The role of biofluid markers in predicting near-term cognitive impairment

Yara Yakoub, Nicholas J. Ashton, Thomas K Karikari, Cherie Strikwerda-Brown, Laia Montoliu-Gaya, Michael Schöll, R. Nathan Spreng, Maiya R. Geddes, Simon Ducharme, Jean-Paul Soucy, Pedro Rosa-Neto, John Breitner, Henrik Zetterberg, Kaj Blennow, Judes Poirier, Sylvia Villeneuve, for the PREVENT-AD Research Group

1.Background

Alzheimer's disease (AD) has been defined as a biological construct that can be determined using *in vivo* biomarkers.[1,2] Recent studies confirmed the clinical value of the AT(N) framework when using Aβ and tau positron emission tomography (PET) scans in predicting progression from cognitively unimpaired to MCI.[3,4] **Our study aims to investigate if plasma biomarkers can yield similar predictive performance to PET biomarkers, and to also compare against cerebrospinal fluid (CSF) biomarkers.**

2.Methods

•Cognitively unimpaired (CU) at the time of measurement.
•At least 12 months of clinical follow-up measurement.

- 291 had plasma $A\beta_{42/40}$ and pTau181 measurements.
- 147 had CSF $A\beta_{42}$ and pTau181 measurements.
- 130 had Aβ- and tau-PET scans.

70-73% Females

Outcome measures

- Progression to MCI from CU is made by clinical consensus blinded to blood, CSF, PET, MRI and *APOE* genotype . (n = 69 MCI in the blood sample; n = 31 MCI in both the CSF and the PET samples).
- Longitudinal Cognition is based on RBANS total score that consists of 5 cognitive domains (immediate memory, delayed memory, attention, language, and visuospatial)

Thresholds

PET thresholds

-Global [^{18}F]NAV4694-PET (Aβ) positivity based on GMM (SUVR threshold = 1.3)
-Entorhinal [^{18}F]AV-1451-PET (tau) positivity based on SUVR ≥ 2 SD from the mean of young participants (SUVR threshold = 1.25).

Blood & CSF thresholds

-Blood and CSF thresholds Aβ and tau positivity based on 80% specificity in identifying individuals with significant levels of Aβ- and tau-PET.

	Blood	CSF
$A\beta_{42/40}$ \| CSF $A\beta_{42}$ (pg/ml)	0.0578 (Sensitivity =25.42%) (Specificity = 80%)	1081.4 (Sensitivity=61.11%) (Specificity = 80%)
pTau181 (pg/ml)	8.238 (Sensitivity = 54.16%) (Specificity = 80%)	65.577 (Sensitivity=64.28%) (Specificity = 80%)

3.Results

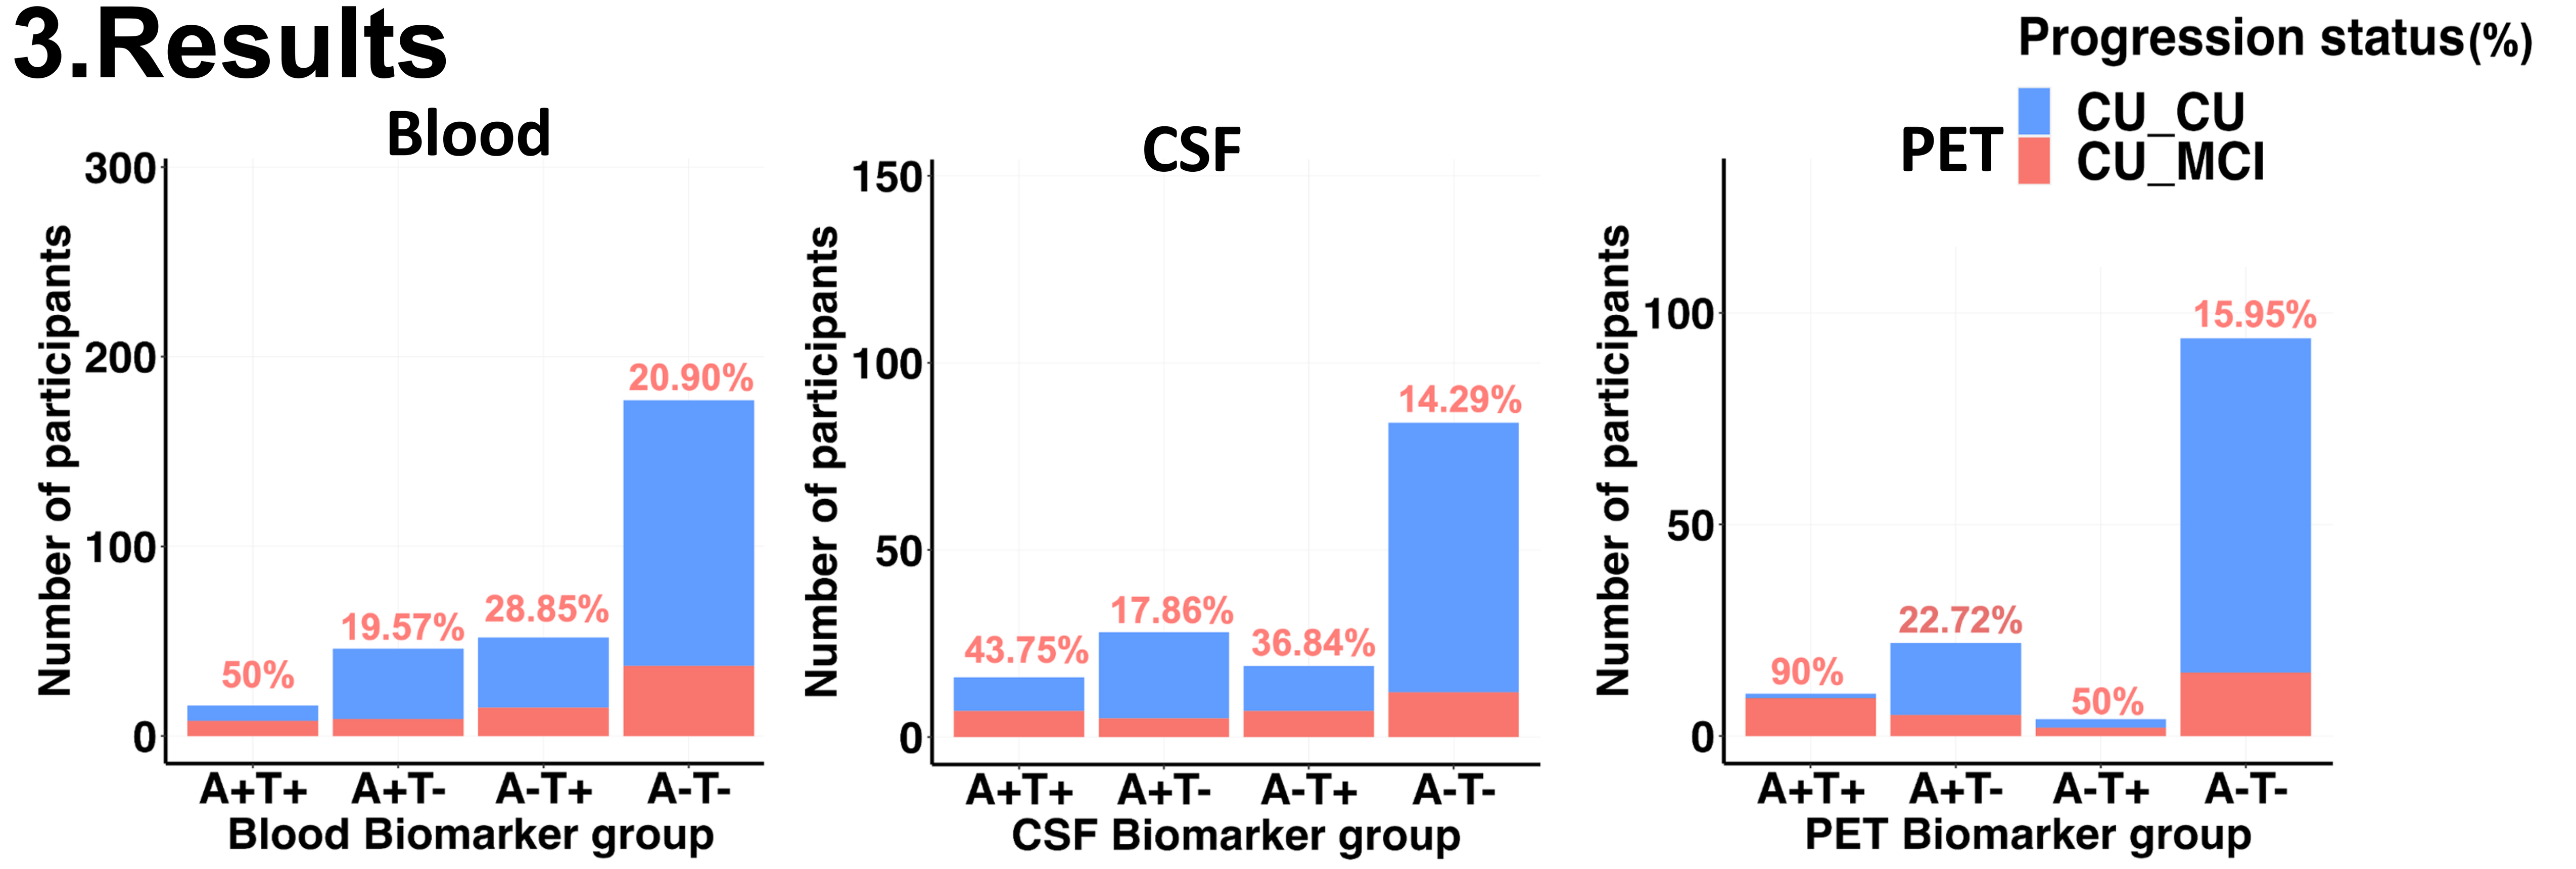
Progression status(%)
CU_CU
CU_MCI
Blood
CSF
PET
Number of participants
50%
19.57%
28.85%
20.90%
43.75%
17.86%
36.84%
14.29%
90%
22.72%
50%
15.95%
A+T+ A+T- A-T+ A-T-
Blood Biomarker group
CSF Biomarker group
PET Biomarker group

Figure 1. Percentage of individuals progressing to MCI across biomarker groups

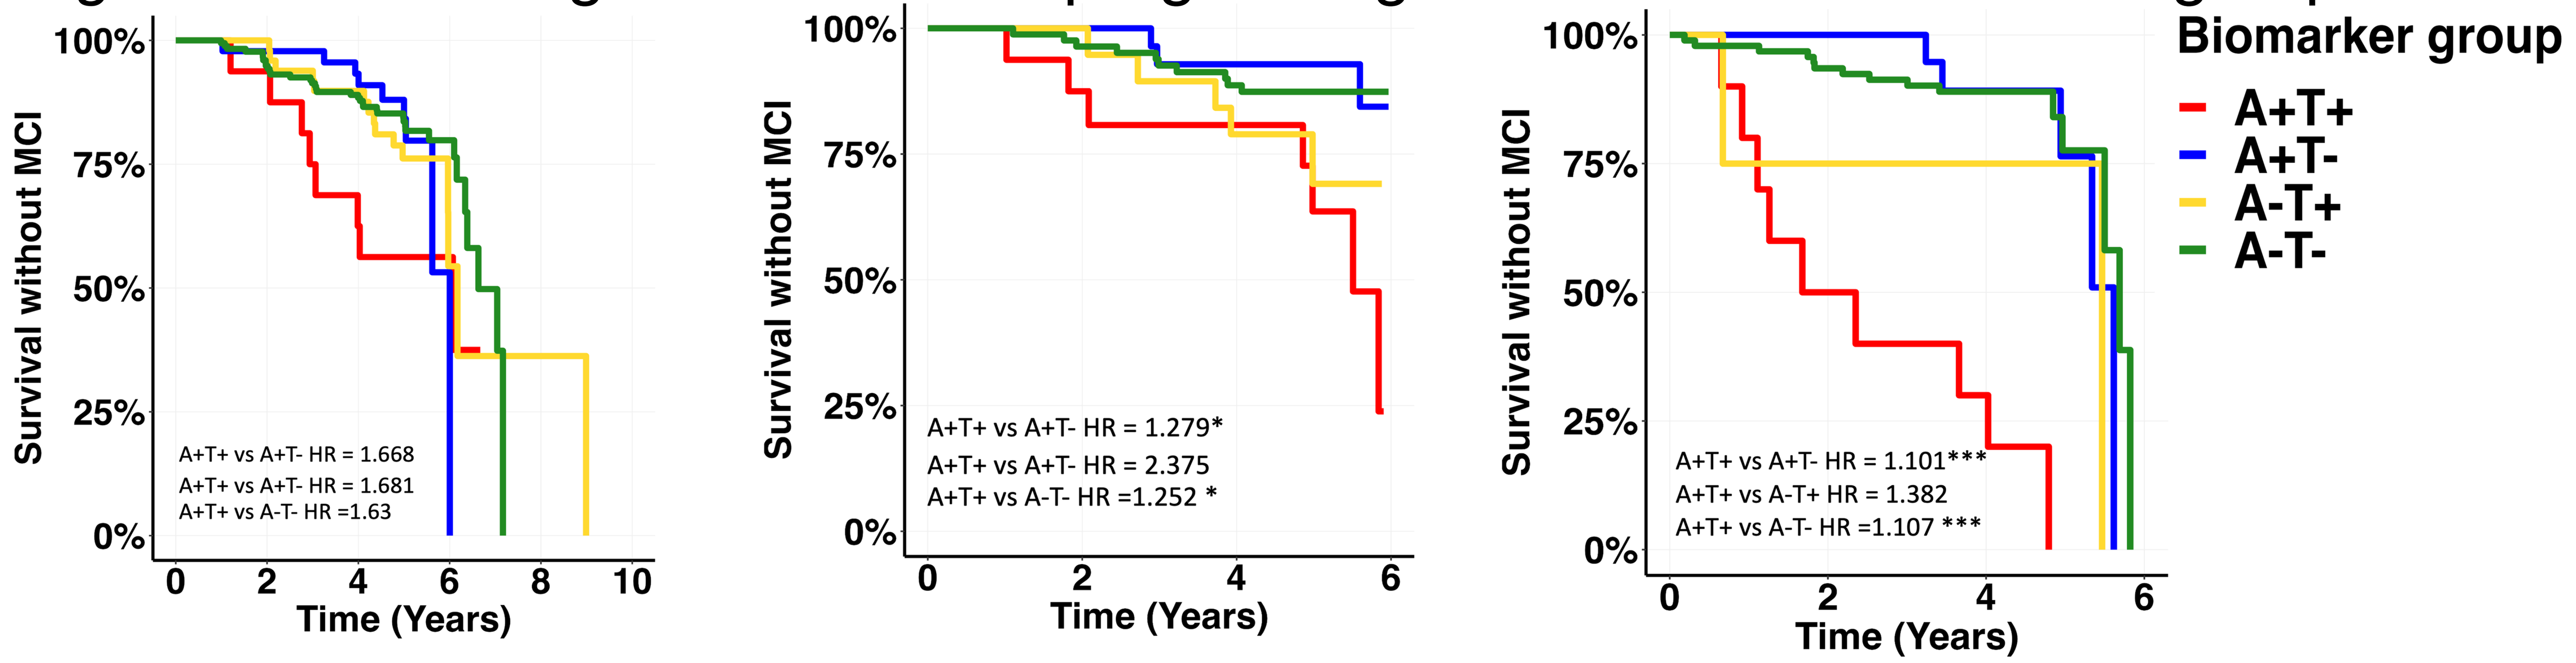
Biomarker group
A+T+
A+T-
A-T+
A-T-
Survival without MCI
Time (Years)
A+T+ vs A+T- HR = 1.668
A+T+ vs A+T- HR = 1.681
A+T+ vs A-T- HR =1.63
A+T+ vs A+T- HR = 1.279*
A+T+ vs A+T- HR = 2.375
A+T+ vs A-T- HR =1.252 *
A+T+ vs A+T- HR = 1.101***
A+T+ vs A-T+ HR = 1.382
A+T+ vs A-T- HR =1.107 ***

Figure 2. Survival curves displaying risk of progression to MCI over time across biomarker groups

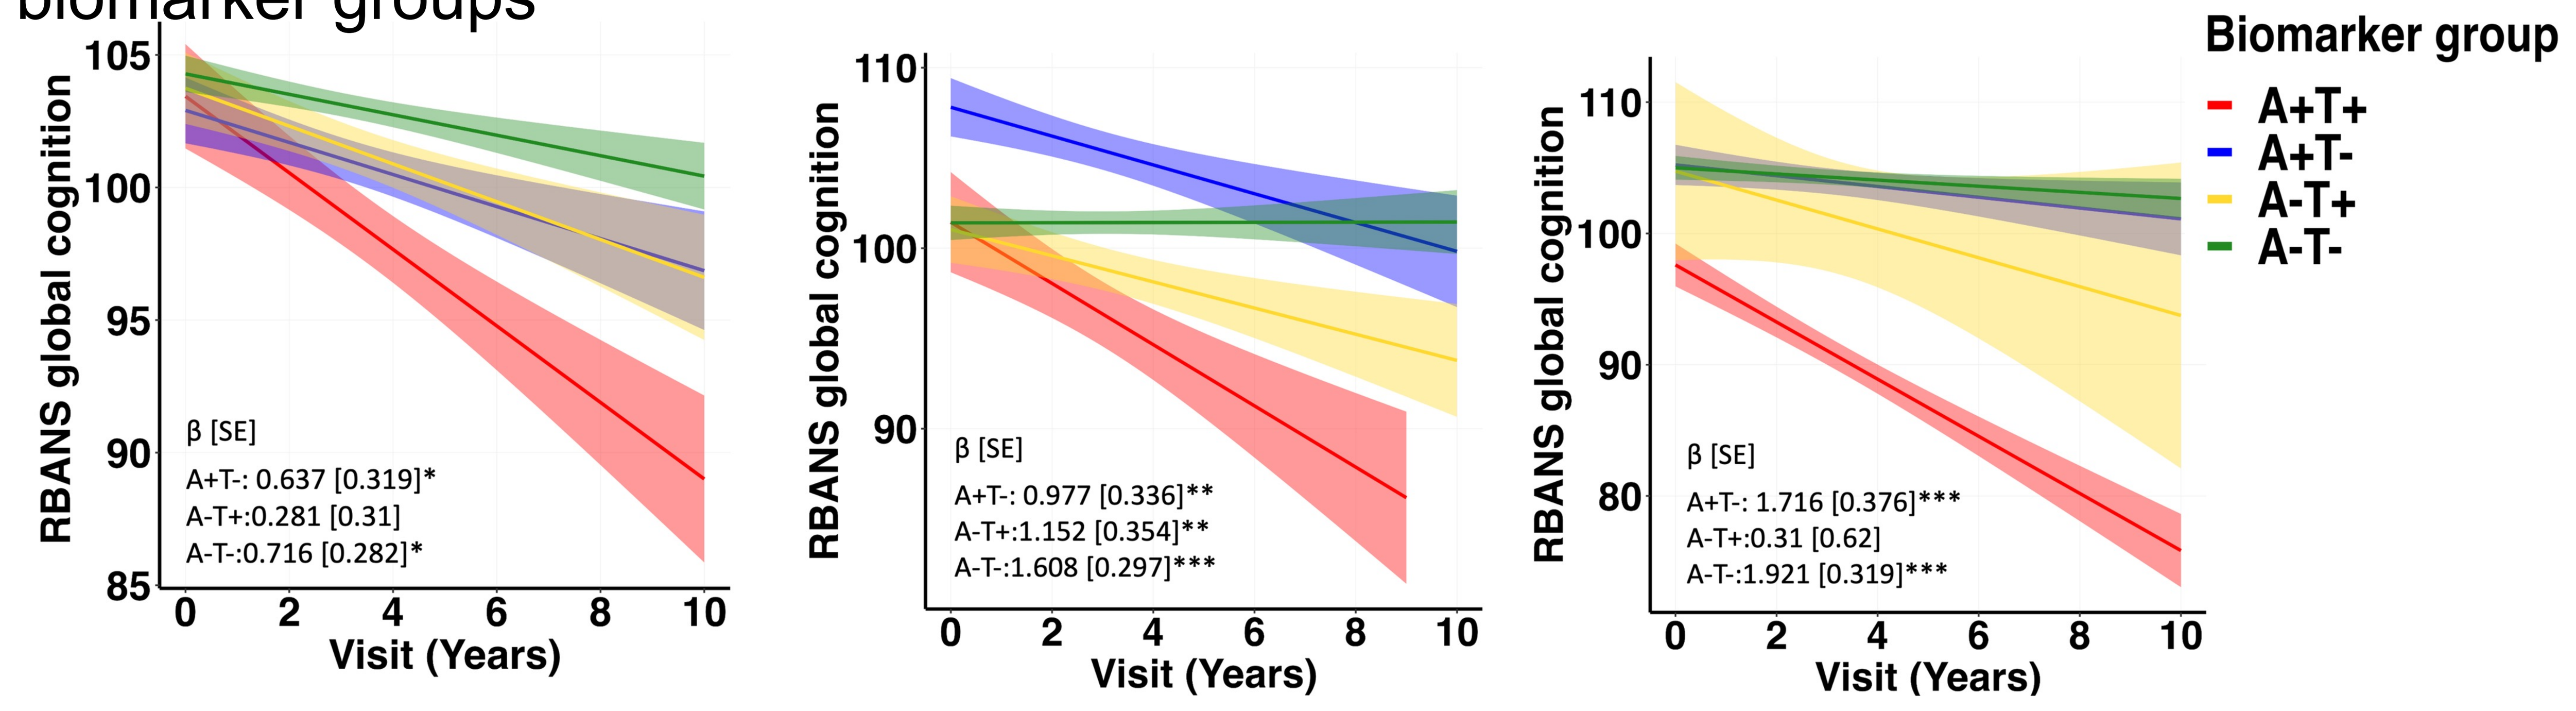
Biomarker group
A+T+
A+T-
A-T+
A-T-
RBANS global cognition
Visit (Years)
β [SE]
A+T-: 0.637 [0.319]*
A-T+:0.281 [0.31]
A-T-:0.716 [0.282]*
β [SE]
A+T-: 0.977 [0.336]**
A-T+:1.152 [0.354]**
A-T-:1.608 [0.297]***
β [SE]
A+T-: 1.716 [0.376]***
A-T+:0.31 [0.62]
A-T-:1.921 [0.319]***

Figure 3. Cognitive performance over time across biomarker groups

Notes: *p < .05; **p < .01;***p <.001

4.Conclusion

- 90% of A+T+ PET group, 50% of A+T+ blood group, and 43.75% CSF A+T+ group progressed to MCI.
- Rates or progression to MCI differed between A+T+ and A-T- as well as A+T- groups in both the blood and PET groups while the only difference in the CSF biomarker groups was between the A+T+ and A-T- groups.
- The A+T+ blood, CSF, and PET groups showed a steeper cognitive decline compared to other biomarker groups.
- **Take Home: Blood biomarkers show a similar performance to CSF biomarkers when predicting near-term progression to MCI. They however showed less sensitivity than PET biomarkers.**

5.Other points of discussion

- Both blood and CSF biomarkers identified higher percentage of A-T+ group compared to PET biomarker groups.
- Soluble pTau levels in CSF and plasma may increase earlier in the disease process and reflect to larger extent Aβ, while tau-PET reflects neurofibrillary tangles buildup in later disease stages as symptoms develop.
- The percentage of A+T+ participants defined with blood or CSF data was relatively small, 5.5% and 10.8% respectively.
- Utilizing 25th percentile to derive a threshold for the blood increased the percentage of A+T+ participants (7.5%) and resulted in a similar percentage of MCI progressors (45.45%).

6. Acknowledgment

References

1.Márquez, F. & M.A. Yassa, Molecular Neurodegeneration, 2019.
2. Jack, C.R., The Lancet Neurology, 2022.
3. Strikwerda-Brown, C., et al., JAMA Neurology, 2022.
4. Ossenkoppele, R., et al., Nature Medicine, 2022.

Author affiliations and an online version of the poster are available at: villeneuvelab.com

Contact
yara.yakoub@mail.mcgill.ca
@YakoubYara

The role of biofluid markers in predicting near-term cognitive impairment

Yara Yakoub, BSc [1]; Nicholas J. Ashton, PhD [2,3,4,5]; Thomas. K. Karikari, PhD [2];

Cherie Strikwerda-Brown, PhD [1]; Laia Montoliu-Gaya, PhD [2]; Michael Schöll, PhD [2,3,6];

Nathan R. Spreng, PhD [7]; Maiya R. Geddes, MD [7]; Simon Ducharme, MD [7,8];

Jean-Paul Soucy, MD [7]; Pedro Rosa-Neto, MD, PhD [7];

John C.S. Breitner, MD, MPH [1,8,9]; Henrik Zetterberg, MD, PhD [2,6,10,11,12,13];

Kaj Blennow, MD, PhD [2,10]; Judes Poirier, PhD [1,8]; Sylvia Villeneuve, PhD [1,8,9];

PREVENT-AD Research Group.

[1] Douglas Mental Health University Institute, Centre for Studies on the Prevention of Alzheimer's Disease (StoP-AD), Montreal, Quebec, H4H 1R3, Canada.

[2] Department of Psychiatry and Neurochemistry, Institute of Neuroscience and Physiology, The

Sahlgrenska Academy, University of Gothenburg, 405 30, Gothenburg, Sweden.

[3] Centre for Age-Related Medicine, Stavanger University Hospital, 4011, Stavanger, Norway.

[4] King's College London, Institute of Psychiatry, Psychology & Neuroscience, Maurice Wohl

Clinical Neuroscience Institute, London, WC2R 2LS, UK.

[5] NIHR Biomedical Research Centre for Mental Health & Biomedical Research Unit for

Dementia at South London & Maudsley NHS Foundation, London, SE5 8AF, UK.

[6] Department of Neurodegenerative Disease, UCL Queen Square Institute of Neurology, University College London, London, WC1N 3BG, UK.

[7] Montreal Neurological Institute, McGill University, Montreal, Quebec, H3A 2B4, Canada.

[8] Department of Psychiatry, McGill University, Montreal, Quebec, H3A 1A1, Canada.

[9] McGill Centre for Integrative Neuroscience, McGill University, Montreal, Quebec, H3A 2B4, Canada.

[10] Clinical Neurochemistry Laboratory, Sahlgrenska University Hospital, Mölndal, 413 45, Sweden.

[11] UK Dementia Research Institute at UCL, London, WC1E 6BT, UK.

[12] Hong Kong Center for Neurodegenerative Diseases, Clear Water Bay, Hong Kong, China.

[13] UW Department of Medicine, School of Medicine and Public Health, Madison, WI, 53705, USA.