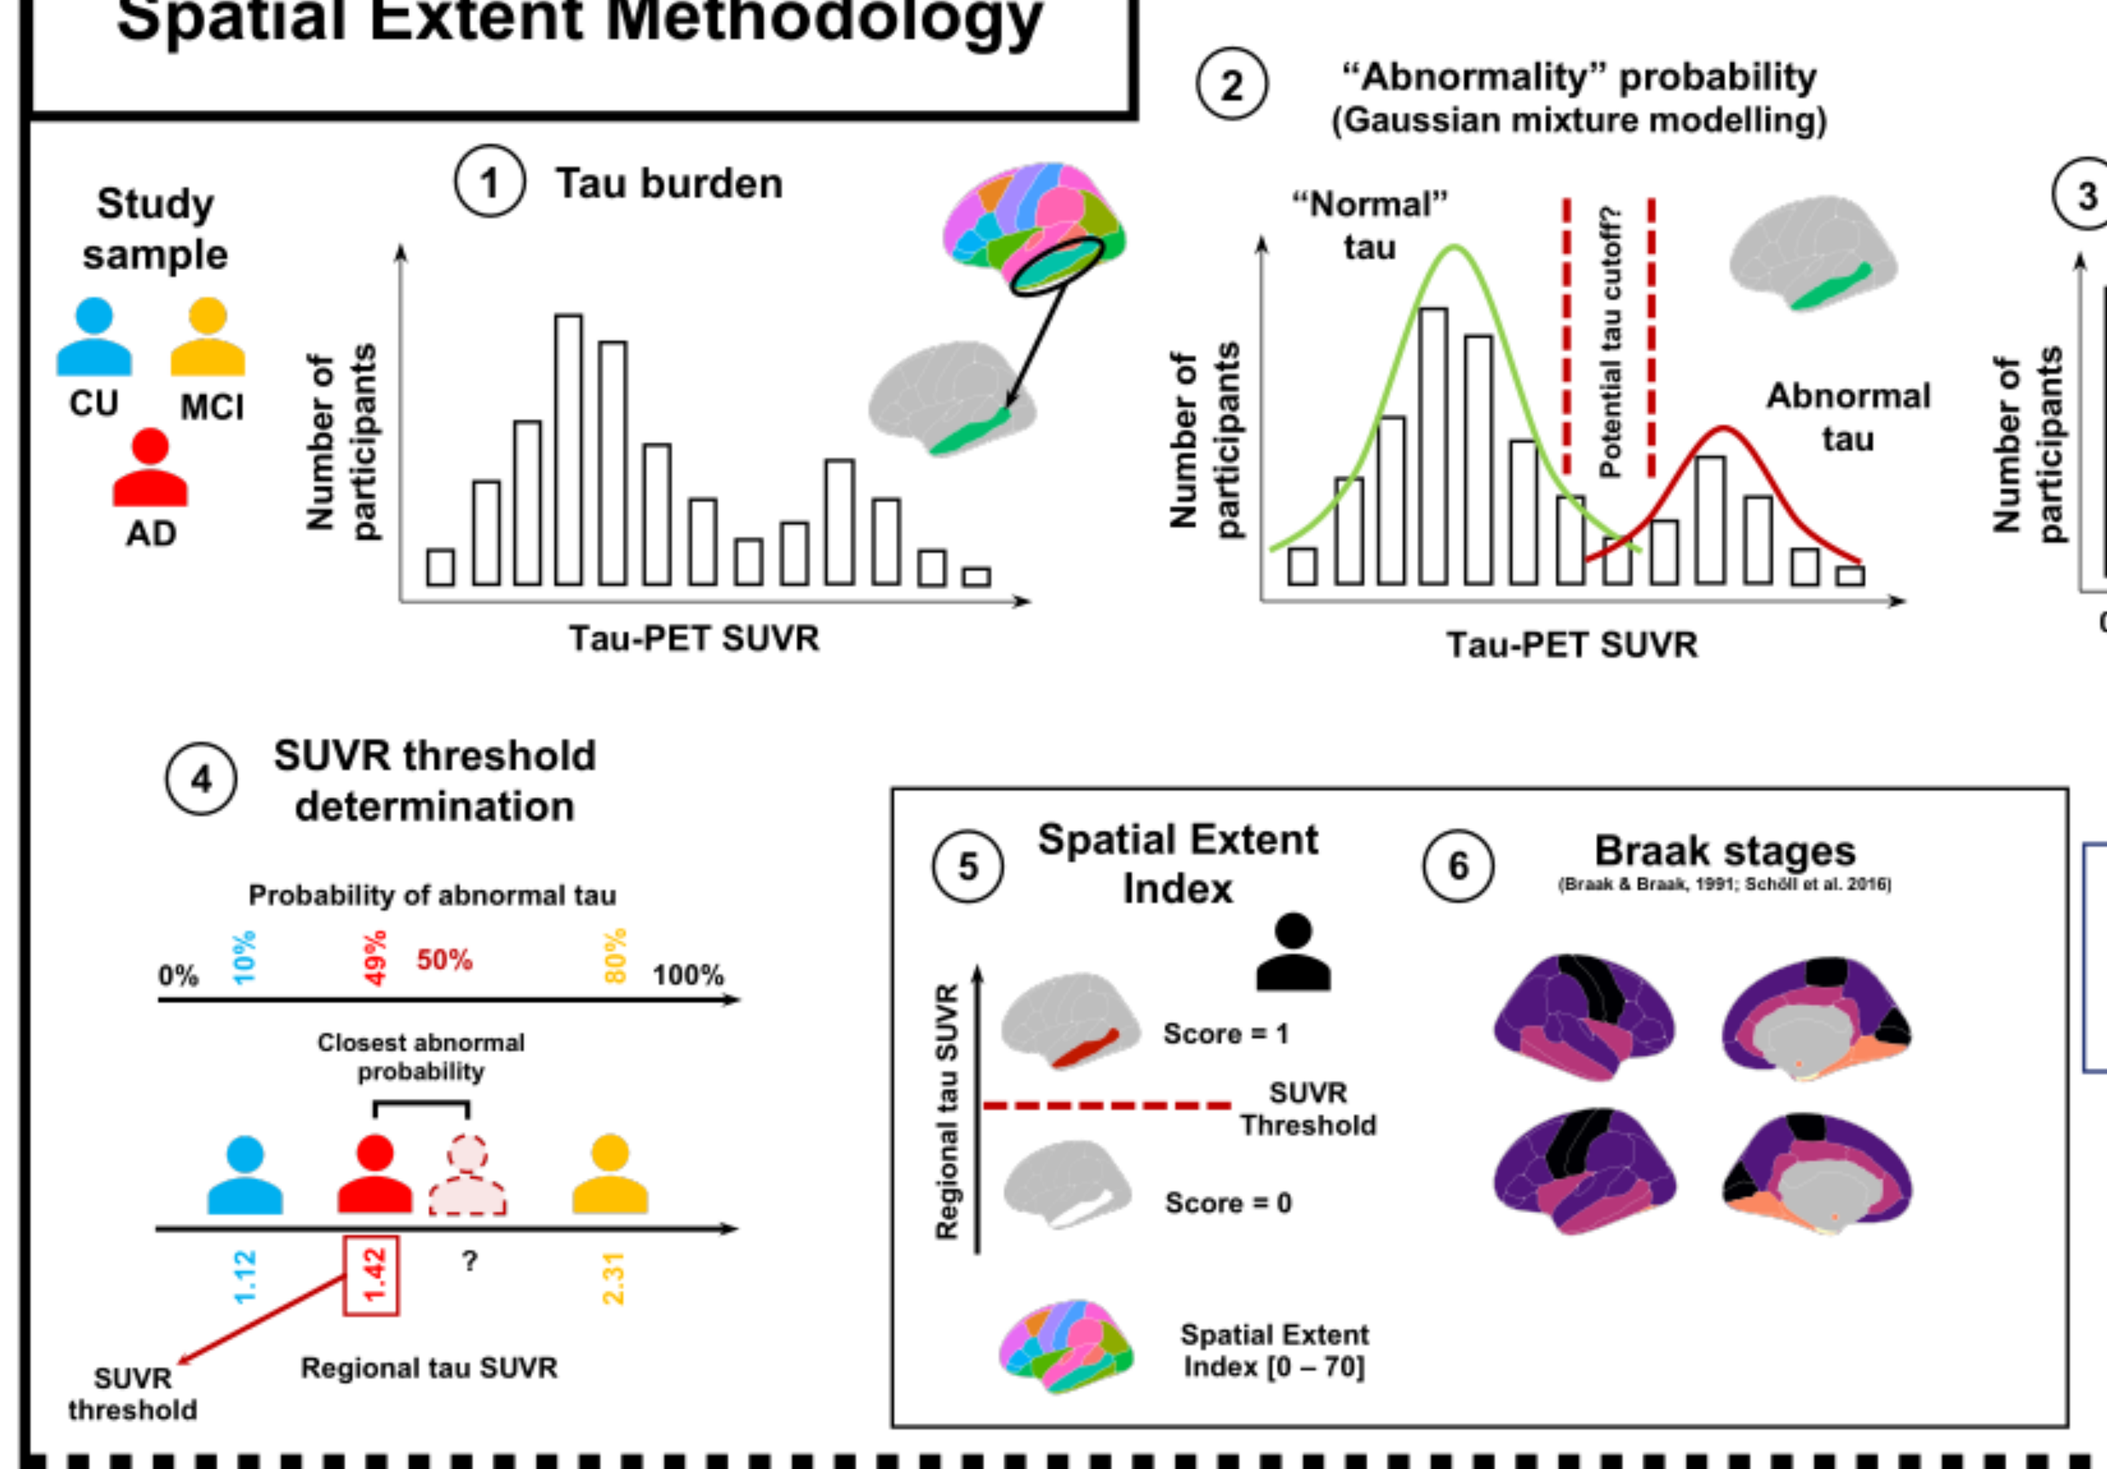

Study sample
CU
MCI
AD
1 Tau burden
Number of participants
Tau-PET SUVR
2 "Abnormality" probability (Gaussian mixture modelling)
"Normal" tau
Potential tau cutoff?
Abnormal tau
3 Abnormal probability threshold
Probability threshold
0%
50%
100%
Probability of abnormal tau
4 SUVR threshold determination
Probability of abnormal tau
0% 10% 49% 50% 80% 100%
Closest abnormal probability
1.12 1.42 ? 2.31
SUVR threshold
Regional tau SUVR
5 Spatial Extent Index
Regional tau SUVR
Score = 1
SUVR Threshold
Score = 0
Spatial Extent Index [0 – 70]
6 Braak stages
(Braak & Braak, 1991; Schöll et al. 2016)

Spatial extent index:
Number of regions at the individual-level with abnormally high levels of tau-PET signal

ADNI Alzheimer's Disease Neuroimaging Initiative

Population

Full cross-sectional sample

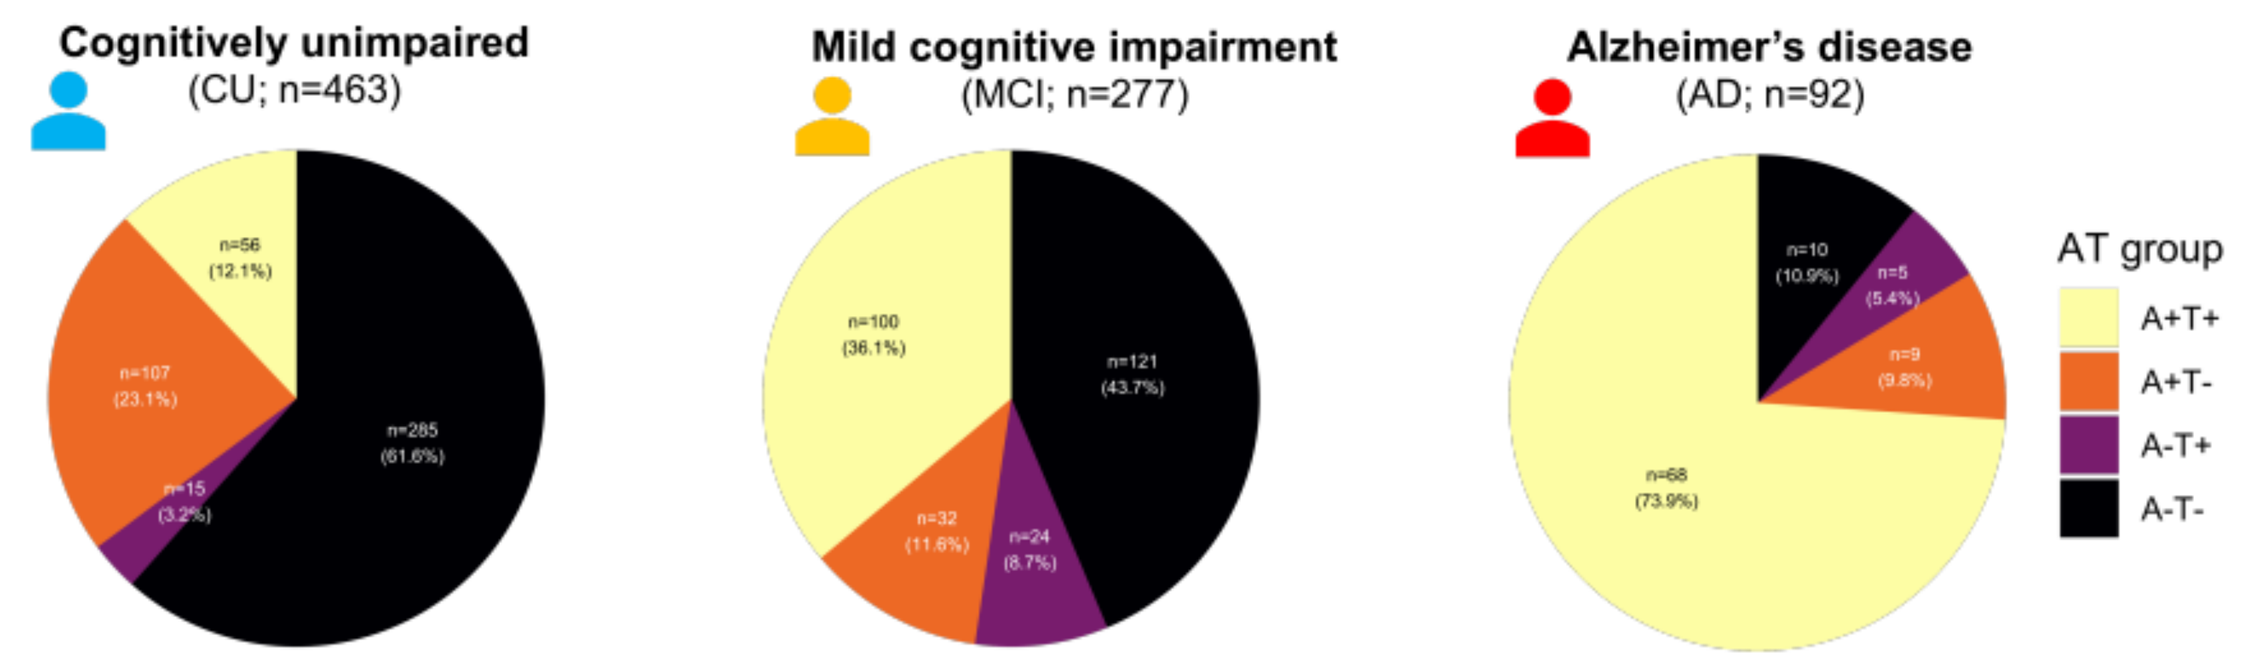

Cognitively unimpaired (CU; n=463)
n=56 (12.1%)
n=107 (23.1%)
n=15 (3.2%)
n=285 (61.6%)
Mild cognitive impairment (MCI; n=277)
n=100 (36.1%)
n=32 (11.6%)
n=24 (8.7%)
n=121 (43.7%)
Alzheimer's disease (AD; n=92)
n=10 (10.9%)
n=5 (5.4%)
n=9 (9.8%)
n=68 (73.9%)
AT group
A+T+
A+T-
A-T+
A-T-

Amyloid-positive sample

	CU (n = 163) (n_{long} = 90)	MCI (n = 132) (n_{long} = 66)	AD (n = 77) (n_{long} = 39)
Age	74.82 years (± 7.57)	74.36 years (± 7.39)	77.35 years (± 8.93)
Sex	58.9% female	49.2% female	41.6% female
APOE4	44.8% APOE4+	53.0% APOE4+	62.3% APOE4+
Education	16.64 years (± 2.34)	15.99 years (± 2.49)	15.55 years (± 2.48)

3. Results

Objective 1: Abnormal progression of tau pathology over time

Figure 1A

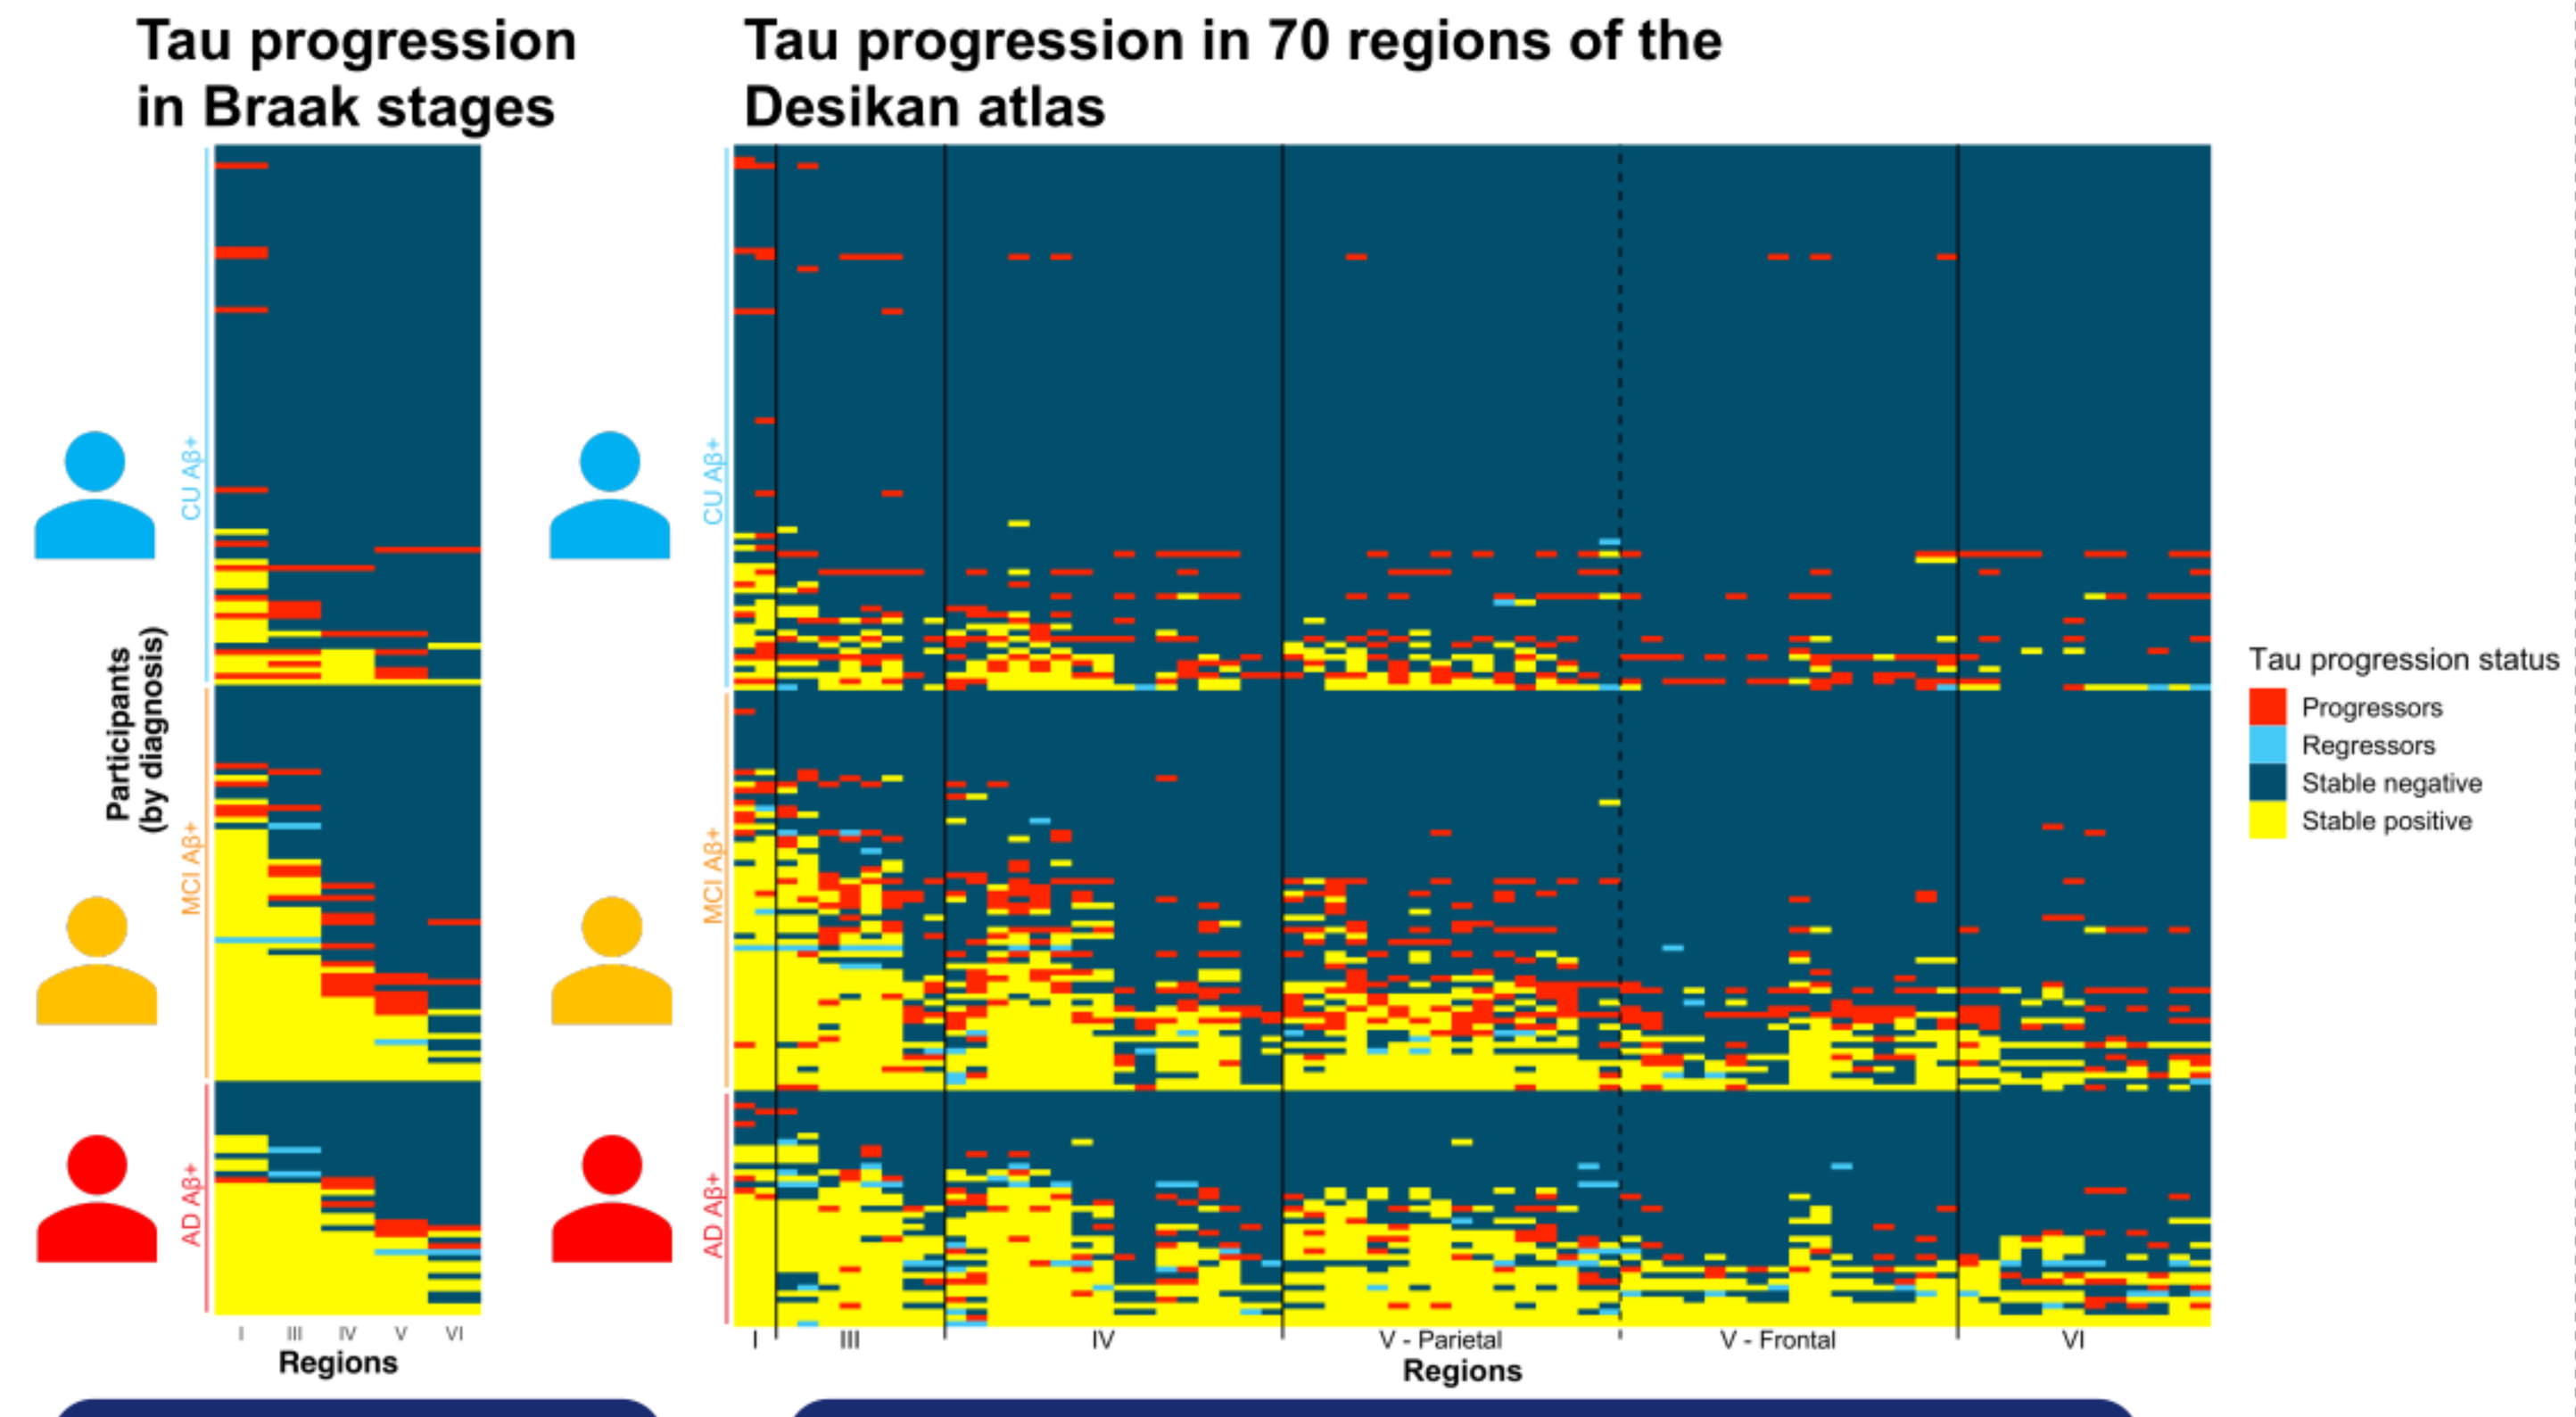

Tau progression in Braak stages
Tau progression in 70 regions of the Desikan atlas
Participants (by diagnosis)
CU Aβ+
MCI Aβ+
AD Aβ+
Regions
I III IV V VI
IV
V - Parietal
V - Frontal
VI
Tau progression status
Progressors
Regressors
Stable negative
Stable positive

>80% follow Braak staging

BUT less than 50% of regions becoming tau abnormal overlap between participants

Jaccard Index

Figure 1B Average SUVR change by region

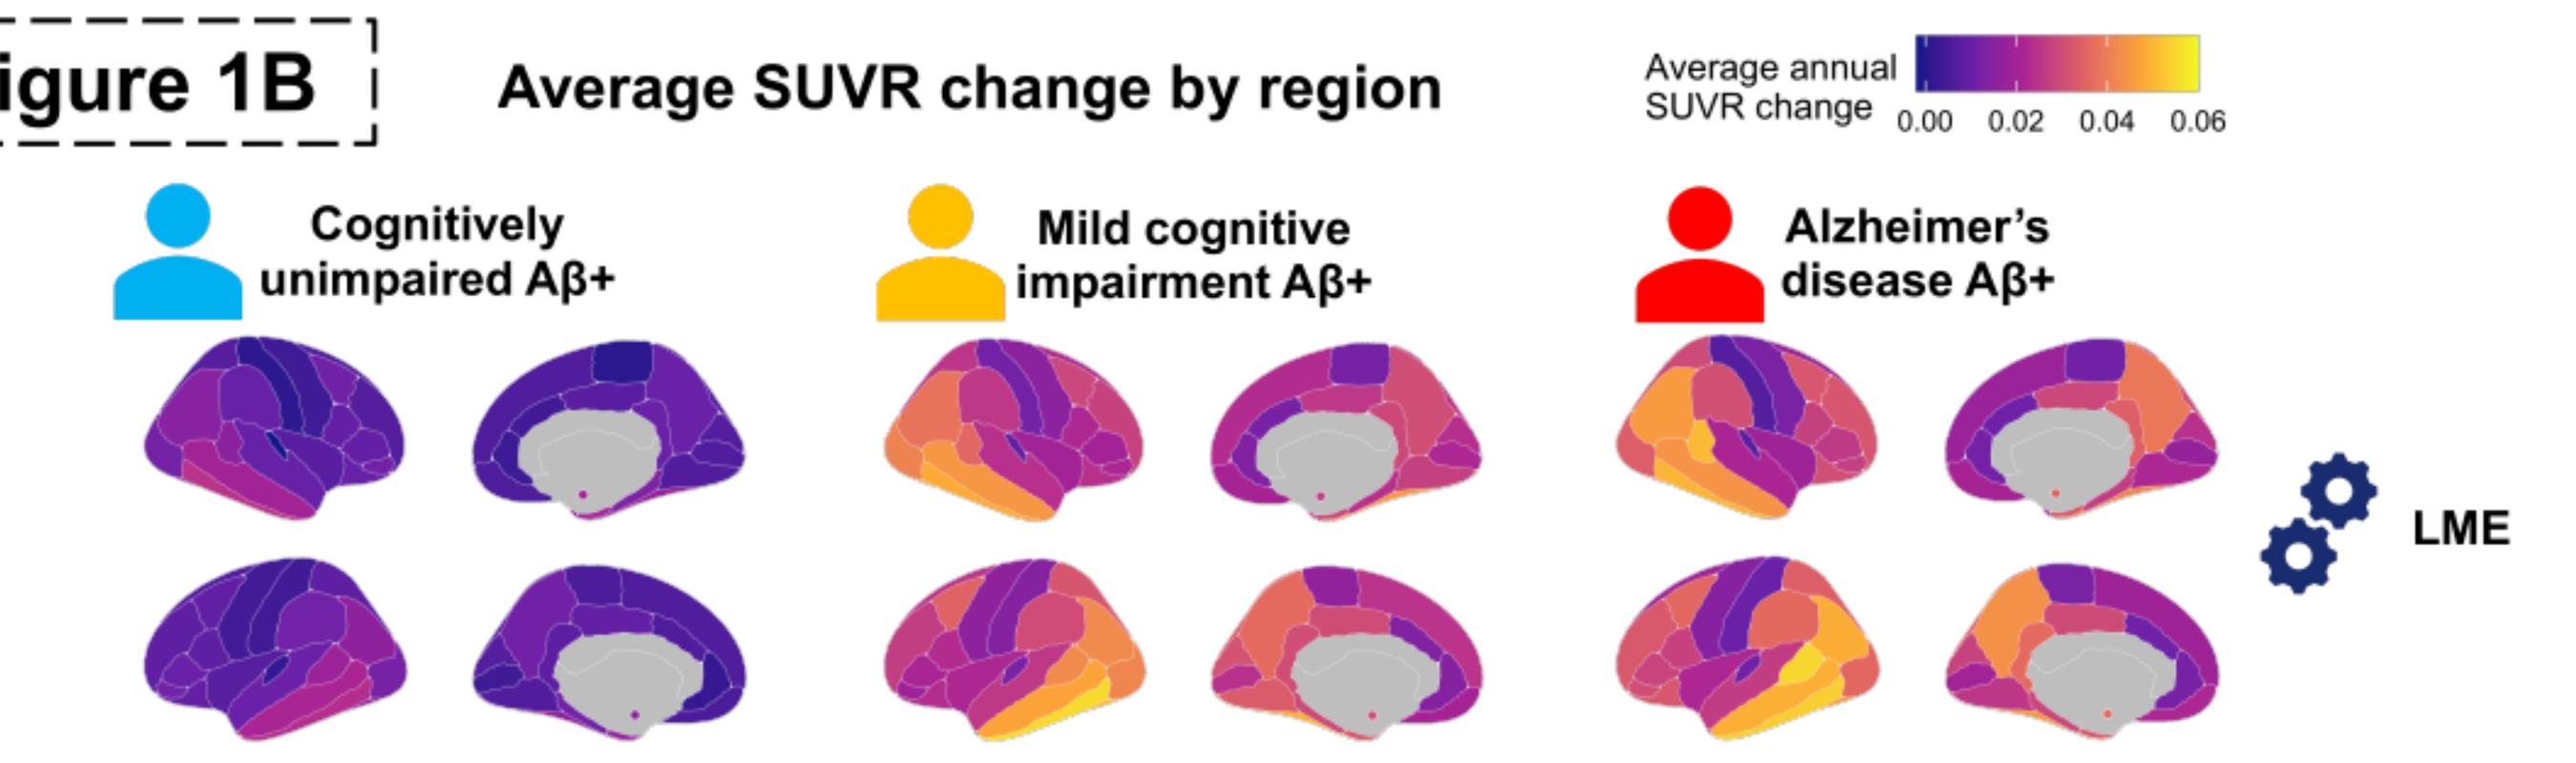

Average annual SUVR change
0.00 0.02 0.04 0.06
Cognitively unimpaired Aβ+
Mild cognitive impairment Aβ+
Alzheimer's disease Aβ+
LME

Tau SUVR accumulation **consistent with Braak staging** (gradient from middle temporal to sensorimotor)

Figure 1C Average abnormality change by region (change in spatial extent)

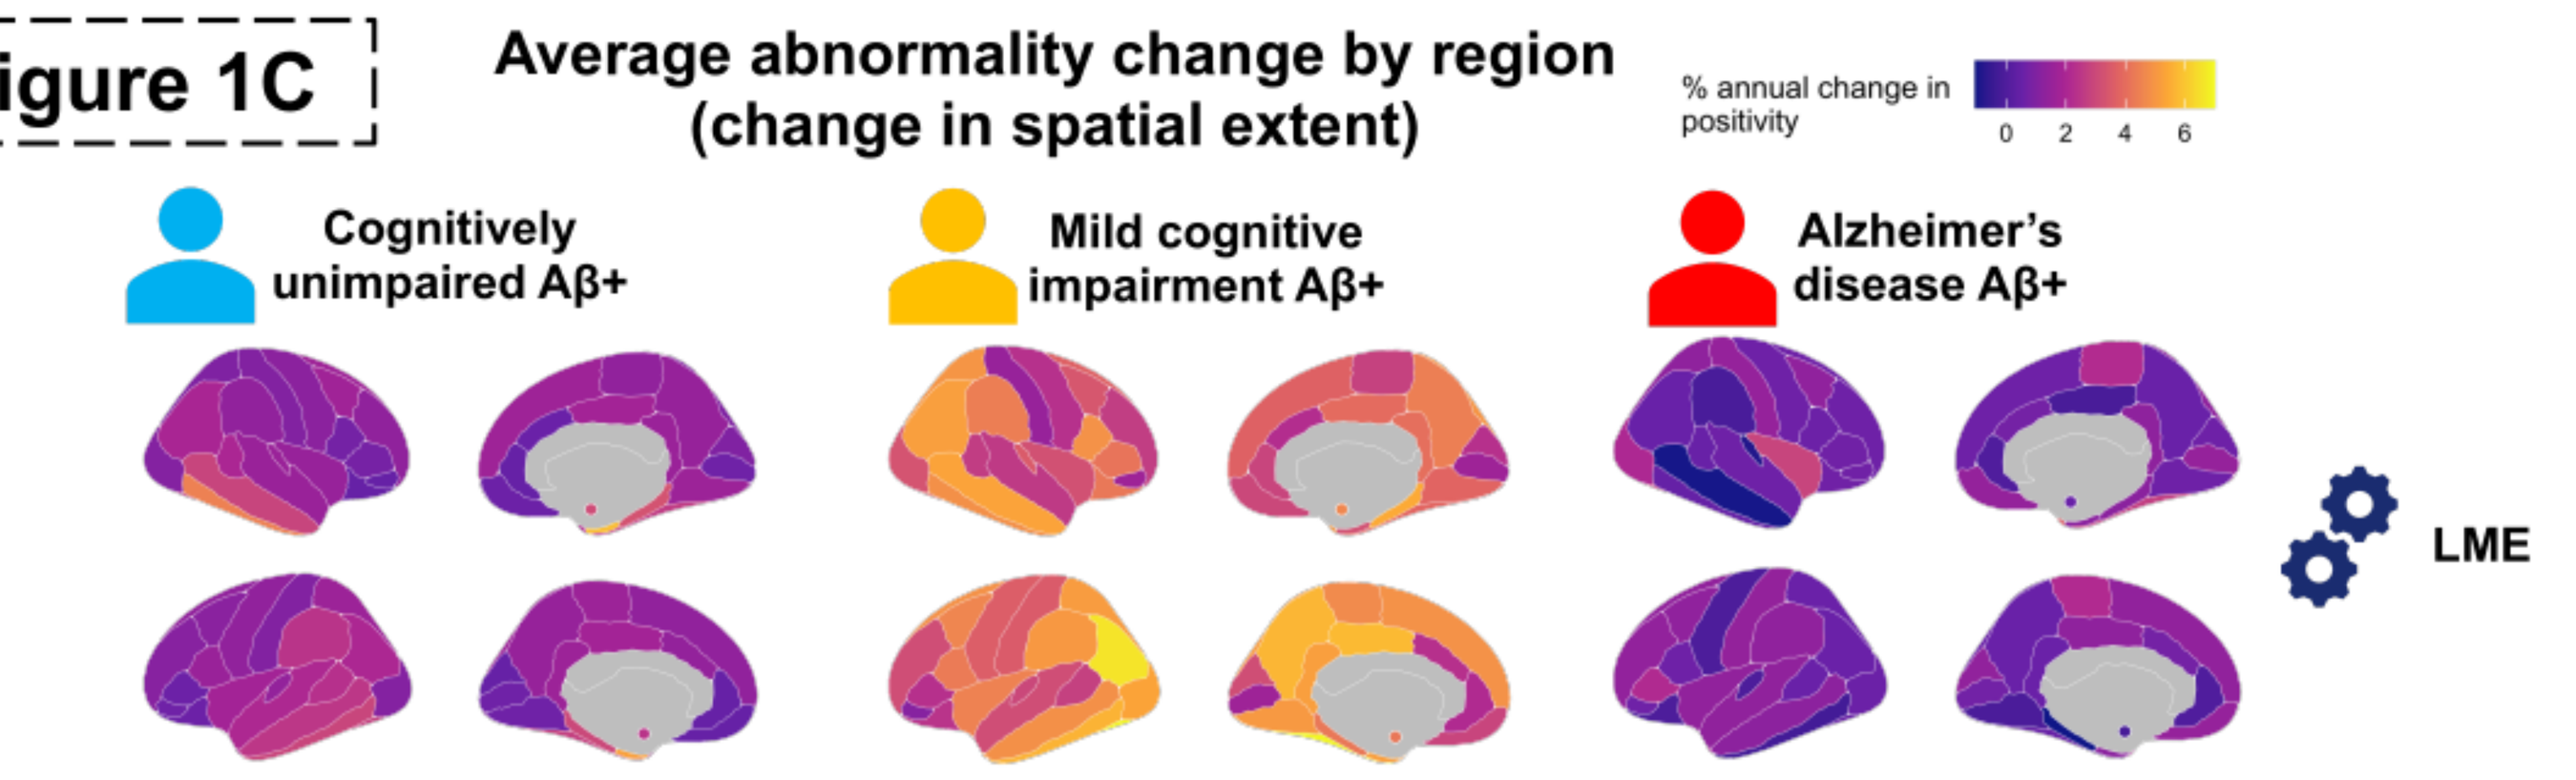

% annual change in positivity
0 2 4 6
Cognitively unimpaired Aβ+
Mild cognitive impairment Aβ+
Alzheimer's disease Aβ+
LME

Tau abnormality **progression is widespread and faster in MCI** (MCI = 2.5 regions/year; CU and AD = 1 region/year)

Objective 2: Associations between regional tau and cognition

Figure 2 Associating baseline regional tau-PET SUVR with…

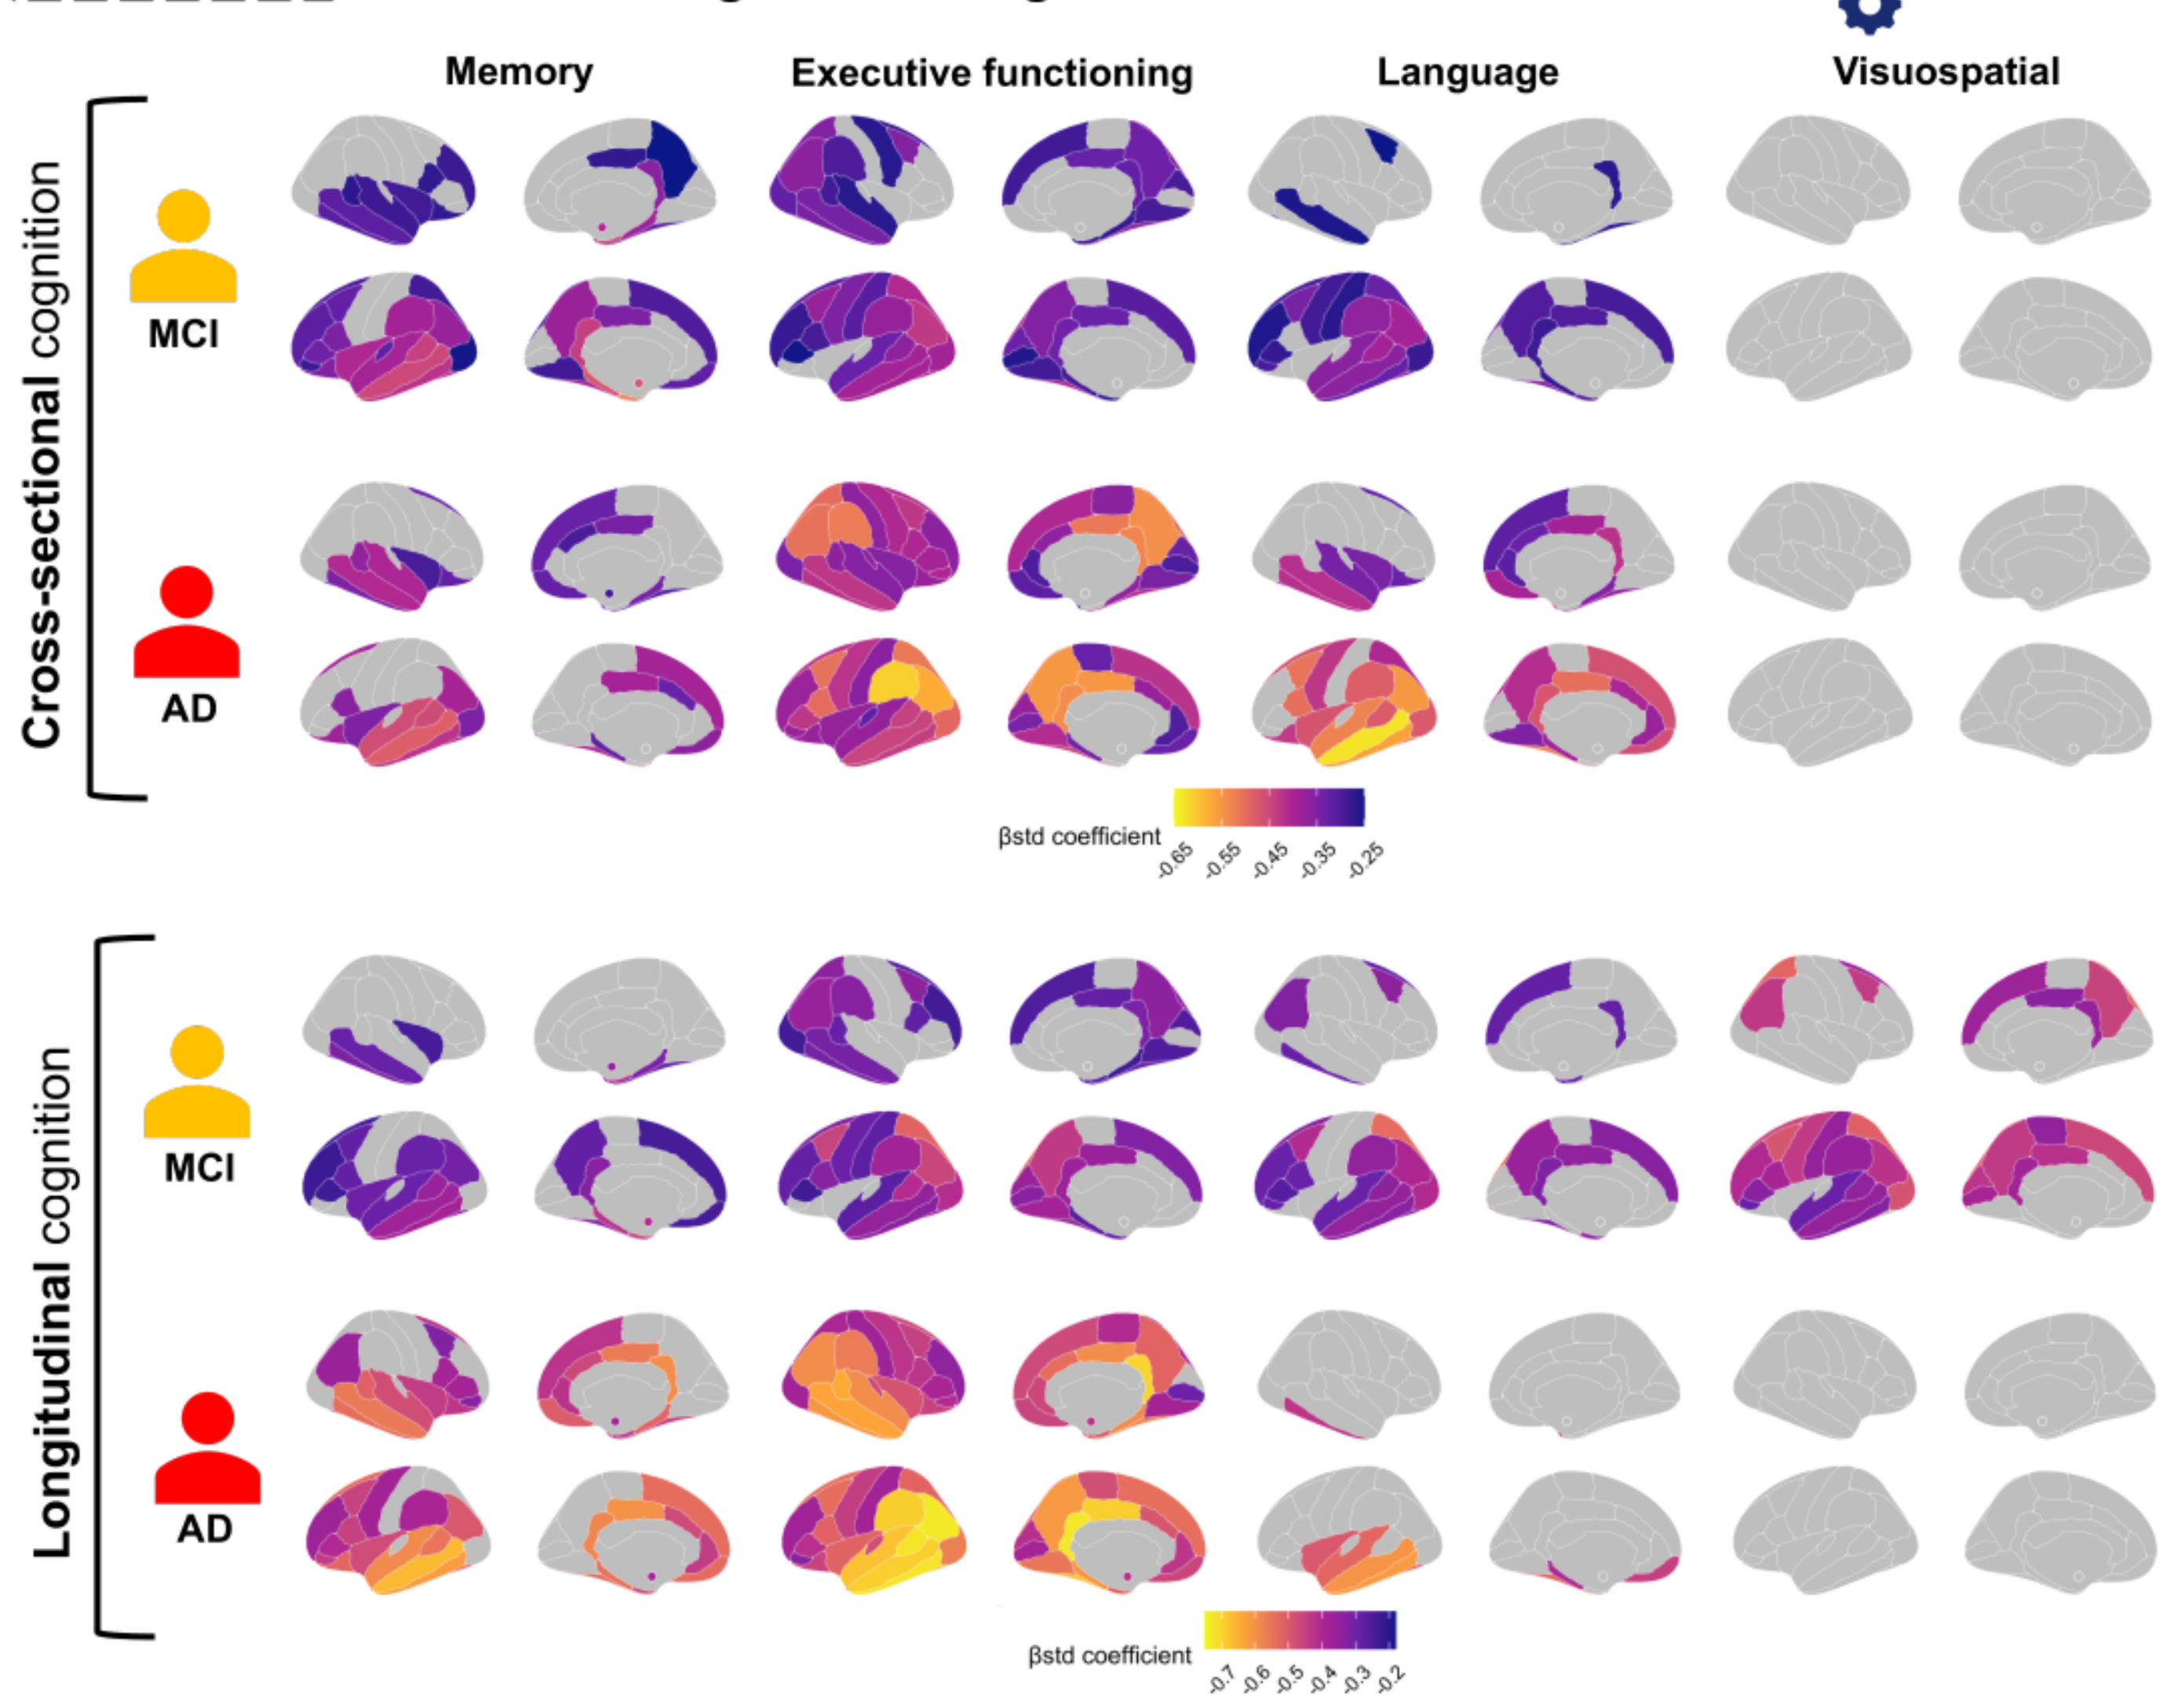

LM
Memory
Executive functioning
Language
Visuospatial
Cross-sectional cognition
MCI
AD
βstd coefficient
-0.65 -0.55 -0.45 -0.35 -0.25
Longitudinal cognition
MCI
AD
βstd coefficient
-0.7 -0.6 -0.5 -0.4 -0.3 -0.2

Tau/Cognition associations are mostly restricted to the temporal lobe for memory and language, **but span the entire cortex for executive functioning**

Figure 3

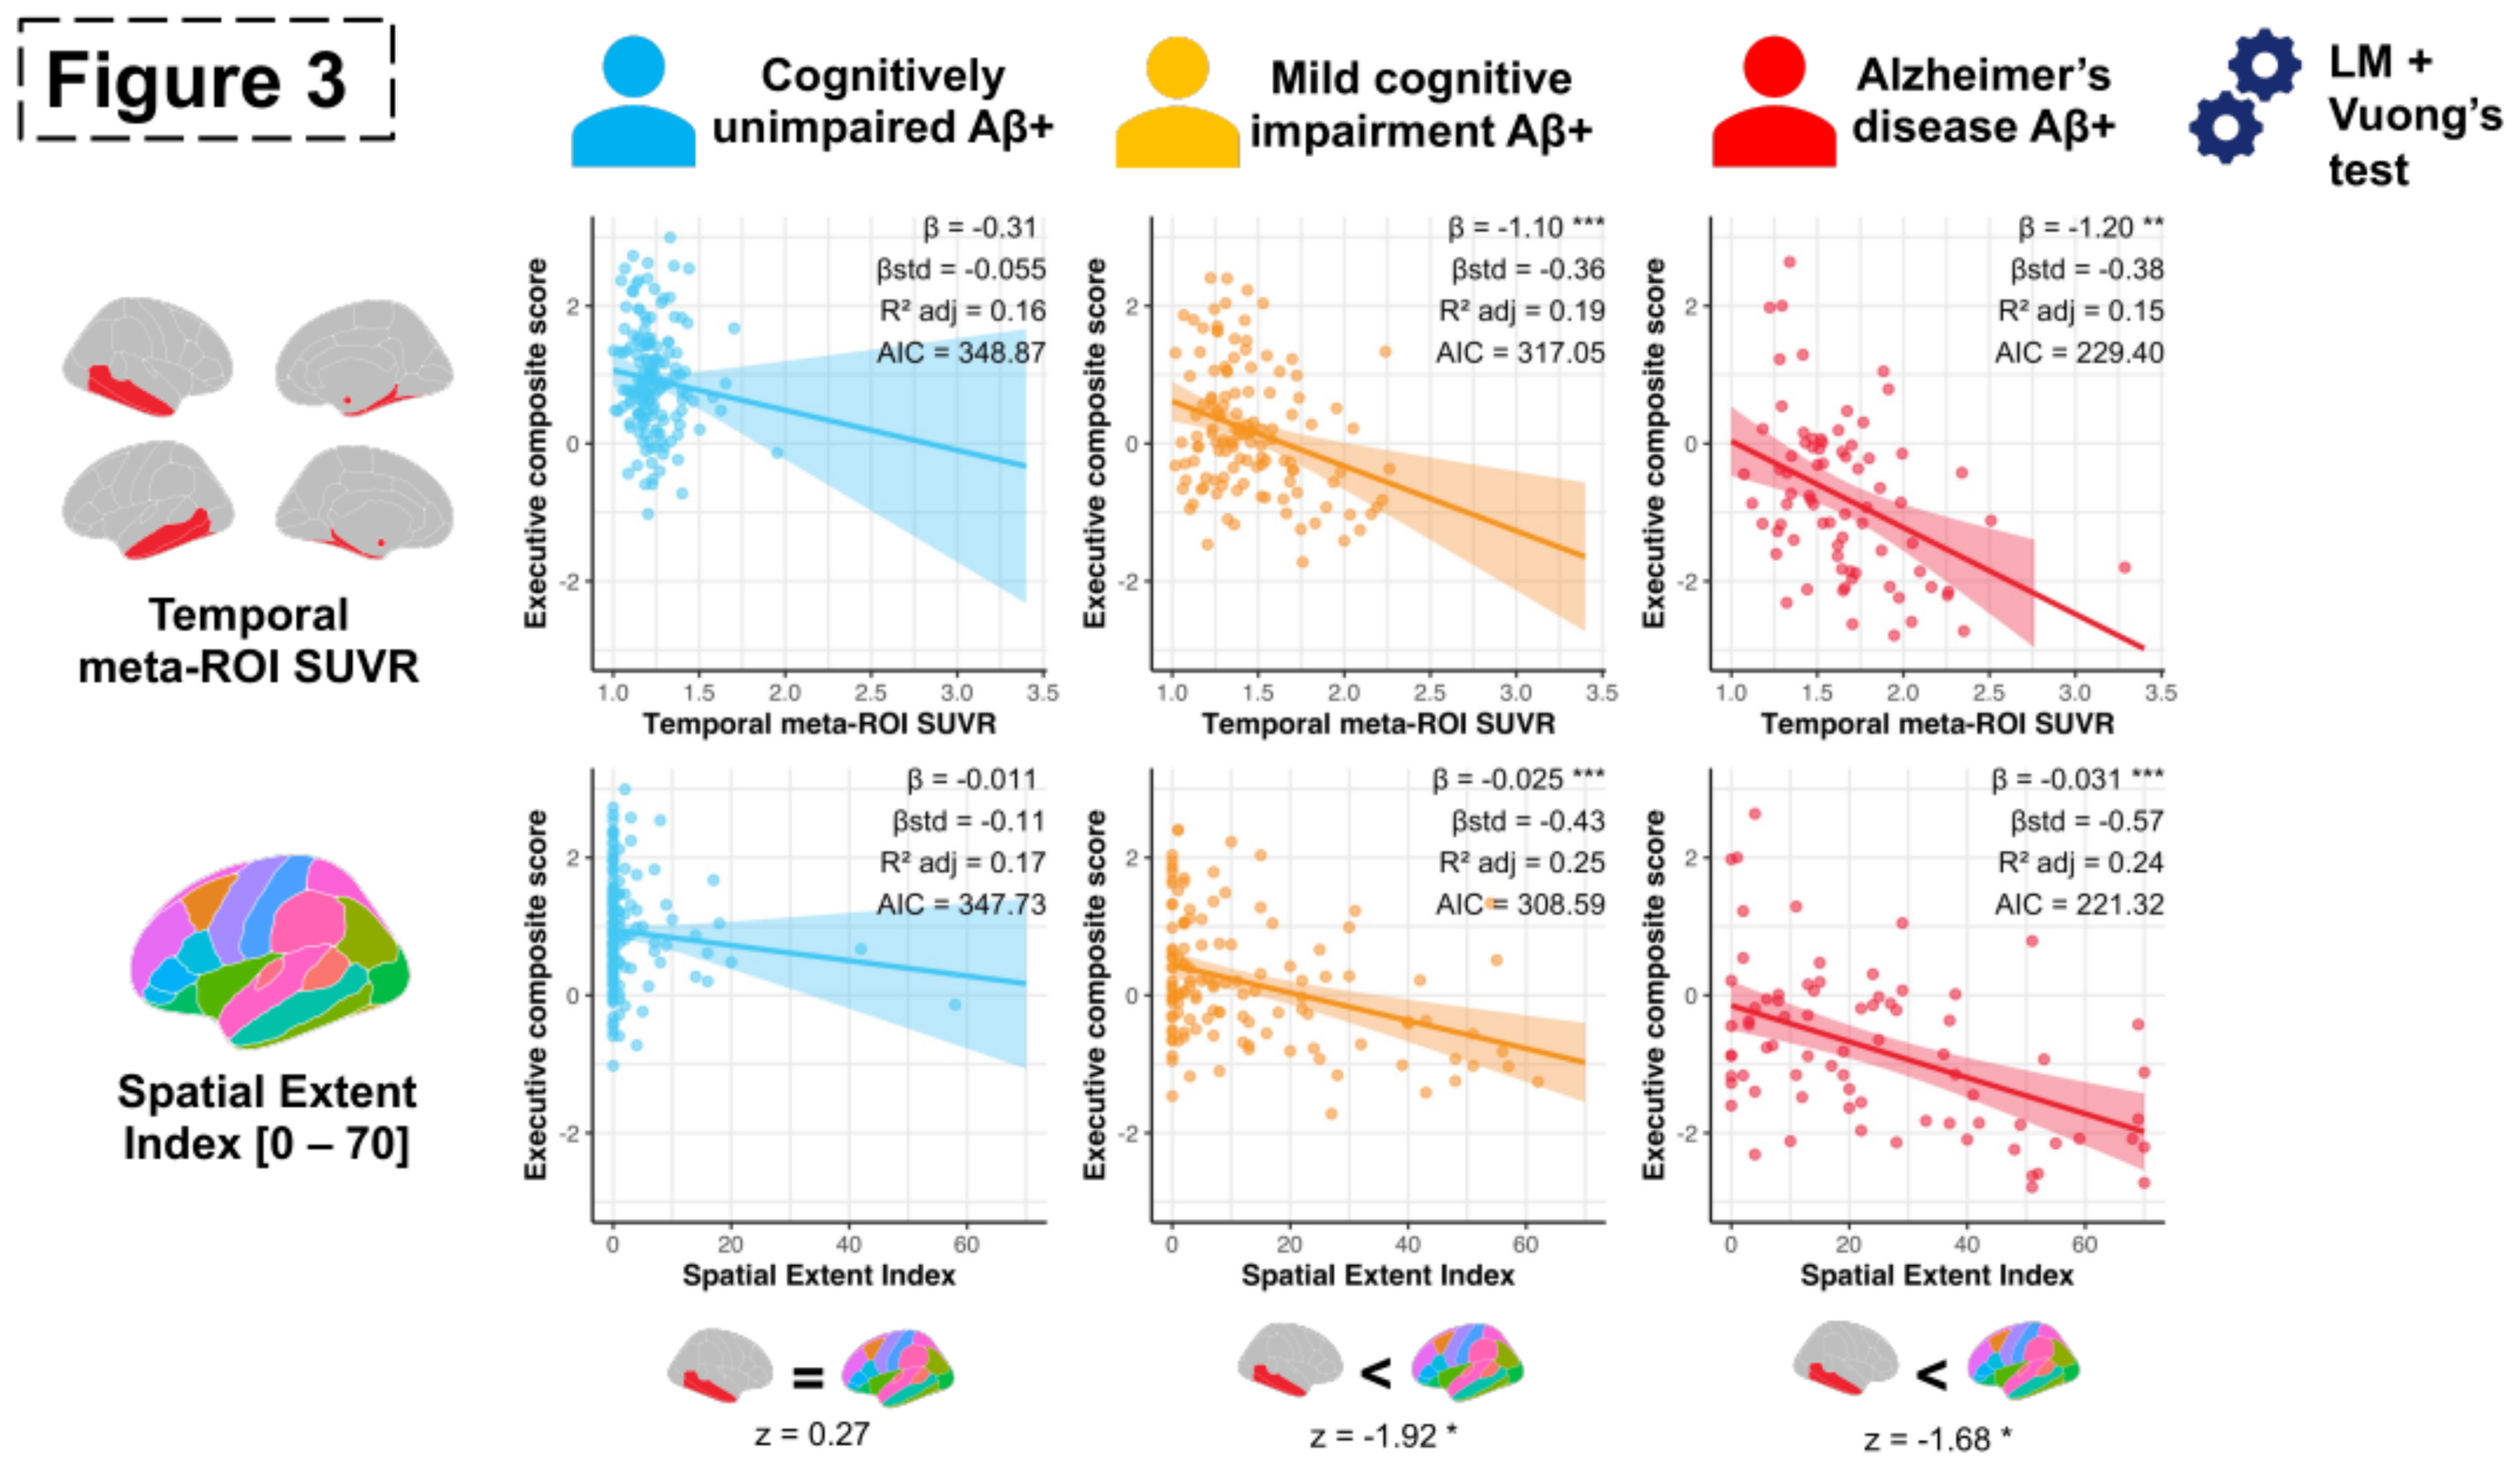

Cognitively unimpaired Aβ+
Mild cognitive impairment Aβ+
Alzheimer's disease Aβ+
LM + Vuong's test
Temporal meta-ROI SUVR
β = -0.31
βstd = -0.055
R² adj = 0.16
AIC = 348.87
β = -1.10 ***
βstd = -0.36
R² adj = 0.19
AIC = 317.05
β = -1.20 **
βstd = -0.38
R² adj = 0.15
AIC = 229.40
Executive composite score
Temporal meta-ROI SUVR
Spatial Extent Index [0 – 70]
β = -0.011
βstd = -0.11
R² adj = 0.17
AIC = 347.73
β = -0.025 ***
βstd = -0.43
R² adj = 0.25
AIC = 308.59
β = -0.031 ***
βstd = -0.57
R² adj = 0.24
AIC = 221.32
Spatial Extent Index
=
z = 0.27
<
z = -1.92 *
<
z = -1.68 *

Whole brain tau-PET spatial extent is better associated with **executive functioning** than tau-PET SUVR in the temporal meta region of interest

All associations in Figure 2 and 3 were corrected for age, sex and education, and corrected for multiple comparison using FDR
In Figure 3, associations with p-values < 0.05 are flanked by *; p-values < 0.01 are flanked by **; p-values < 0.001 are flanked by ***

Braak stages are followed by participants over time **BUT significant regional heterogeneity exists**

Regional heterogeneity of tau could **better explain cognitive deficits beyond memory**

medRχiv THE PREPRINT SERVER FOR HEALTH SCIENCES

@fredstong3

Fonds de recherche Santé Québec

This research received the following travel awards: a Conference Fellowship from the AAIC, a Merit award from the OHBM and a Roger J. Paiement Outreach award from the Douglas Research Center

SIHNpy