Tracing the Spatial Extent of Cortical Tau Over Time in Alzheimer's Disease

Frédéric St-Onge[1,2], Alexa Pichet Binette[3], Marianne Chapleau[4], John CS Breitner[2,5], Sylvia Villeneuve[2,5,6]

1. Integrated program in Neuroscience, Faculty of Medicine, McGill University, Montreal, Canada; 2. Research Center of the Douglas Mental Health University Institute, Montreal Canada; 3. Clinical Memory Research Unit, Faculty of Medicine, Lund University, Lund, Sweden; 4. Faculty of medicine, University of California in San Francisco, United States; 5. Department of Psychiatry, Faculty of Medicine, McGill University, Montreal, Canada; 6. McConnell Brain Imaging Centre, Montreal Neurological Institute, Montreal, Canada

McGill

Douglas CENTRE DE RECHERCHE RESEARCH CENTRE

Villeneuve Lab
Multimodal Imaging of the Aging Brain

frederic.st-onge@mail.mcgill.ca

1. Introduction

Tau progression in the brain follows…

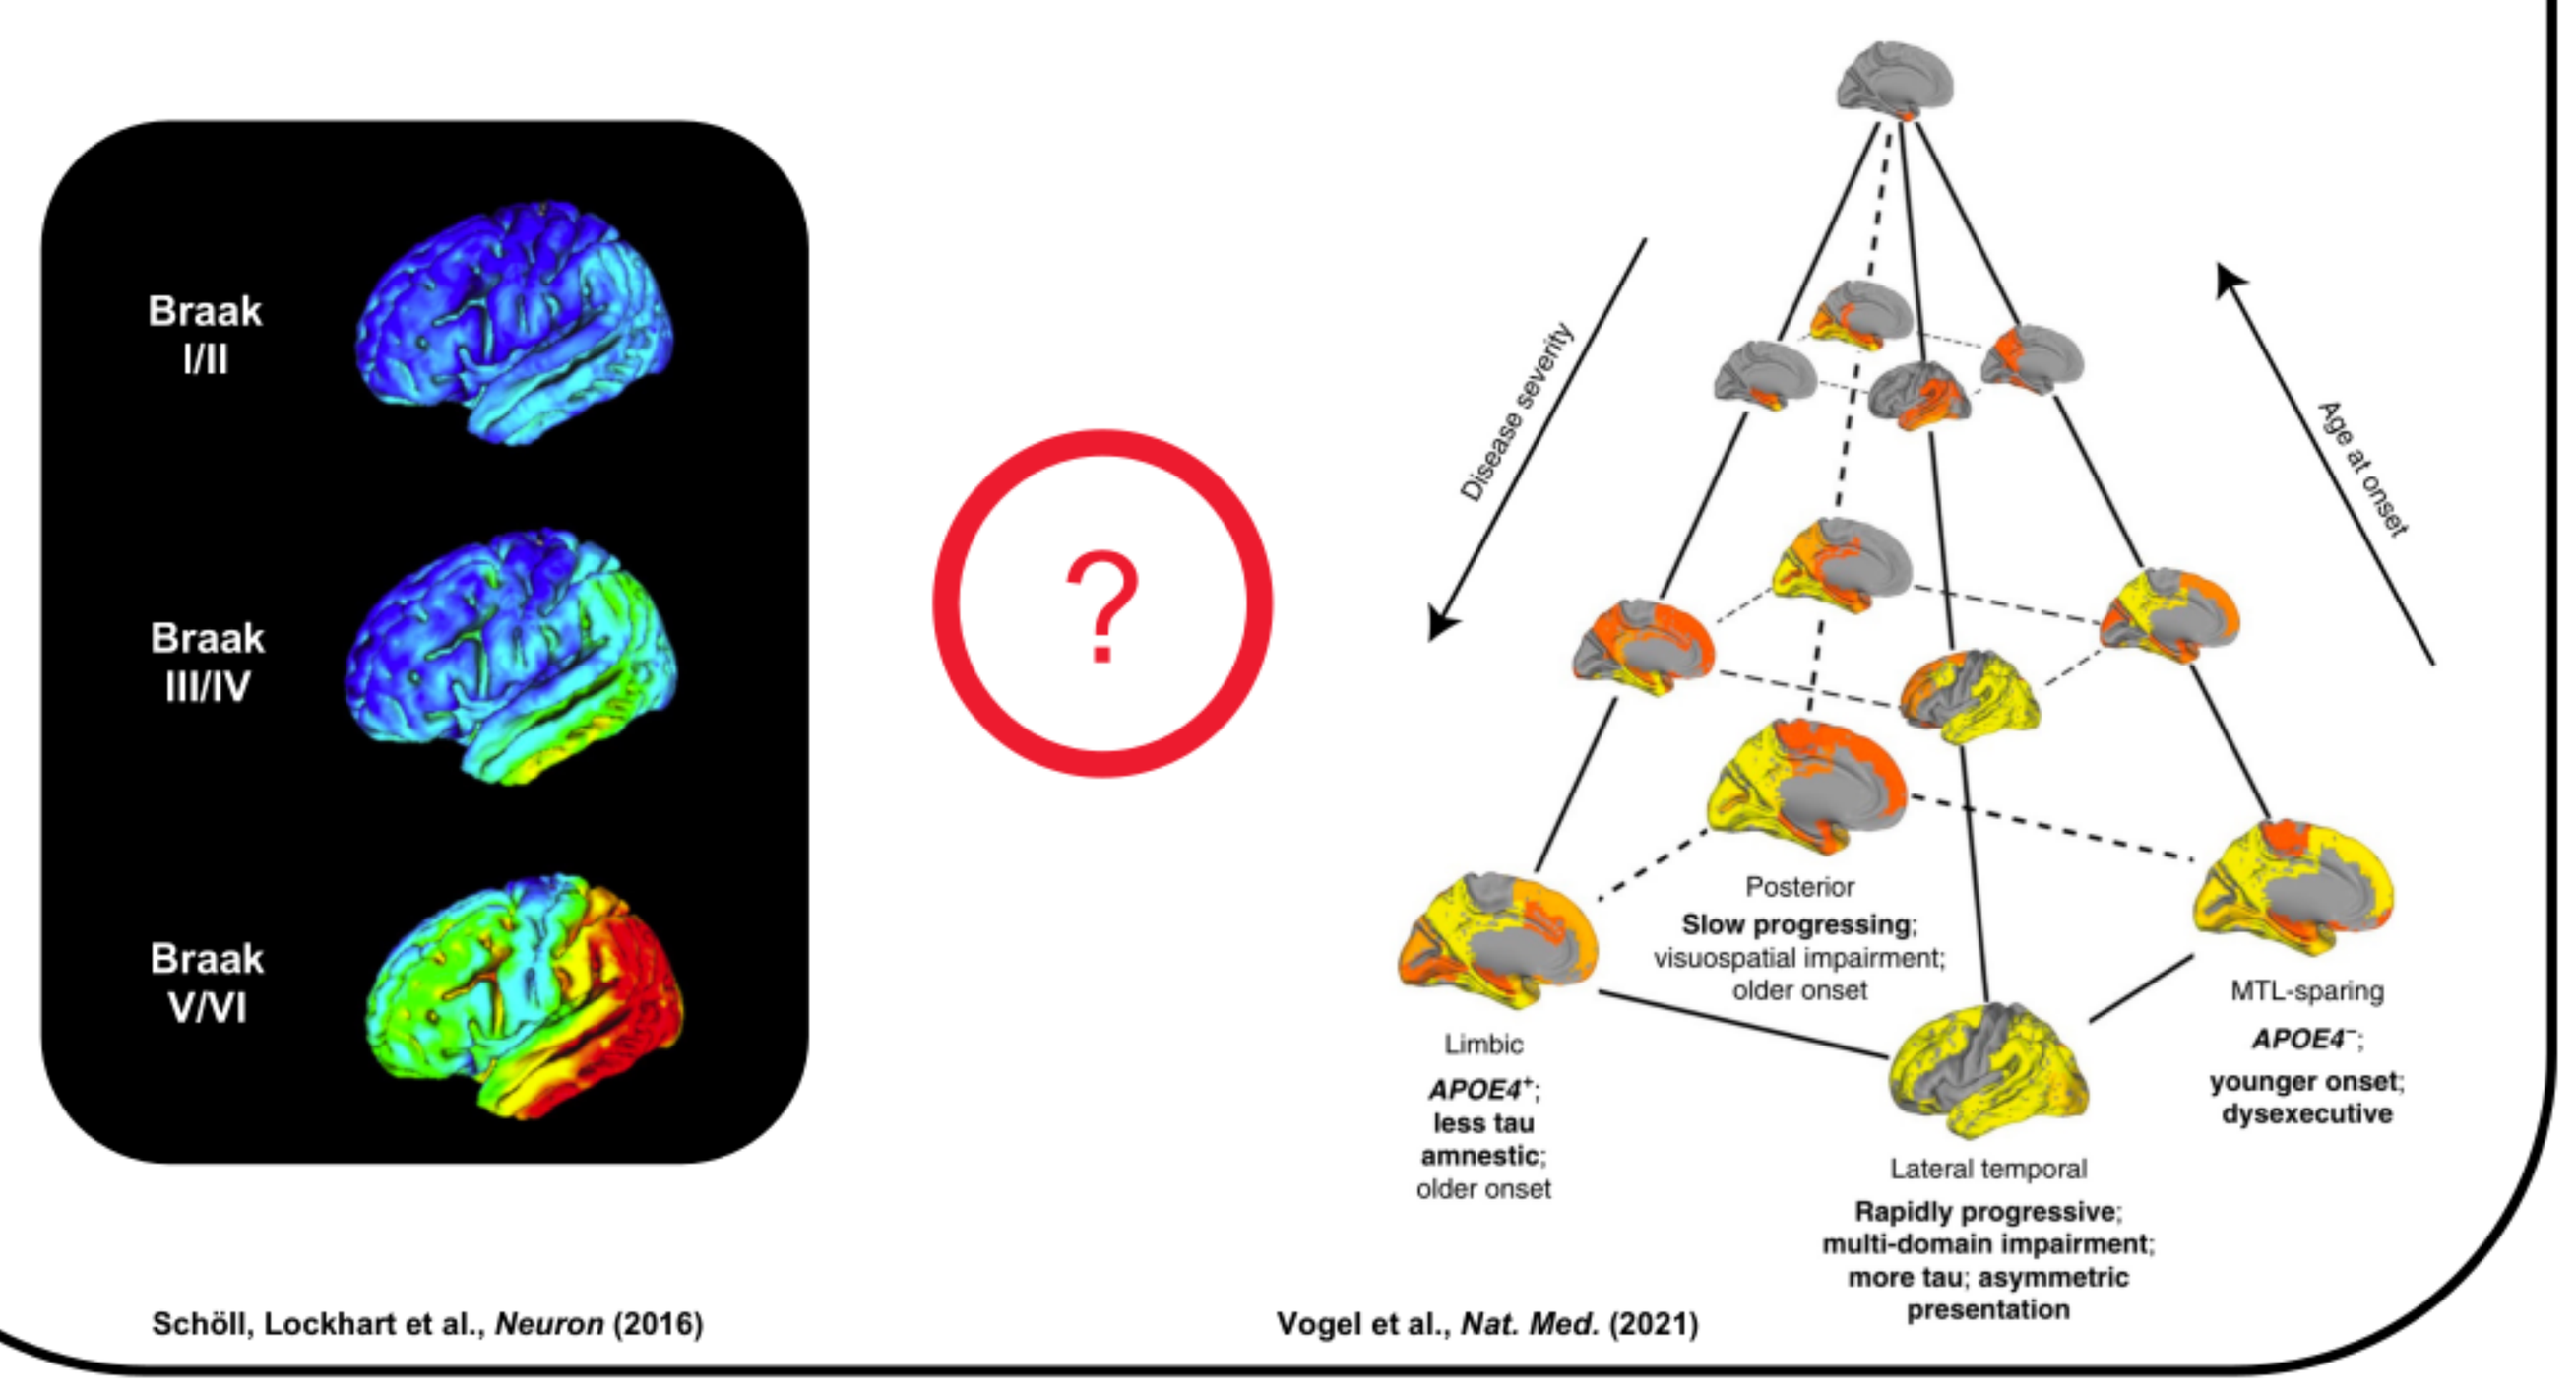
Braak I/II
Braak III/IV
Braak V/VI
?
Disease severity
Age at onset
Posterior
Slow progressing; visuospatial impairment; older onset
Limbic
APOE4+; less tau amnestic; older onset
MTL-sparing
APOE4−; younger onset; dysexecutive
Lateral temporal
Rapidly progressive; multi-domain impairment; more tau; asymmetric presentation

Schöll, Lockhart et al., *Neuron* (2016) Vogel et al., *Nat. Med.* (2021)

2. Methods

Spatial Extent Methodology

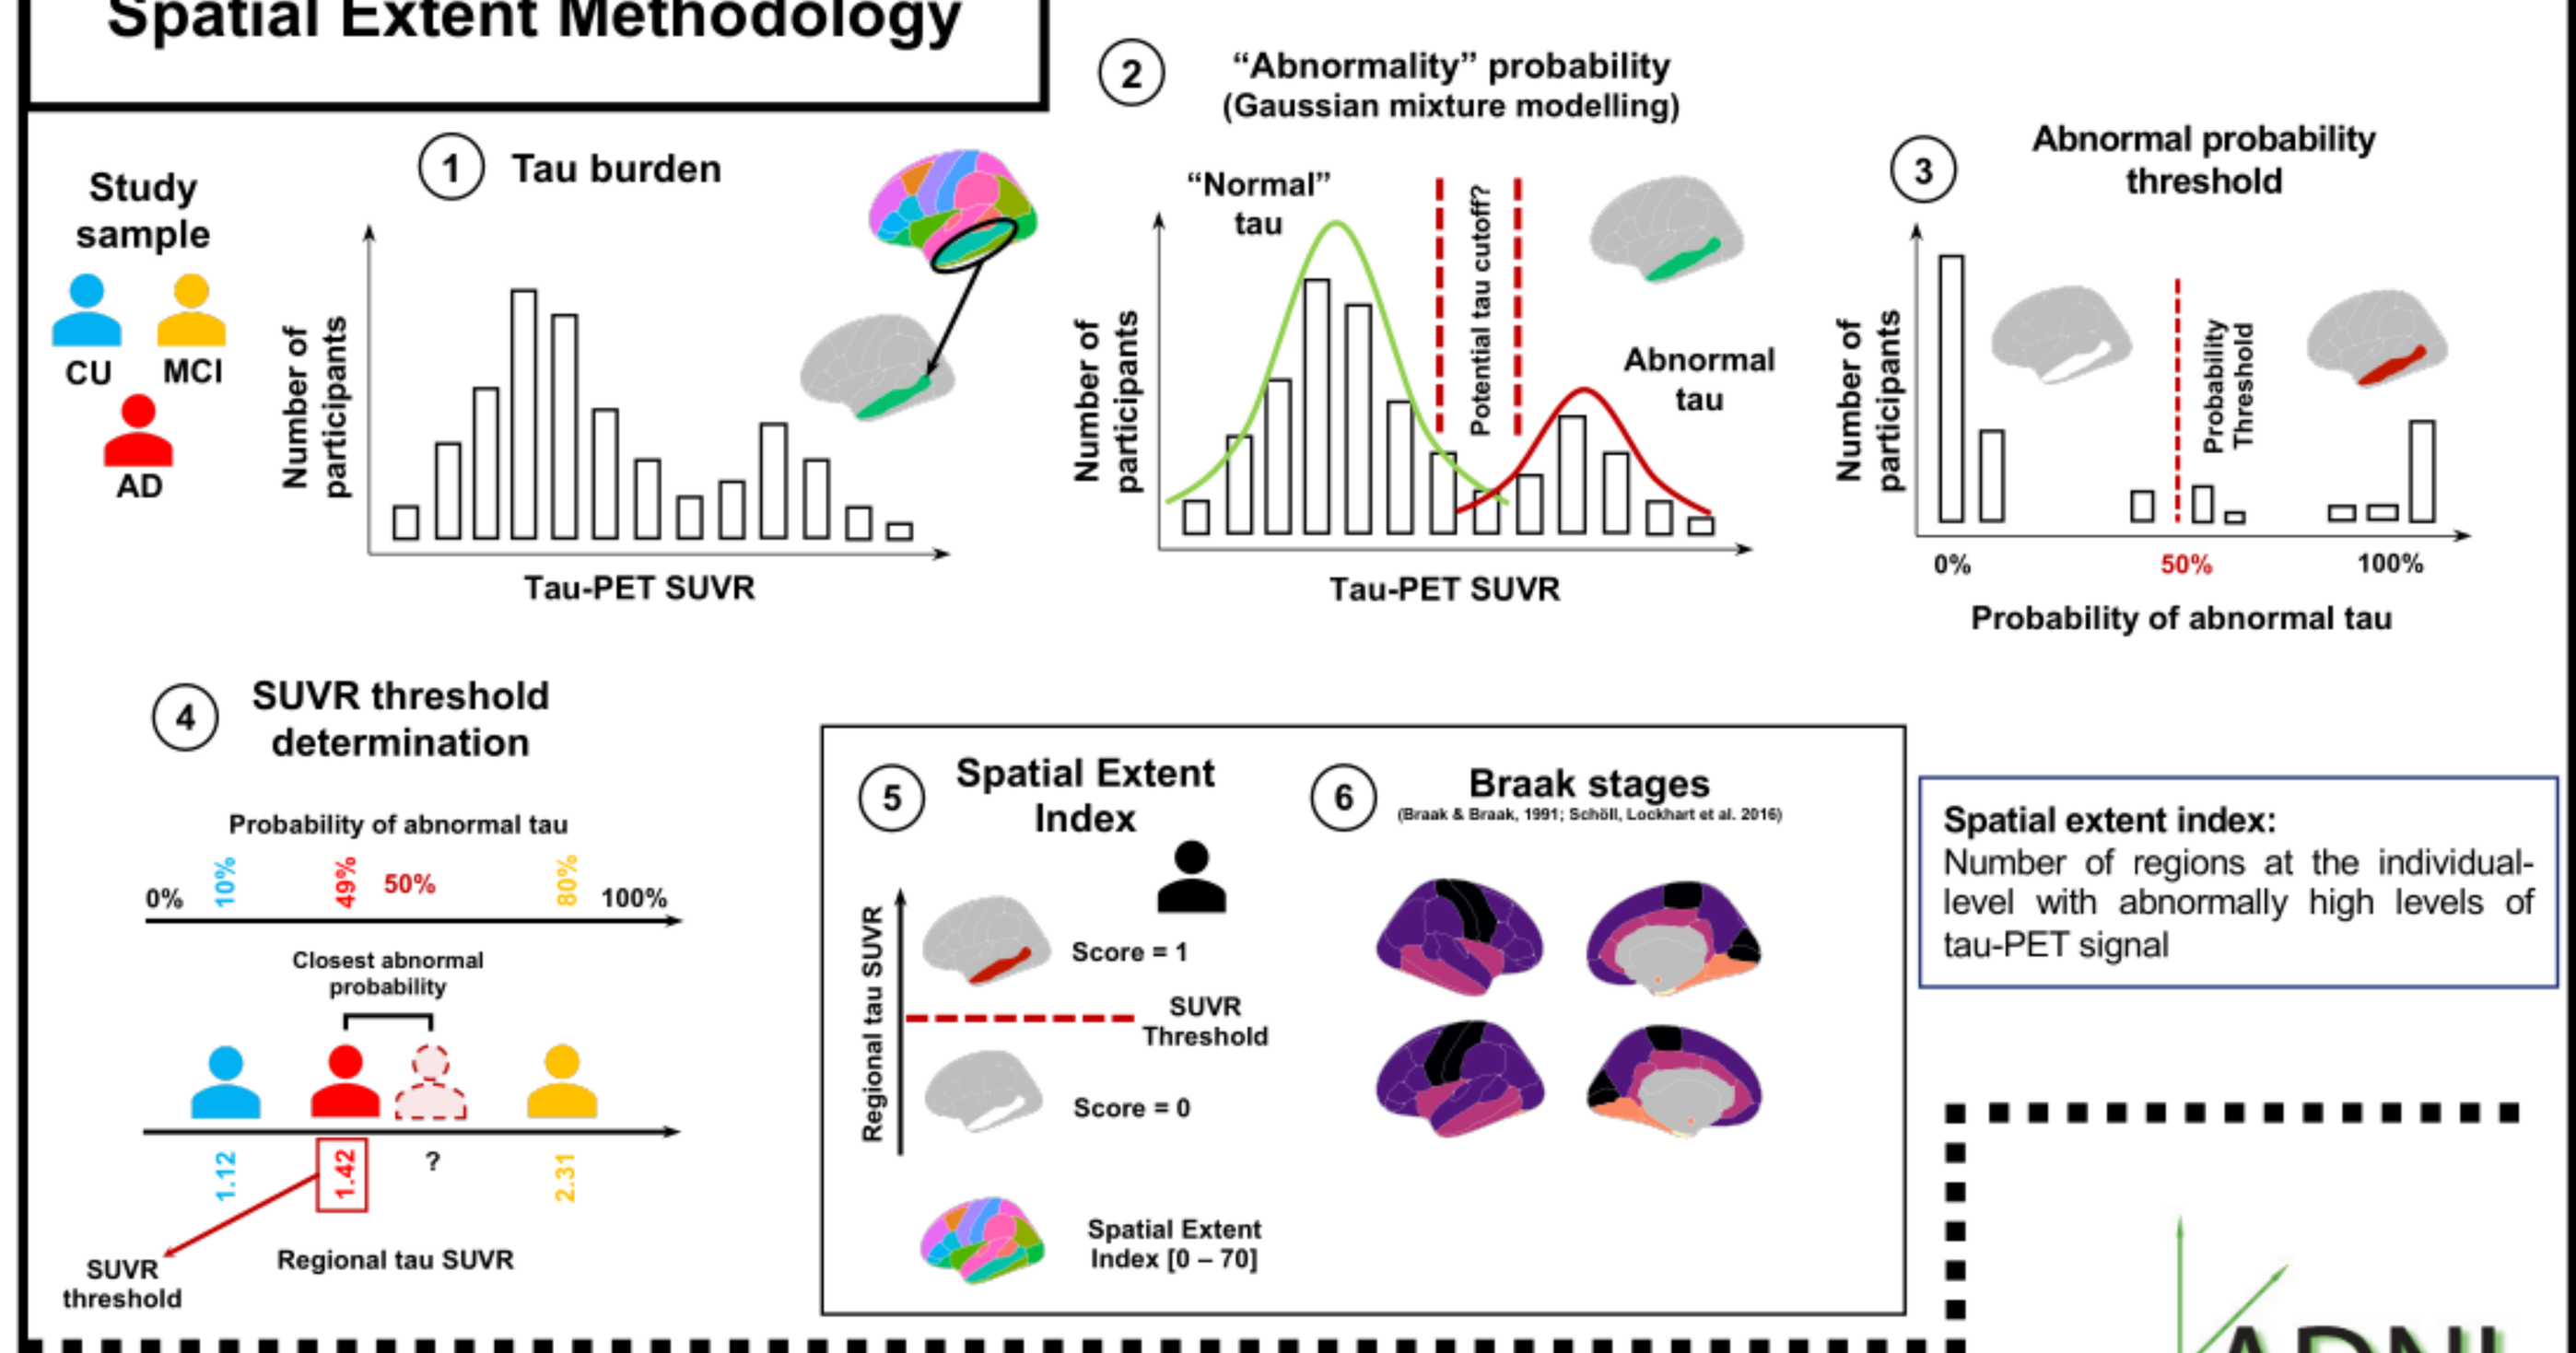
Study sample
CU MCI AD
1 Tau burden
Number of participants
Tau-PET SUVR
2 "Abnormality" probability (Gaussian mixture modelling)
"Normal" tau
Potential tau cutoff?
Abnormal tau
3 Abnormal probability threshold
Probability Threshold
0% 50% 100%
Probability of abnormal tau
4 SUVR threshold determination
Probability of abnormal tau
0% 10% 49% 50% 80% 100%
Closest abnormal probability
1.12 1.42 ? 2.31
SUVR threshold
Regional tau SUVR
5 Spatial Extent Index
Regional tau SUVR
Score = 1
SUVR Threshold
Score = 0
Spatial Extent Index [0 – 70]
6 Braak stages
(Braak & Braak, 1991; Schöll, Lockhart et al. 2016)

Spatial extent index: Number of regions at the individual-level with abnormally high levels of tau-PET signal

ADNI Alzheimer's Disease Neuroimaging Initiative

Population

Full cross-sectional sample

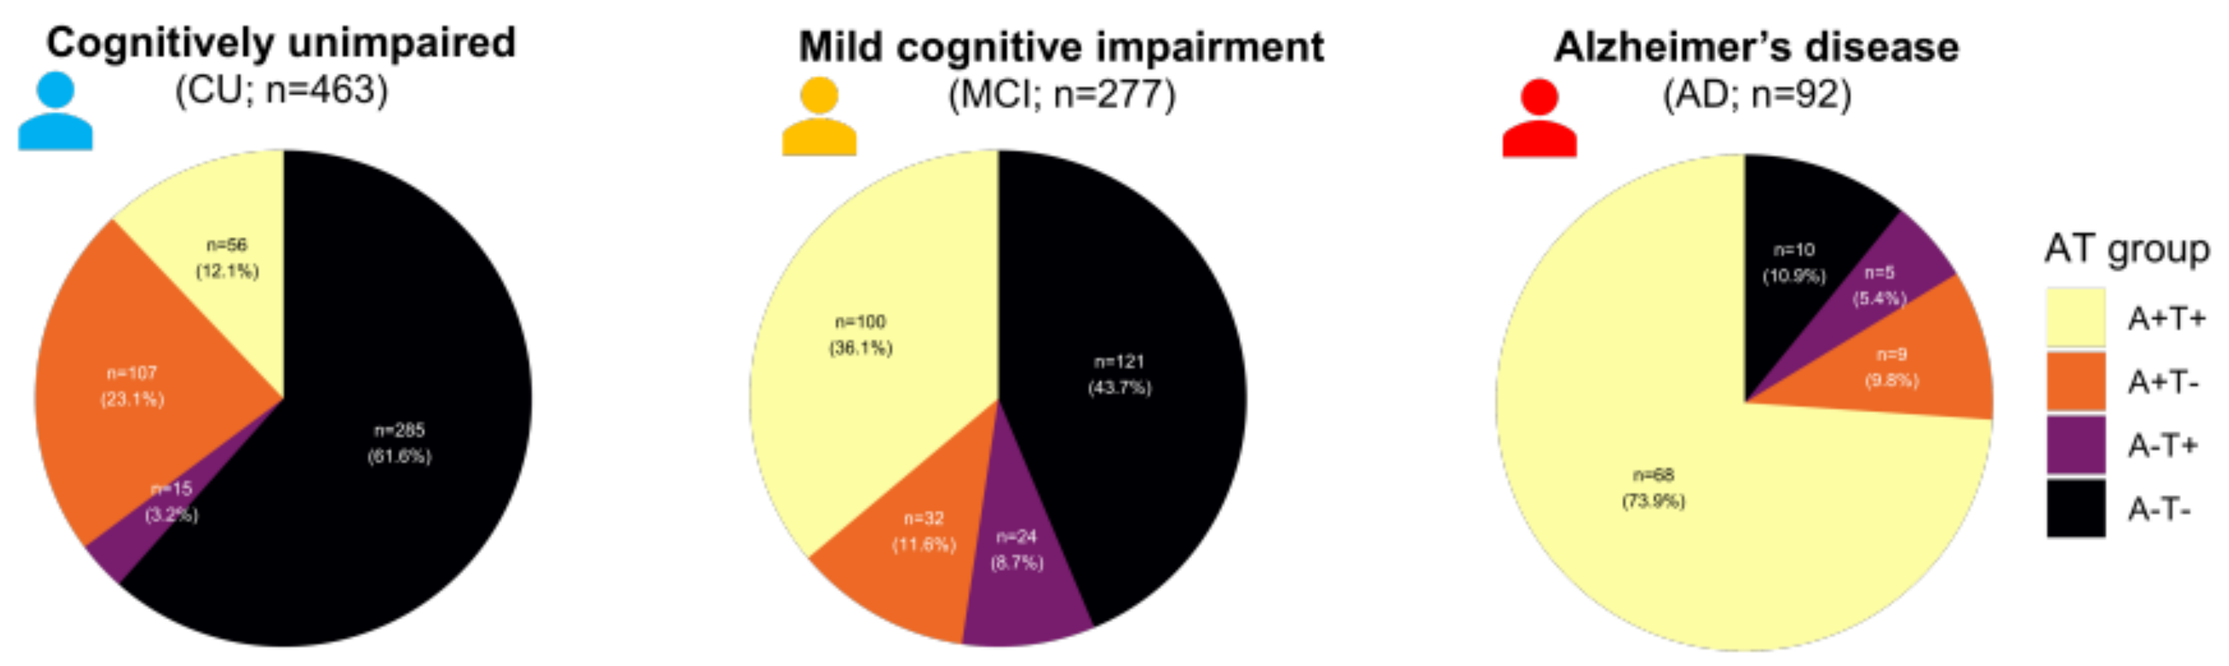
Cognitively unimpaired (CU; n=463)
n=56 (12.1%)
n=107 (23.1%)
n=15 (3.2%)
n=285 (61.6%)
Mild cognitive impairment (MCI; n=277)
n=100 (36.1%)
n=32 (11.6%)
n=24 (8.7%)
n=121 (43.7%)
Alzheimer's disease (AD; n=92)
n=10 (10.9%)
n=5 (5.4%)
n=9 (9.8%)
n=68 (73.9%)
AT group
A+T+
A+T-
A-T+
A-T-

Amyloid-positive sample

	CU (n = 163) (n_{long} = 90)	MCI (n = 132) (n_{long} = 66)	AD (n = 77) (n_{long} = 39)
Age	74.82 years (± 7.57)	74.36 years (± 7.39)	77.35 years (± 8.93)
Sex	58.9% female	49.2% female	41.6% female
APOE4	44.8% APOE4+	53.0% APOE4+	62.3% APOE4+
Education	16.64 years (± 2.34)	15.99 years (± 2.49)	15.55 years (± 2.48)

3. Results

Objective 1: Abnormal deposition of tau pathology at baseline

Figure 1A

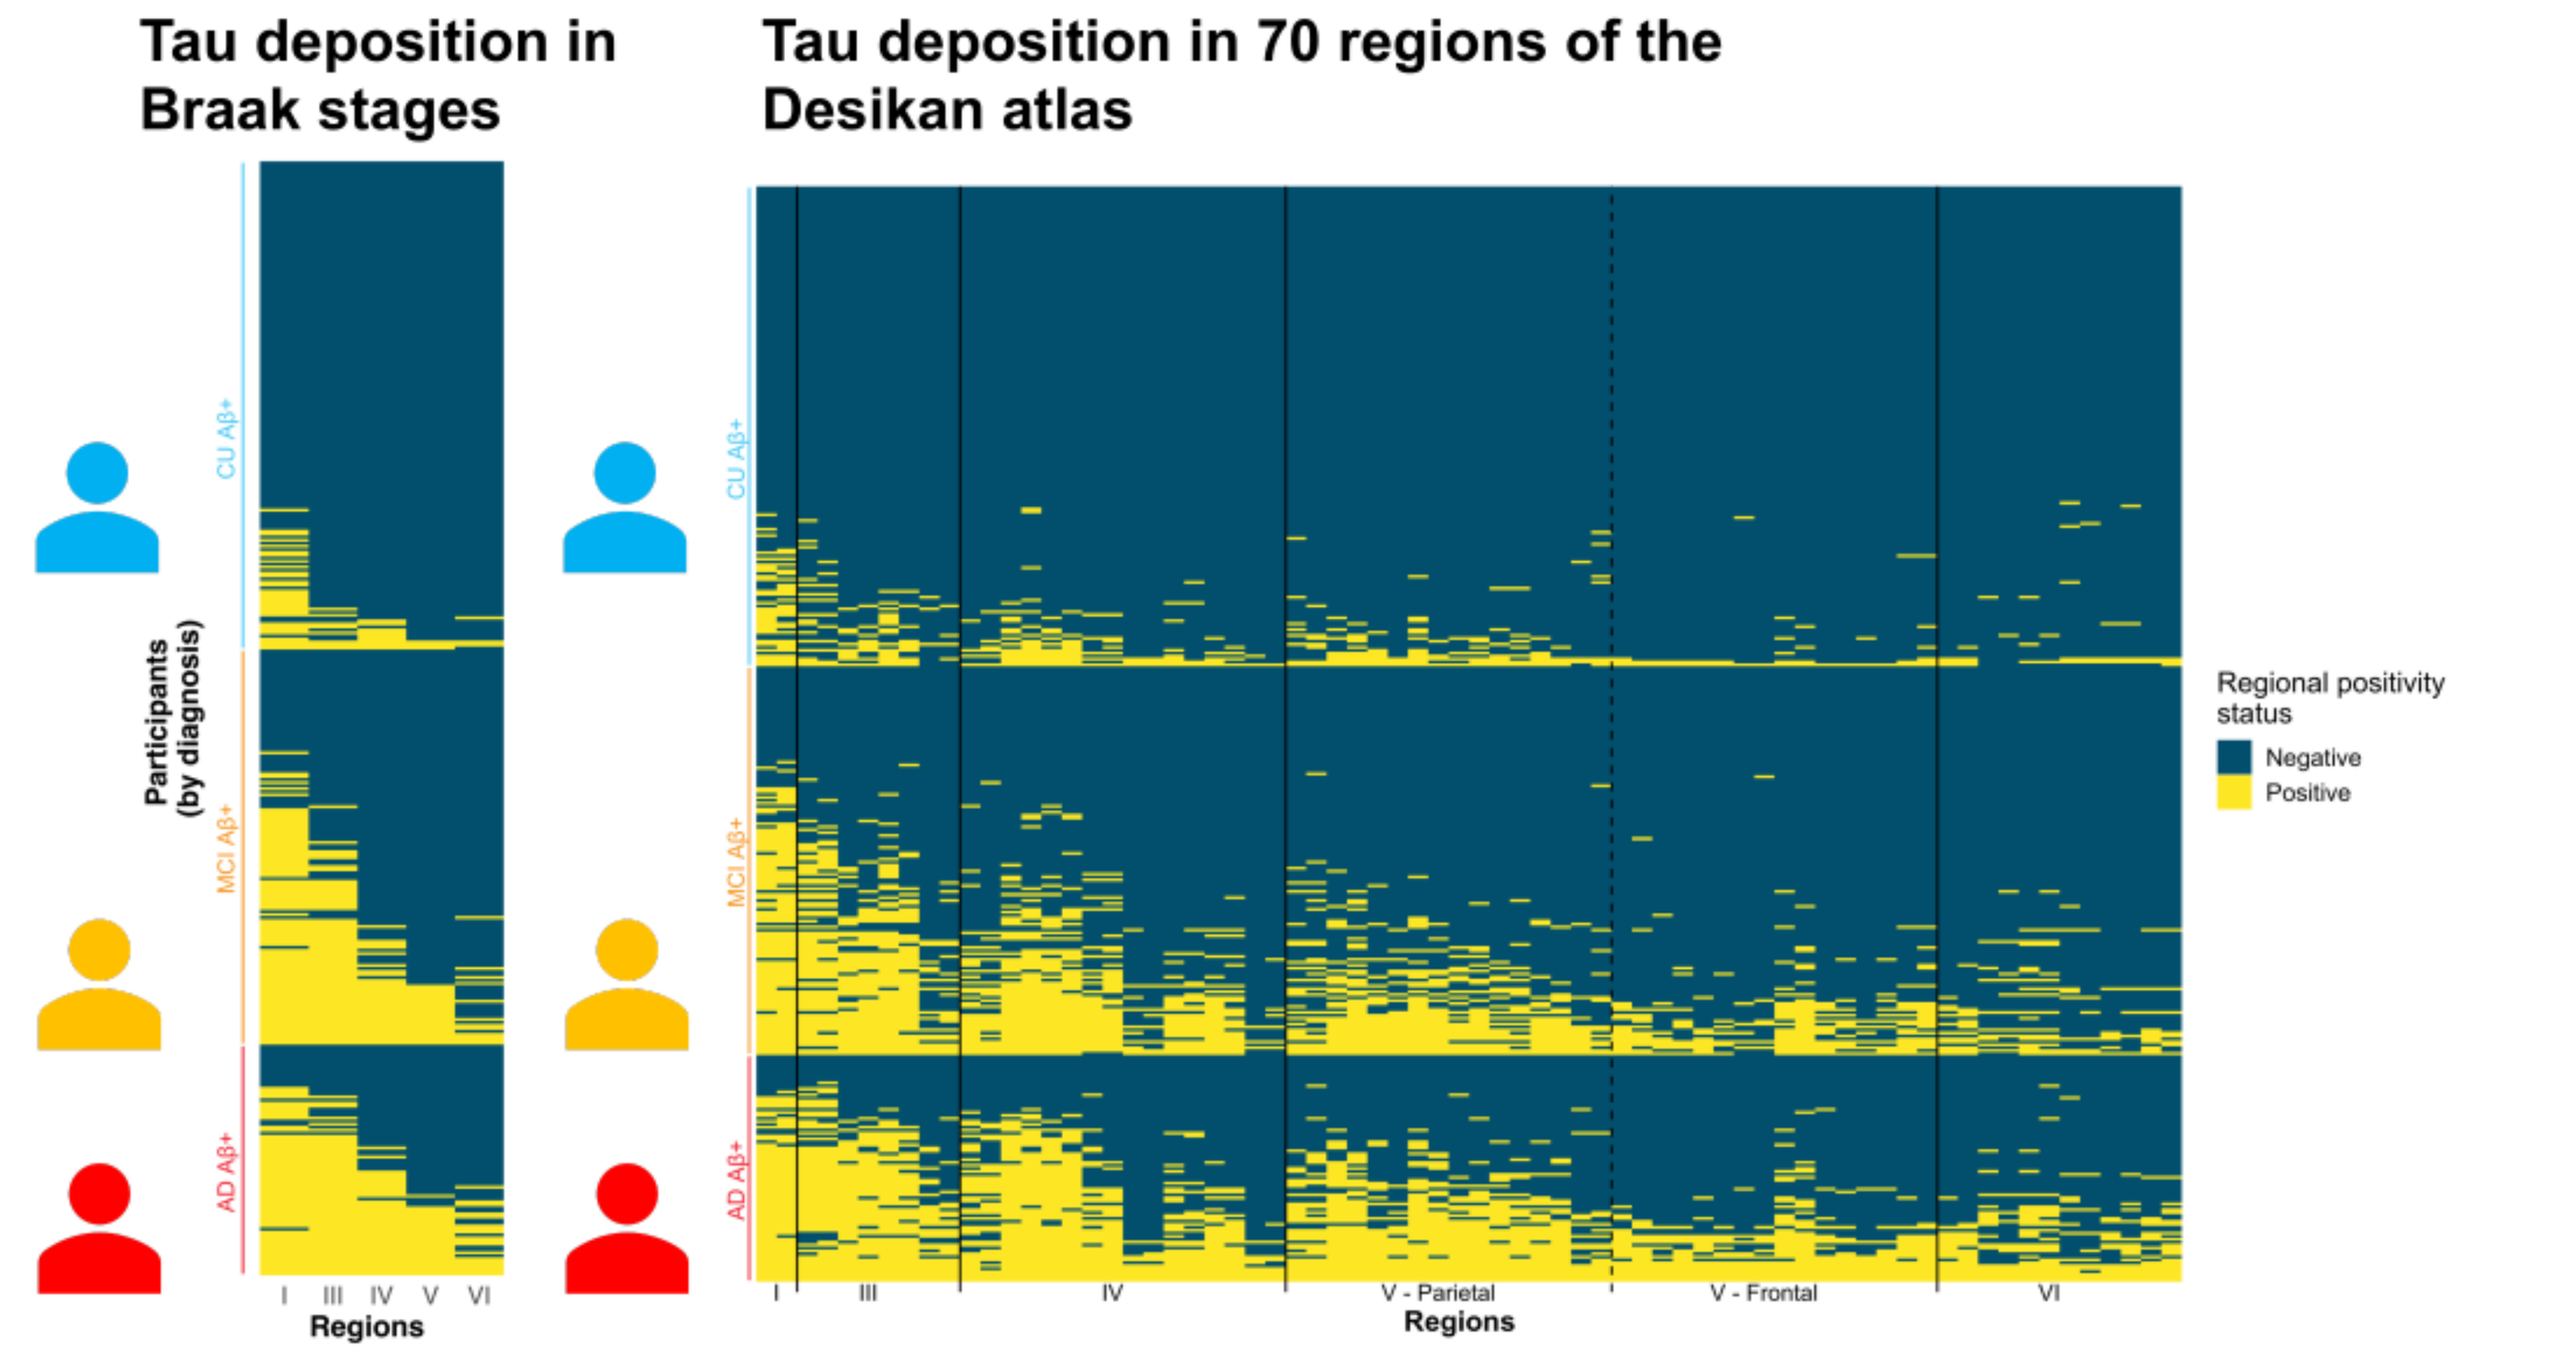
Tau deposition in Braak stages
Tau deposition in 70 regions of the Desikan atlas
Participants (by diagnosis)
CU Aβ+
MCI Aβ+
AD Aβ+
I III IV V VI
Regions
IV
V - Parietal
V - Frontal
VI
Regional positivity status
Negative
Positive

>90% follow Braak staging

BUT less than 50% of regions being tau abnormal overlap between participants

Jaccard Index

Figure 1B Average SUVR by region

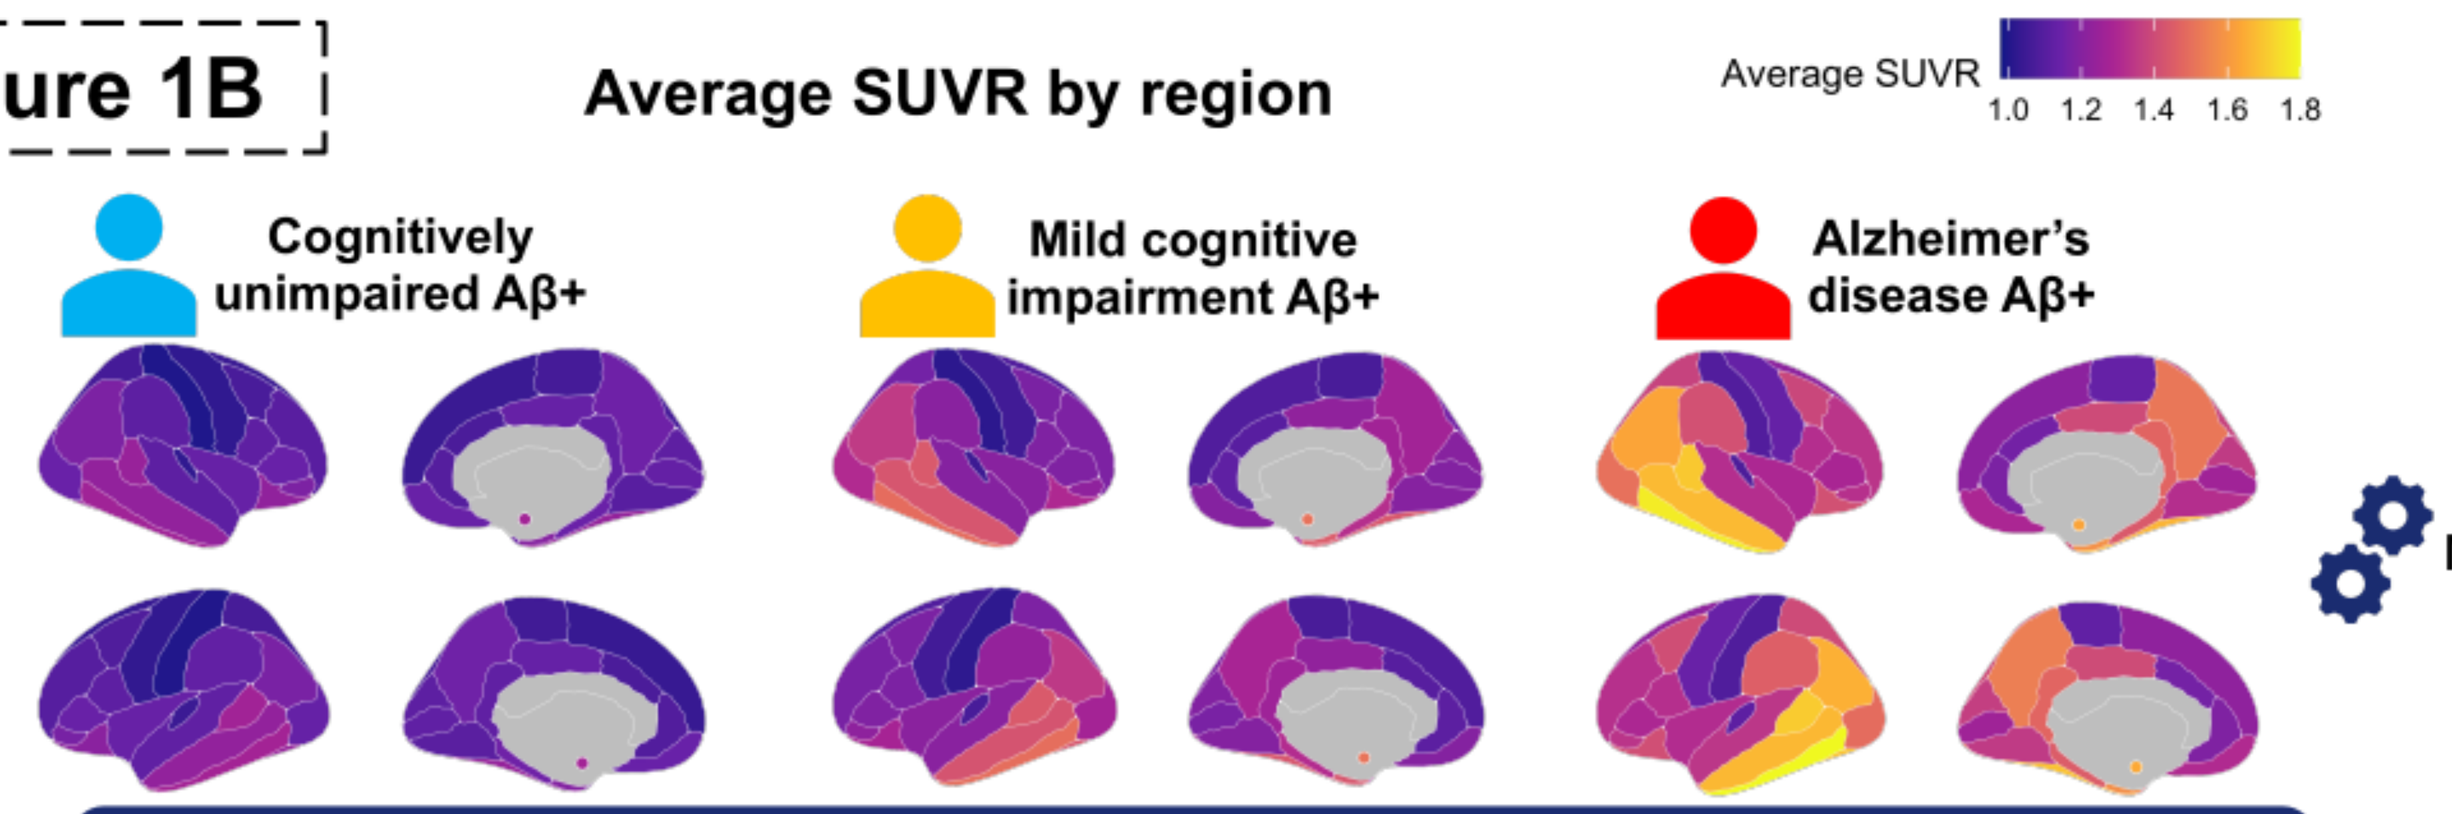
Average SUVR
1.0 1.2 1.4 1.6 1.8
Cognitively unimpaired Aβ+
Mild cognitive impairment Aβ+
Alzheimer's disease Aβ+

Mean

Tau SUVR deposition **consistent with Braak staging**
(gradient from middle temporal to sensorimotor)

Figure 1C Average tau abnormality by region (spatial extent)

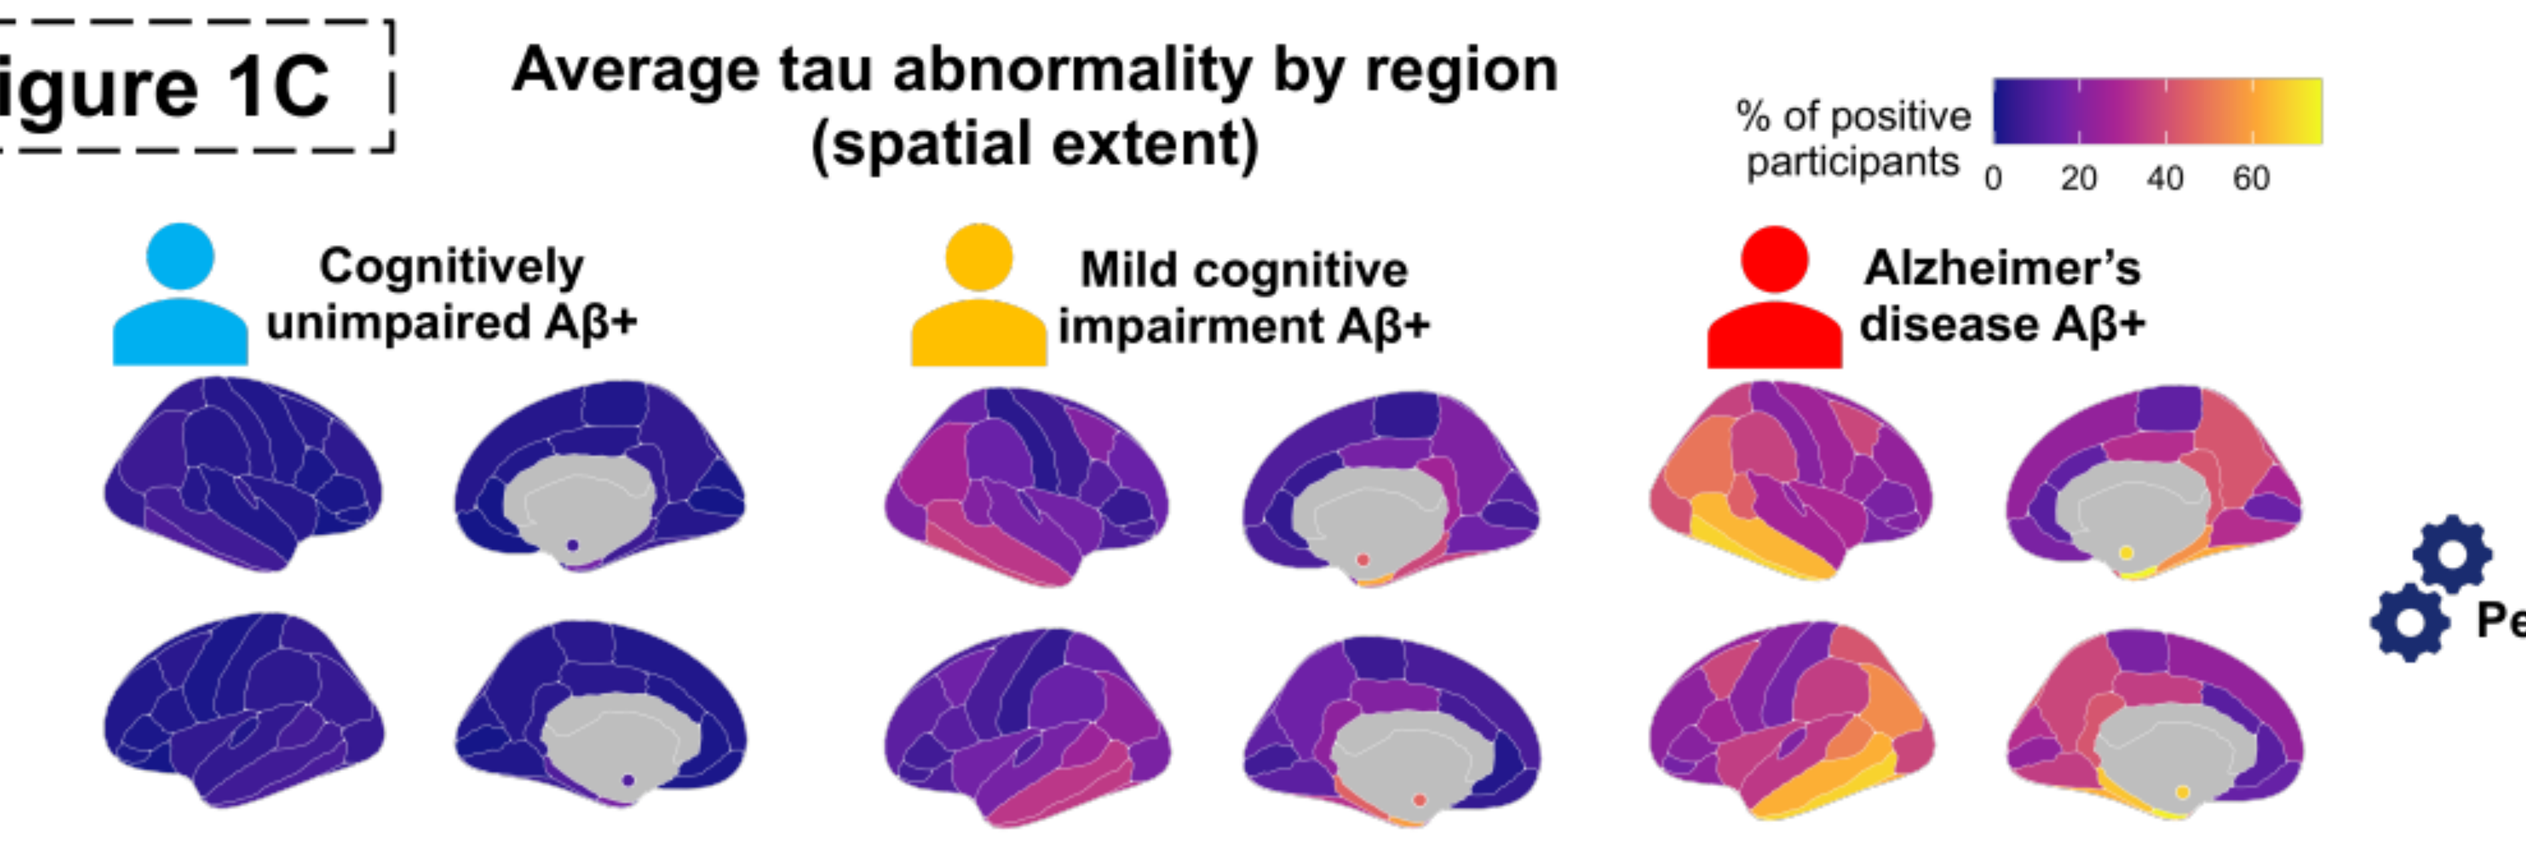
% of positive participants
0 20 40 60
Cognitively unimpaired Aβ+
Mild cognitive impairment Aβ+
Alzheimer's disease Aβ+

Percentage

Tau **abnormality** at baseline **consistent with Braak staging**

Objective 2: Abnormal progression of tau pathology over time

Figure 2A

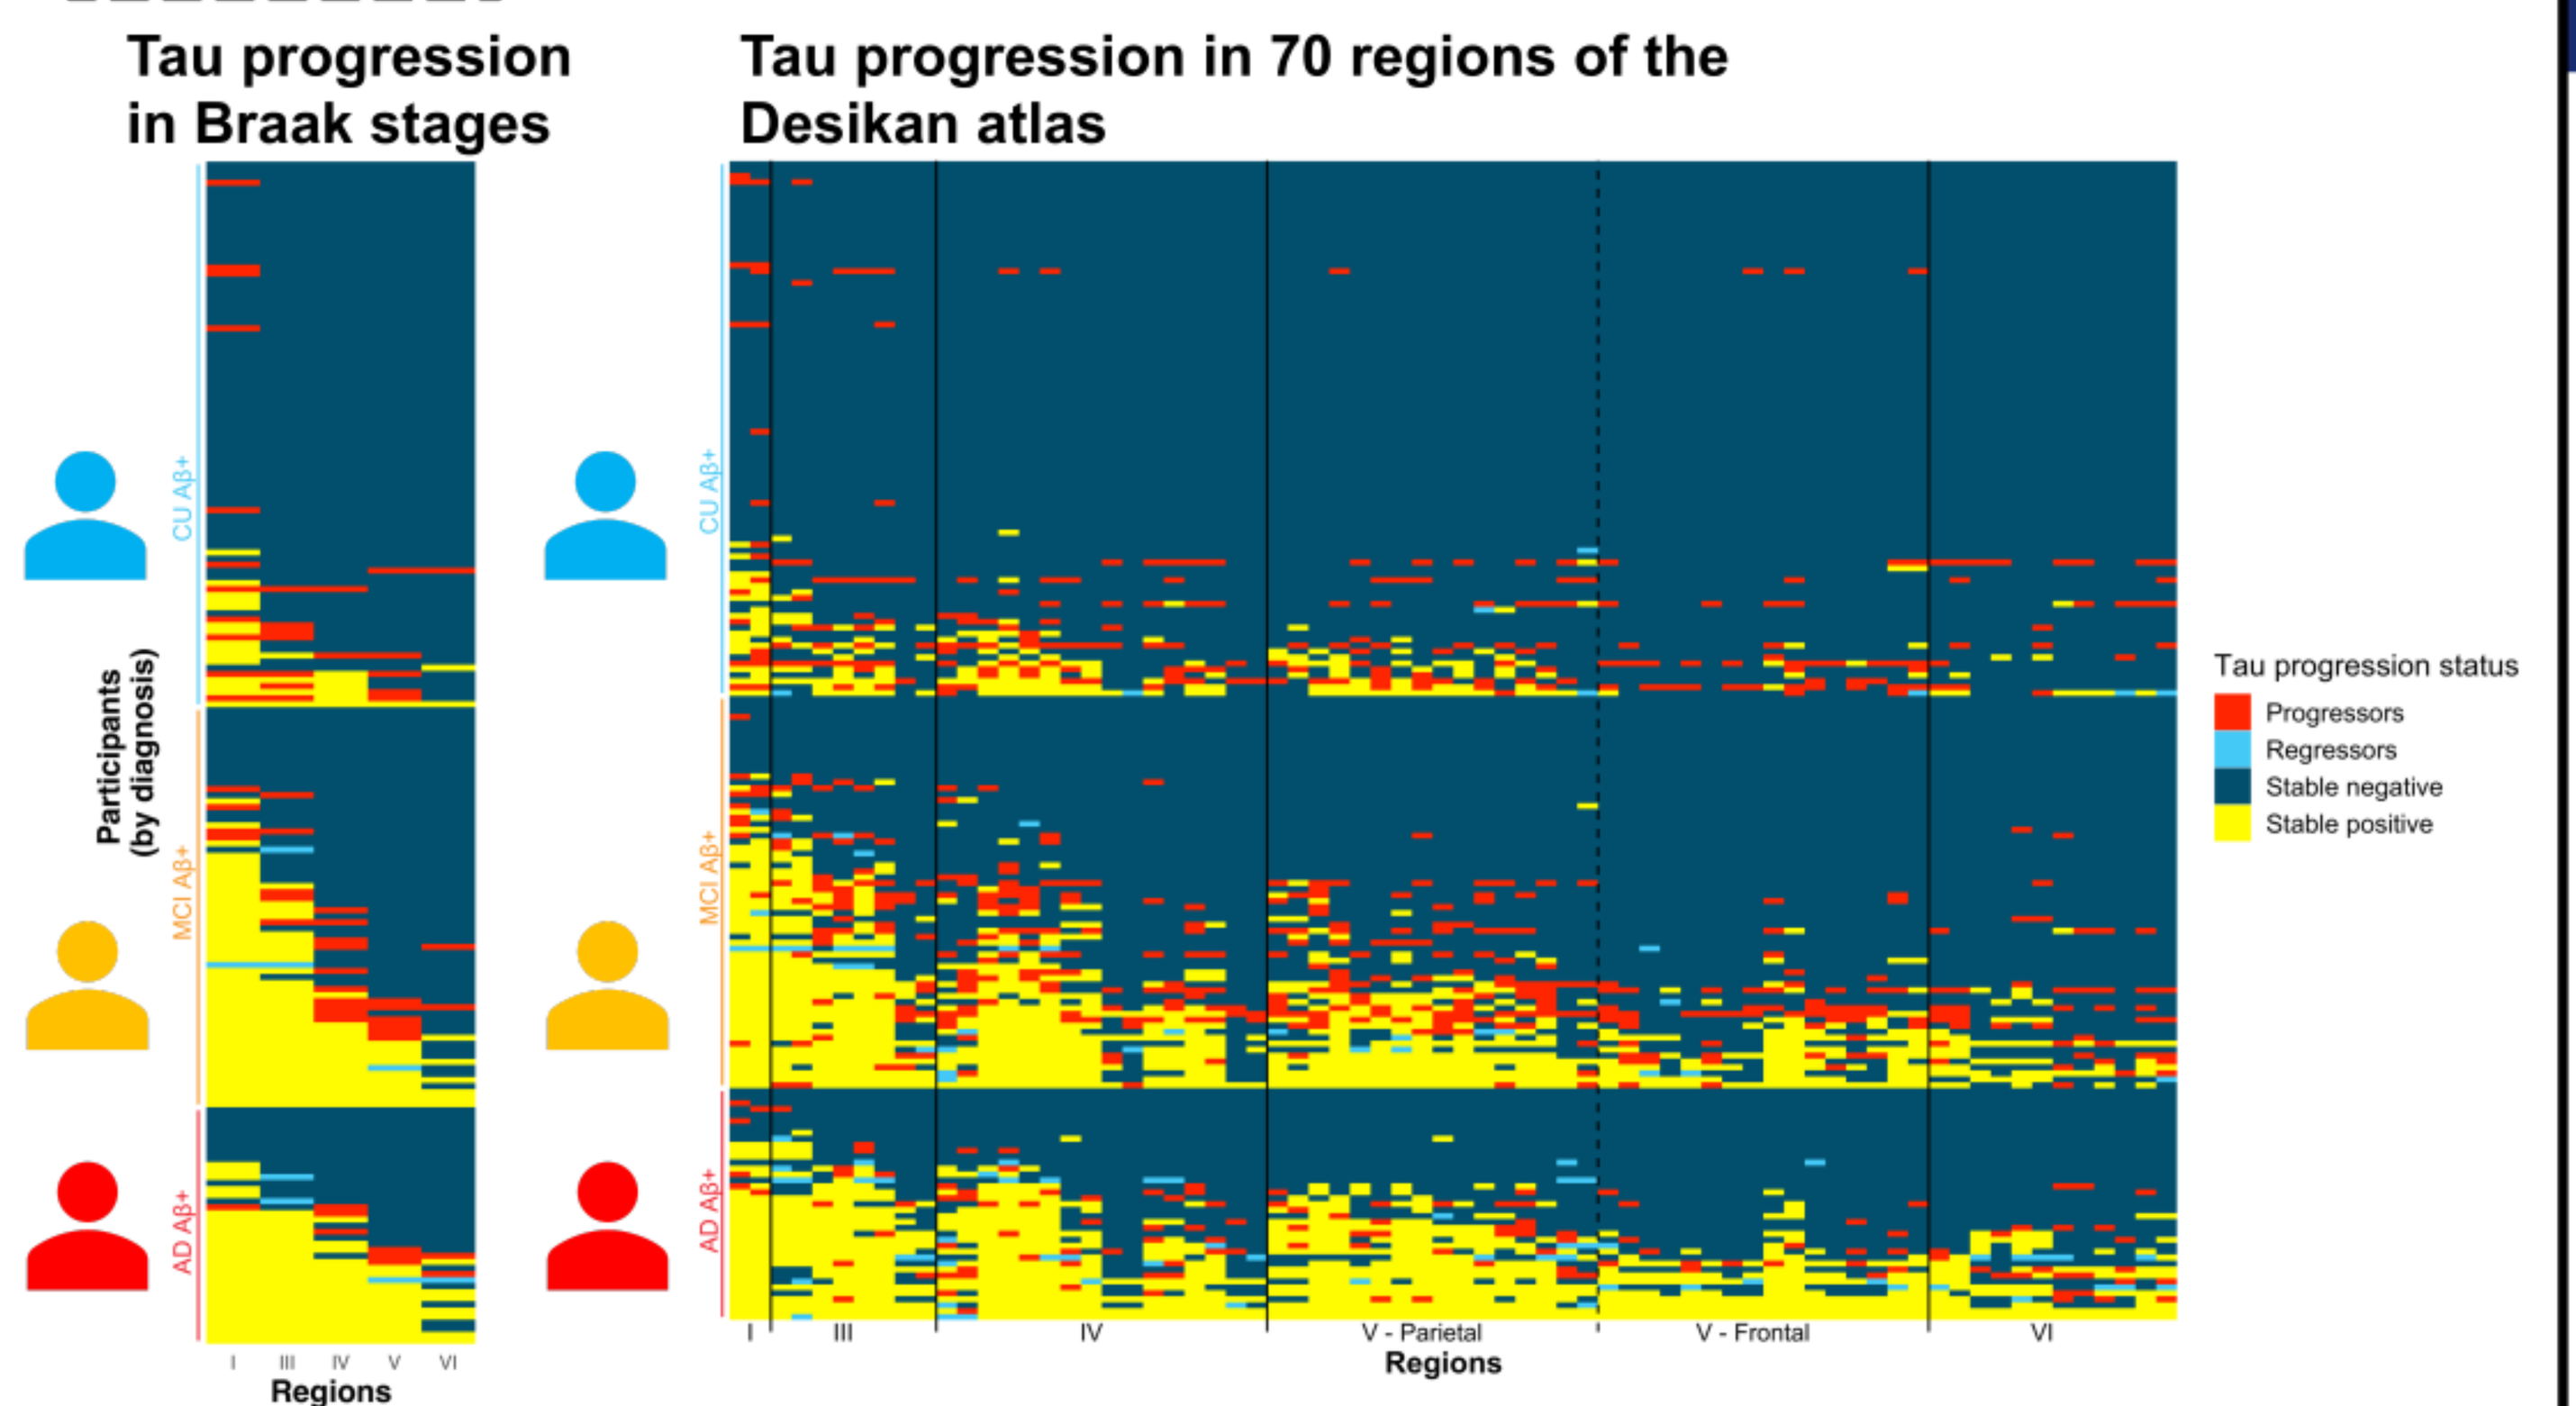
Tau progression in Braak stages
Tau progression in 70 regions of the Desikan atlas
Participants (by diagnosis)
CU Aβ+
MCI Aβ+
AD Aβ+
I III IV V VI
Regions
IV
V - Parietal
V - Frontal
VI
Tau progression status
Progressors
Regressors
Stable negative
Stable positive

>80% follow Braak staging

BUT less than 50% of regions becoming tau abnormal overlap between participants

Jaccard Index

Figure 2B Average SUVR change by region

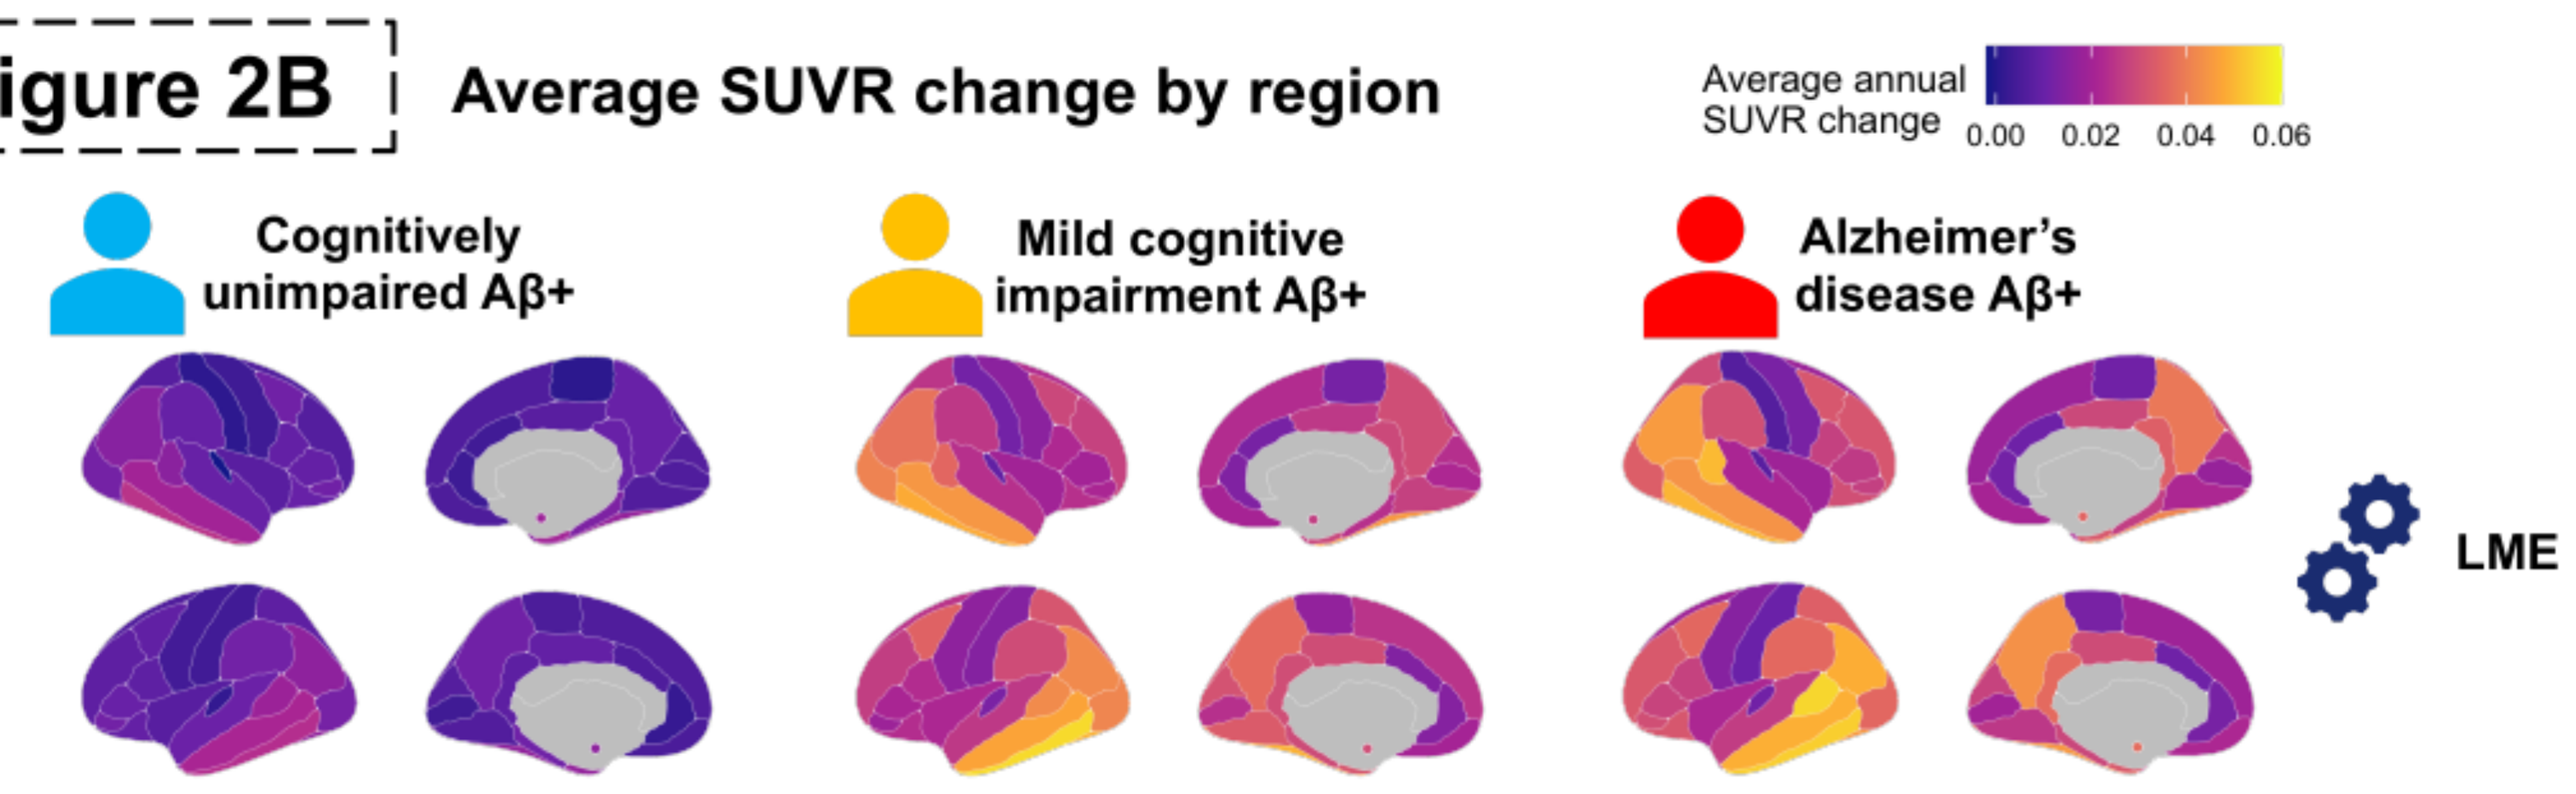
Average annual SUVR change
0.00 0.02 0.04 0.06
Cognitively unimpaired Aβ+
Mild cognitive impairment Aβ+
Alzheimer's disease Aβ+

LME

Tau SUVR accumulation **consistent with Braak staging**
(gradient from middle temporal to sensorimotor)

Figure 2C Average abnormality change by region (change in spatial extent)

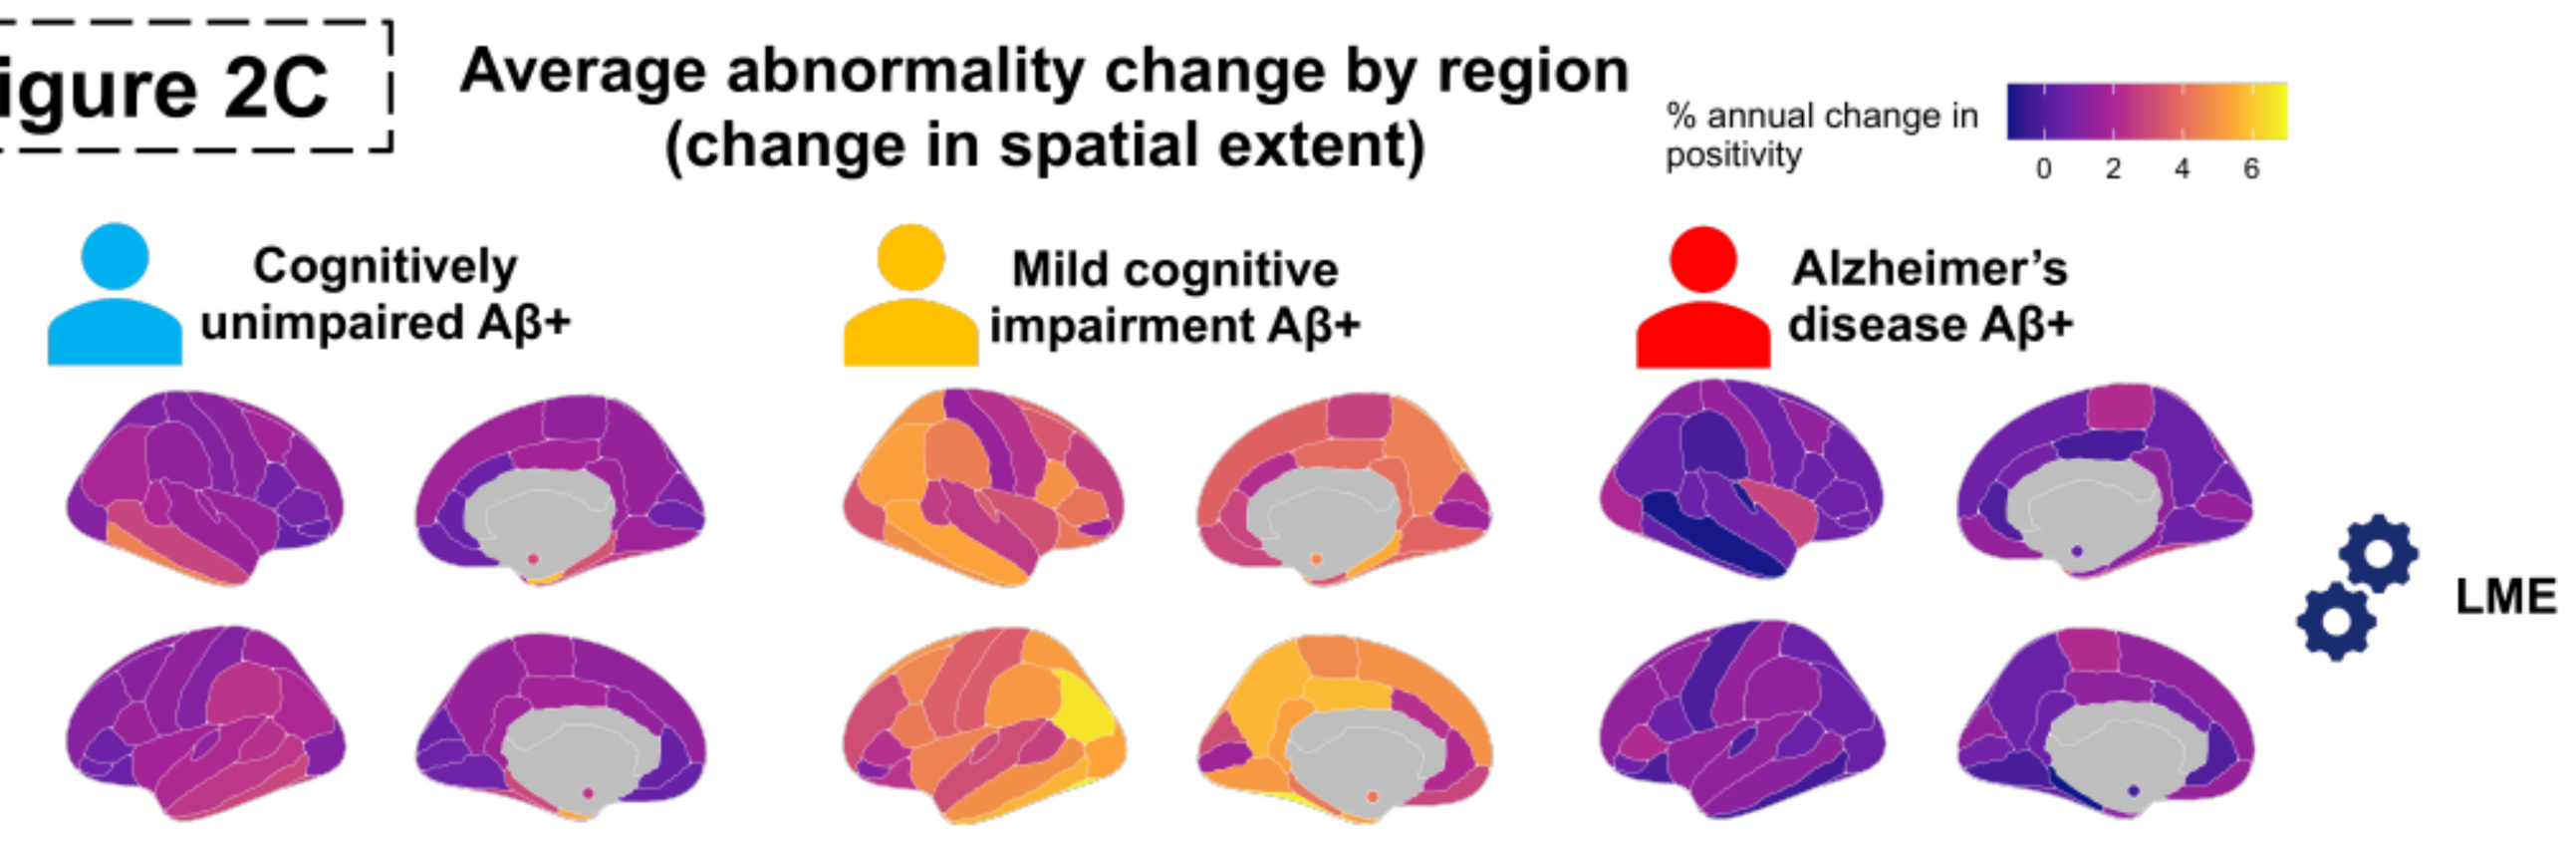
% annual change in positivity
0 2 4 6
Cognitively unimpaired Aβ+
Mild cognitive impairment Aβ+
Alzheimer's disease Aβ+

LME

Tau abnormality **progression is widespread and faster in MCI**
(MCI = 2.5 regions/year; CU and AD = 1 region/year)

Braak stages are followed by participants **cross-sectionally** and **over time**

BUT significant regional heterogeneity exists, particularly in the progression of **MCI participants**

medRχiv THE PREPRINT SERVER FOR HEALTH SCIENCES

@ fredstong3

Fonds de recherche Santé Québec

This research received the following travel awards: a Conference Fellowship from the AAIC, a Merit award from the OHBM and a Roger J. Paiement Outreach award from the Douglas Research Center

SIHNpy